48th Annual ICAAC/IDSA 46th Meeting, Washington, DC, USA, October 25–28, 2008

Poster H-1221

Weighted OBT Susceptibility Score (wOBTSS) is a Stronger Predictor of Virologic Response at 48 Weeks than Baseline Tropism Result in MOTIVATE 1 and 2

H Valdez[1], M Lewis[2], C Delogne[2], P Simpson[2], D Chapman[1], L McFadyen[2], E Coakley[3], E van der Ryst[2] and M Westby[2]

[1]*Pfizer Inc, New York, USA;* [2]*Pfizer Global Research and Development, Sandwich, UK;* [3]*Monogram Biosciences, South San Francisco, USA*

For further information, please contact Hernan Valdez at Hernan.Valdez@pfizer.com

Introduction

- Pooled Week 48 data from the Phase 3 MOTIVATE 1 and 2 studies showed significantly greater virologic and immunologic responses in treatment-experienced (TE) patients with R5 virus who received maraviroc (MVC) either twice-daily (BID) or once-daily (QD) with an optimized background therapy (OBT) than in those who received OBT with placebo (PBO).[1]
- Patients receiving MVC BID had a mean reduction in viral load from baseline of –1.84 $\log_{10}$ copies/mL (46% <50 copies/mL) and a mean increase in CD4+ cell count of 124 cells/mm^3. By comparison, subjects on PBO had a –0.78 $\log_{10}$ copies/mL reduction (17% <50 copies/mL) and 61 cells/mm^3 increase.[1]
- It is known that the number of active drugs in the regimen of TE patients is strongly linked to the degree and duration of virologic response.[2,3] However, the method of scoring activity will have a significant effect on the degree of correlation.
- OBT scoring in TE studies is typically undertaken by counting all on-study OBT drugs to which a subjects' baseline virus is considered sensitive by resistance testing. This approach has several conceptual limitations. In particular:
 - Counting active drugs assumes equal potency for each agent, but evidence suggests that active NRTIs contribute only about 50% of the activity of other drug classes in TE patients.[4]
 - Drugs continued from a failing pre-study (baseline) regimen, even where considered active, cannot contribute to observed *post-baseline* response, and their inclusion overestimates the activity of the OBT as a correlate of virologic outcome.
 - Changes to the OBT during active on-study treatment are not usually controlled for in activity-response analyses.
- In order to better identify patients most likely to respond to MVC the following *post-hoc* analysis was undertaken to examine the predictive value of both the standard drug-counting approach to OBT scoring, and an alternative simple weighted methodology (previously used to explore antiretroviral phenotypic clinical cutoffs[5]) on Week 48 virologic response to MVC-containing therapy in MOTIVATE 1 and 2, along with other baseline covariates.

Methods

Study populations

- The MOTIVATE studies both enrolled HIV-1-infected adults with R5-only virus, and ≥6 months experience of, or resistance to, three antiretroviral classes. All subjects had a screening viral load >5,000 HIV-1 RNA copies/mL and received an OBT of 3–6 approved agents.
 - Raltegravir, darunavir, and etravirine were not permitted. Tipranavir was allowed following availability of drug–drug interaction data in early 2006.
 - Patients were randomized 2:2:1 to receive MVC QD, MVC BID, or PBO in addition to OBT.
- The full analysis set of MOTIVATE 1 and 2 comprised all randomized subjects who received at least one dose of study medication (N=1,049).
- In order to study only patients who started all regimen components simultaneously, for this analysis of OBT activity and virologic outcome an OBT window was defined as 6 days before to 7 days after the date of administration of the first randomized therapy. The final OBT composition must have been fixed within this window without further changes during the first 48 weeks in order to be included in the final analysis.
- A sub-population of the full analysis set, the Virologic Outcomes (VO) population, was defined to exclude both non-virologic failures and those whose baseline OBT score did not accurately reflect the drugs received throughout their period of randomized treatment.
- Figure 1a shows the allowed patterns of OBT drug use for subjects included in the VO population. Figure 1b gives the derivation of the VO population from the full analysis set.

Figure 1. a) Allowed patterns of antiretroviral drug use in the Virologic Outcomes populations, and b) Patients included in the analyses

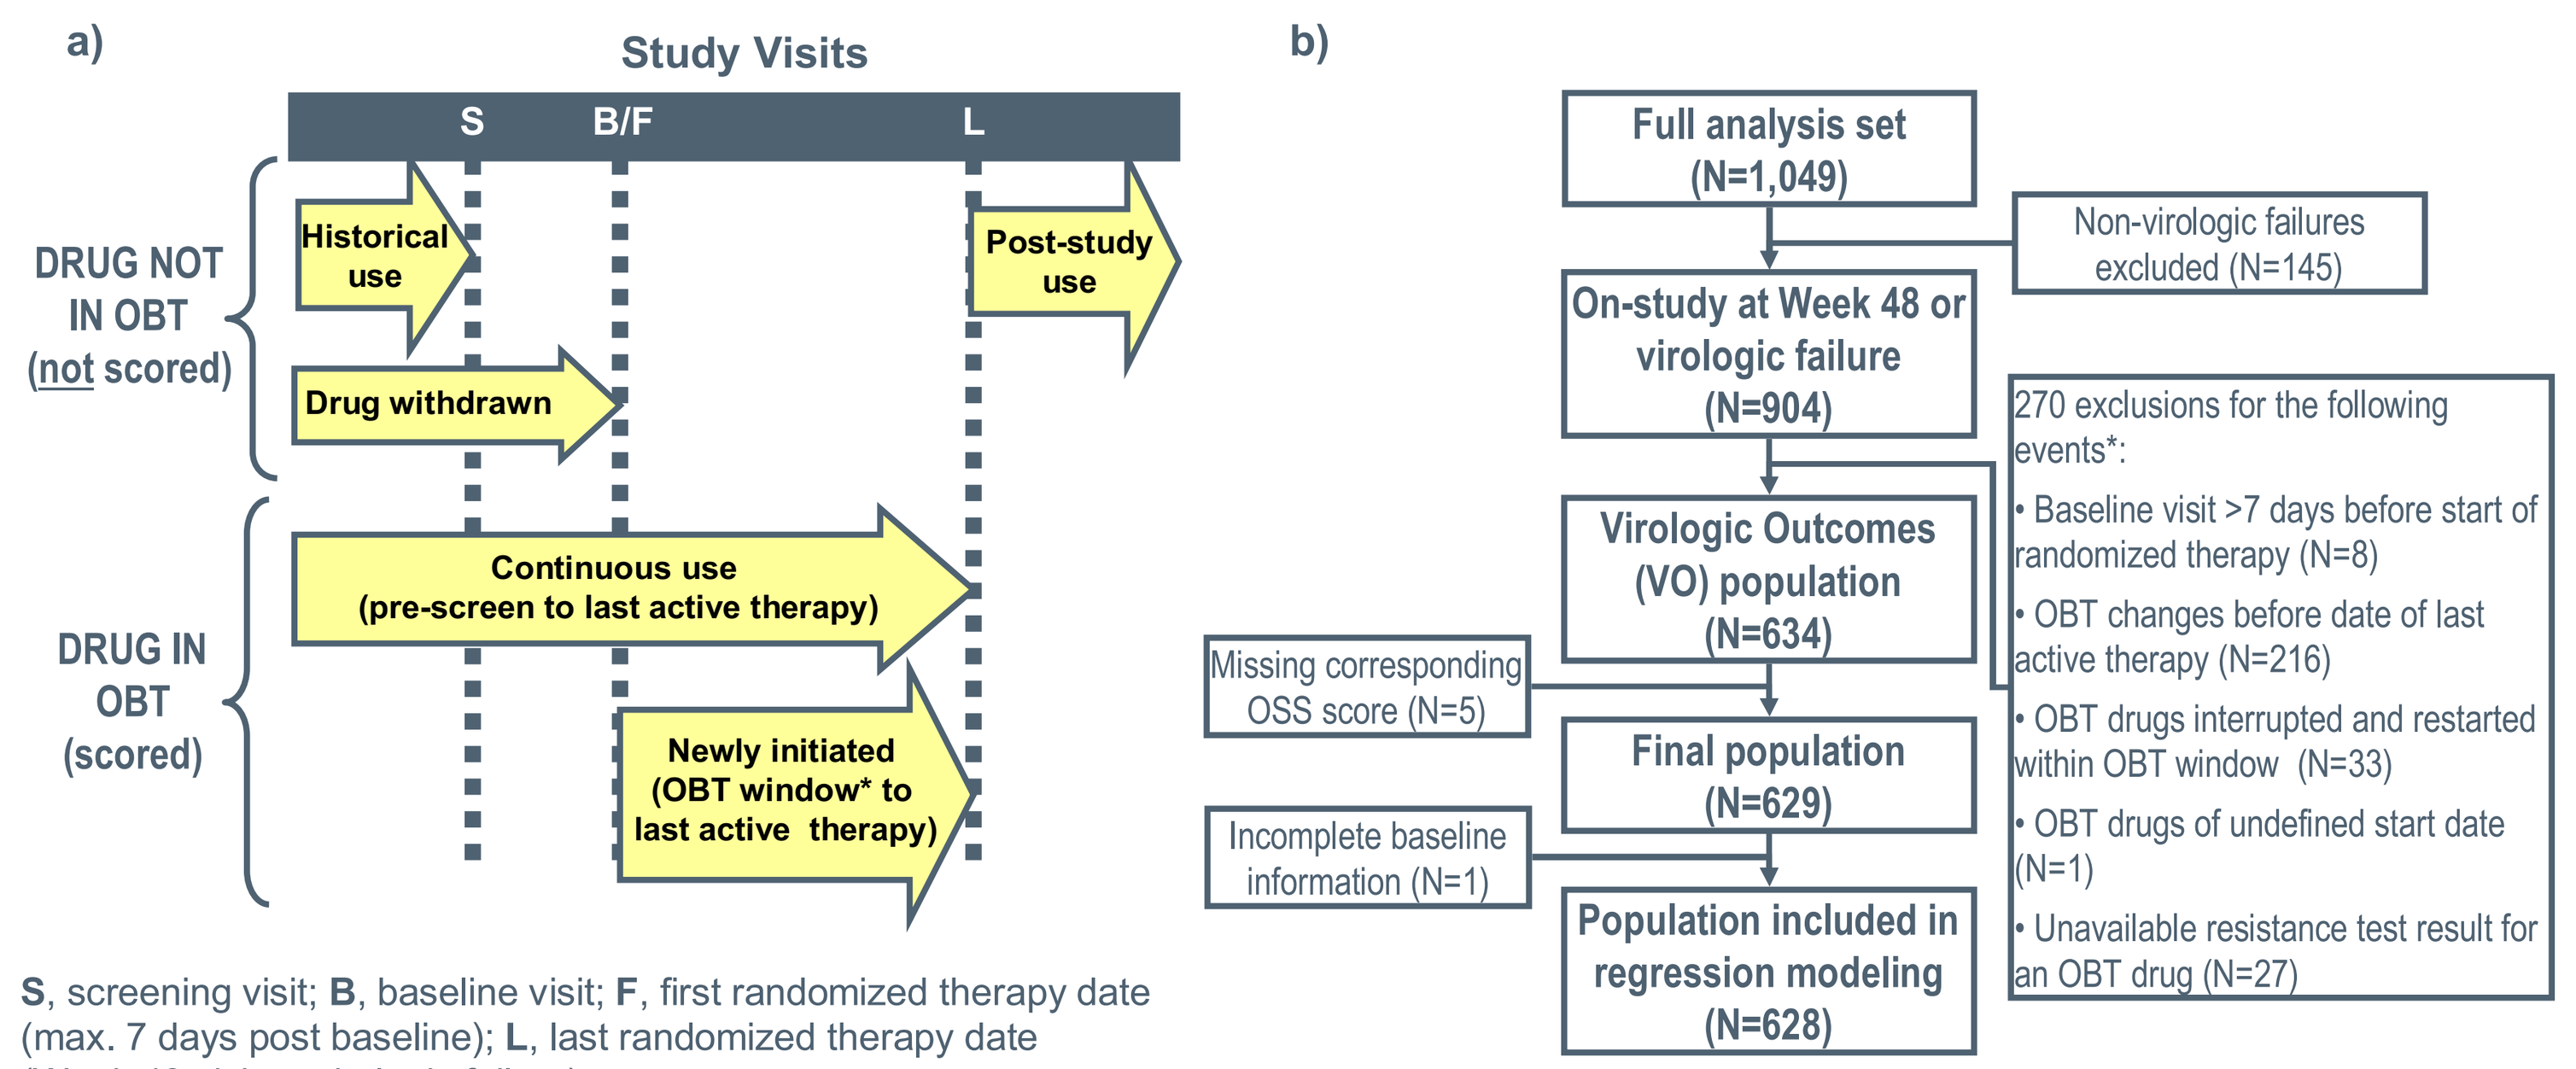

a)
Study Visits
S
B/F
L
DRUG NOT IN OBT (not scored)
Historical use
Post-study use
Drug withdrawn
DRUG IN OBT (scored)
Continuous use (pre-screen to last active therapy)
Newly initiated (OBT window* to last active therapy)
b)
Full analysis set (N=1,049)
Non-virologic failures excluded (N=145)
On-study at Week 48 or virologic failure (N=904)
270 exclusions for the following events*:
• Baseline visit >7 days before start of randomized therapy (N=8)
• OBT changes before date of last active therapy (N=216)
• OBT drugs interrupted and restarted within OBT window (N=33)
• OBT drugs of undefined start date (N=1)
• Unavailable resistance test result for an OBT drug (N=27)
Virologic Outcomes (VO) population (N=634)
Missing corresponding OSS score (N=5)
Final population (N=629)
Incomplete baseline information (N=1)
Population included in regression modeling (N=628)

S, screening visit; **B**, baseline visit; **F**, first randomized therapy date (max. 7 days post baseline); **L**, last randomized therapy date (Week 48 visit or virologic failure)
* OBT window = –6 days to +7 days from **F**

* More than one event could have occurred in a given subject

OBT scoring algorithms

- Both OBT scores were based on a conservative binary definition of sensitivity which assumed virus to be either fully sensitive or fully resistant to a given ARV.
- Both scores excluded booster doses of ritonavir.
- Weighted OBT sensitivity score (wOBTSS; Table 1)
 - Drugs in continuous use from the screening to the treatment phase of the trial were scored as 0.
 - Viral sensitivity or resistance was defined by phenotype only (PhenoSense™ HIV assay, Monogram Biosciences) or by genotype for enfuvirtide (BC Centre for Excellence in HIV/AIDS, Vancouver, Canada).
 - Active NRTIs score 0.5 each, other active agents score 1.0.
- Overall sensitivity score (OSS; Table 2)
 - All drugs in OBT were scored as shown, including those in continuous use as it is usually done.
 - Sensitivity or resistance for enfuvirtide was assessed genotypically as for the wOBTSS.
 - Sensitivity or resistance to other drugs was defined by a net assessment combining genotypic and phenotypic data according to a proprietary commercial methodology ('PhenoSense GT': Monogram Biosciences).
 - All active drugs scored 1.0.

Table 1. Weighted OBT sensitivity score

Any drug in continuous use, score = 0	$IC_{50}FC$	
	S *	R †
PI	1	0
NRTI	0.5	0
NNRTI	1	0
	Genotype	
	S	R
ENF	1	0

Table 2. Overall sensitivity score

	Net assessment	
	Yes	No
NRTI, NNRTI, PI	1	0
	Genotype	
	S	R
ENF	1	0

S, sensitive; **R**, resistant; * IC_{50} fold-change is less than or equal to the lower clinical cut-off; † IC_{50} fold-change is greater than the lower clinical cut-off

- Note that conceptually the OSS counts the number of 'active' drugs in the OBT whereas the wOBTSS is an estimate of relative post-baseline OBT activity that will not necessarily correspond to the number of active drugs in use.

Statistical methods

- Virologic success was defined as Week 48 HIV-1 viremia <50 RNA copies/mL.
- Logistic regression analysis (Logit function) was used to assess predictors of virologic success for two different methods of scoring OBT activity along with fixed baseline covariates common to both models.
 - CD4+ cell count (arithmetic mean of screening and baseline visits).
 - Viral load (geometric mean of up to three visits at screening, randomization, and baseline).
 - Tropism result (at baseline visit).
 - Treatment (MVC QD or BID, combined, vs placebo).
- Goodness-of-fit was assessed primarily by the Hosmer and Lemeshow (H&L) test,[6] supported by other estimates of fit and predictive power including the Pearson test, Deviance, co-efficient of determination (R^2), max-rescaled R^2 (adj-R^2), Akaikes' Information Criterion (AIC), Somers' D, area under the curve (c), and the Schwarz criterion (SC).

Results

- OBT activity score distributions in the VO population are shown in Figure 2a for those who were or were not on a drug holiday prior to screening for MOTIVATE 1 or 2 (defined as off all ARVs for ≥7 days at screening visit). Figure 2b shows the proportion of subjects in each arm with a given OSS or wOBTSS estimate.
- Median scores by OSS were consistently higher by ~1 unit than with the wOBTSS for the corresponding population.
- For both OSS and wOBTSS, subjects not on therapy at screening had a median score ~1 unit higher than those who were on therapy, consistent with reversion to a wild-type virus population following withdrawal of drug selection pressure.
- Comparing wOBTSS with OSS, there was an 82% reduction in subjects estimated to have received a fully active regimen in the absence of MVC (OSS or wOBTSS ≥3) and a 128% increase in those estimated to have received a weak or ineffective OBT (OSS or wOBTSS <1).
- All subsequent analyses shown here are on the full VO population of subjects both on and off treatment at screening.

Figure 2. OSS method overestimates OBT drug activity. a) Higher median active drug scores for OSS vs wOBTSS, and b) Infrequent estimates of weak or inactive OBT (score <1) and high estimates of fully-active triple therapy (score ≥3) from OBT alone for OSS vs wOBTSS

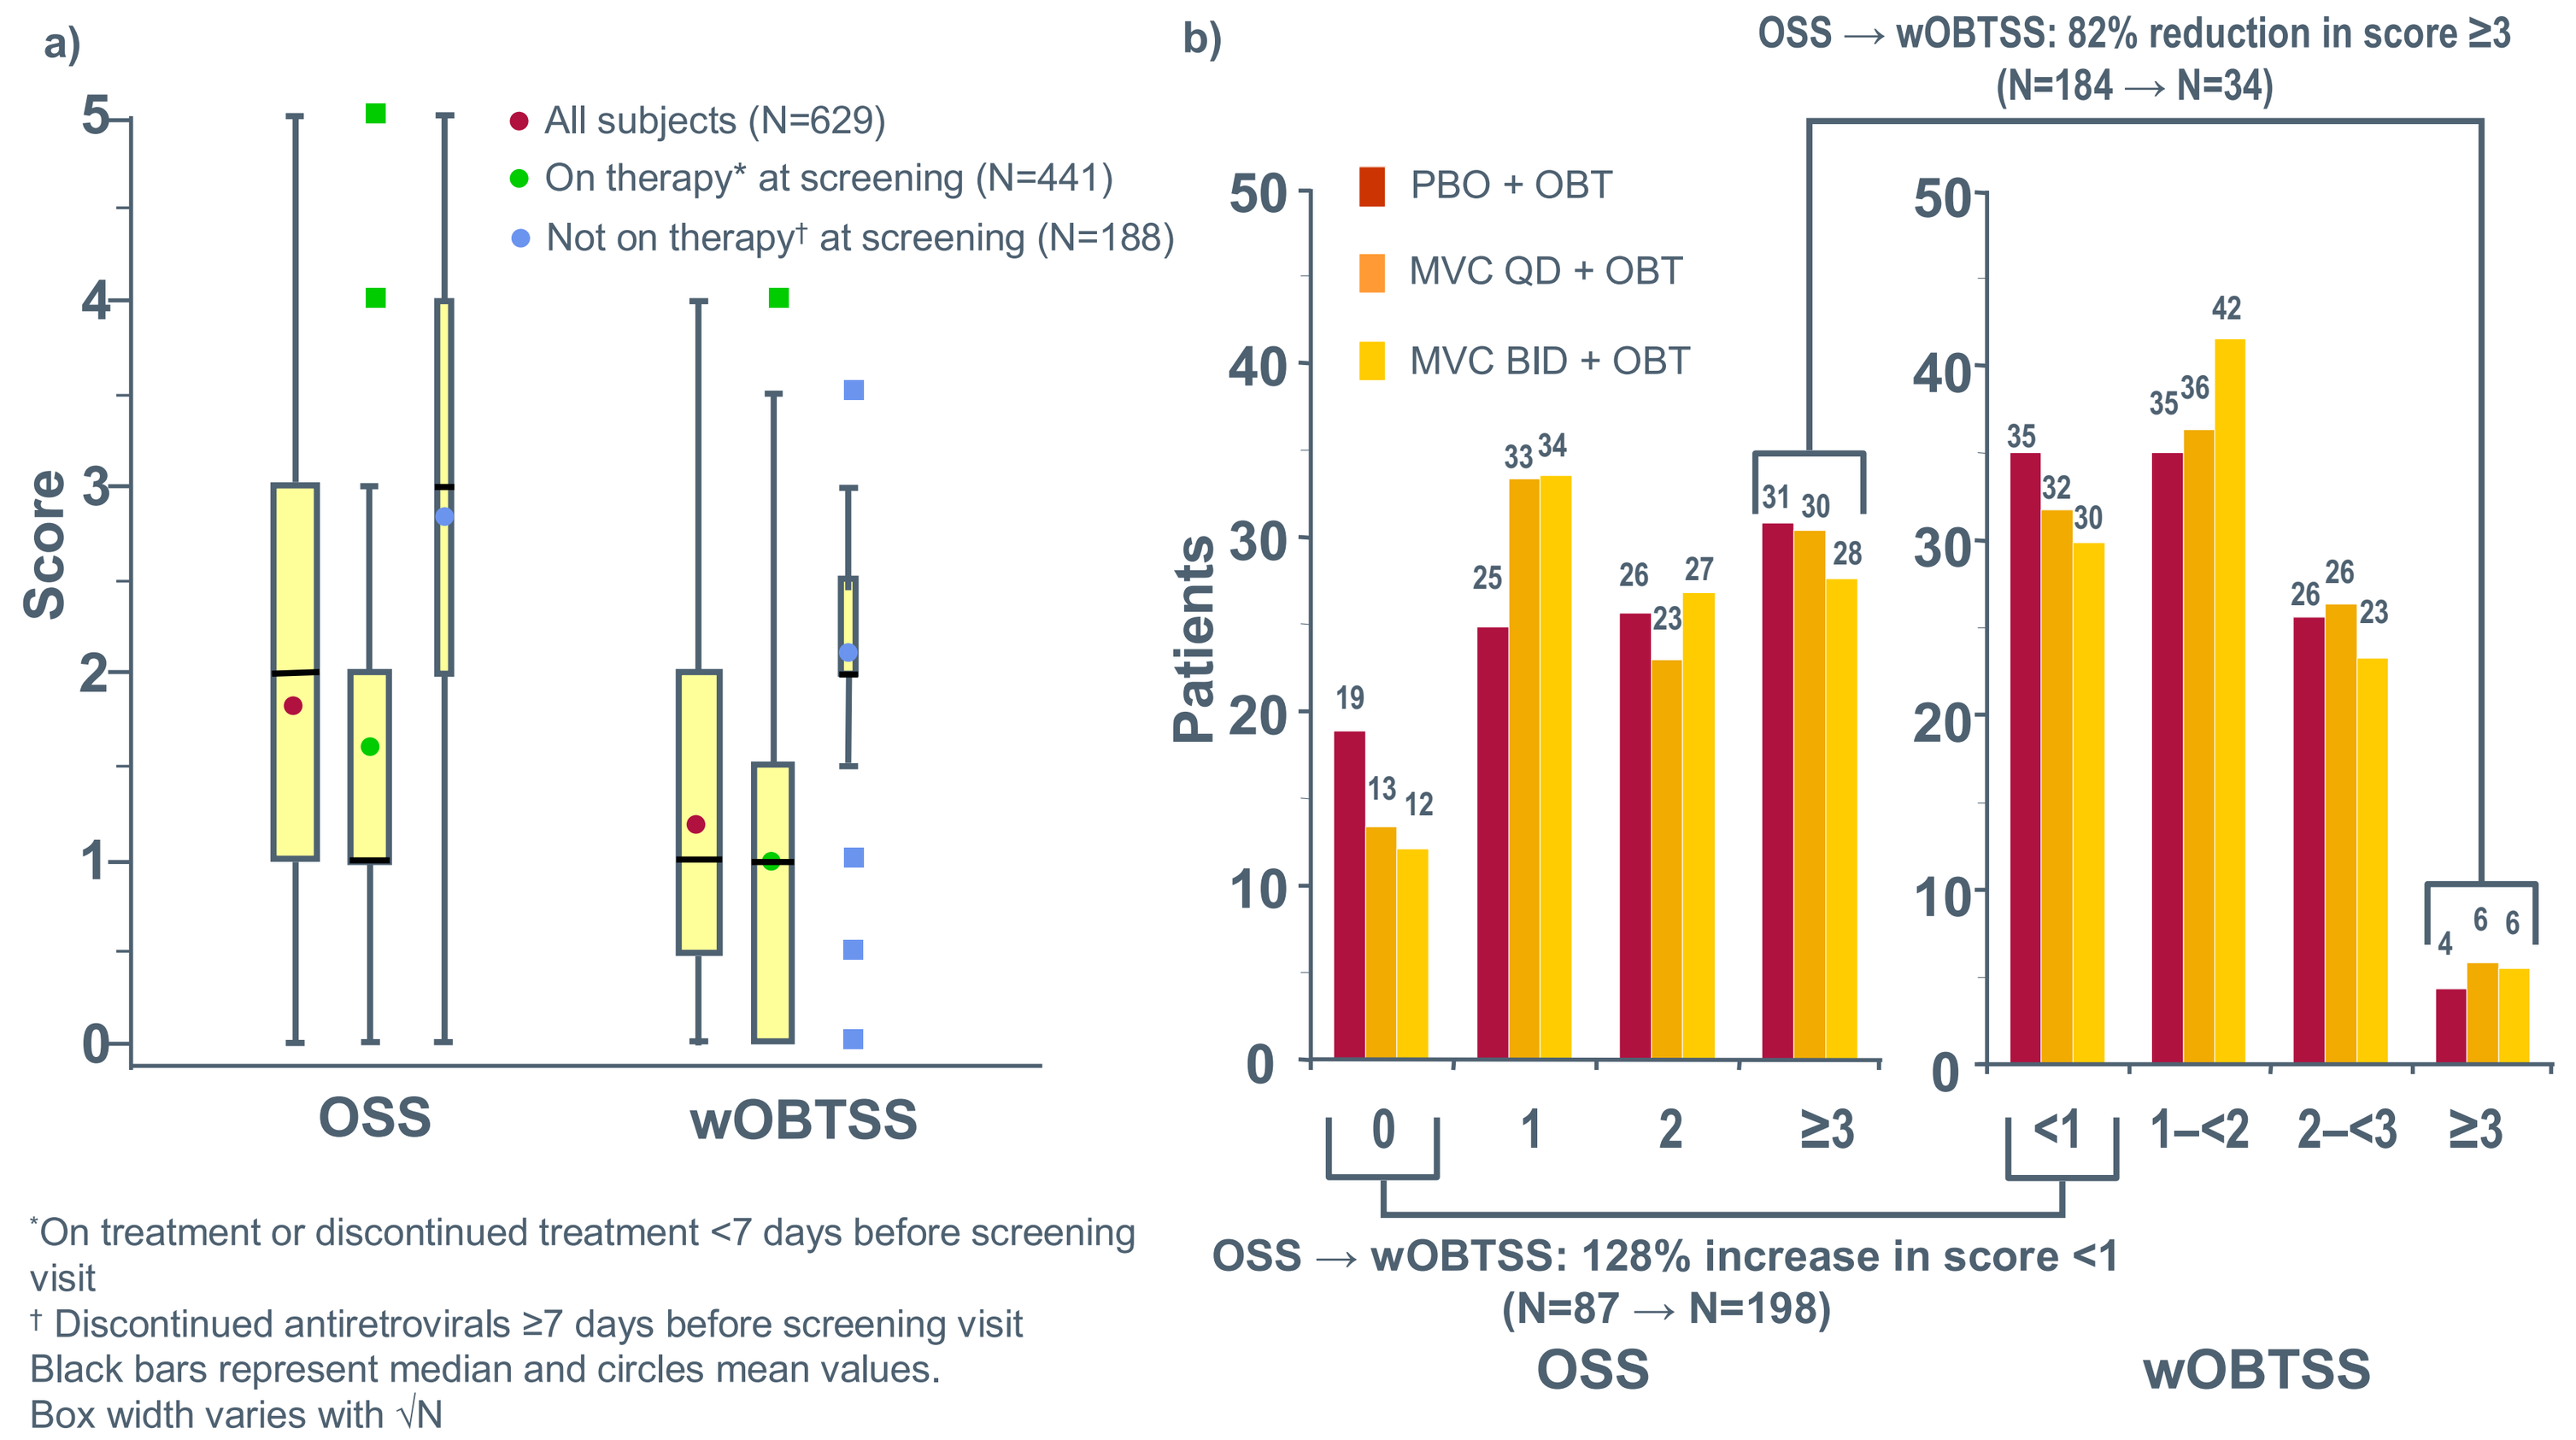

a)
Score
All subjects (N=629)
On therapy* at screening (N=441)
Not on therapy† at screening (N=188)
OSS
wOBTSS
b)
Patients
PBO + OBT
MVC QD + OBT
MVC BID + OBT
OSS → wOBTSS: 82% reduction in score ≥3
(N=184 → N=34)
OSS → wOBTSS: 128% increase in score <1
(N=87 → N=198)
OSS
wOBTSS

*On treatment or discontinued treatment <7 days before screening visit
† Discontinued antiretrovirals ≥7 days before screening visit
Black bars represent median and circles mean values.
Box width varies with √N

- Median scores by OSS were consistently higher by ~1 unit than with the wOBTSS for the corresponding population.
- Compared to the OSS method, wOBTSS scoring was associated with:
 - Incremental increases in response in the placebo arm across each wOBTSS stratum from 0–≥3 (Figure 3).
 - Higher response rates for subjects receiving MVC at each wOBTSS stratum than for the placebo arm at the next highest stratum – ie, greater response for a given total number of active drugs when MVC was included in the regimen (Figure 3).
- Since very few subjects had a wOBTSS of ≥3, subsequent analyses for both OSS and wOBTSS used a highest score stratum of ≥2.

Figure 3. Response (<50 HIV-1 RNA copies/mL at Week 48) to OBT + PBO increases incrementally for increasing wOBTSS but does not for increasing OSS (VO population)

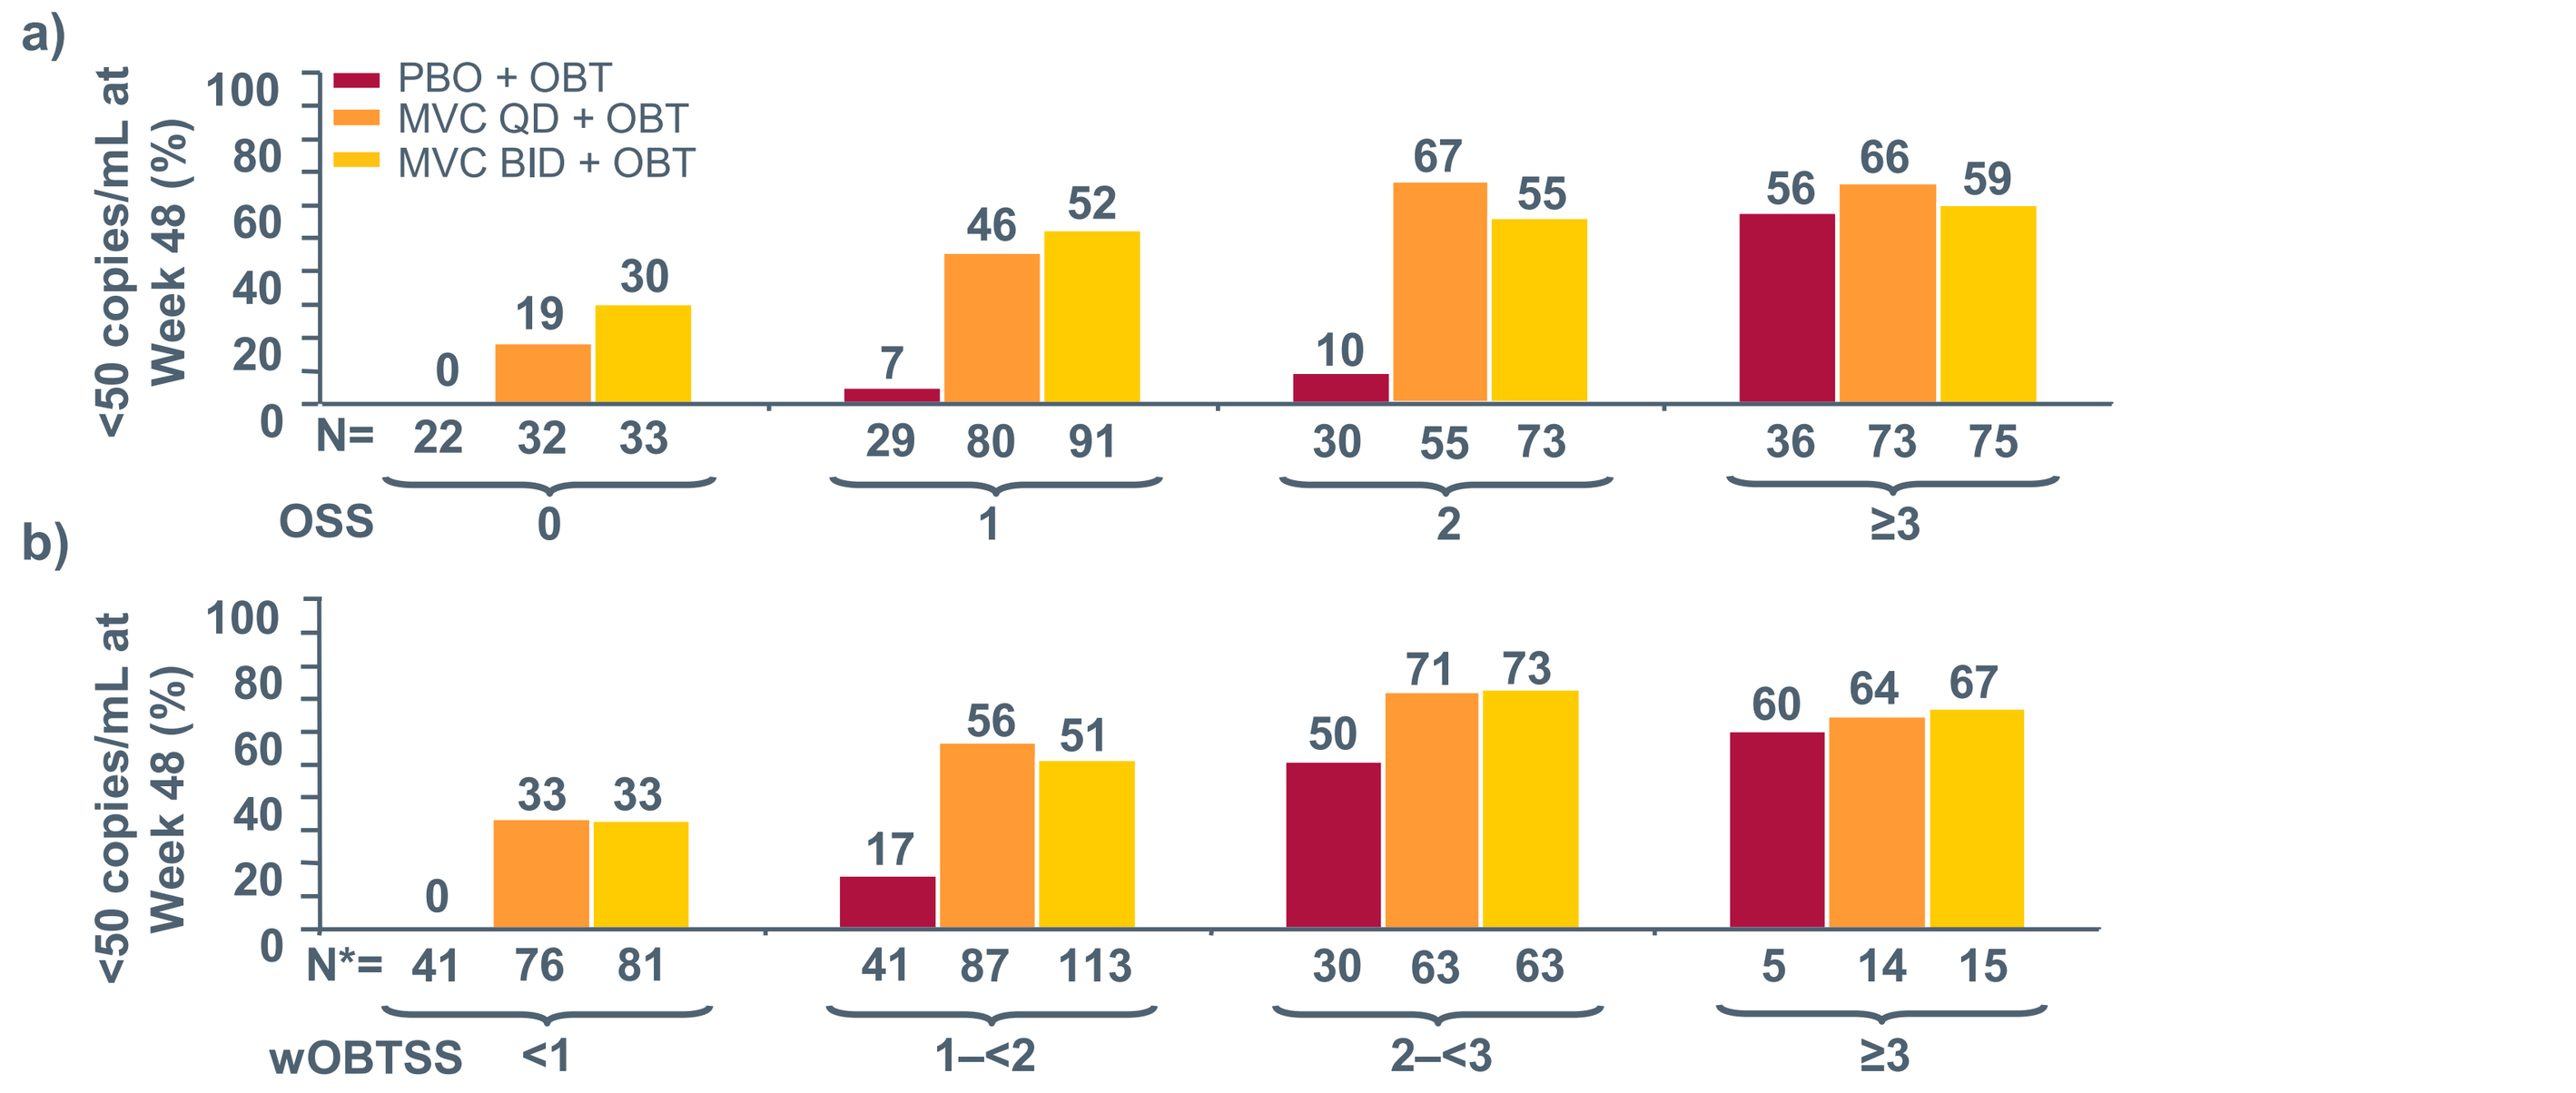

a)
<50 copies/mL at Week 48 (%)
PBO + OBT
MVC QD + OBT
MVC BID + OBT
OSS
b)
wOBTSS

*For each subgroup this is the number of subjects with a post-baseline observation used to calculate the percentage

Figure 4. Goodness-of-fit and other statistical correlation measures indicate that a model containing wOBTSS is better in predicting virologic success than one containing OSS:
Odds ratios (95% confidence intervals) for predictors of virologic success in MOTIVATE 1 and 2 for models using a) OSS, and b) wOBTSS assessment of OBT activity (N=628)

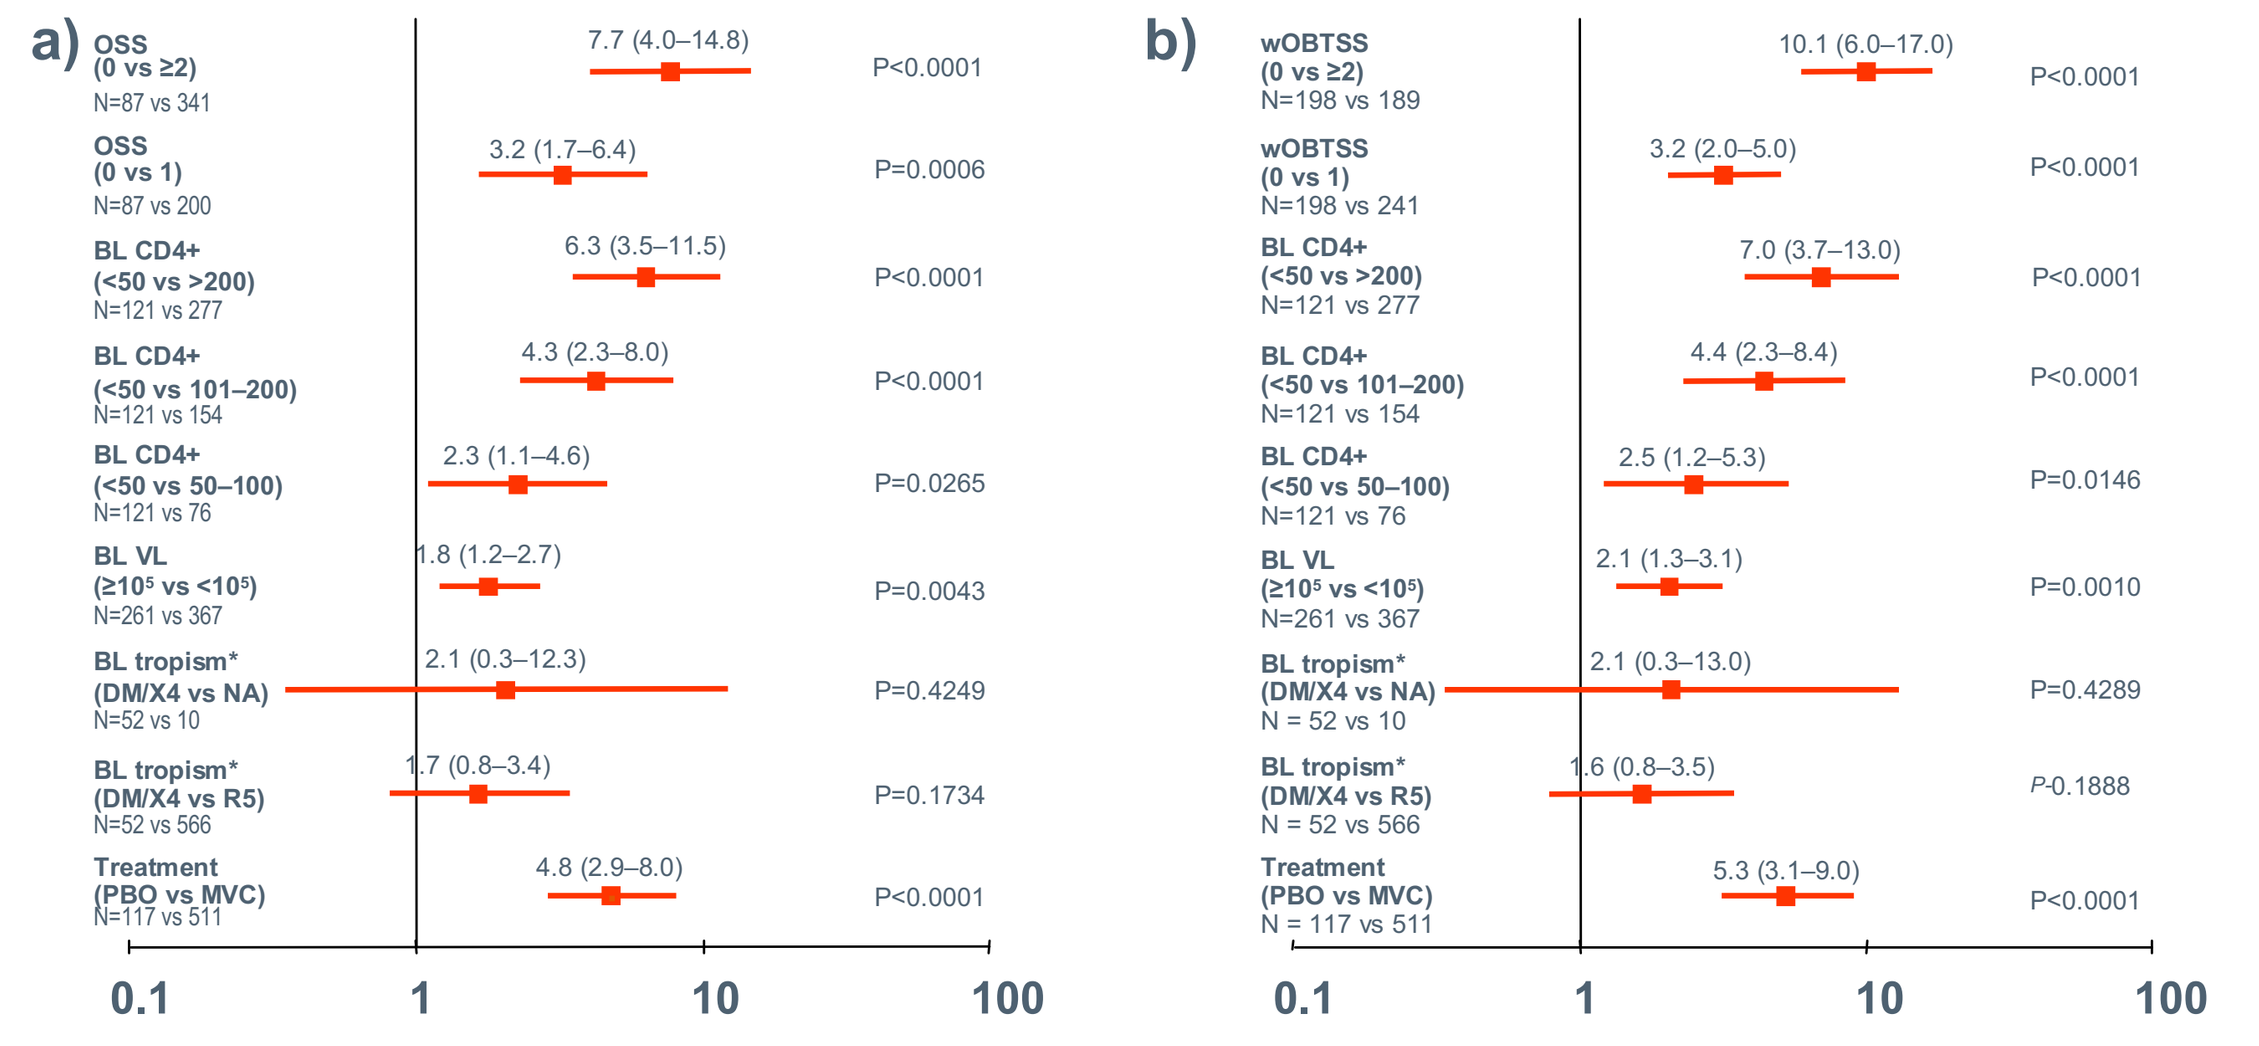

a)
b)
Odds Ratio
Odds Ratio

a) H&L: 0.0950; R2: 0.2406; Adj-R2: 0.3212; Pearson: 0.1881; AIC: 715.221; Somers' D: 0.568; Deviance: 0.1754; SC: 759.647; c: 0.784

b) H&L: 0.2082; R2: 0.2820; Adj-R2: 0.3766; Deviance: 0.5779; AIC: 679.955; Somers' D: 0.623; Pearson: 0.7290; SC: 724.380; c: 0.812

* All subjects previously screened with R5 virus.
Fractional wOBTSS values were rounded down.
BL, baseline; VL, viral load; DM/X4, dual/mixed or X4; NA, tropism result not available (non-reportable, non-phenotypeable, or VL below assay limit); PBO, placebo; MVC, maraviroc.

- Keeping other covariates the same, the wOBTSS appears to fit the model better than the OSS method, as shown by an improved H & L score (0.2082 vs 0.0950), and other statistical test results as presented (Figure 4). Subsequent analyses used the wOBTSS only.
- Baseline CD4+ cell count >50 cells/mm^3 and a wOBTSS or OSS result >0 were both strong predictors of virologic success, as was a baseline viral load <10^5 copies/mL.
- An OSS or wOBTSS value of ≥2 (vs a score of 0) and a baseline CD4+ cell count >200 cells/mm^3 (vs <50 cells/mm^3) were the strongest predictors of success. Decreasing baseline CD4+ was associated with decreasing probability of success. Lower viral load at baseline (<10^5 copies/mL) and use of MVC were also significant independent predictors of success in both models.
- In these patients, who had R5 virus at screening, an R5 vs non-R5 result at baseline was not a significant predictor of success in either model.
- To better explore the importance of these variables on MVC response, the wOBTSS model was run again after exclusion of VO subjects receiving PBO (Figure 5).

Figure 5. wOBTSS and baseline CD4+ cell count are the strongest predictors of response to MVC: Odds ratios (95% confidence intervals) for predictors of virologic success in subjects receiving MVC QD or BID (VO population excluding subjects on PBO; N=511).

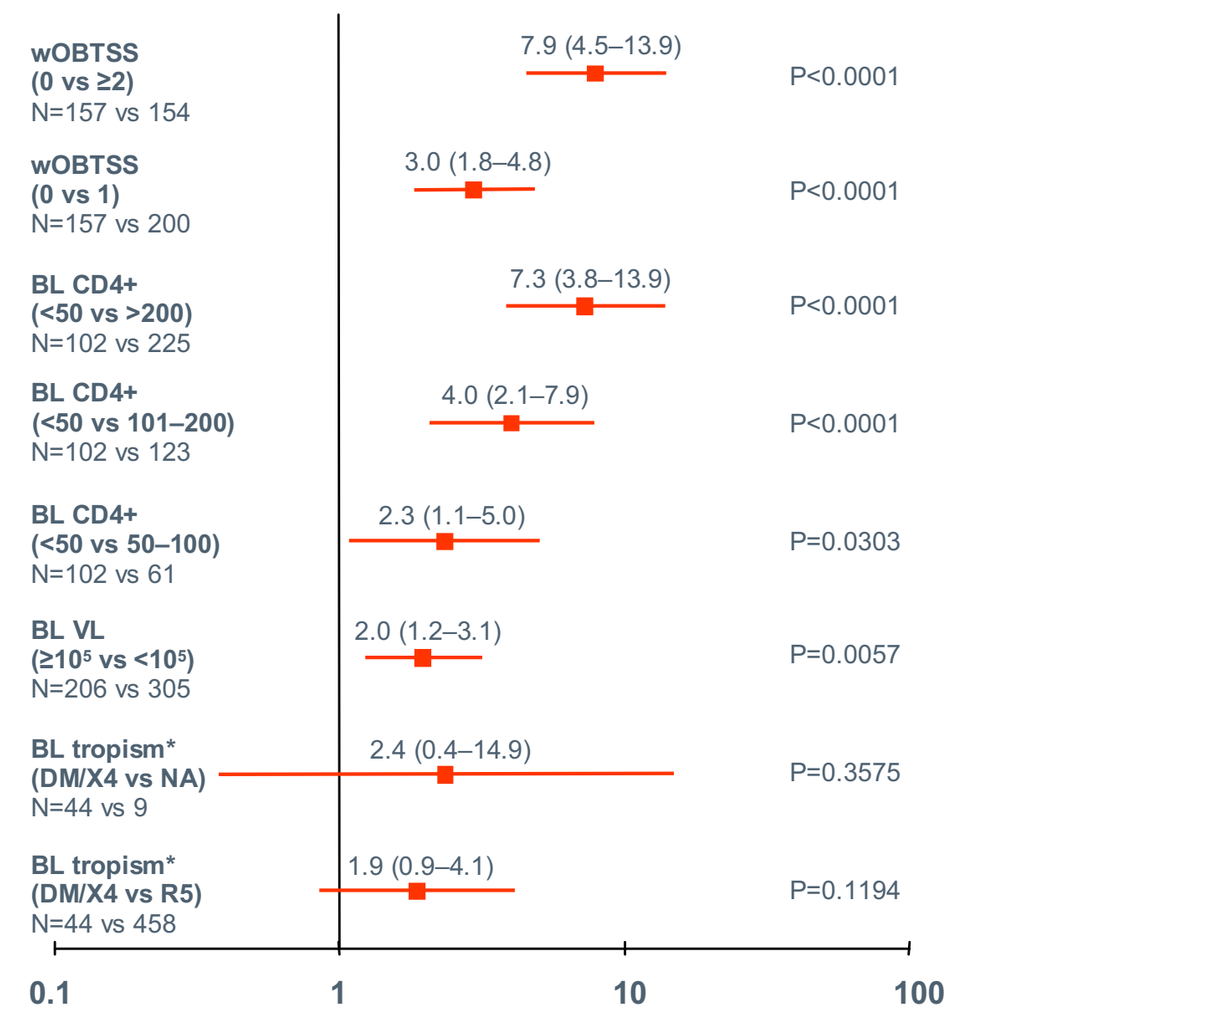

H&L: 0.2472; R^2: 0.2443; Adj-R^2: 0.3259; Deviance: 0.3027; AIC: 582.242; Somers' D: 0.583; Pearson: 0.4925; SC: 620.369; c: 0.792

* All subjects previously screened with R5 virus.
Fractional wOBTSS values were rounded down.
BL, baseline; VL, viral load;
DM/X4, dual/mixed or X4;
NA, tropism result not available (non-reportable, non-phenotypeable, or VL below assay limit).

- Predictors of virologic success and odds ratios were similar when subjects receiving PBO were excluded from the model.
- Among MVC recipients and in multivariate models controlled for other relevant clinical variables the HIV-1 RNA responses among those with an R5 tropism result at screening but non-R5 at baseline were not different from those with an R5 result at both screening and baseline.
- Clinical trial data from subjects who screened with DM/X4 virus (Study 4001029) show no virologic benefit for MVC vs placebo.[7]
- Lack of significance in the VO population analysis may therefore have been due to small population size.

Figure 6. Proportion of subjects with virologic success (<50 HIV-1 RNA copies/mL at Week 48) according to wOBTSS for a) the full VO population and b) subjects with baseline CD4+ cell count ≥50 cells/mm³

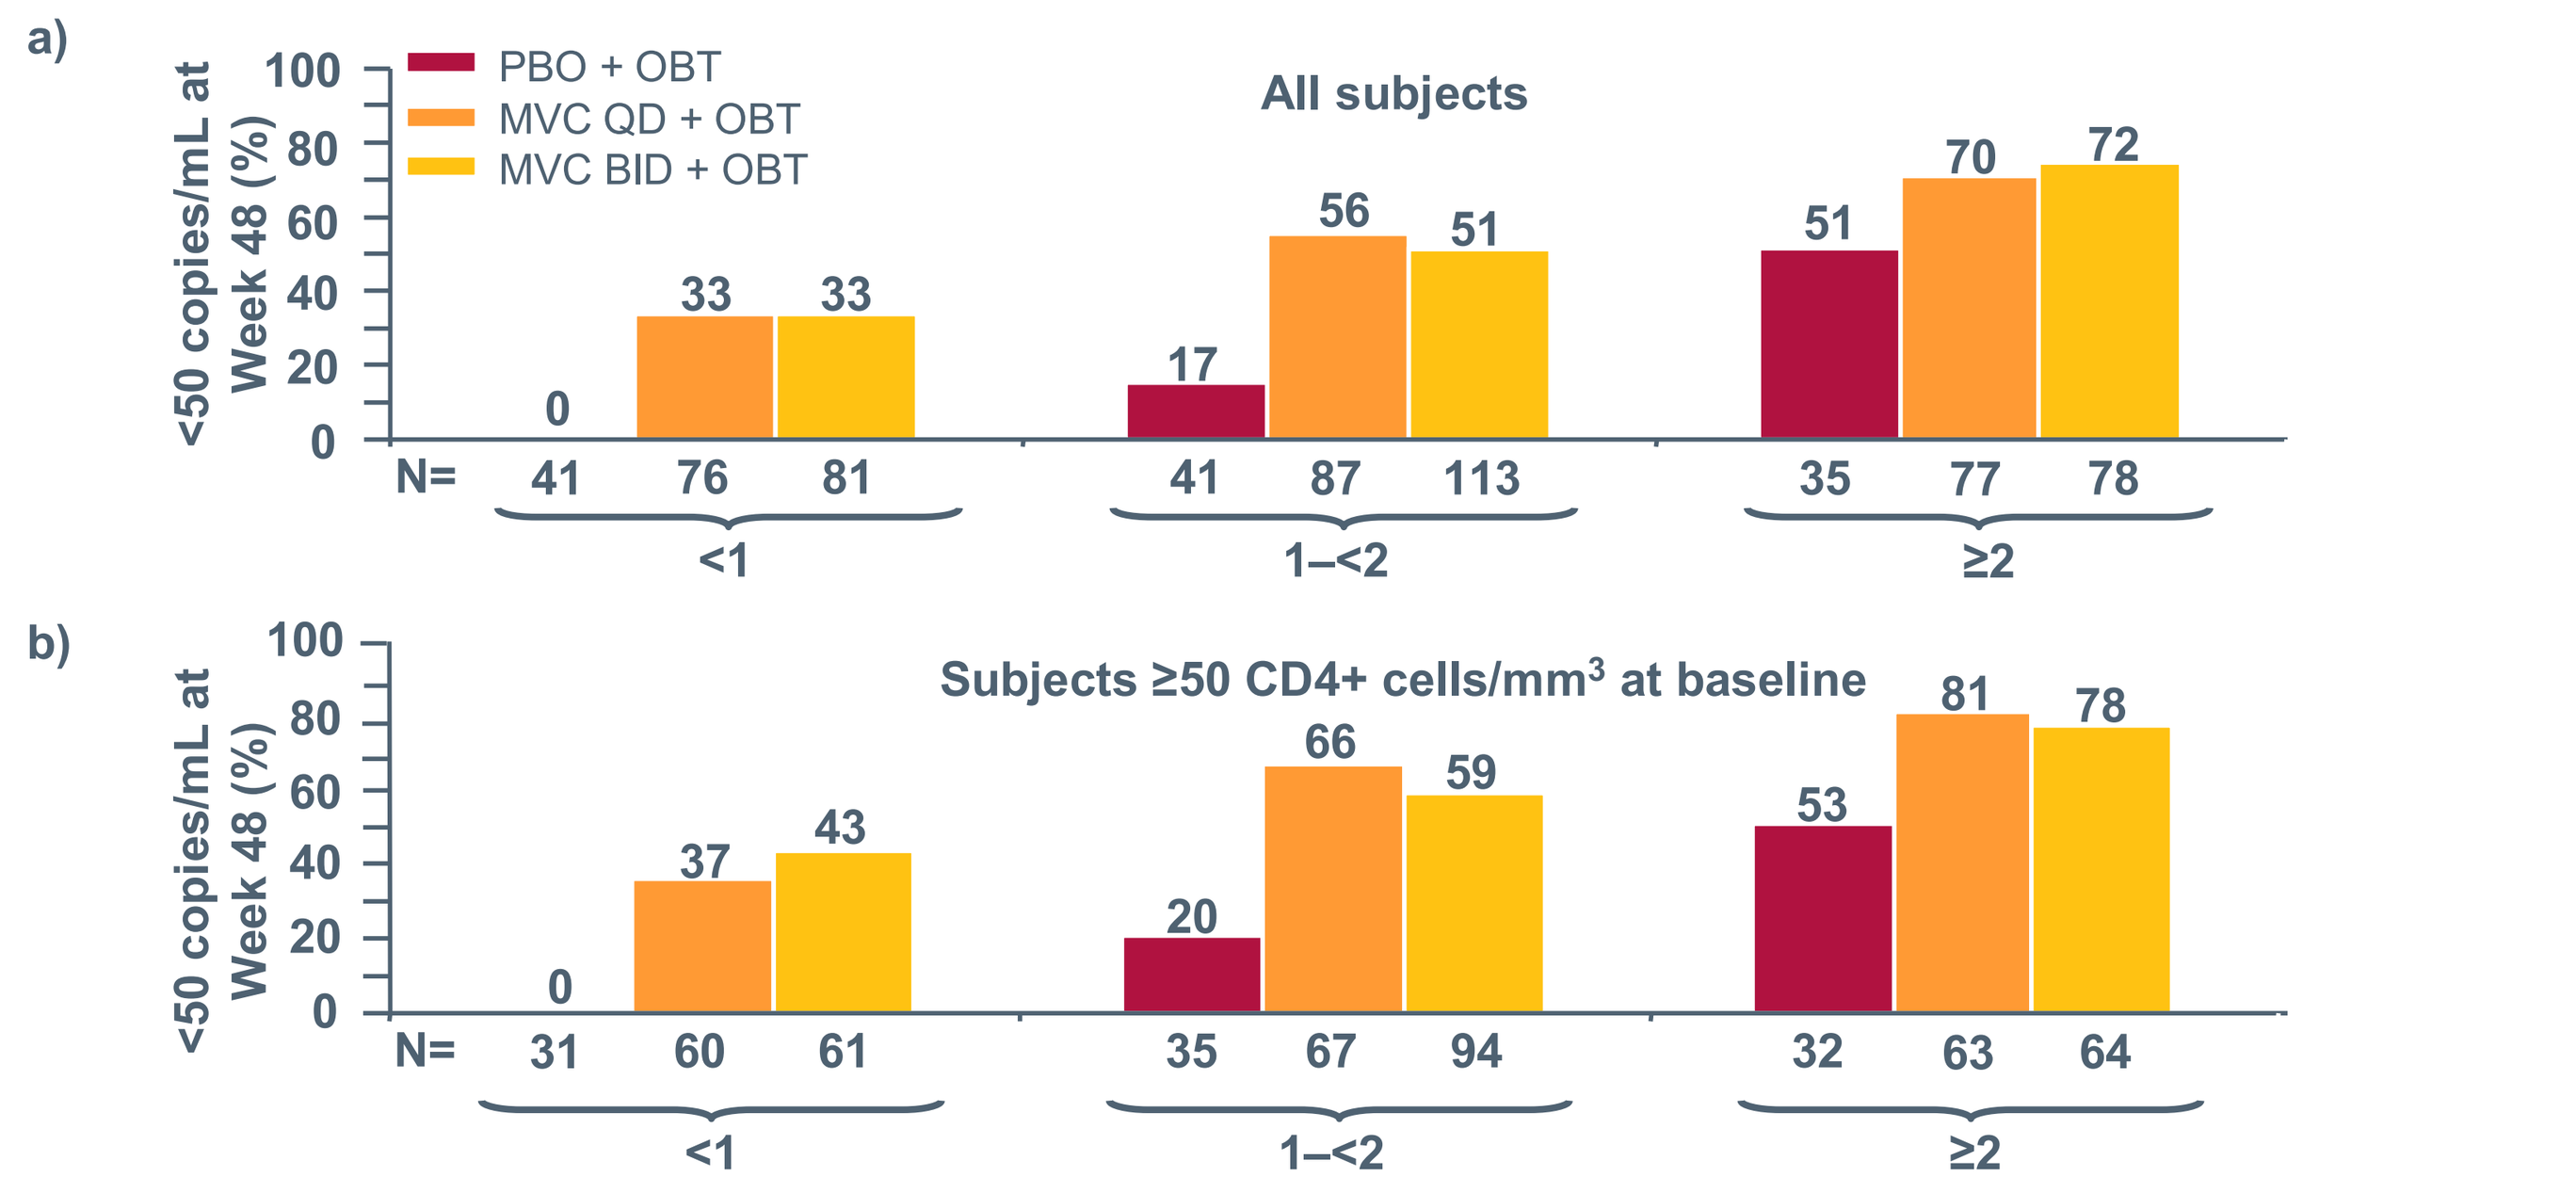

a)
All subjects
<50 copies/mL at Week 48 (%)
PBO + OBT
MVC QD + OBT
MVC BID + OBT
b)
Subjects ≥50 CD4+ cells/mm³ at baseline

- Approximately 80% of subjects receiving MVC (QD or BID) with a wOBTSS of ≥2 and ≥50 CD4+ cells/mm^3 at baseline achieved undetectable viremia, compared to 53% of similar subjects who did not receive MVC.
- Subjects receiving MVC (QD or BID) with a wOBTSS of ≥2 also displayed low rates of CXCR4-using virus at virologic failure (Figure 7).

Figure 7. With wOBTSS ≥2 the percentage of patients who achieve virologic success is higher with MVC while the percentage failing with CXCR4-using virus is similar to placebo

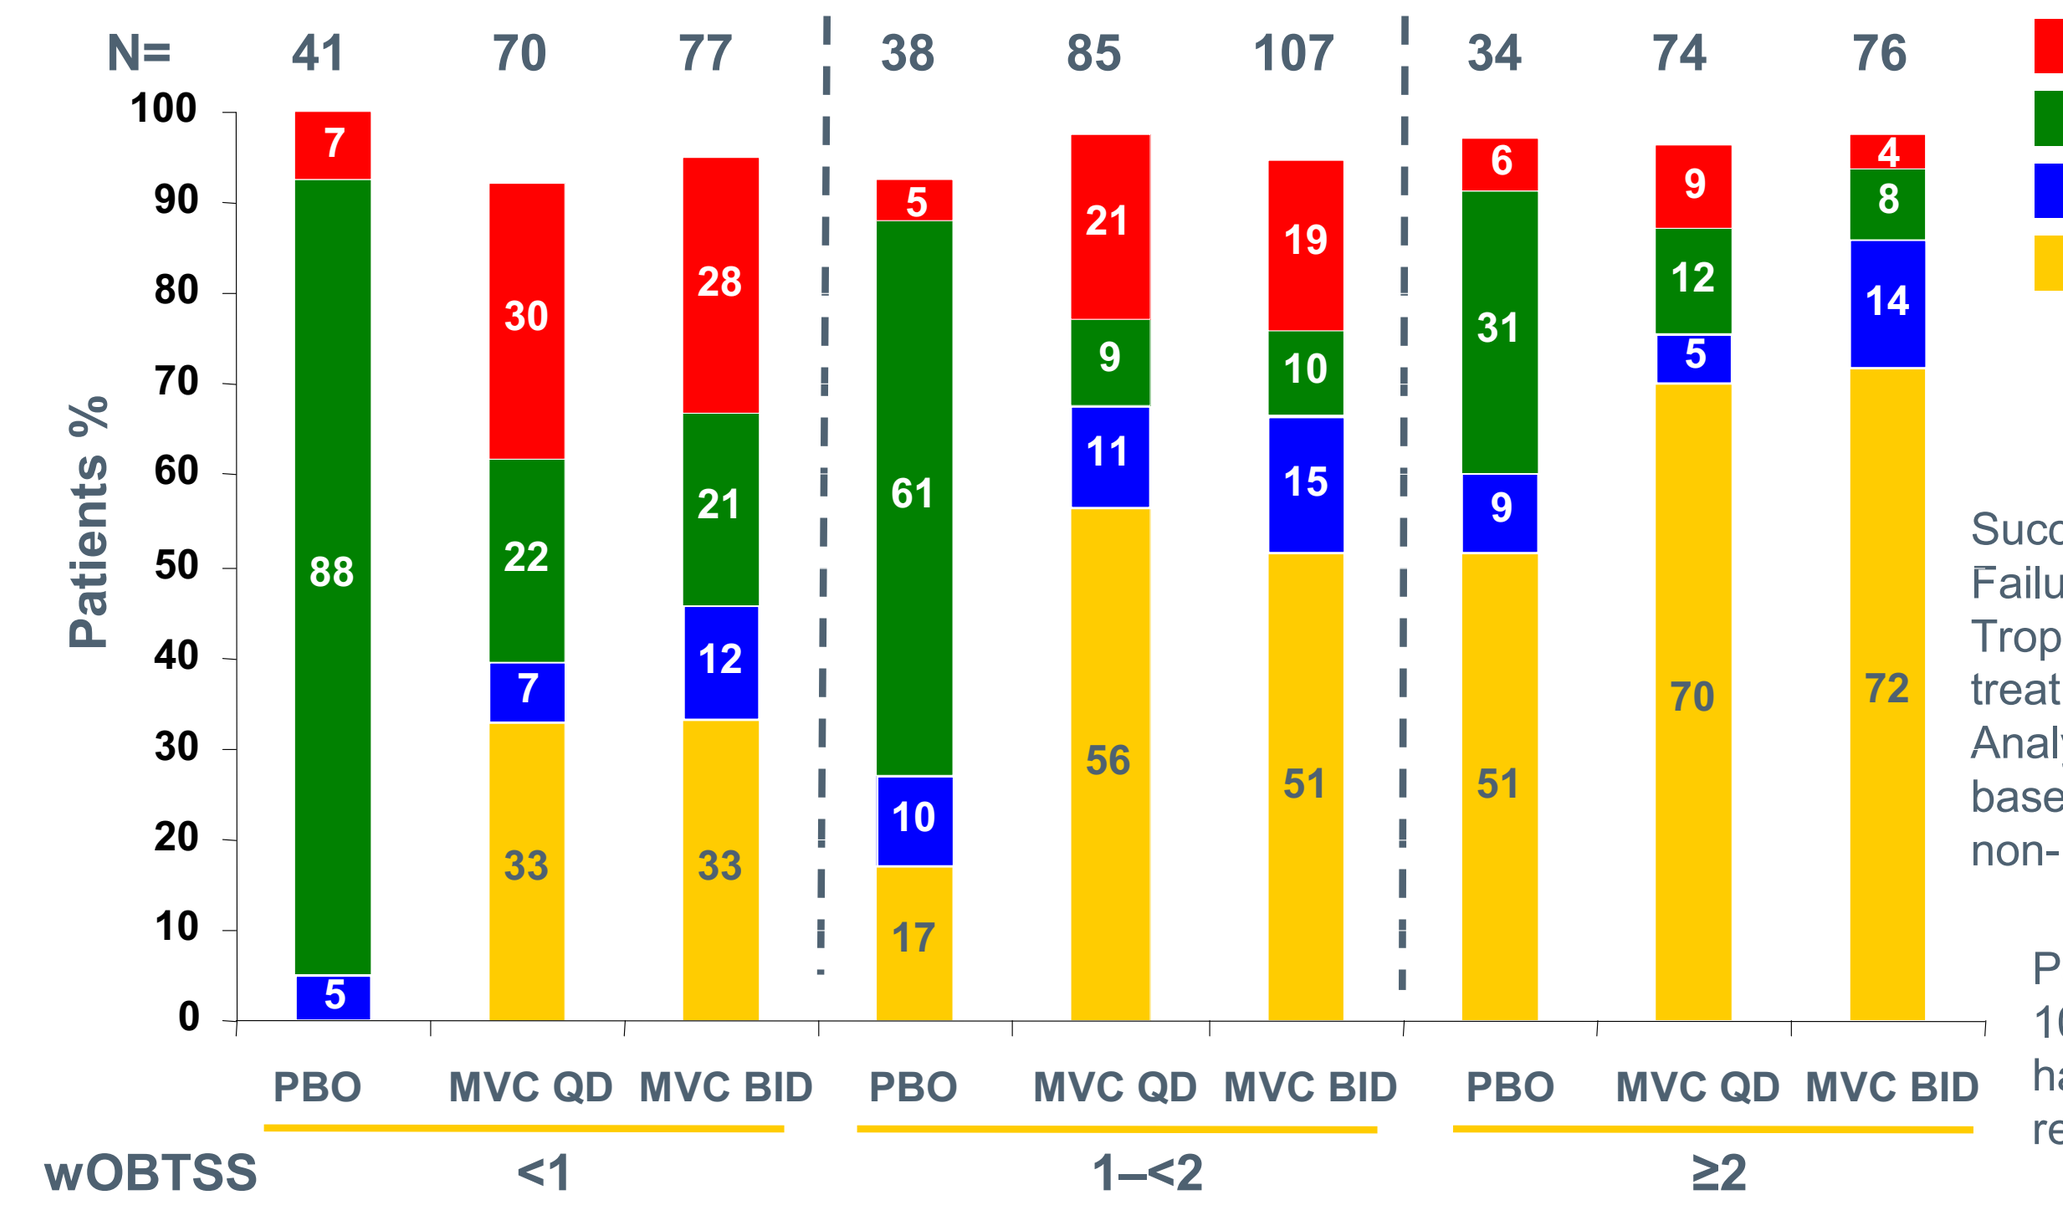

Patients %
Fail with CXCR4-using virus
Fail with R5 virus
BLQ Failure (VL<500 copies/mL)
Success
PBO
MVC QD
MVC BID
wOBTSS

Success = <50 RNA copies/mL at week 48.
Failure = ≥50 RNA copies/mL at week 48.
Tropism was retrieved at the last on-treatment assessment.
Analysis only includes subjects with a post-baseline observation and who had a non-missing wOBTSS score.

Percentages do not add up to 100 because 0–8% of patients had a non-reportable tropism result at failure

Conclusions

- In this defined treatment-experienced population the wOBTSS provided a better measure of OBT activity than counting active drugs.
- A wOBTSS ≥2 and a baseline CD4+ cell count >50 cells/mm^3 are the strongest predictors of virologic response to a MVC-containing regimen.
- Approximately 80% of subjects receiving MVC (BID or QD) with wOBTSS ≥2 and >50 CD4+ cells/mm^3 at initiation had undetectable viremia at Week 48.
- These findings are consistent with earlier data in treatment-experienced patients showing baseline CD4+ cell count, viral load, number of active OBT agents, and prior experience of fewer ARVs as being strongly predictive of virologic response on enfuvirtide.[3]
- The percentage of patients experiencing virologic failure associated with CXCR4-using HIV among patients with a wOBTSS of ≥2 were similar in the MVC and placebo arms.

References

1. Hardy D, *et al. 15th CROI.* Boston, USA. February 3–6, 2008; Abstract 792.
2. Deeks SG, *et al. J Infect Dis* 1999; **179**:1375–1381.
3. Clotet B, *et al. AIDS 2004*; **18**:1137–1146.
4. Swanstrom R, *et al. J Infect Dis* 2004; **190**:886–893.
5. Coakley EP, *et al. Antivir Ther* 2006; **11**:S81.
6. Hosmer DW & Lemeshow S, *Applied Logistic Regression, 2nd Edn.*, 2000; New York, NY, USA: John Wiley and Sons, Inc.
7. Goodrich JM, *et al. 45th ICAAC.* Washington, DC, December 16–19, 2007; Abstract LB-2.