48th Annual ICAAC/IDSA 46th Annual Meeting, Washington, DC, USA, October 25–28, 2008

Poster H-4055

A Pharmacokinetic Study to Evaluate an Interaction between Maraviroc and Raltegravir in Healthy Adults

E Andrews, P Glue, J Fang, P Crownover, R Tressler, and B Damle

Pfizer Inc, New York, USA

For further information, please contact Emma Andrews at Emma.N.Andrews@pfizer.com

Background

- Maraviroc (marketed as Celsentri®/Selzentry™) and raltegravir (marketed as Isentress®) are two new antiretroviral drugs approved by the FDA and EMEA for use in treatment-experienced patients with HIV-1 infection. Maraviroc is the first approved CCR5 antagonist and raltegravir is the first approved integrase inhibitor.
- Maraviroc is a substrate for CYP3A4 and P-gp, and is not an inducer or inhibitor of cytochrome P450 enzymes.[1]
- Raltegravir is a substrate for UGT 1A1 and P-gp.[2,3] It is not a substrate for cytochrome P450 enzymes and is neither an inducer or inhibitor of these enzymes.[2]
- As there is interest in using these two agents as part of a combination antiretroviral drug regimen, we evaluated the two-way pharmacokinetic (PK) interaction between maraviroc and raltegravir.

Objectives

- To estimate the effect of coadministration of maraviroc (MVC) and raltegravir (RAL) on the pharmacokinetics of MVC and RAL.
- To evaluate the short-term safety and tolerability of MVC and RAL when administered alone and concomitantly.

Methods

- This was a Phase 4 open-label, multiple dose, fixed-sequence study (see Figure 1) performed in healthy adult volunteers at a single US site.
- MVC 300 mg and/or RAL 400 mg was administered q12hr orally (Figure 1) on an empty stomach (no food for 1 hour before and after dosing).
- PK evaluation was performed on Days 3 (RAL), 11 (MVC), and 14 (RAL + MVC).
- On PK sampling days (Days 3, 11, and 14) blood samples for PK analysis were collected prior to the morning dose (immediately pre-dose) and at 0.5, 1, 1.5, 2, 3, 4, 6, 8, and 12 hours post-dose.
- MVC and RAL plasma levels were assayed using validated LC/MS/MS assays.[4] Calibration standard responses were linear over the range of 0.500 to 500 ng/mL for MVC and 2.00 to 1,000 ng/mL for RAL.
- PK was determined by non-compartmental analyses.
- Point estimates of the geometric mean ratios (GMRs) and 90% confidence intervals (CIs) were determined for PK parameters (C_{max}, AUC_τ, and C_{12}).
- Safety was assessed by monitoring adverse events (AEs), safety laboratory tests, vital signs, and ECGs.

Figure 1. Study design

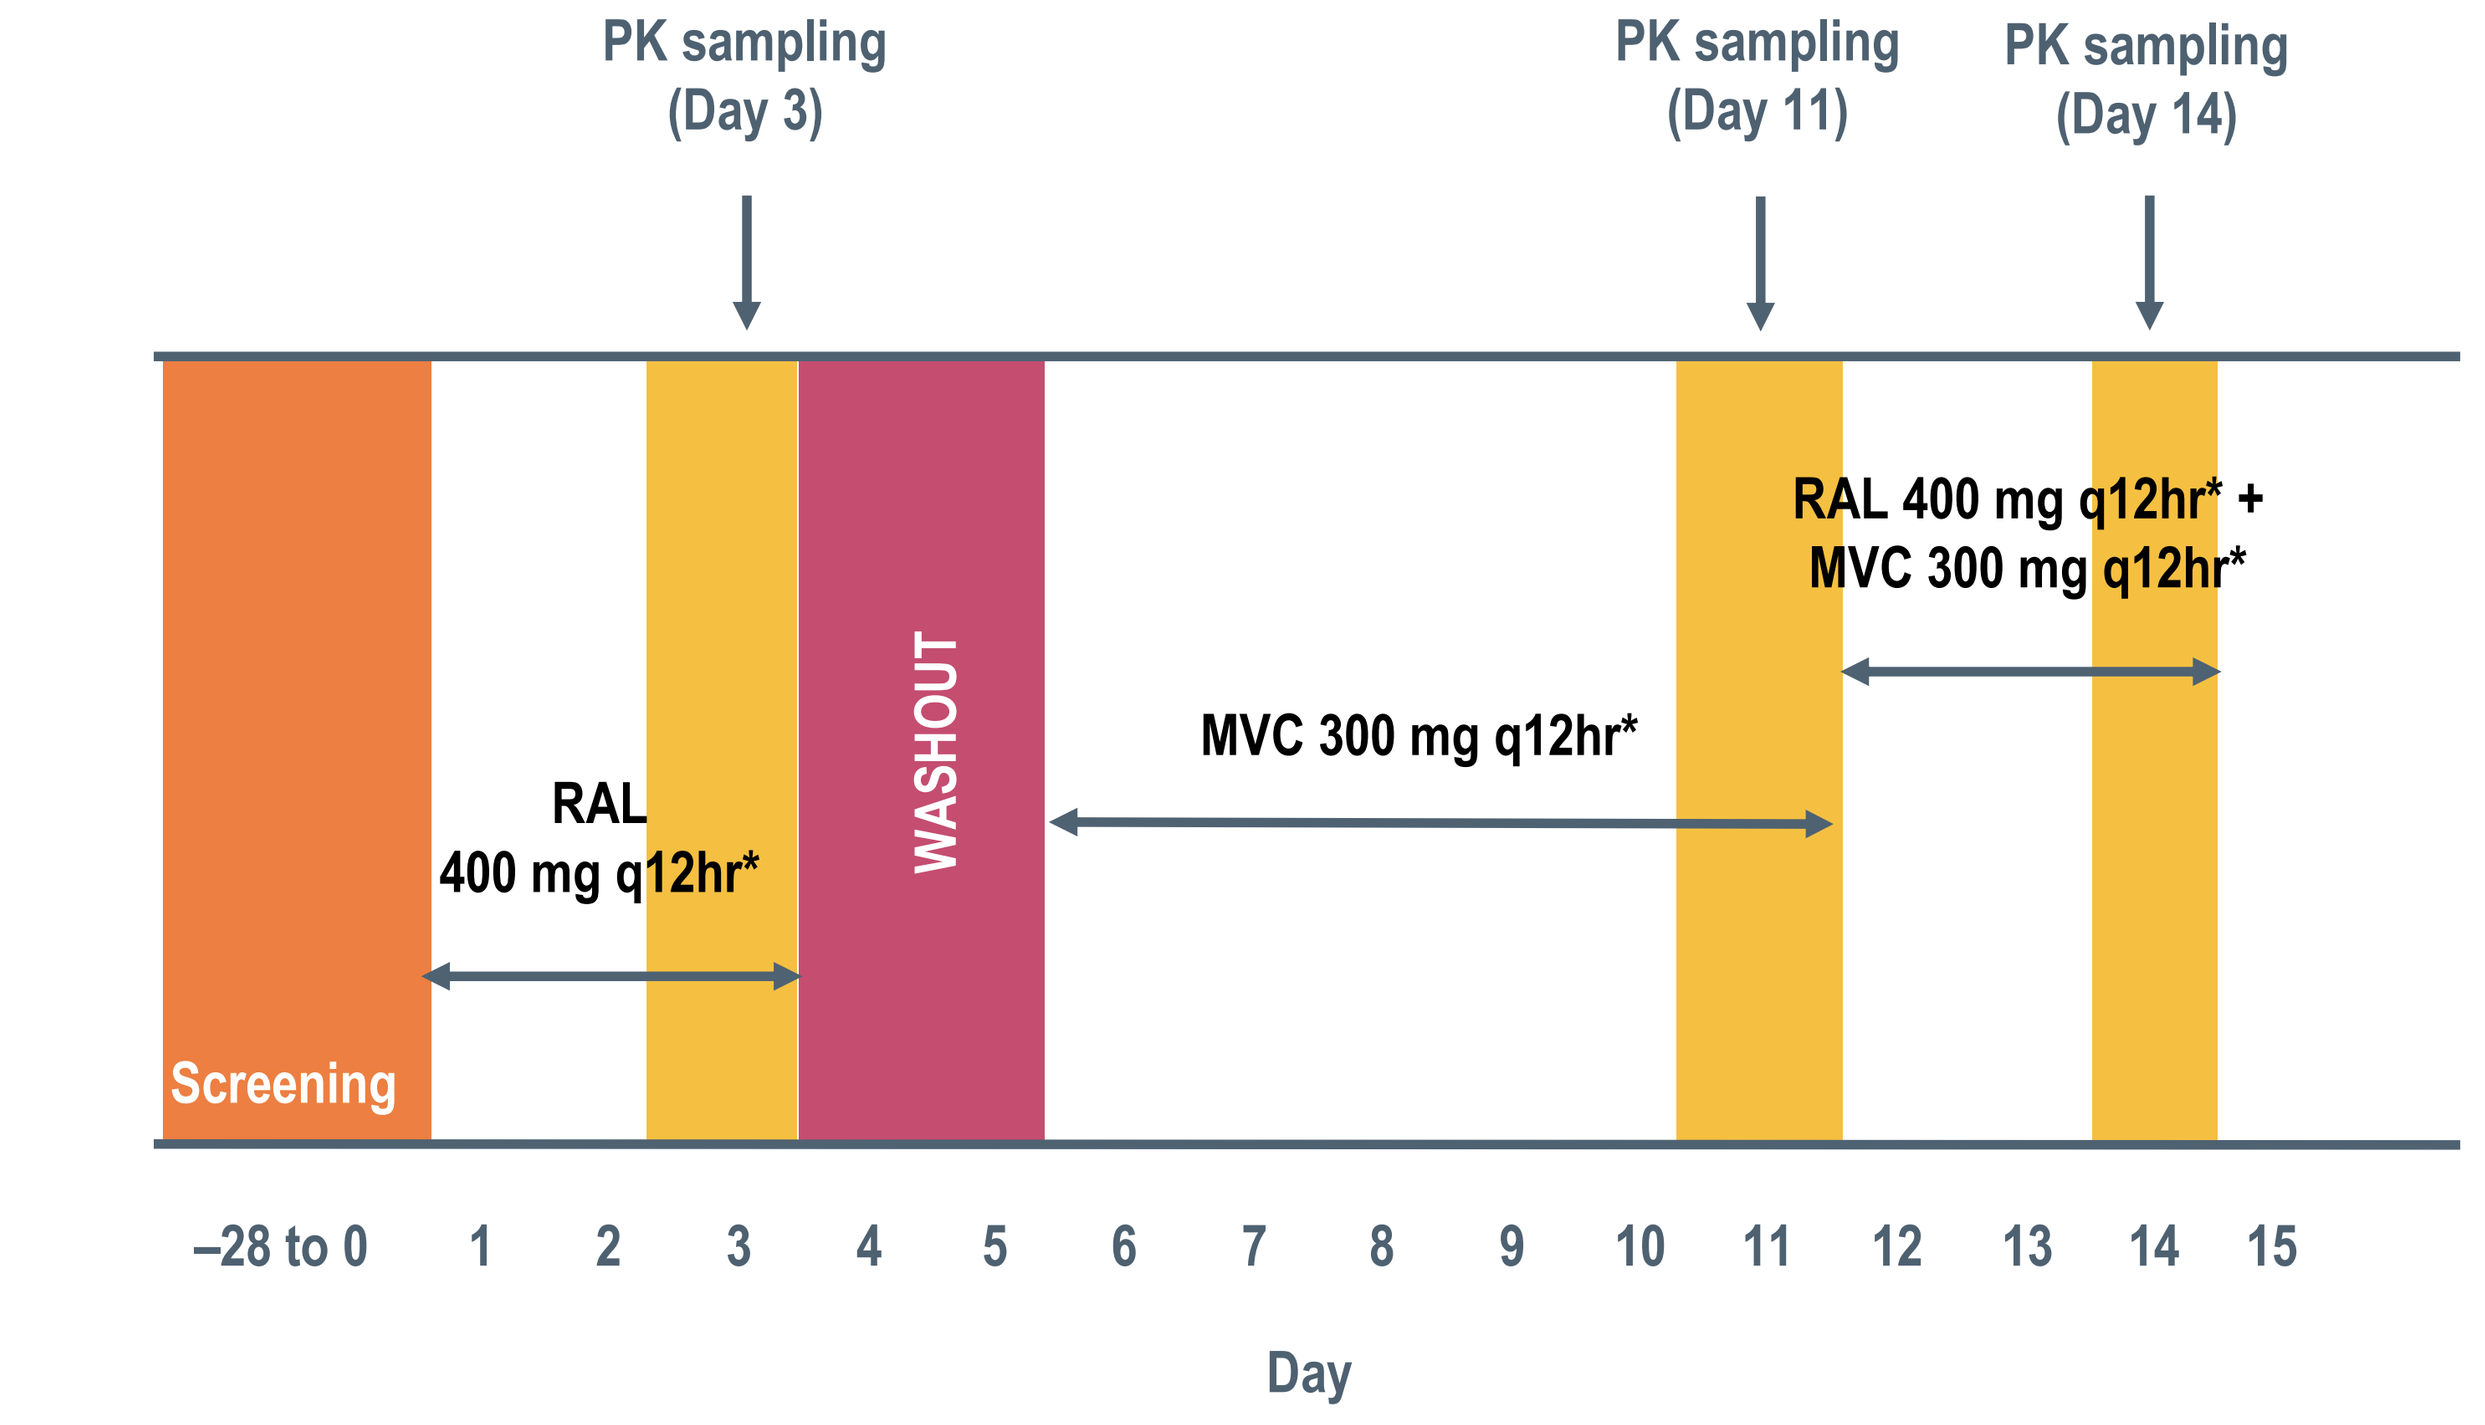

PK sampling (Day 3)
PK sampling (Day 11)
PK sampling (Day 14)
Screening
RAL 400 mg q12hr*
WASHOUT
MVC 300 mg q12hr*
RAL 400 mg q12hr* + MVC 300 mg q12hr*
–28 to 0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15
Day

* No evening dose administered on Days 3 and 14

Results

- Eighteen subjects (16 male, 2 female) were enrolled in this study, with an age range of 25–54 years. Race was recorded as black for nine subjects, white for six, and 'other' for three. BMI ranged from 19.9 to 30.4 kg/m^2, with a mean of 26.2 kg/m^2.
- Seventeen subjects completed the study. One subject withdrew consent during the study for personal reasons and was discontinued on Day 10 prior to the full PK day (Day 11) while receiving MVC.

Pharmacokinetics

- Median plasma concentration-time plots are presented in Figure 2 (MVC) and Figure 3 (RAL).

Figure 2. MVC median plasma concentration-time profiles in the presence and absence of RAL

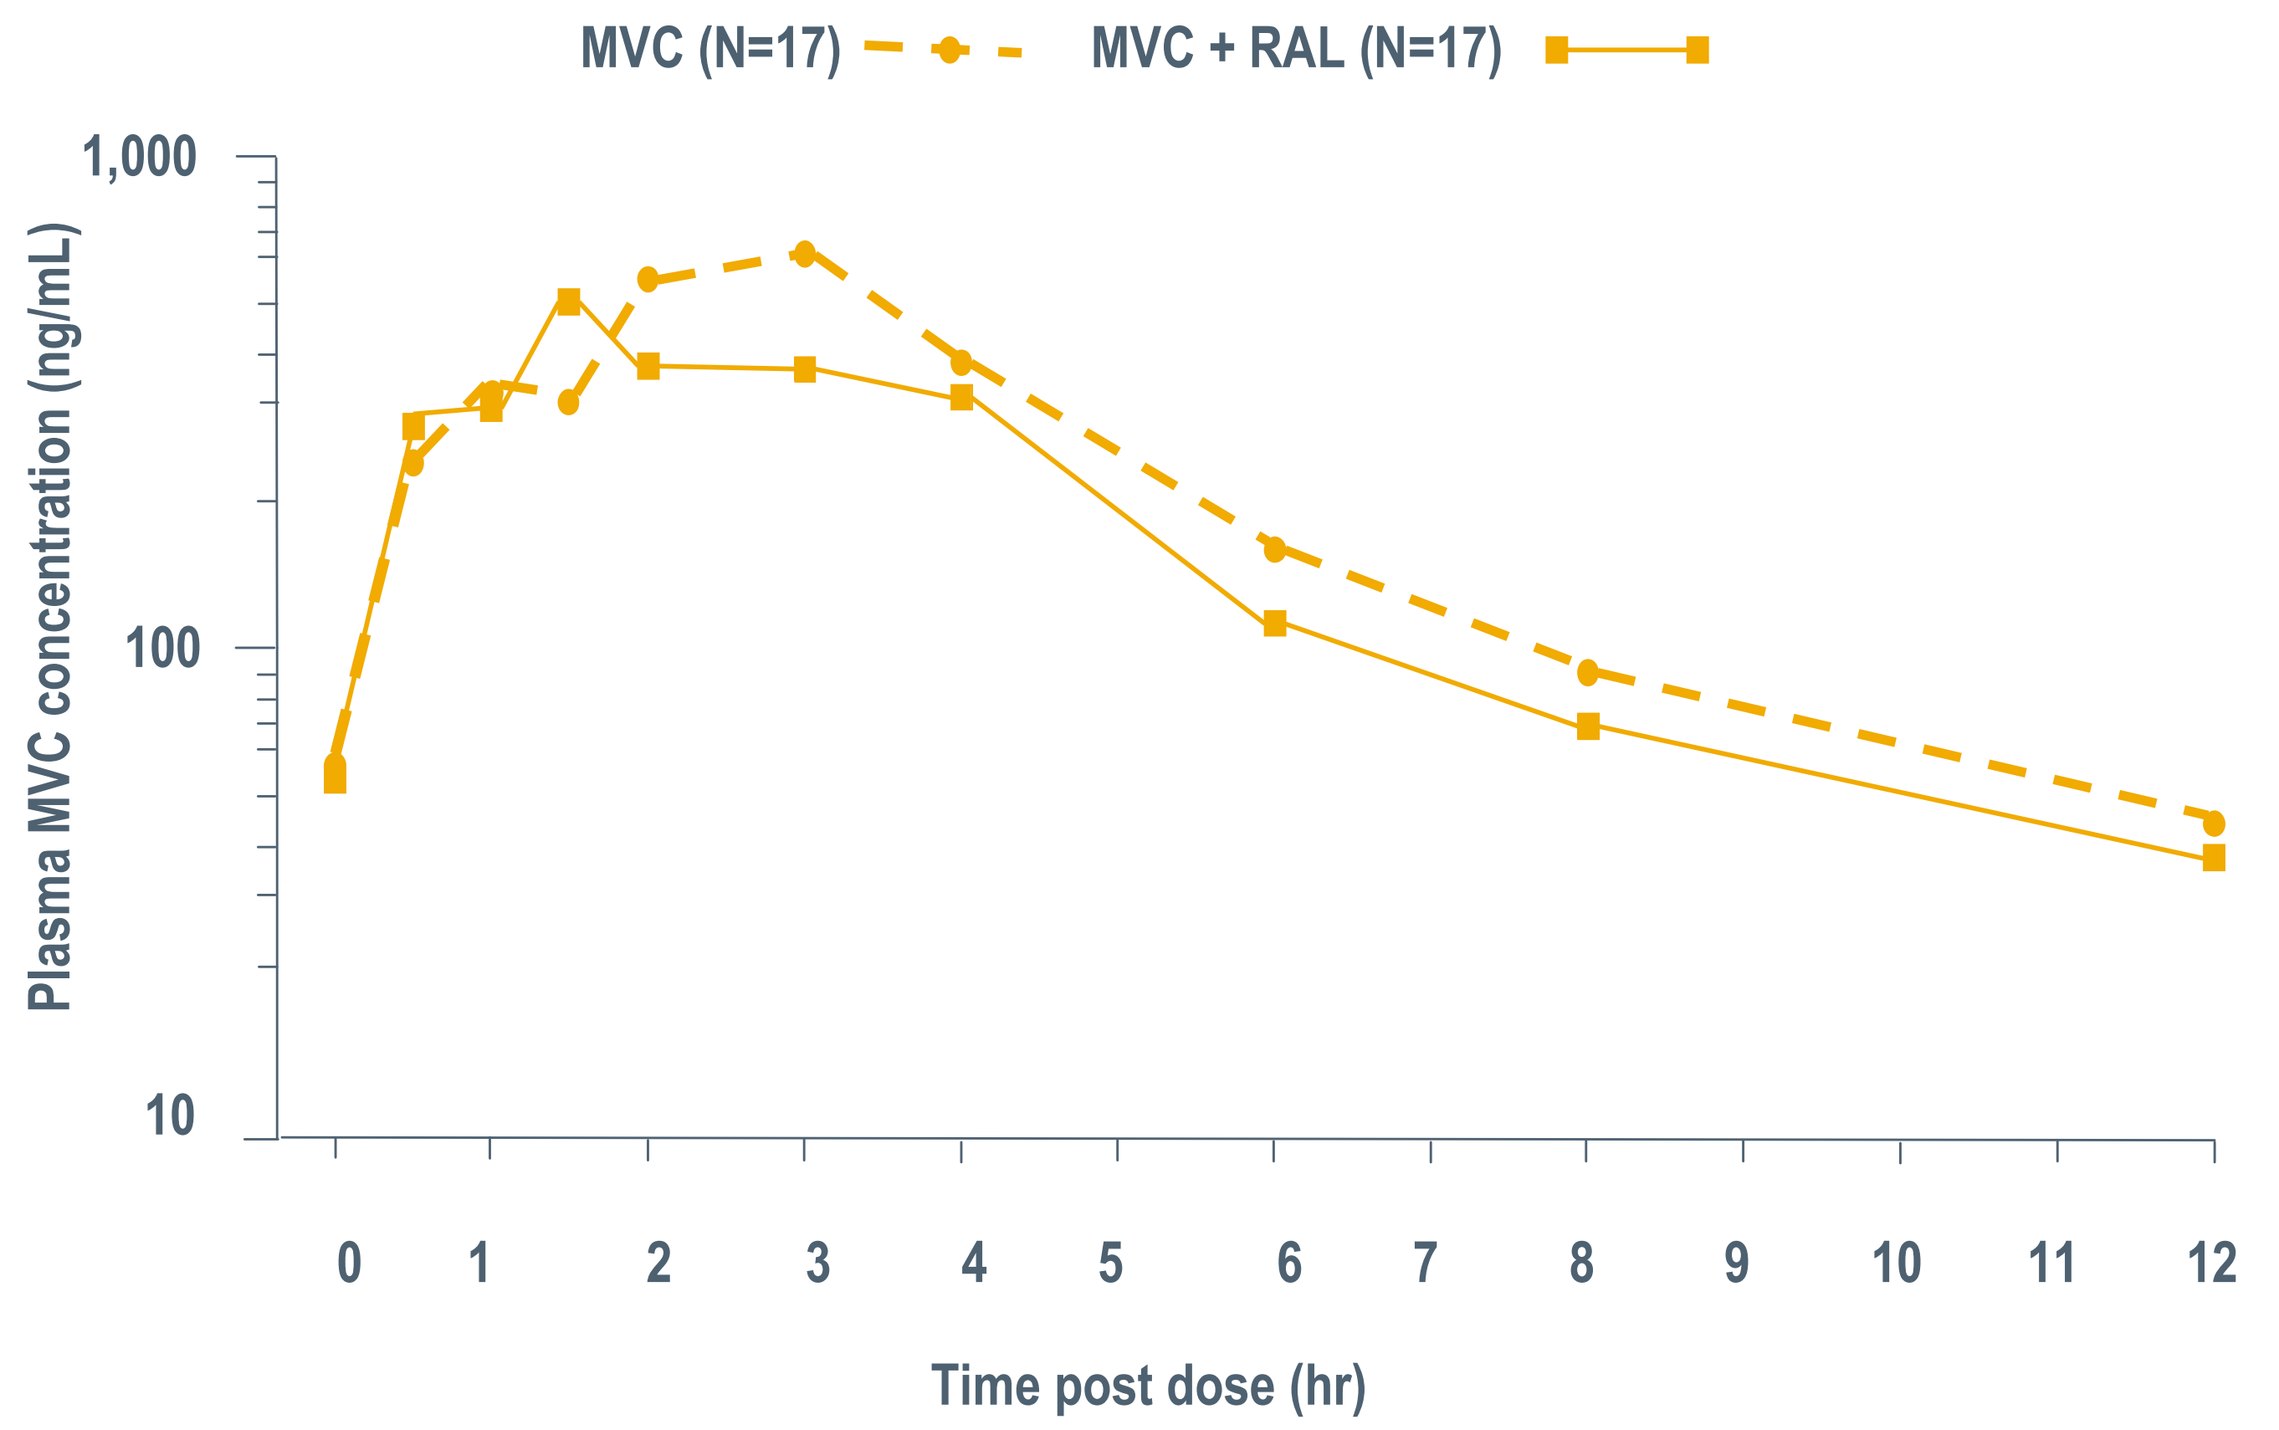

MVC (N=17)
MVC + RAL (N=17)
Plasma MVC concentration (ng/mL)
1,000
100
10
0 1 2 3 4 5 6 7 8 9 10 11 12
Time post dose (hr)

Figure 3. RAL median plasma concentration-time profiles in the presence and absence of MVC

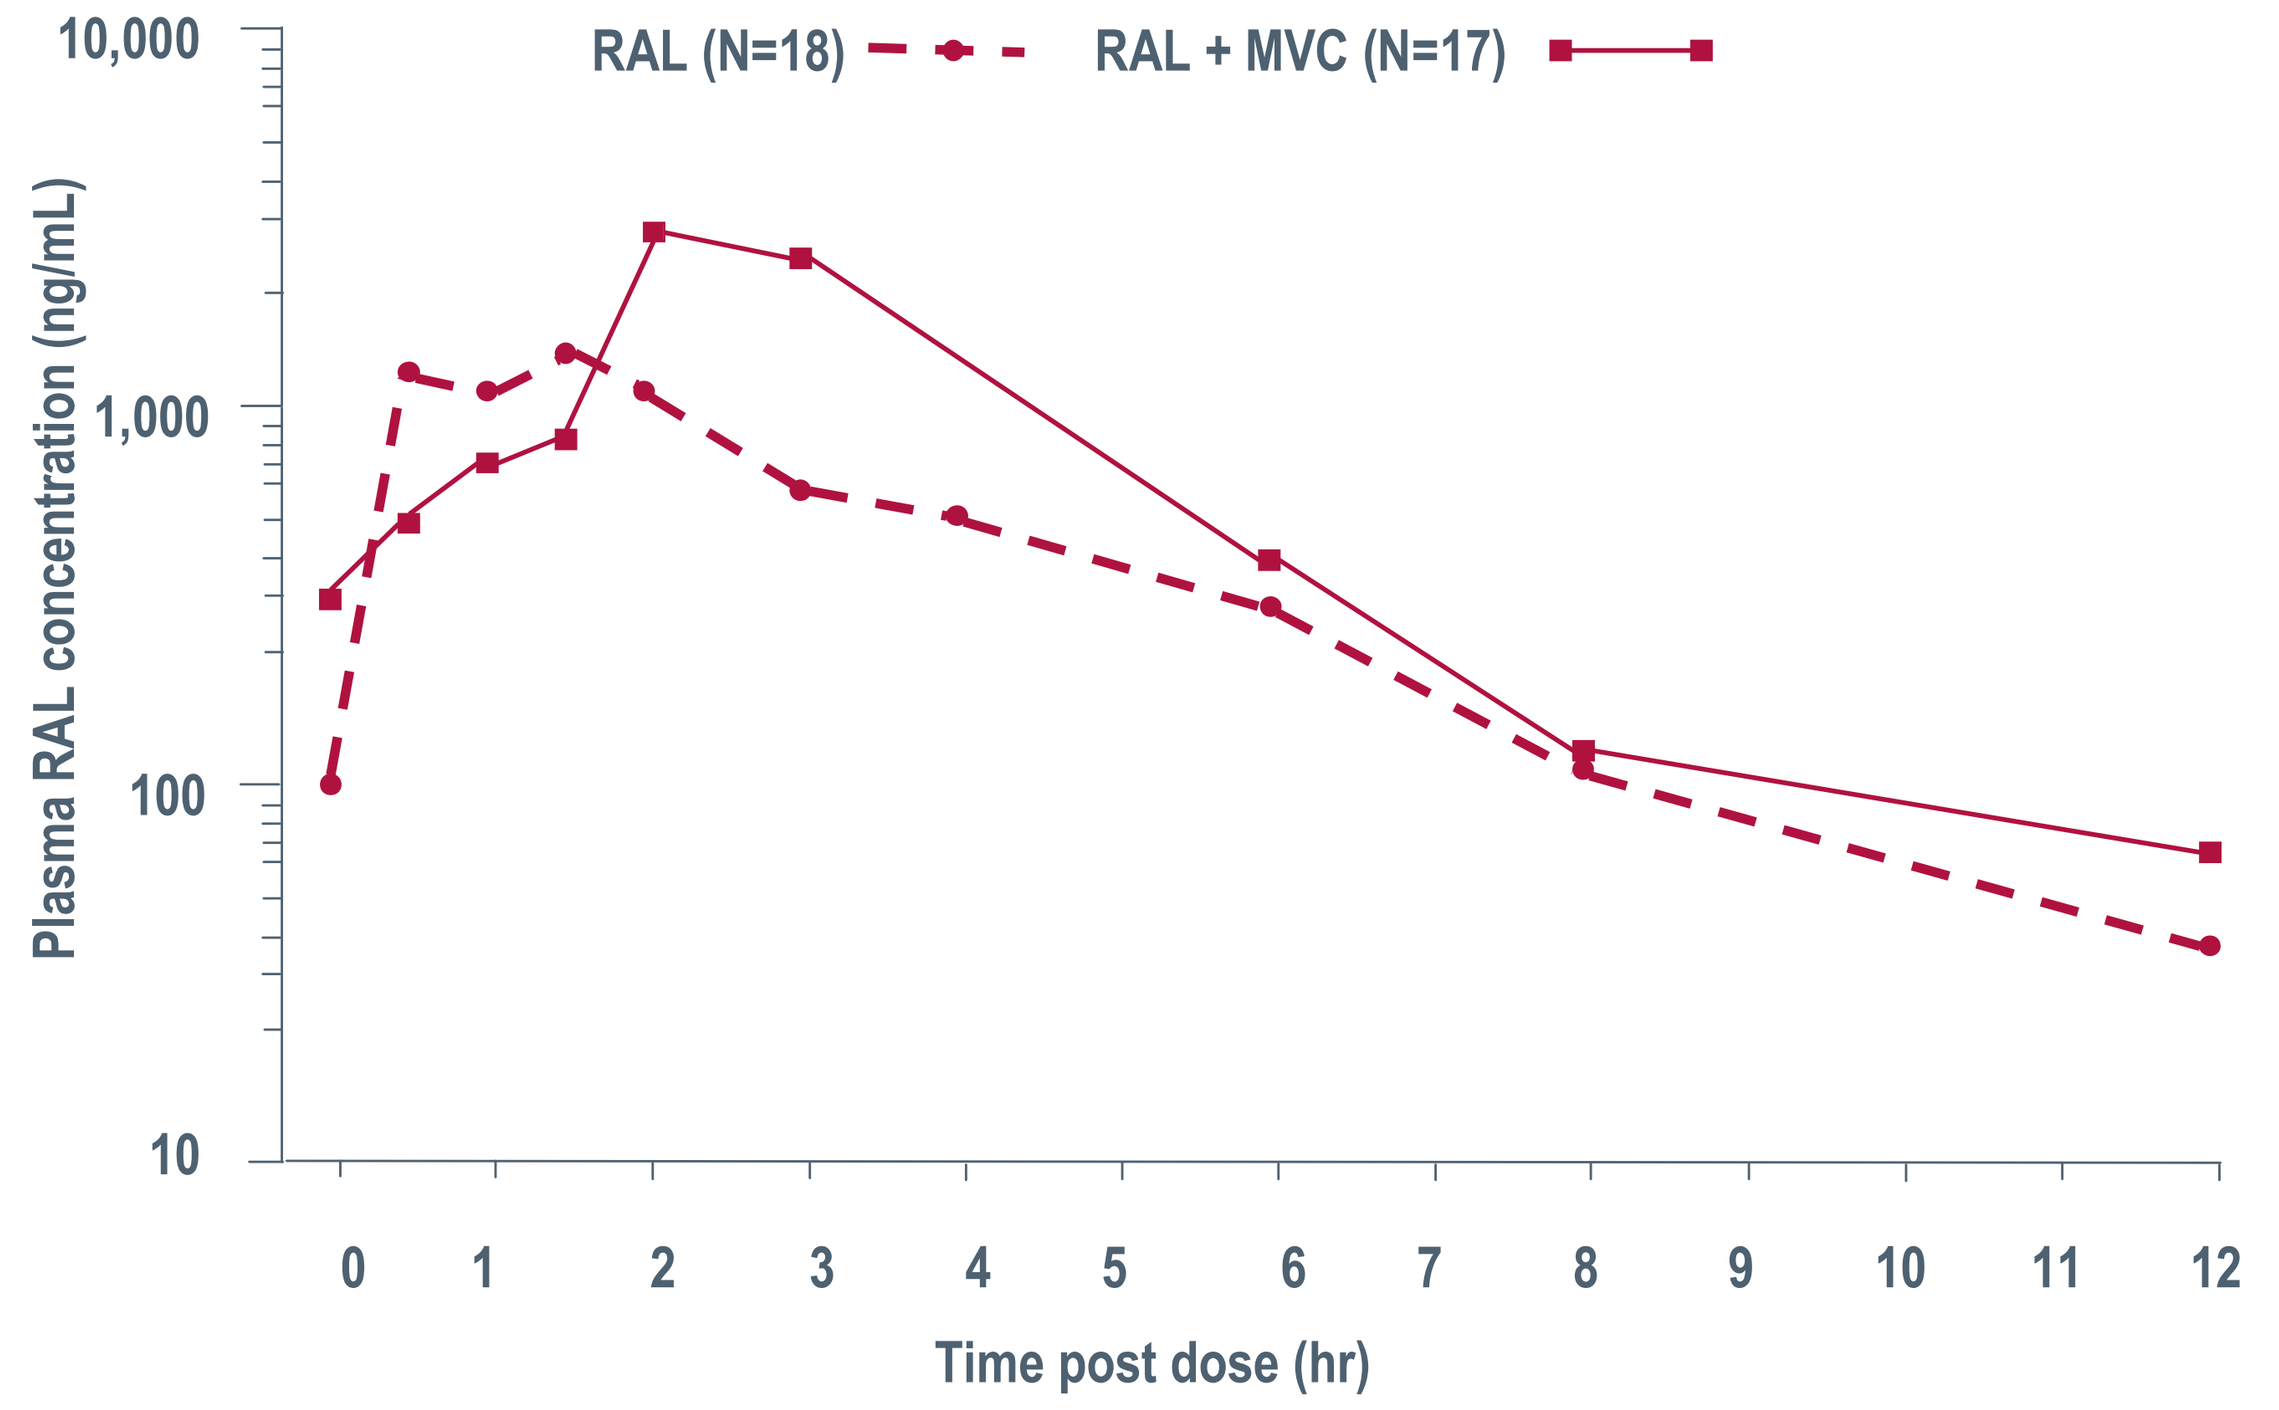

RAL (N=18)
RAL + MVC (N=17)
Plasma RAL concentration (ng/mL)
10,000
1,000
100
10
0 1 2 3 4 5 6 7 8 9 10 11 12
Time post dose (hr)

- Geometric mean values for plasma PK parameters are presented in Tables 1 and 3 and statistical analyses are presented in Tables 2 and 4 for MVC and RAL, respectively.
- When coadministered with RAL, MVC AUC_τ was reduced by an average of 14% relative to monotherapy. However, MVC average concentration (AUC_τ divided by 12) was >100 ng/mL in all subjects. RAL coadministration reduced MVC C_{max} by an average of 21% relative to MVC monotherapy. Median T_{max} was similar for MVC given together with RAL compared to MVC given alone.
- When coadministered with MVC, RAL AUC_τ was reduced by an average of 37% relative to monotherapy. MVC coadministration reduced RAL C_{12} values by an average of 28% relative to RAL monotherapy. Median T_{max} was 1.5 hours shorter for RAL given together with MVC compared to RAL given alone.

Table 1. Plasma PK parameters of MVC in the presence and absence of RAL

Parameter (units)	MVC + RAL N=17	MVC alone N=17
AUC_τ (ng·hr/mL)	2,447 (31)	2,853 (28)
C_{max} (ng/mL)	624 (47)	785 (41)
C_{12} (ng/mL)	46.6 (28)	51.6 (26)
T_{max} (hr)	2.0 [0.5–4.0]	2.0 [0.5–4.0]

Data expressed as geometric mean (% coefficient of variation), or median [range] for T_{max}

Table 2. Statistical analyses of plasma PK parameters of MVC

Parameter (units)	Adjusted geometric means		Geometric mean ratio (90% CI) (%)
	MVC + RAL	MVC alone	
AUC_τ (ng·hr/mL)	2,447	2,853	85.76 (79.89, 92.07)
C_{max} (ng/mL)	624	785	79.48 (67.19, 94.02)
C_{12} (ng/mL)	46.6	51.6	90.33 (85.28, 95.68)

Geometric mean values obtained from mixed model (ANOVA)

Table 3. Plasma PK parameters of RAL in the presence and absence of MVC

Parameter (units)	MVC + RAL N=17	RAL alone N=18
AUC_τ (ng·hr/mL)	4,819 (63)	7,356 (58)
C_{max} (ng/mL)	1,485 (72)	2,116 (67)
C_{12} (ng/mL)	47.0 (44)	66.2 (49)
T_{max} (hr)	1.0 [0.0–2.0]	2.5 [1.0–6.0]

Data expressed as geometric mean (% coefficient of variation), or median [range] for T_{max}

Table 4. Statistical analyses of plasma PK parameters of RAL

Parameter (units)	Adjusted geometric means		Geometric ratio (90% CI) (%)
	MVC + RAL	RAL alone	
AUC_τ (ng·hr/mL)	4,653	7,356	63.25 (44.27, 90.39)
C_{max} (ng/mL)	1,413	2,116	66.77 (41.22, 108.15)
C_{12} (ng/mL)	48.0	66.2	72.42 (57.82, 90.71)

Geometric mean values obtained from mixed model (ANOVA)

Safety results

- There were no serious adverse events, discontinuations, or dose reductions due to AEs.
- At least one treatment-emergent AE was experienced by three subjects (16.7%) during treatment with raltegravir, four subjects (22.2%) during treatment with maraviroc, and four subjects (23.5%) during treatment with maraviroc plus raltegravir. All events were mild and treatment related, and included constipation, dyspepsia, folliculitis, back pain, headache, peripheral neuropathy, and somnolence.

Discussion

- In healthy volunteers, relative to monotherapy, coadministration of MVC and RAL was associated with reductions in MVC C_{max} and RAL C_{12}, as well as a reduction in AUC_τ for both agents.
- Of all PK parameters investigated in a recent analyses of the exposure-response (efficacy) relationship for MVC in treatment-naive[5] and treatment-experienced[6] patients, MVC exposure (C_{ave}) appeared to correlate most closely with virologic success.
- The analysis in treatment-experienced patients suggested a greater than 80% net probability of success at an MVC average concentration (AUC_τ divided by 12) of >100 ng/mL, in subjects likely to respond to MVC.[6]
- In the present study, this MVC C_{ave} threshold value was achieved in all subjects when MVC was coadministered with RAL, which suggests that coadministration is unlikely to affect the antiretroviral activity of MVC (Figure 4a).
- Historically, considerable inter- and intra-subject variability has been observed in the pharmacokinetics of RAL. For observed C_{12} in pivotal Phase 3 studies (BENCHMRK 1 and 2), the coefficient of variation for inter-subject variability was 212% and for intra-subject variability was 122%.[2] Based on available clinical experience from Phase 1 and 2 studies, a drug–drug interaction-associated increase in exposure of up to 2-fold is considered to be not clinically relevant.[3]
- In a recent evaluation of the correlation of a range of RAL PK parameters with antiretroviral activity in Phase 2 and 3 studies in treatment-experienced patients, no statistically significant relationship was observed between RAL C_{12} and virologic response.[7] Based on available clinical experience from Phase 1 and 2 studies, a drug–drug interaction-associated decrease in RAL C_{12} of less than 60% is considered to be not clinically relevant.[3]
- When given with MVC, RAL C_{12} values were reduced by an average of 28% (90% CI: 9, 42). Two of 17 subjects had a decrease in C_{12} >60% (76% for one subject and 69% for the other; Figure 4b). However, given the variable PK associated with RAL, we believe this is unlikely to be clinically significant.
- Safety and efficacy data from studies of patients receiving regimens that include both MVC and RAL will be helpful in understanding the role of this combination in the treatment of HIV-1 infection.

Figure 4. Stick plots showing a) Change in MVC C_{ave} when coadministered with RAL, and b) Change in RAL C_{12} when coadministered with MVC

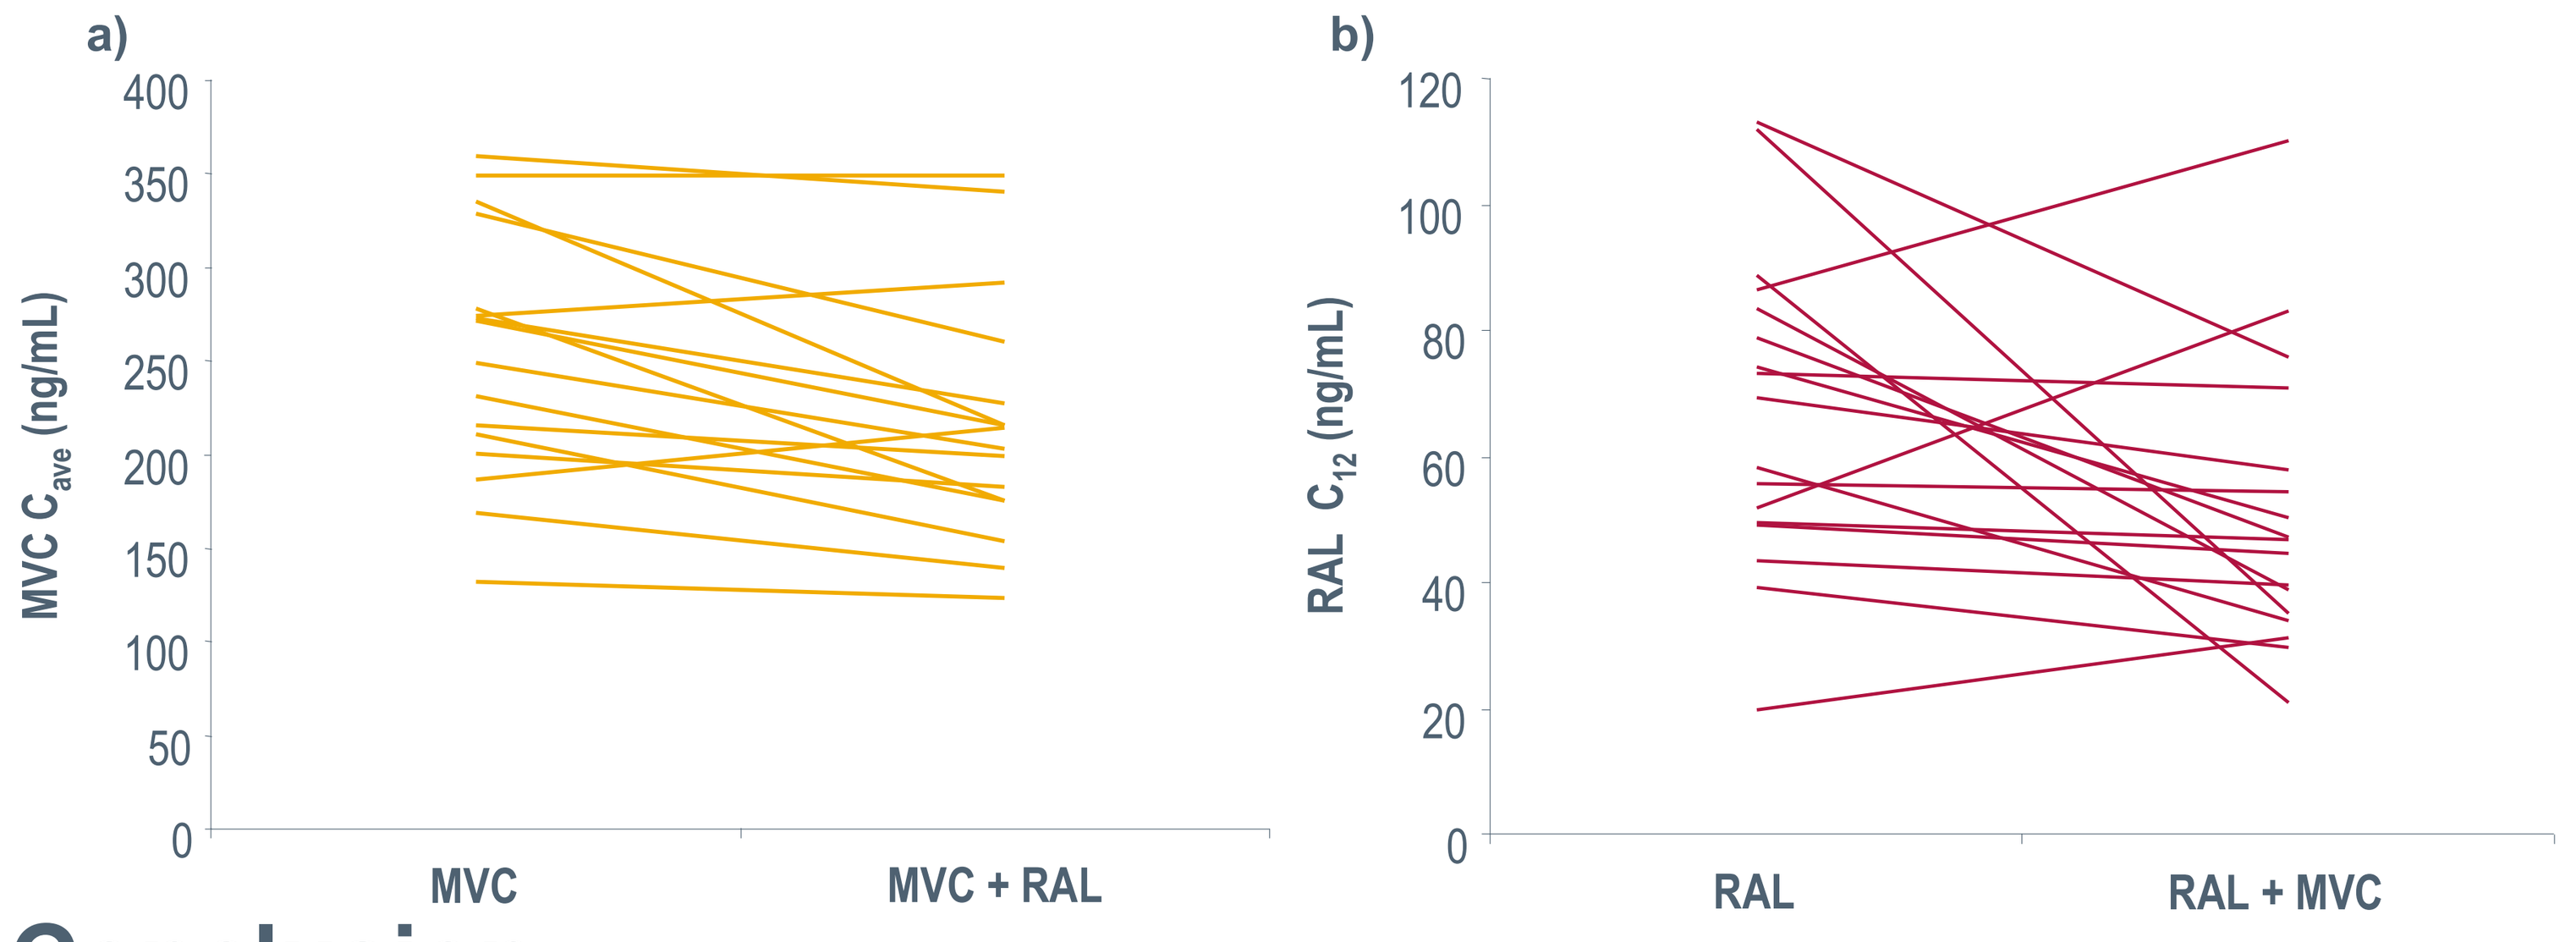

a)
MVC Cave (ng/mL)
400
350
300
250
200
150
100
50
0
MVC
MVC + RAL
b)
RAL C12 (ng/mL)
120
100
80
60
40
20
0
RAL
RAL + MVC

Conclusion

- When MVC and RAL were coadministered in healthy adults, MVC C_{max}, MVC AUC_τ, RAL C_{12} and RAL AUC_τ were reduced compared to monotherapy.
- These changes are not likely to be clinically relevant as exposure to both drugs appeared to be above pharmacokinetic–pharmacodynamic targets.
- Thus, no dose adjustment may be required when MVC and RAL are coadministered.
- MVC and RAL when administered alone and concomitantly were generally safe and well tolerated.

References

1. Pfizer Inc. *Selzentry prescribing information.* 2007.
2. Merck & Co, Inc. *Isentress prescribing information.* 2008.
3. Merck. *Isentress™ (raltegravir) 400 mg For Treatment of HIV. (NDA 22-145). Briefing Document. Presented as part of the FDA Antiviral Drugs Advisory Committee meeting, held September 5, 2007. Available at: www.fda.gov/ohrms/dockets/ac/07/briefing/2007-4314b1-01-Merck.pdf.*
4. Merschman SA, *et al. J Chromatogr B Analyt Technol Biomed Life Sci* 2007; **857**:15–24.
5. McFadyen L, *et al. XVII International AIDS Conference.* Mexico City, Mexico, 3–8 August, 2008; Poster TUPE0053.
6. McFadyen L, *et al. 11th European AIDS Conference.* Madrid, Spain, 24–27 October, 2007; Poster P4.1/06.
7. Wenning L, *et al. 9th International Workshop on Clinical Pharmacology of HIV Therapy.* New Orleans, USA, 7–9 April 2008.

Pfizer HIV/AIDS