Poster Number
H-2307
48th Annual ICAAC / IDSA 46th Annual Meeting
October 25-28, 2008
Washington, DC, USA

The Impact of Tenofovir on Insulin Sensitivity and Lipids in Healthy Volunteers

P Randell,[1] A Jackson,[1] L Zhong,[2] K Yale,[2] and G Moyle[1]

[1]Chelsea and Westminster Hosp., London, UK; [2]Gilead Sciences, Inc, Foster City, CA, USA

Gilead Sciences, Inc.
333 Lakeside Drive
Foster City, CA 94404
Tel: (650)574-3000
Fax: (650)578-9264

Introduction

- Effective antiretroviral therapy has resulted in a dramatic improvement in HIV associated morbidity and mortality
- Certain antiretrovirals are associated with metabolic side effects including dyslipidaemia, body fat changes and insulin resistance[1]
- These effects are variable not only between different drug classes but between drugs in the same class

Background

- The hyperinsulinaemic euglycaemic clamp[2] is considered the gold standard for investigating insulin sensitivity and a number of trials have used the clamp methodology to investigate antiretrovirals
- The development of insulin resistance in healthy volunteers has been seen with lopinavir/ritonavir[3], ritonavir[4], indinavir[5] and stavudine[6] but not with amprenavir[4] or atazanavir[3]
- Insulin resistance associated with protease inhibitors appears to be mediated through the inhibition of the glucose transporter GLUT4[7]
- Mitochondrial toxicity is implicated in the development of insulin resistance with nucleoside reverse transcriptase inhibitors[6]

Objectives

- To investigate the impact of tenofovir disoproxil fumarate (TDF) on insulin sensitivity as assessed by the peripheral glucose disposal rate during the last hour of the hyperinsulinaemic euglycaemic clamp
- To investigate the impact of TDF on lipid parameters

Methods

- Single centre, randomised, double blinded, placebo-controlled study that utilised a 2 sequence, 2 period crossover design
- HIV negative subjects were recruited and randomised 1:1 as shown (fig 1)

Methods (cont'd)

Figure 1. Study Design

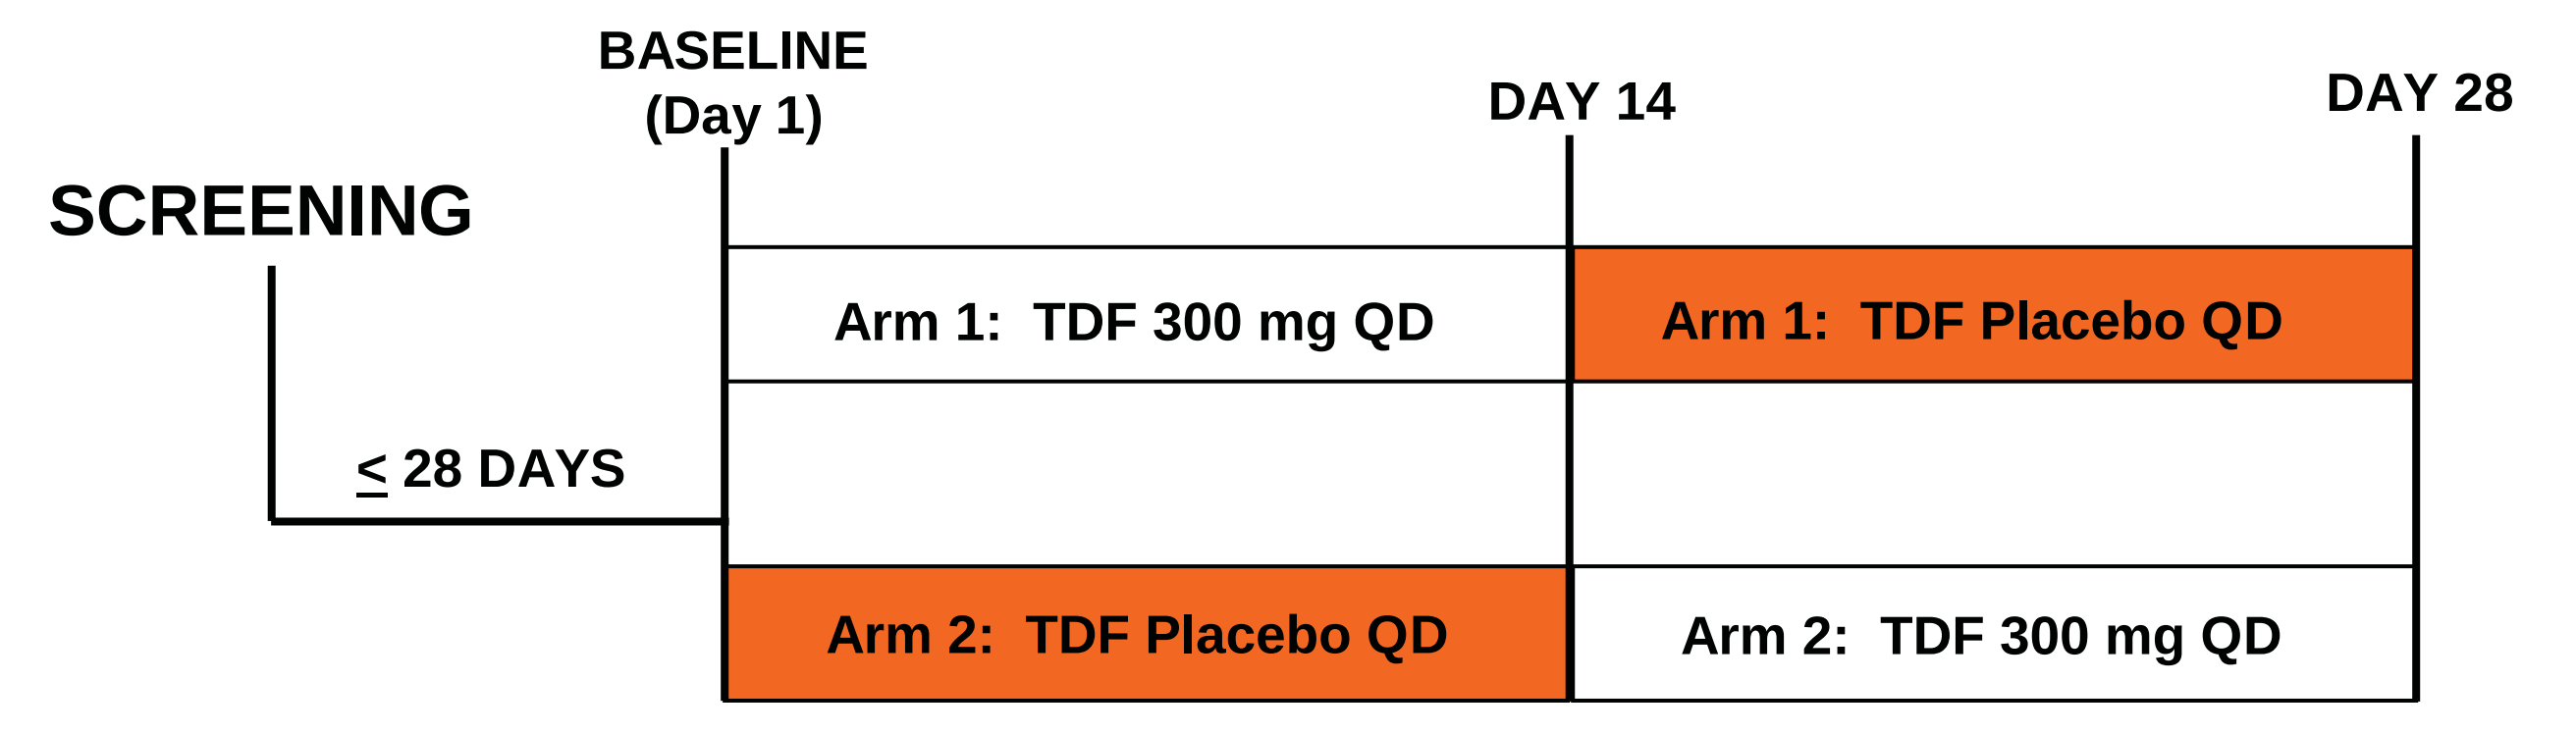
SCREENING
BASELINE (Day 1)
DAY 14
DAY 28
≤ 28 DAYS
Arm 1: TDF 300 mg QD
Arm 1: TDF Placebo QD
Arm 2: TDF Placebo QD
Arm 2: TDF 300 mg QD

- Important exclusion criteria were BMI >28kg/m^2, waist:hip ratio >0.97 and any medication or disease process likely to cause a marked disturbance in glucose and lipid homeostasis. Fasting glucose, cholesterol and triglycerides also had to be within normal limits
- A hyperinsulinaemic euglycaemic clamp and fasting bloods were performed at baseline, after 2 weeks of treatment and after 4 weeks of treatment (2 weeks after treatment switch)
- Peripheral glucose disposal rate was assessed during the last hour (120-180 min) of the clamp

Results

- 19 subjects were randomised. 16 male subjects completed all 3 clamp procedures (3 withdrew prior to baseline visit)
- Subjects had mean age of 27.4 years and mean BMI of 23.02 kg/m^2
- 7 subjects received TDF followed by placebo and 9 received placebo followed by TDF
- No significant change from baseline in the mean peripheral glucose disposal rate was seen after 2 weeks of TDF (fig 2b) or placebo (fig 2a)
 - No significant difference between placebo and TDF
- Statistically significant decreases from baseline in total cholesterol and LDL cholesterol were seen after 2 weeks of TDF (fig 3)

Results (cont'd)

Figure 2. Glucose Disposal Rate

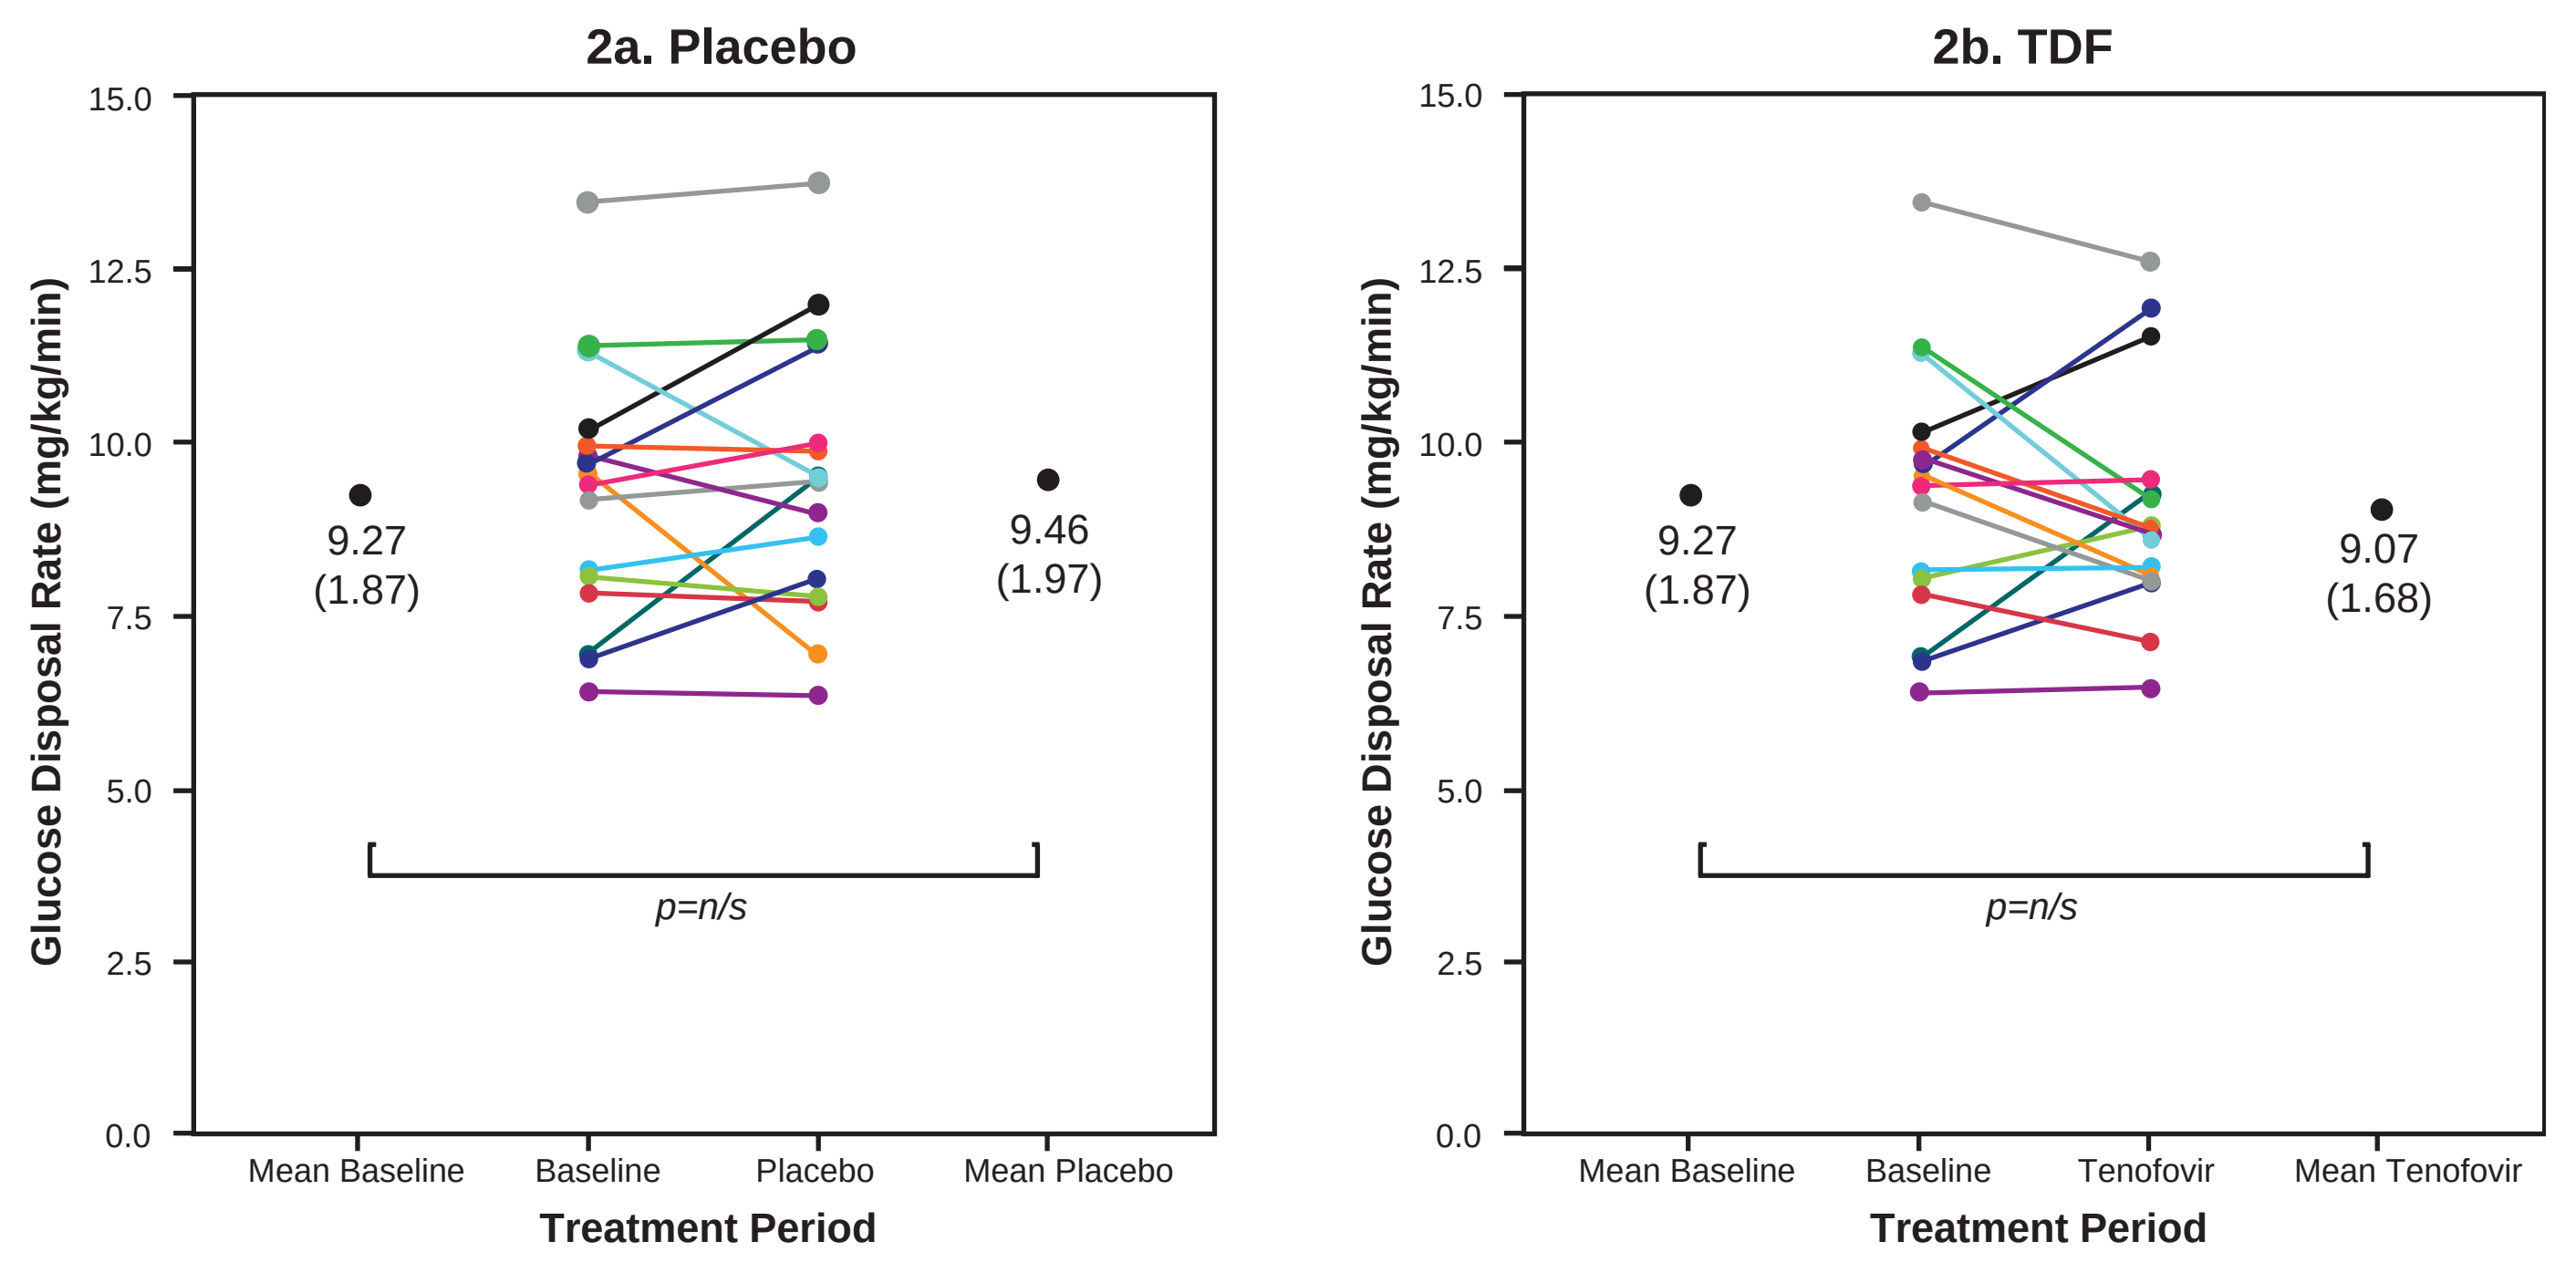
2a. Placebo
2b. TDF
Glucose Disposal Rate (mg/kg/min)
15.0
12.5
10.0
7.5
5.0
2.5
0.0
9.27 (1.87)
9.46 (1.97)
9.07 (1.68)
p=n/s
Mean Baseline
Baseline
Placebo
Mean Placebo
Tenofovir
Mean Tenofovir
Treatment Period

Mean values are presented with (SD). p-value obtained using Wilcoxon signed rank test

Figure 3. Fasting Lipid Parameters

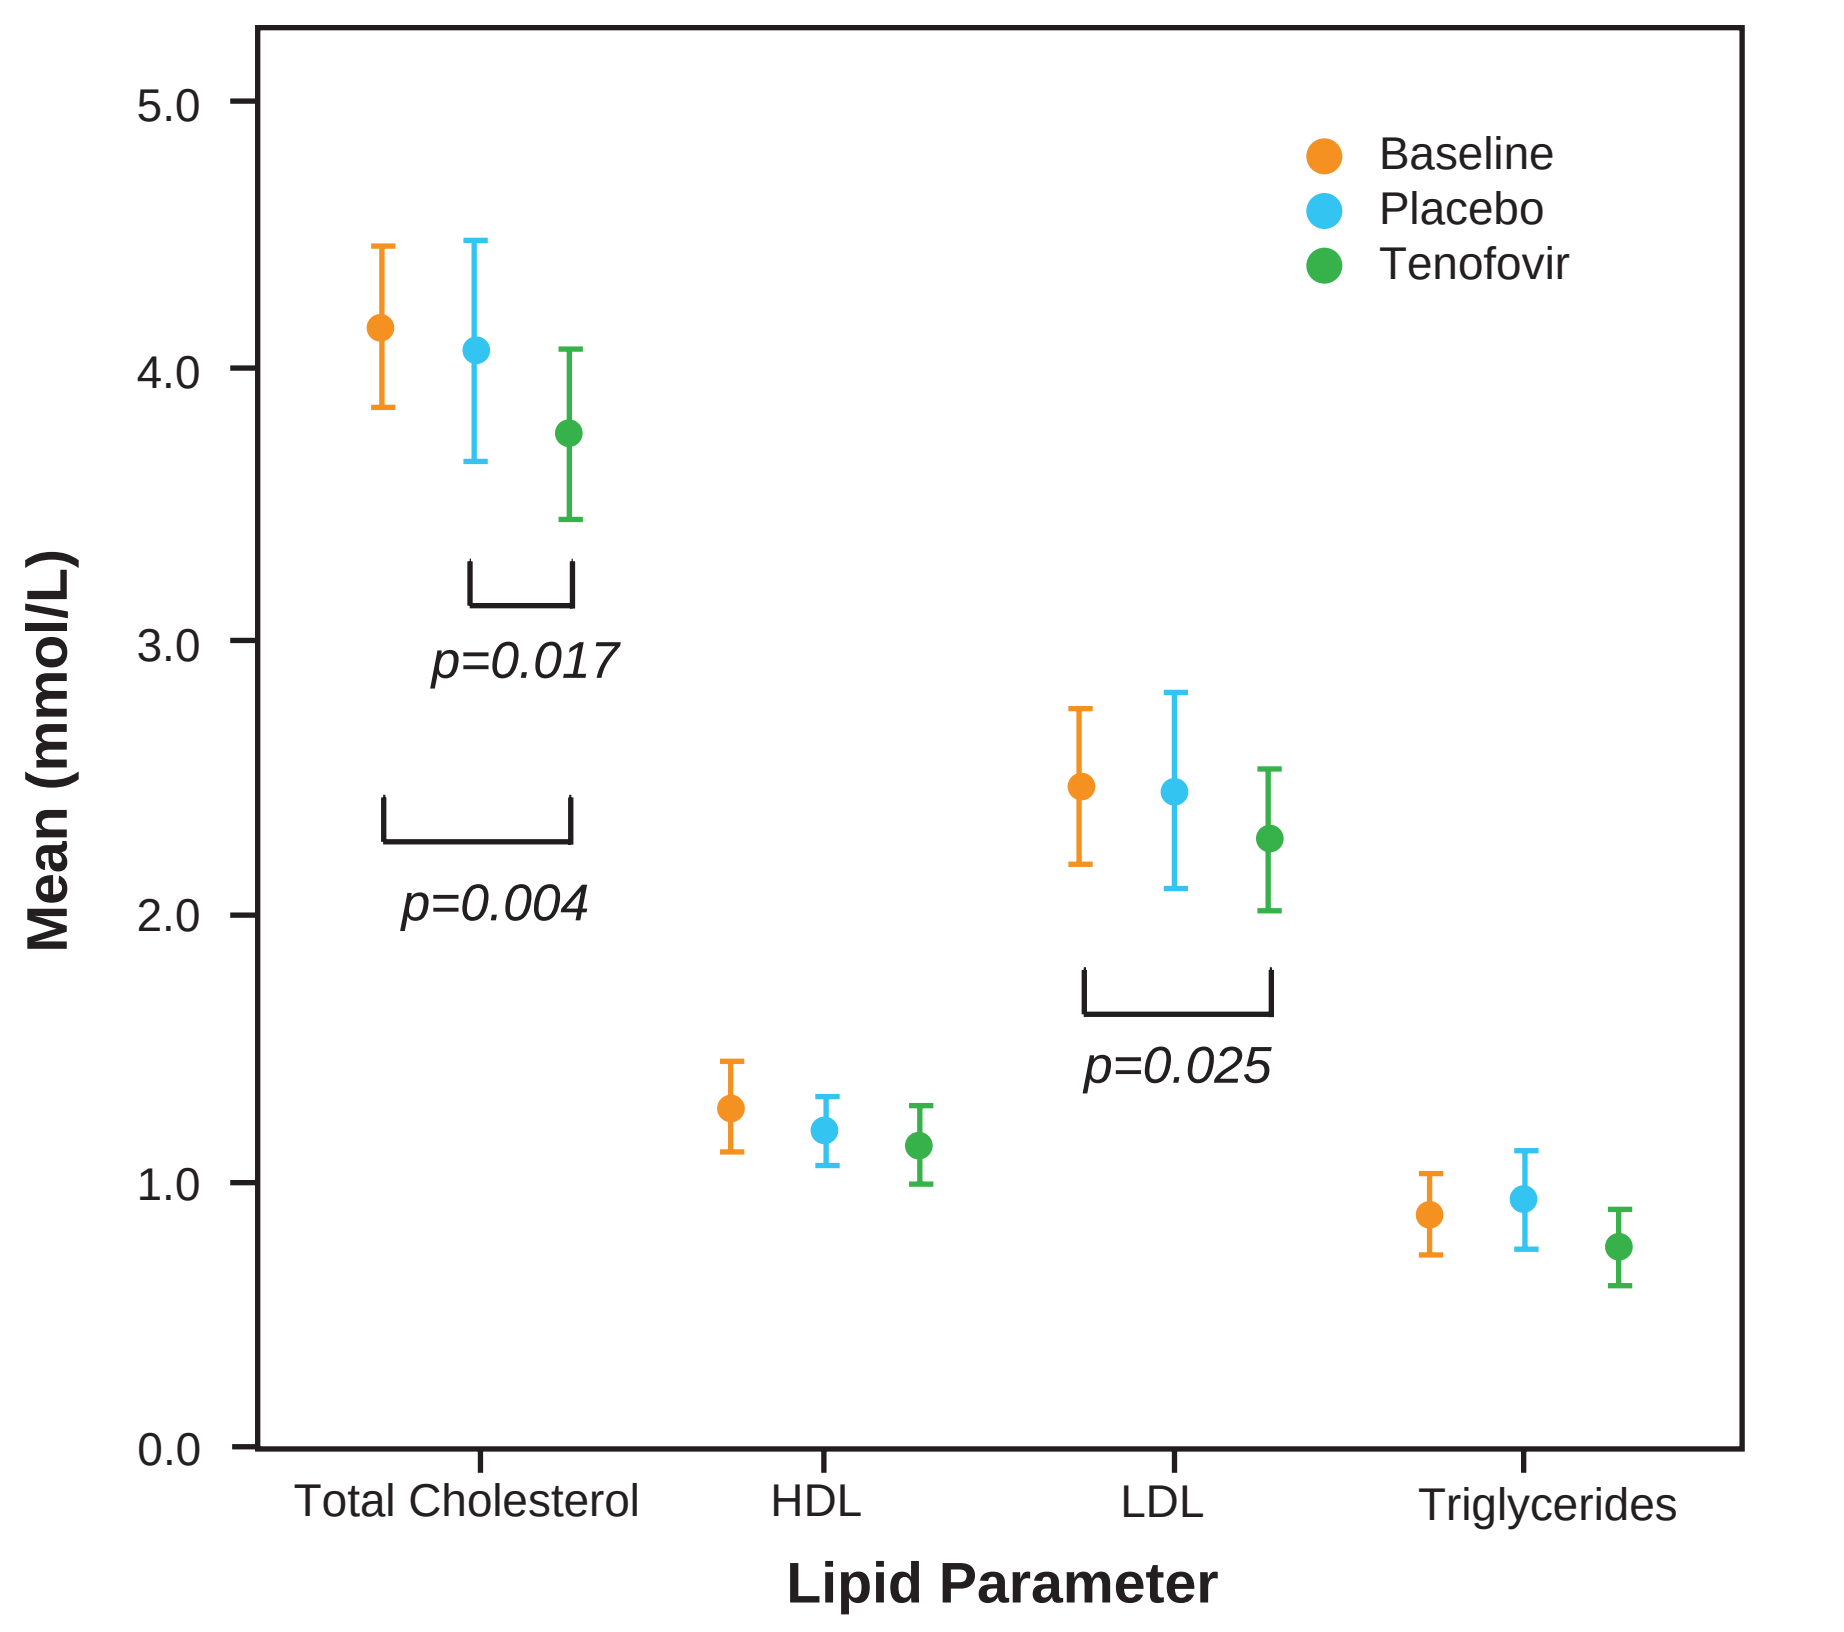
Baseline
Placebo
Tenofovir
Mean (mmol/L)
5.0
4.0
3.0
2.0
1.0
0.0
p=0.017
p=0.004
p=0.025
Total Cholesterol
HDL
LDL
Triglycerides
Lipid Parameter

Error Bars: 95% CI. p-values obtained using Wilcoxon signed rank test.
Non statistically significant changes not shown.

Conclusions

- **Two-week dosing with TDF did not have a significant impact on insulin sensitivity**
- **Statistically significant reductions in total cholesterol and LDL cholesterol were seen after 2 weeks of TDF**
- **These study results are consistent with previous observations that no significant mitochondrial toxicity was associated with TDF[8]**

References

1. Grinspoon S, Carr A. Cardiovascular risk and body-fat abnormalities in HIV-infected adults. N Engl J Med 2005;352:48-62.
2. DeFronzo RA, Tobin JD, Andres R. Glucose clamp technique: a method for quantifying insulin secretion and resistance. Am J Physiol. 1979;237(3):E214-E23.
3. Noor MA, Parker RA, O'Mara E, et al. The effects of HIV protease inhibitors atazanavir and lopinavir/ritonavir on insulin sensitivity in HIV-seronegative healthy adults. AIDS. 2004;18(16):2137–44.
4. Lee GA, Rao M, Mulligan K, et al. Effects of ritonavir and amprenavir on insulin sensitivity in healthy volunteers. AIDS. 2007;21(16):2183–90.
5. Noor MA, Seneviratne T, Aweeka FT, et al. Indinavir acutely inhibits insulin-stimulated glucose disposal in humans: A randomized, placebo-controlled study. AIDS. 2002;16(5):F1-F8.
6. Fleischman A, Johnsen S, Systrom DM, et al. Effects of a nucleoside reverse transcriptase inhibitor, stavudine, on glucose disposal and mitochondrial function in muscle of healthy adults. Am J Physiol Endocrinol Metab. 2007;292:E1666–E73.
7. Murata H, Hruz PW, Mueckler M. The mechanism of insulin resistance caused by HIV protease inhibitor therapy. The Journal of Biological Chemistry. 2000;275(27):20251-4.
8. Birkus B, Hitchcock M, Cihlar T. Assessment of mitochondrial toxicity in human cells treated with tenofovir: comparison with other nucleoside reverse transcriptase Inhibitors. Antimicrobial Agents and Chemotherapy. 2002;46(3):716-723.