48th Annual ICAAC/IDSA 46th Meeting, Washington, DC, USA, October 26–28, 2008

Poster H-1248

CD4+ Cell Increases at 48 Weeks in the Maraviroc Treatment-naive MERIT Trial

A Lazzarin[1], M Battegay[2], DA Cooper[3], JG Sieera-Madero[4], JB Angel[5], I Frank[6] R Wood[7], JM Goodrich[8], J Heera[8], N Rajicic[9], R Tressler[9], H Valdez[9] and H Mayer[8]

[1]San Raffaele Scientific Institute, Milan Italy; [2]Universitätsspital Basel, Switzerland; [3]University of New South Wales, Sydney, Australia; [4]Instituto Nacional de Ciencias Médicas y Nutrición Salvador Zubirán, Mexico City, Mexico; [5]University of Ottawa, Canada; [6]University of Pennsylvania, Philadelphia, USA; [7]The Desmond Tutu HIV Centre, Observatory, South Africa; [8]Pfizer Global Research and Development, New London, USA; [9]Pfizer Inc, New York, USA

For further information, please contact Hernan Valdez at Hernan.Valdez@pfizer.com

Background

- Treatment-induced increases in CD4+ cell count are associated with clinical benefits beyond decreasing the occurrence of AIDS-defining illnesses:
 - Mortality rates for AIDS-defining malignancies (ADM) were 20.1 vs 0.1 per 1,000 patient-years and for non-ADM were 6.0 vs 0.6 per 1,000 patient-years when the current CD4+ cell count was <50 vs >500 cells/mm³, respectively.[1]
 - HIV-infected adults who achieve CD4+ cell counts of ≥500 cells/mm³ for ≥6 years have a survival rate similar to that of HIV-negative individuals.[2]
- Immunologic response to HAART in previously ARV-naive patients has been shown to be an important predictor of clinical outcome regardless of virologic response.[3]
 - A lack of immunologic response 3–9 months after HAART initiation in patients who respond virologically has been shown to be significantly associated with earlier development of an opportunistic infection or death.[4]
 - In the CPCRA FIRST trial, following initiation of HAART and after a median follow-up of 5 years, each 100 cell/mm³ higher CD4+ cell count during follow-up was independently associated with a 14% lower risk for non-AIDS diseases (liver disease, cardiovascular disease, renal disease, and non-ADM).[5] A poor initial CD4+ recovery (<50 or 100 cells/mm³ after 8 months of virologically effective HAART), was associated with increased risk for a composite outcome of AIDS, non-AIDS disease, or death.[6]
- The MERIT study is an ongoing, randomized, double-blind study designed to compare the safety and efficacy of maraviroc (MVC; marketed as Celsentri®/Selzentry™) 300 mg BID vs efavirenz (EFV) 600 mg QD, both administered with Combivir® (CBV; a fixed-dose combination of zidovudine and lamivudine), in treatment-naive patients with R5 HIV-1 (Trofile™ assay, Monogram Biosciences, South San Francisco, CA).
 - As previously presented,[7] analysis of Week 48 data demonstrated that 65% of patients receiving MVC achieved an HIV-1 RNA level of <50 copies/mL compared to 69% for EFV.[7] More patients discontinued from MVC for lack of efficacy compared to EFV (11.9% vs 4.2%), whereas fewer patients discontinued MVC due to adverse events (4.2% vs 13.6%).[7]

Objectives

- The objectives of these *post-hoc* analyses are:
 - To describe CD4+ cell count increases through 48 weeks and the incidence of discordant virologic–immunologic responses after 6 months in treatment-naive patients receiving MVC or EFV in the MERIT study.
 - To identify predictors of CD4+ cell count increases in treatment-naive patients receiving MVC vs EFV.
 - To identify factors associated with the development of Category C (AIDS-defining) events.

Methods

- The design of the MERIT study has been presented previously.[7]
- For this *post-hoc* analysis, missing CD4+ cell count values were handled using a method of last observation carried forward (LOCF).

Data summaries and statistical analyses

- Descriptive summaries.
- Comparison of frequency distributions of concordant and discordant immunologic and virologic responses using a Pearson's chi-squared test.
- Summary of number of patients exceeding clinically meaningful CD4+ cell count thresholds.
- Repeated-measures model of CD4+ cell counts over time that included treatment arm and visit, with interaction, as model terms.
- A multivariate regression model of Week 48 CD4+ cell count change from baseline that included baseline CD4+ cell count and HIV-1 RNA level, Week 48 HIV-1 RNA change from baseline, age, gender, and treatment arm as model predictors.
- A Cox proportional hazard regression model to analyze time to Category C event, using baseline CD4+ cell count, baseline HIV-1 RNA level, time-adjusted CD4+ cell count, achieving HIV-1 RNA <50 copies/mL by week 48, age, gender, and treatment arm as variables.

Results

- Baseline characteristics of the MERIT study population are summarized in Table 1.
- Significantly greater and faster increases in CD4+ cell count were observed in patients receiving MVC compared to EFV (Figure 1).
 - The median (IQR) change from baseline in CD4+ cell count at Week 48 was +151 (+76, +236) cells/mm³ in the MVC arm compared to +122 (+54, +204) cells/mm³ in the EFV arm (P=0.002).
 - The difference between treatment arms in CD4+ cell counts responses was apparent as early as Week 4, when patients in the MVC arm had experienced a median (IQR) change from baseline of +77 (+24, +130) cells/mm³ compared to +47 (0, +111) cells/mm³ for patients in the EFV arm (P=0.0001).
- 79% of patients receiving MVC who started the trial with <200 CD4+ cells/mm³ achieved ≥200 cells/mm³ by Week 48, compared to 71% of patients receiving EFV (Figure 2).

Table 1. Baseline characteristics

Randomized: N=740 Treated: N=721	EFV + CBV N=361	MVC + CBV N=360
Mean age, years (range)	37.4 (18–77)	36.7 (20–69)
Male, n (%)	259 (71.7)	256 (71.1)
Mean (SD) HIV-RNA, log_{10} copies/mL	4.9 (0.6)	4.8 (0.7)
Median (IQR) CD4+ cell count, cells/mm³	259 (189, 327)	244 (176, 320)
CD4+ cell count at baseline, n (%):		
<50 cells/mm³	9 (3)	13 (4)
50-99 cells/mm³	14 (4)	17 (5)
100-199 cells/mm³	84 (23)	94 (26)
200-349 cells/mm³	184 (51)	164 (46)
≥350 cells/mm³	69 (19)	71 (20)
Median (IQR) CD8+ cell count, cells/mm³*	860 (612, 1,142)	792 (602, 1,150)

SD, standard deviation; IQR, interquartile range from 25th to 75th percentile
Baseline CD4+ count values missing for one patient in each treatment arm; baseline CD8+ count values missing for one patient in the EFV arm

Figure 1. Significantly greater and faster increases in CD4+ cell count were observed in patients receiving MVC + CBV compared to EFV + CBV

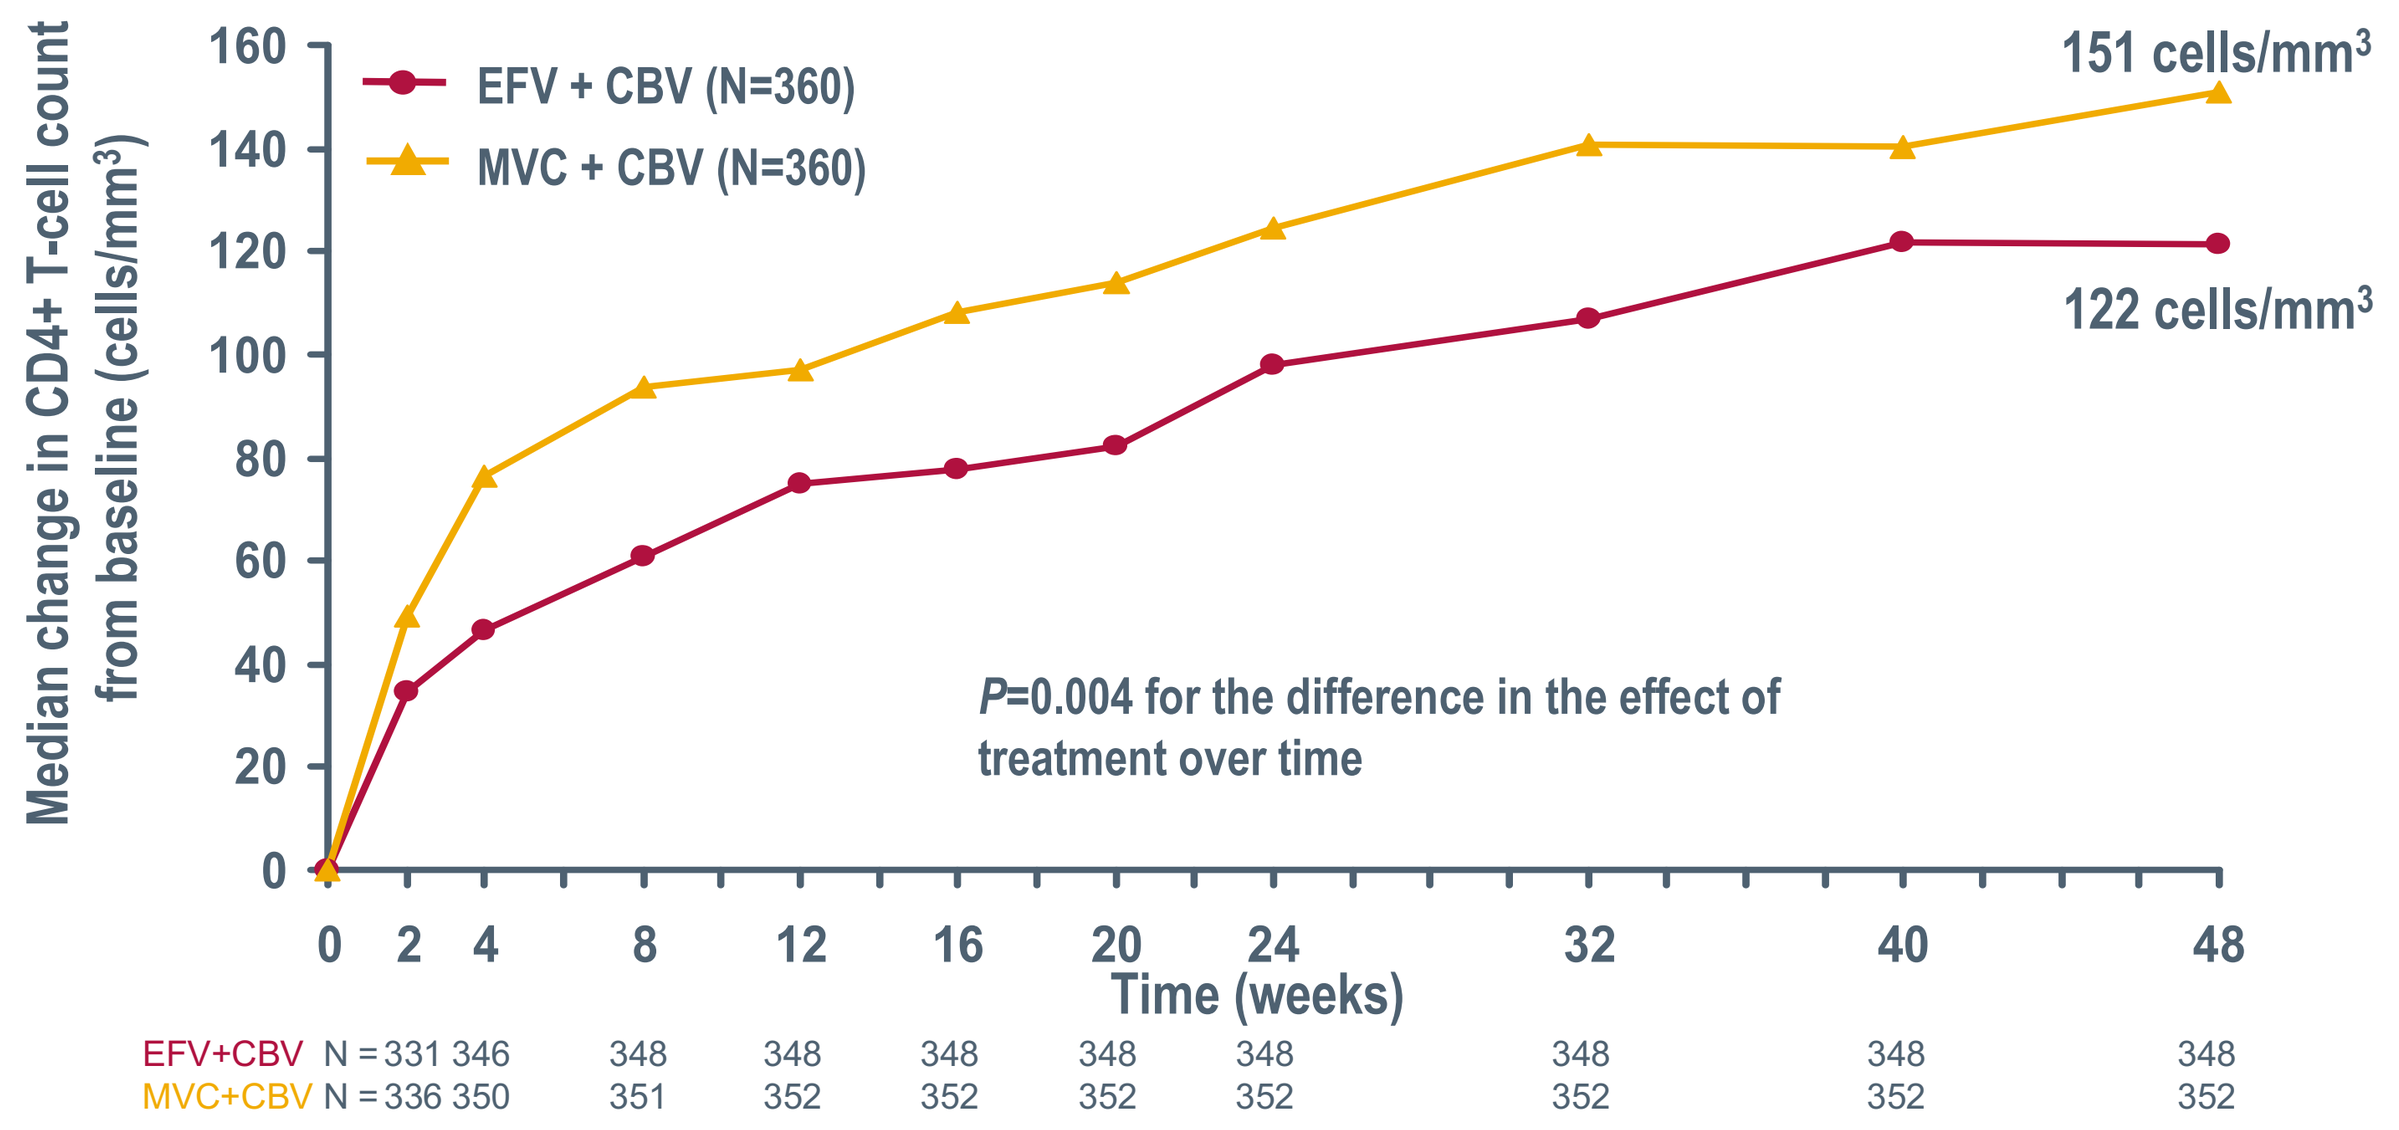
Median change in CD4+ T-cell count from baseline (cells/mm³)
EFV + CBV (N=360)
MVC + CBV (N=360)
151 cells/mm³
122 cells/mm³
P=0.004 for the difference in the effect of treatment over time
Time (weeks)
0 2 4 8 12 16 20 24 32 40 48
EFV+CBV N = 331 346 348 348 348 348 348 348 348 348
MVC+CBV N = 336 350 351 352 352 352 352 352 352 352

Includes all patients who received at least one dose of study medication and had a baseline CD4+ cell count value (LOCF)

Figure 2. A larger percentage of patients in the MVC arm exceeded clinically meaningful CD4+ cell count thresholds by Week 48 compared to those in the EFV arm

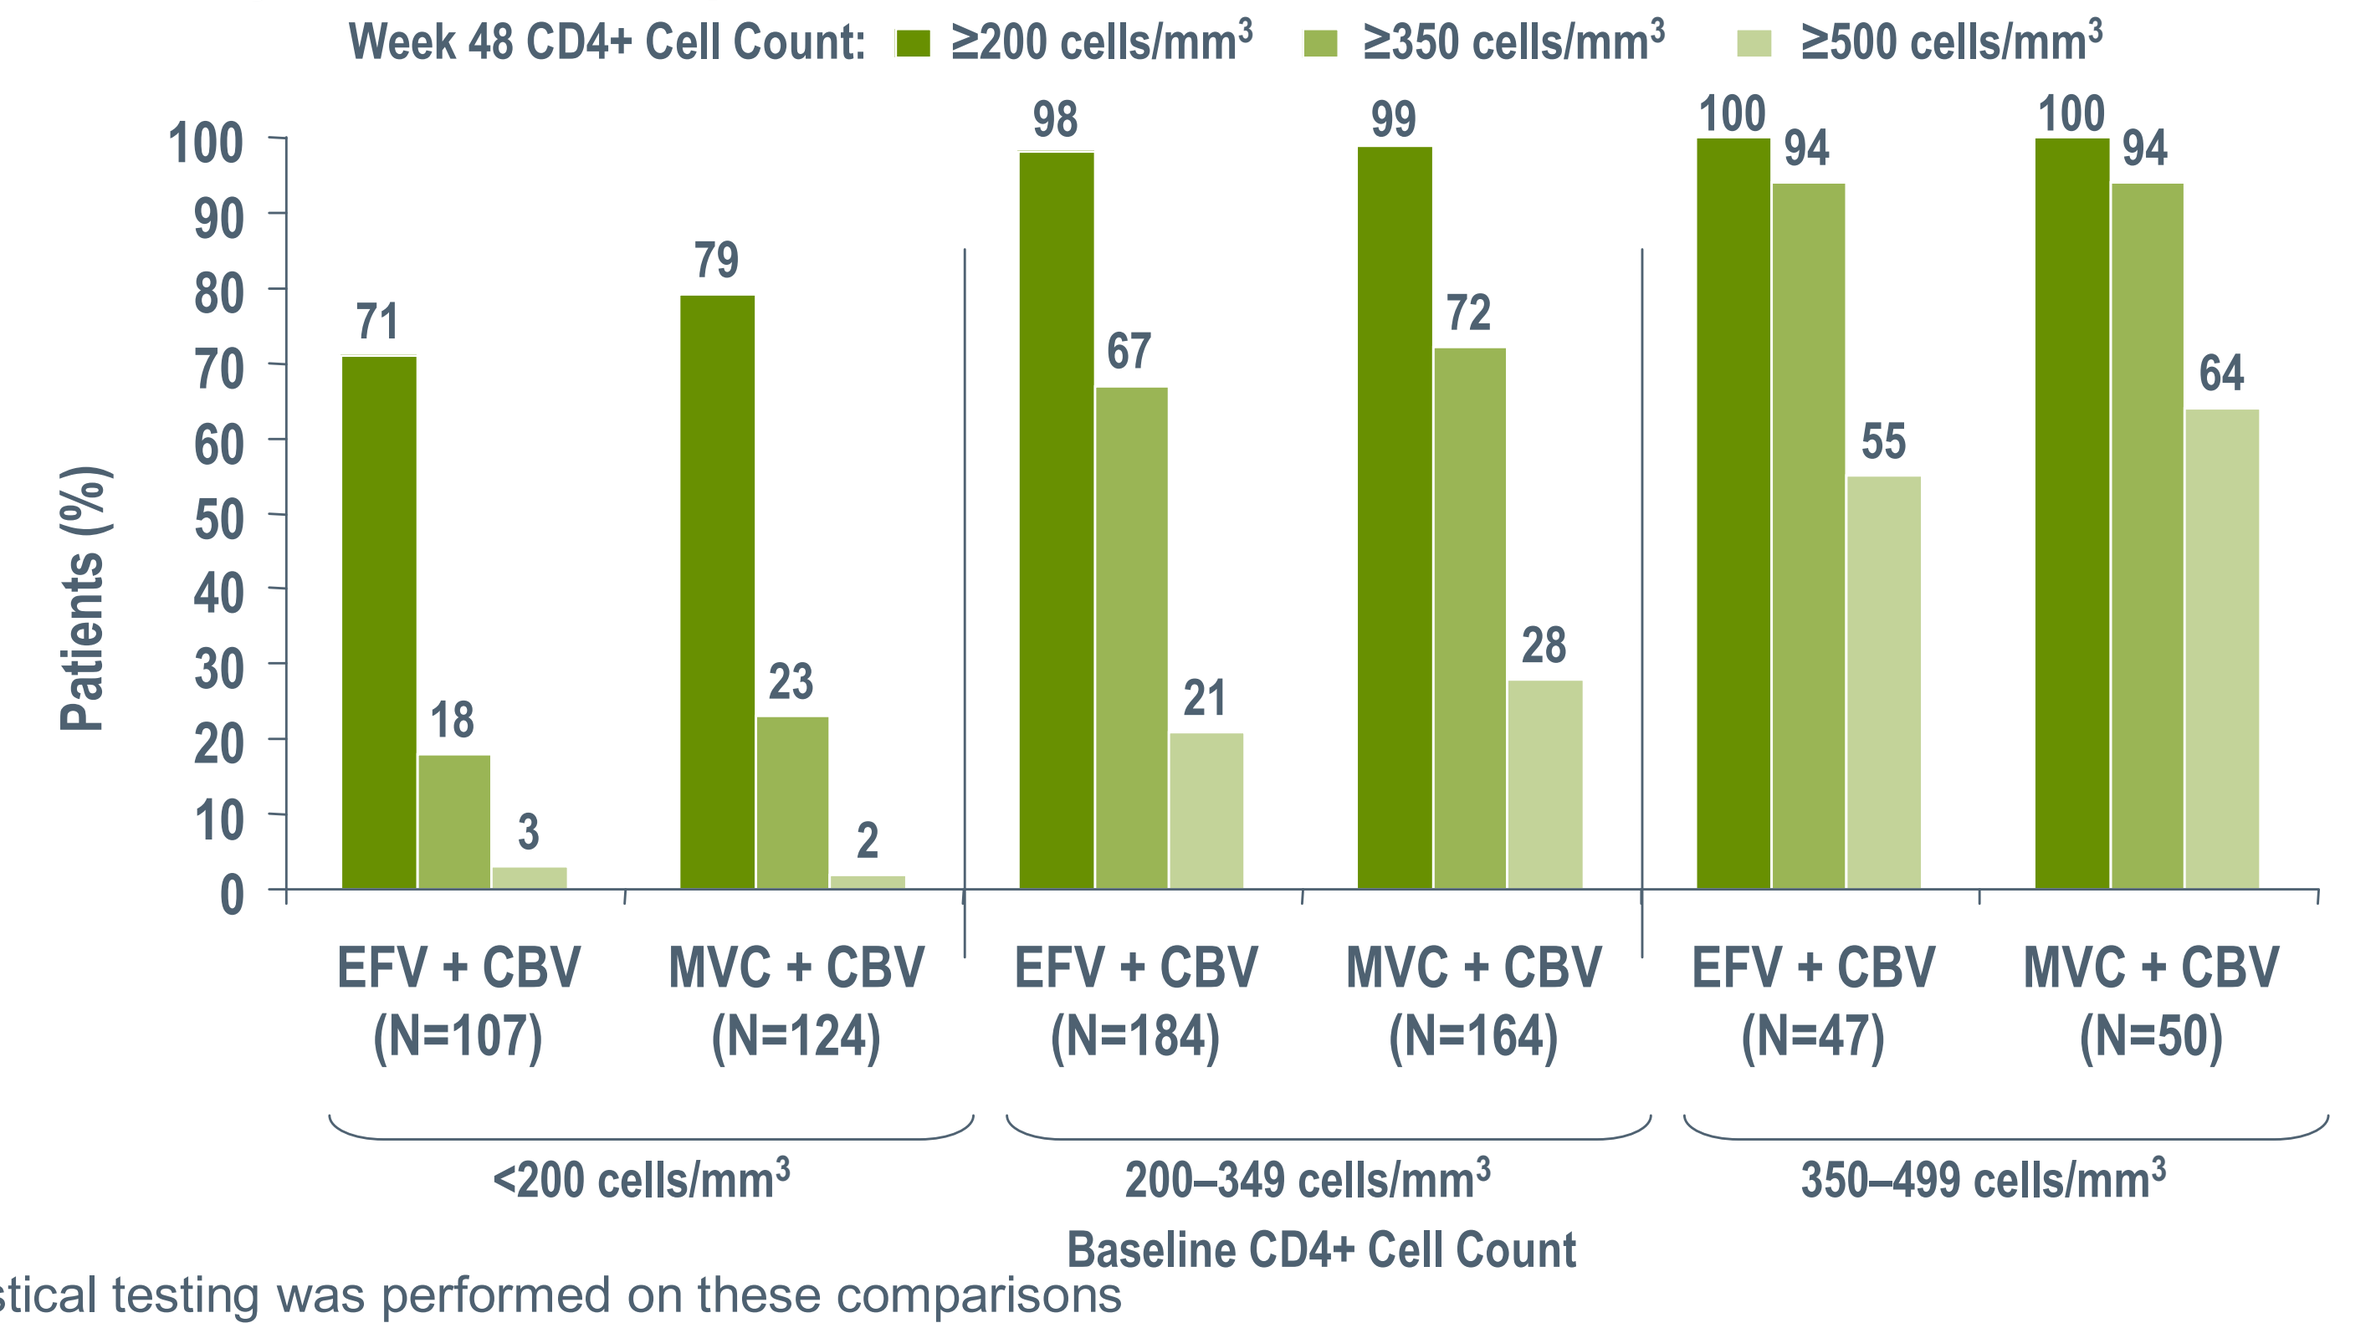
Week 48 CD4+ Cell Count: ≥200 cells/mm³ ≥350 cells/mm³ ≥500 cells/mm³
Patients (%)
71 18 3
79 23 2
98 67 21
99 72 28
100 94 55
100 94 64
EFV + CBV (N=107)
MVC + CBV (N=124)
EFV + CBV (N=184)
MVC + CBV (N=164)
EFV + CBV (N=47)
MVC + CBV (N=50)
<200 cells/mm³
200–349 cells/mm³
350–499 cells/mm³
Baseline CD4+ Cell Count

Note: No statistical testing was performed on these comparisons

Discordant virologic–immunologic responses

- After 6 months of study treatment, more patients in the MVC arm than in the EFV arm experienced concordant responses to study treatment, according to thresholds of <400 copies/mL for virologic response and >100 cells/mm³ increase in CD4+ cell count from baseline for immunologic response (Figure 3).
- Among virologic responders a higher proportion of patients in the EFV arm experienced a discordant immunologic response vs those in the MVC arm (Figure 3).
 - The percentage of patients achieving <400 copies/mL who did not experience a >100 cells/mm³ increase in CD4+ cell count from baseline was 36% for the EFV vs 29% for the MVC arm.
- Similar results were seen when response rates were observed for patients achieving <50 copies/mL who did not experience a >50 cells/mm³ increase in CD4+ cell count from baseline; P<0.001).

Figure 3. More concordant responders and a lower percentage of immunologic non-responders were observed among MVC-treated patients at 24 weeks

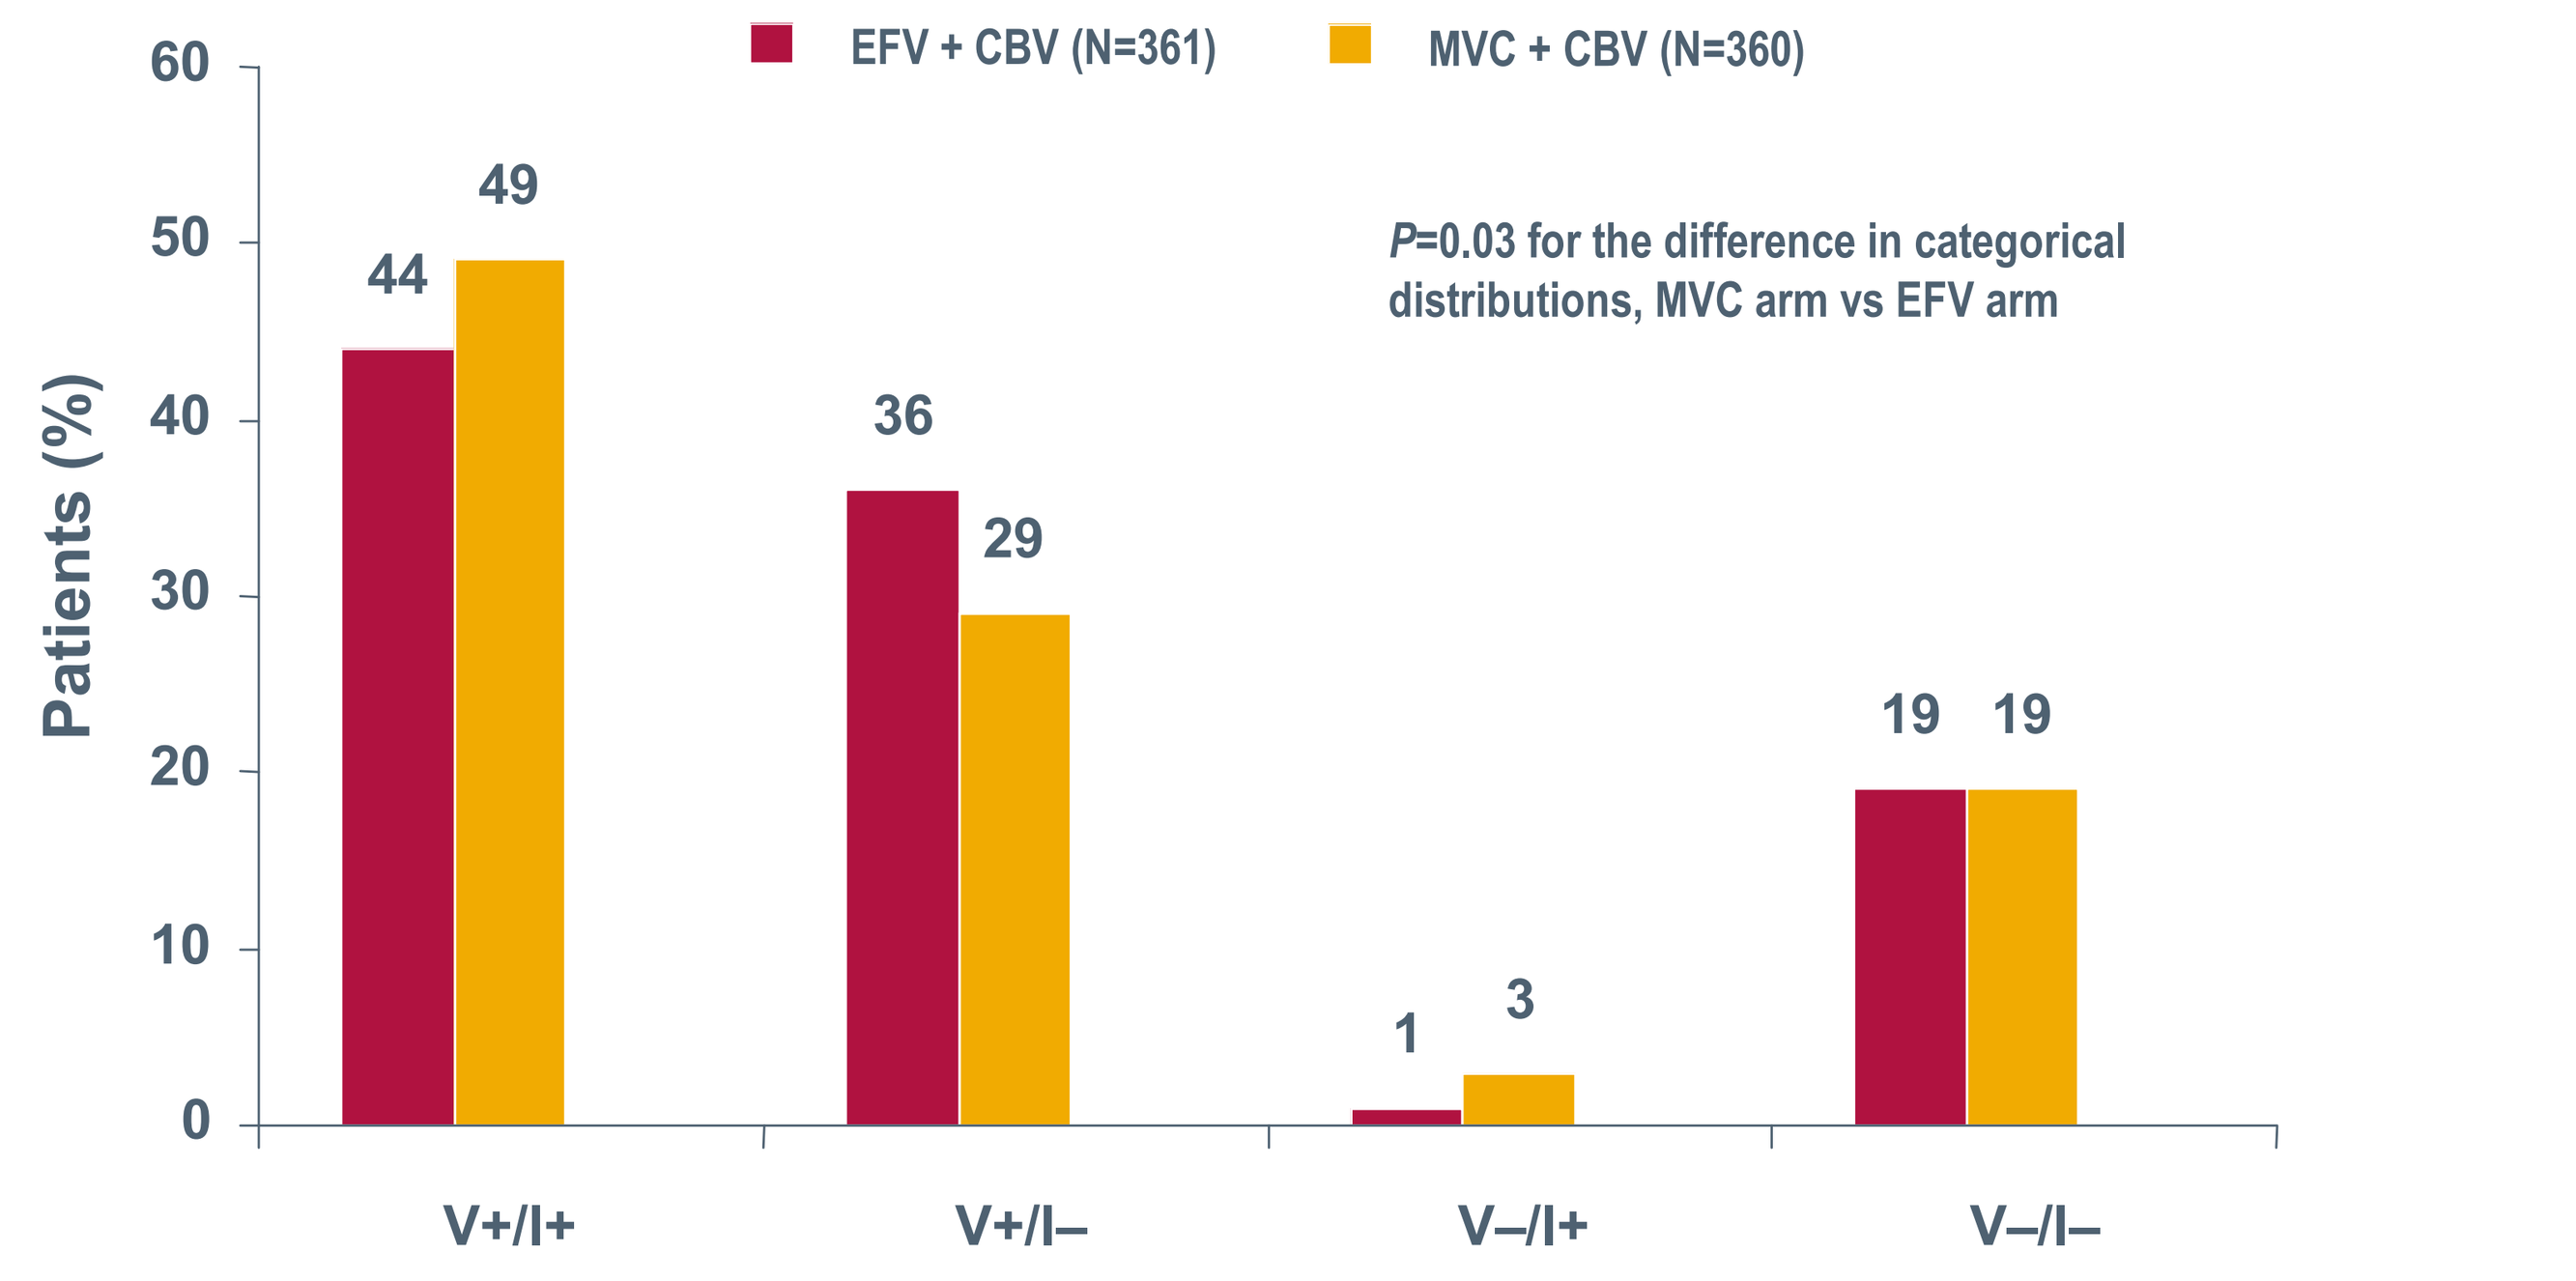
EFV + CBV (N=361)
MVC + CBV (N=360)
Patients (%)
P=0.03 for the difference in categorical distributions, MVC arm vs EFV arm
44 49
36 29
1 3
19 19
V+/I+
V+/I–
V–/I+
V–/I–

I, immunologic response, defined as increase in CD4+ count from baseline of >100 cells/mm³; V, virologic response, defined as HIV-1 RNA <400 copies/mL; +, positive response; –, negative response

- An early increase in median CD8+ cell count was observed in the MVC arm, which subsequently declined back towards baseline at Week 16 through to Week 48 (Figure 4). In contrast, a gradual decline in CD8+ cell counts was observed from baseline in the EFV arm, with a median change at Week 48 of –110 cells/mm³.
- At Week 48, there was an increase from baseline in CD4% (CD4+ cell count as a percentage of the total lymphocyte count) of similar size in both treatment groups (mean of +8.1% and +9.5% in the MVC and EFV treatment groups, respectively).

Figure 4. Median change from baseline in CD8+ cell count by visit

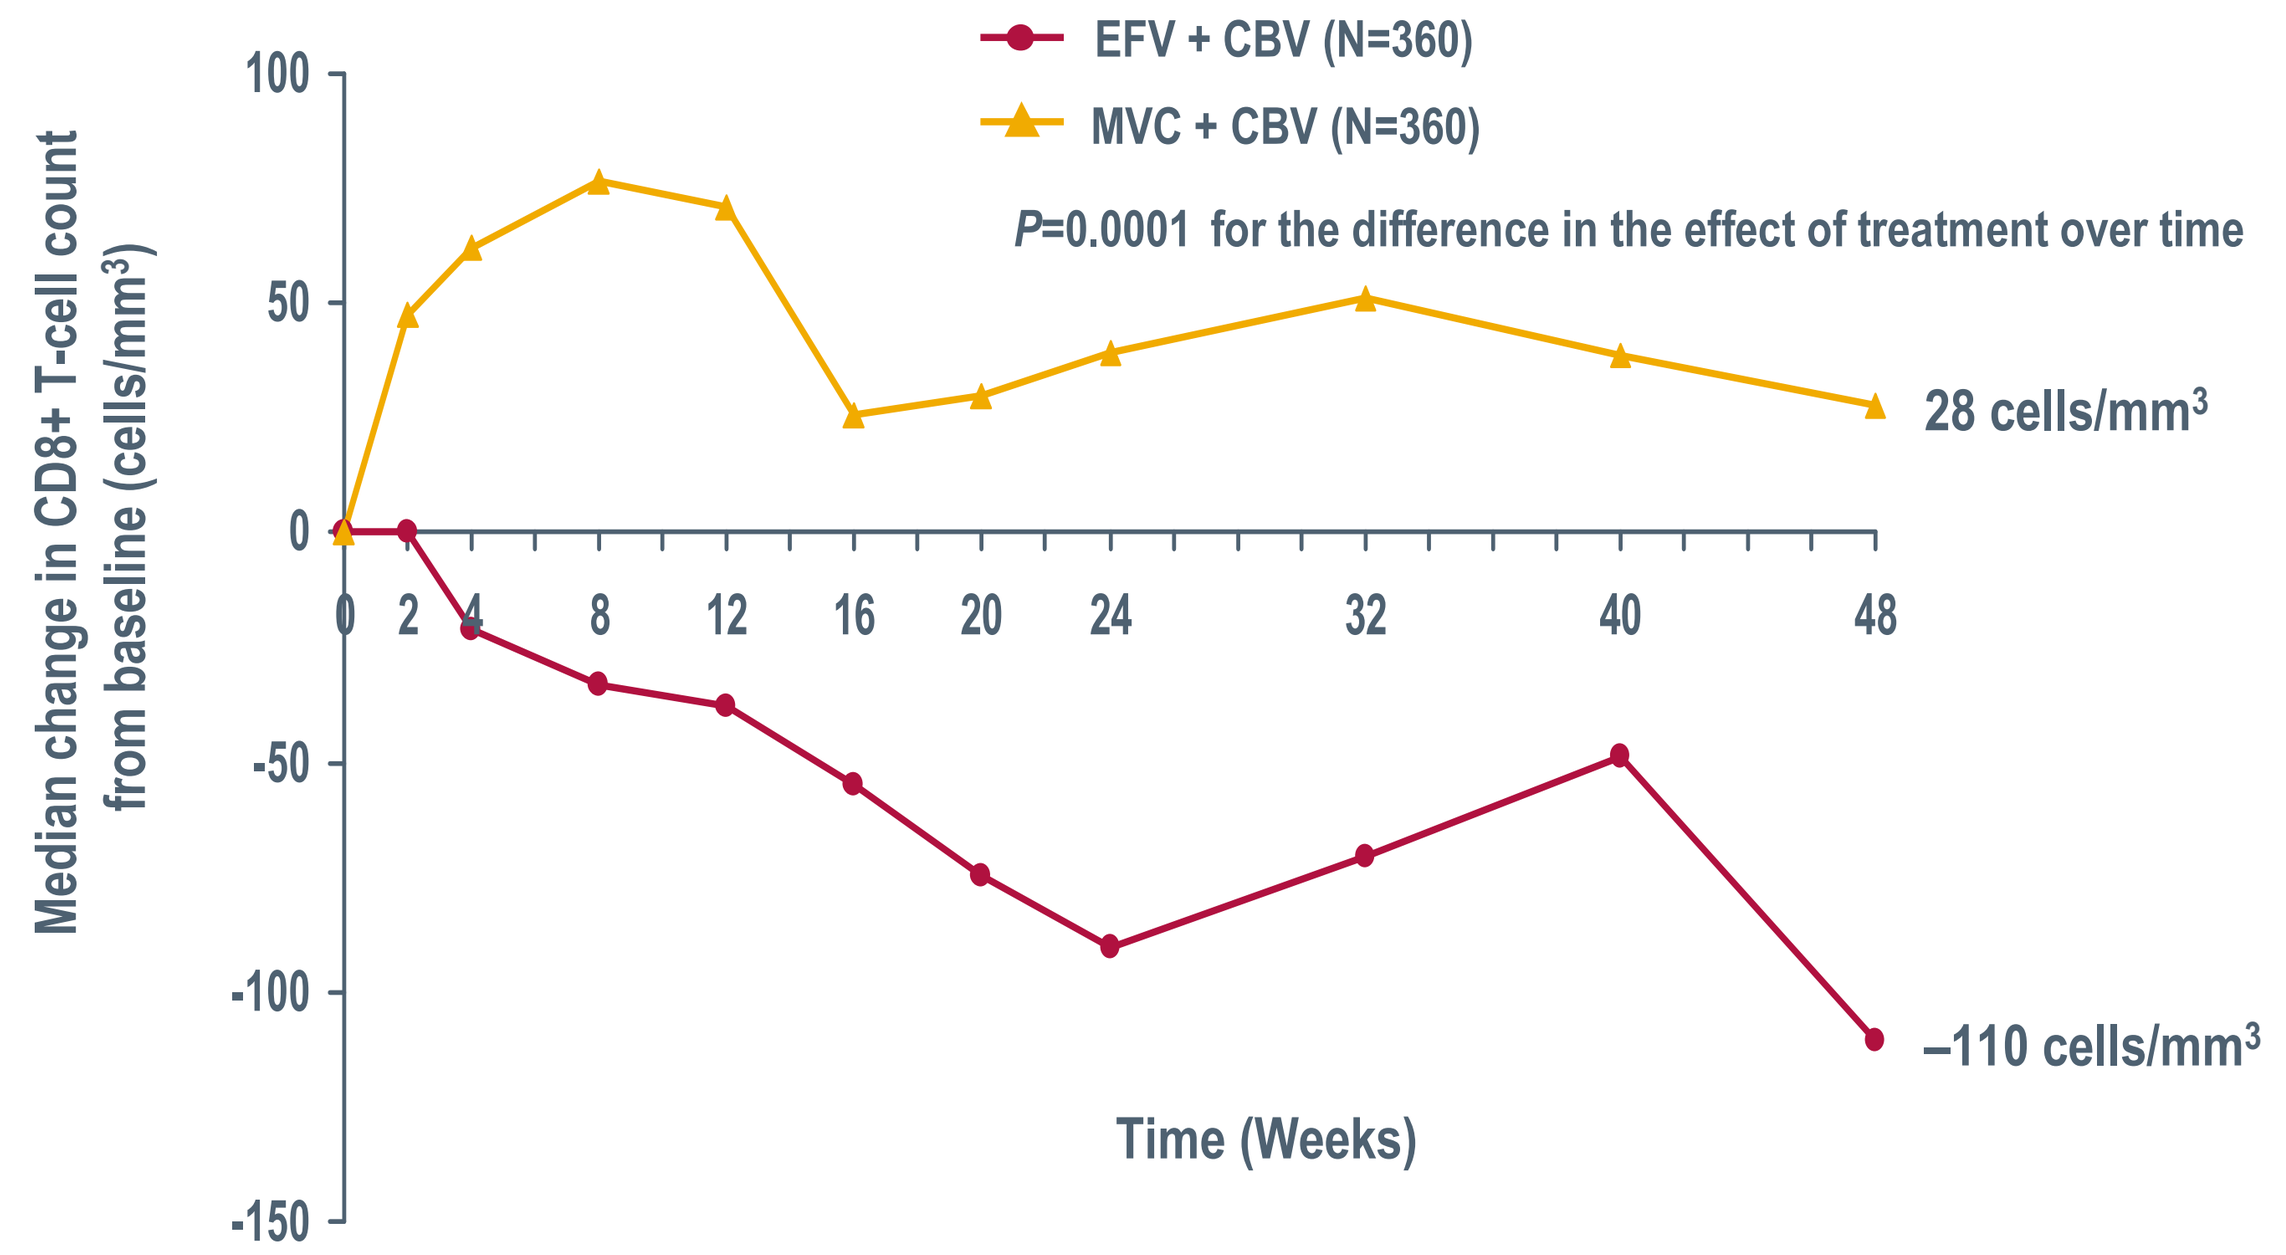
EFV + CBV (N=360)
MVC + CBV (N=360)
P=0.0001 for the difference in the effect of treatment over time
Median change in CD8+ T-cell count from baseline (cells/mm³)
28 cells/mm³
–110 cells/mm³
0 2 4 8 12 16 20 24 32 40 48
Time (Weeks)

Includes all patients who received at least one dose of study medication (LOCF)

Predictors of CD4+ cell count increases

- A multivariate regression model of predictors of Week 48 CD4+ cell count increases from baseline identified significant associations with:
 - Baseline CD4+ cell count: Higher baseline CD4+ cell counts were associated with greater increases in CD4+ cell count (P=0.0005).
 - Week-48 change in HIV-1 RNA level: Larger decreases from baseline in HIV-1 RNA at Week 48 were associated with greater increases in CD4+ cell count (P<0.0001).
 - Treatment arm: Patients randomized to MVC + CBV experienced greater increases in CD4+ cell count than those randomized to EFV + CBV (P<0.0001).
- Variables found not to be associated with Week 48 CD4+ cell count increase were age, gender, and baseline HIV-1 RNA level.

Predictors of Category C events

- As previously reported, twice as many patients in the EFV arm vs the MVC arm experienced a Category C event on treatment (12 [3.3%] vs 6 [1.7%], respectively).[7]
- Figure 5 shows a Kaplan–Meier plot of time to Category C event occurrence.

Figure 5. Occurrence of Category C events over time by treatment arm

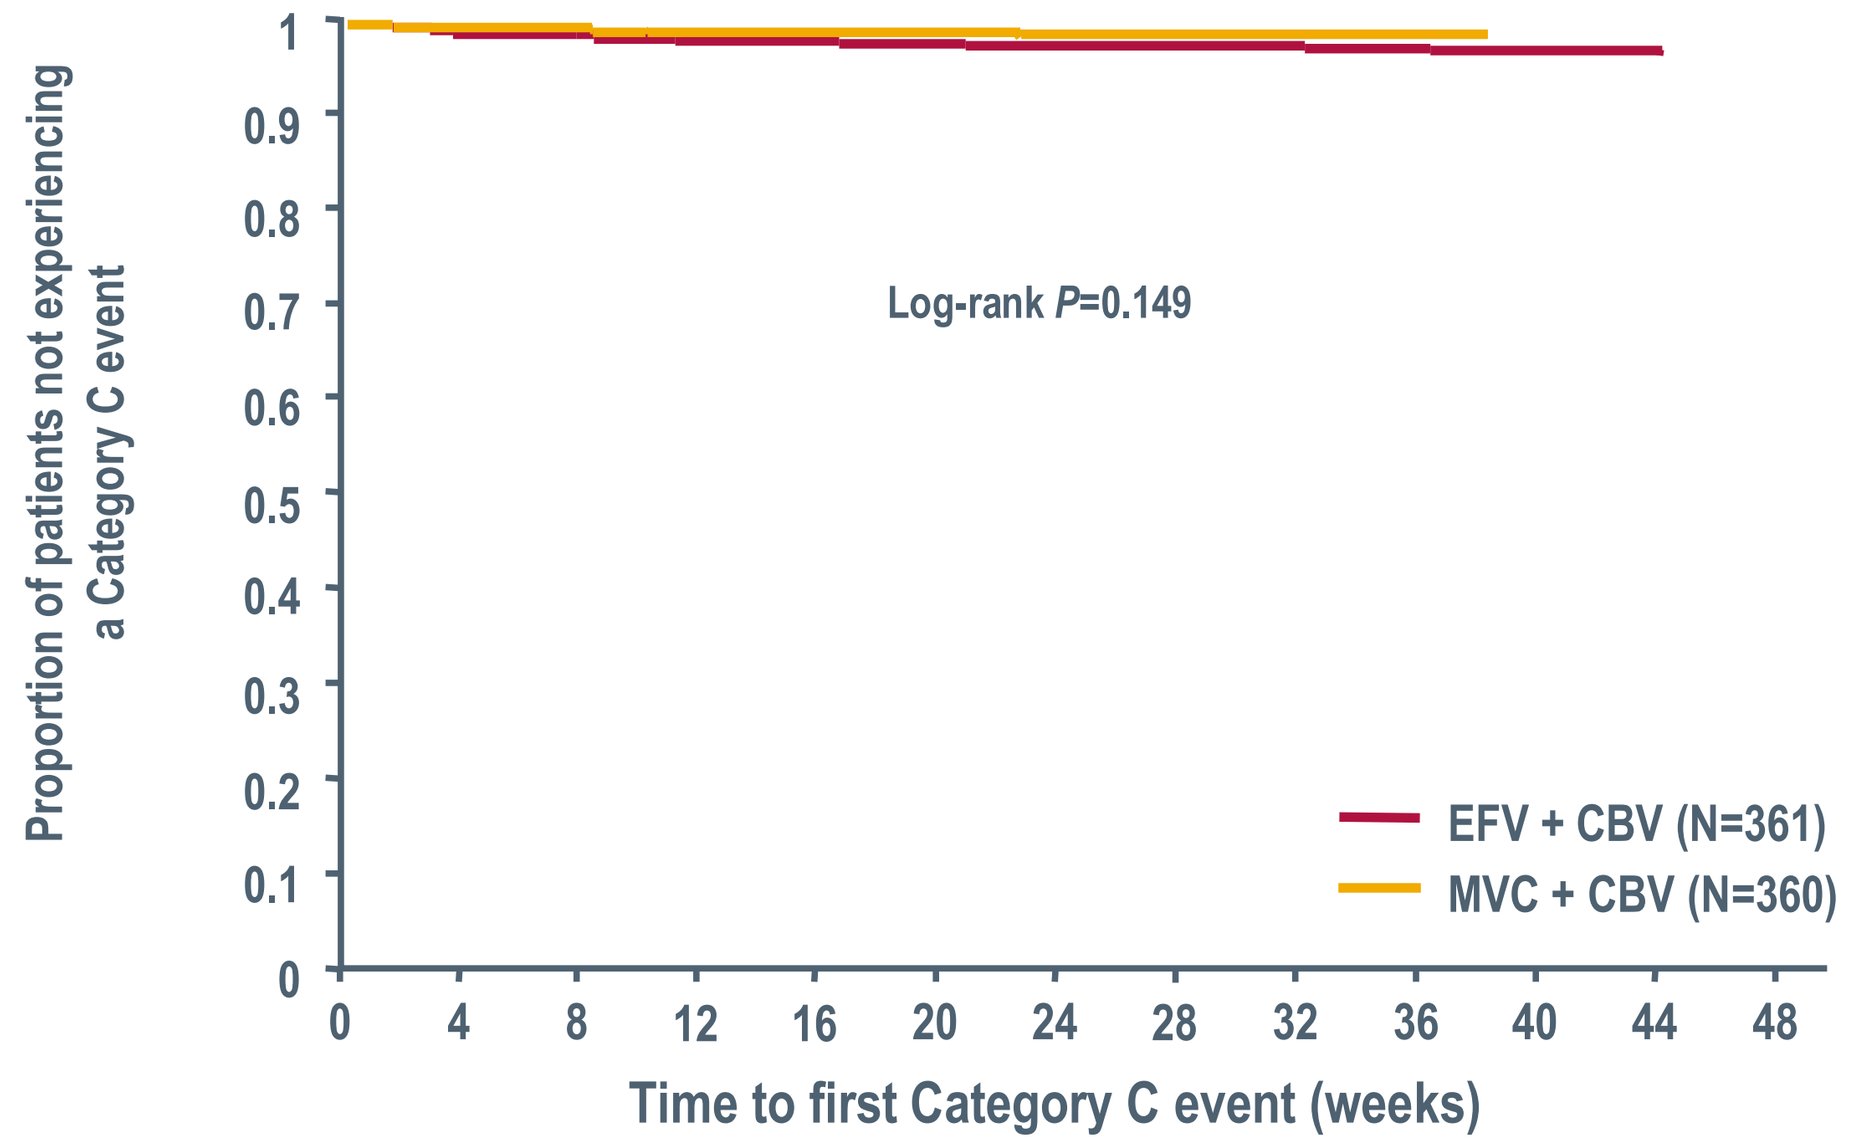
Proportion of patients not experiencing a Category C event
Log-rank P=0.149
EFV + CBV (N=361)
MVC + CBV (N=360)
0 4 8 12 16 20 24 28 32 36 40 44 48
Time to first Category C event (weeks)

- Three significant factors associated with time to Category C event occurrence (Cox proportional hazard modeling) were identified:
 - On-treatment CD4+ cell count (per 50 cells/mm³ increase from baseline): HR=0.8 (P=0.040).
 - Baseline HIV-1 RNA level (per log higher): HR=2.4 (P=0.030).
 - Achieving an HIV-1 RNA <50 copies/mL by Week 48: HR= 0.08 (P<0.0001).
- Longer time to Category C event was associated with lower baseline HIV-1 RNA level, higher CD4+ cell counts on treatment, and achieving <50 HIV-1 RNA copies/mL by Week 48.
- Baseline CD4+ cell count, age, and gender were not significant predictors of time to Category C event in the multivariate model.
- In a univariate analysis, treatment arm was not significantly associated with the time to Category C event (P=0.149). However, among patients who did not achieve HIV-1 RNA <50 copies/mL at Week 48, fewer MVC recipients (1/104 [1%]) developed a Category C event compared to EFV recipients (7/75 [9%]; P=0.007). The median (IQR) CD4+ cell count changes from baseline in these non-responder subgroups were 64 (4.5, 142.5) and 0 (–7.75, 76.5) cells/mm³, respectively.
- When treatment arm was included in the multivariate model, MVC treatment was associated with longer time to development of a Category C event, in addition to baseline HIV-1 RNA level, on-treatment CD4+ cell count, and achieving HIV-1 RNA <50 copies/mL at Week 48.

Conclusions

- In the MERIT study, treatment-naive patients receiving MVC + CBV experienced early, rapid, and persistently greater CD4+ cell count increases vs those receiving EFV + CBV through Week 48 (P=0.004).
- The percentage of patients with a discordant immunologic response to study treatment was lower with MVC + CBV than with EFV + CBV.
- An early, rapid but transient CD8+ cell count rise was observed with MVC + CBV, while with EFV + CBV CD8+ cell count declined between baseline and Week 48.
- MVC + CBV was associated with a larger increase in CD4+ cell count than EFV + CBV, even after adjusting for change in HIV-1 RNA.
- Higher on-treatment CD4+ cell counts were associated with a longer time to Category C event occurrence.
 - This supports the premise that the additional CD4+ cells conferred by MVC + CBV were functional, and suggests that antagonism of the CCR5 co-receptor by MVC was not associated with any deleterious effect as measured by Category C events at 48 weeks.
 - Among patients who did not achieve an undetectable viral load, MVC + CBV recipients had a significantly lower risk of developing a Category C event, and experienced significantly greater CD4+ cell count increases compared to EFV + CBV recipients.

References

1. Monforte A, *et al. AIDS* 2008; **22**:2143–2153.
2. Lewden C, *et al. J Acquir Immune Defic Syndr* 2007; **46**:72–77.
3. Grabar S, *et al. Ann Intern Med* 2000; **133**:401–410.
4. Tan R, *et al. J Acquir Immune Defic Syndr* 2008; **47**:553–558.
5. Baker JV, *et al. AIDS* 2008; **22**:841–848.
6. Baker JV, *et al. J Acquir Immune Defic Syndr* 2008; **48**:541–546.
7. Saag M, *et al. 4th IAS Conference on HIV Pathogenesis, Treatment and Prevention*. Sydney, Australia, 22-25 July 2007; Abstract WESS104.

Acknowledgements

- Patients participating in the MERIT study.
- Investigators and study site staff.
- Pfizer MERIT study team.