Estimated Number of Births to HIV-Positive Women, United States, 2006

Suzanne K. Whitmore, DrPH; Xinjian Zhang, PhD; Allan W. Taylor, MD, MPH

Division of HIV/AIDS Prevention
National Center for HIV/AIDS, Viral Hepatitis, STD, and TB Prevention
Centers for Disease Control and Prevention, Atlanta, GA

Introduction

- Current strategies for the elimination of perinatal HIV infections (<1% transmission rate) call for targeting areas of missed opportunities for HIV prevention among HIV+ women of child-bearing age.
- Yet enumeration of HIV+ women of childbearing age for the U.S. is lacking due to incomplete reporting of perinatally HIV-exposed cases from the 50 states and the District of Columbia. Currently, approximately 34 jurisdictions collect data on perinatal HIV exposure, which include reporting of HIV+ infants and infants that serorevert after birth.
- The total number of infants perinatally exposed to HIV is needed to determine overall transmission rates for perinatal HIV infection in the U.S. Without a denominator for determining the number of women of childbearing age and information on the demographic, clinical, and behavioral characteristics of HIV exposed infants, targeting national goals and strategies to eliminate the numbers to <1% would be problematic.
- This study applies a variety of methods to determine the number of exposed infants in the United States in 2006. The number of HIV-exposed infants is approximately equal to the number of HIV+ women of child-bearing age giving birth.

Methods

- The model used in the current study was based on the abstract from Fleming et al. in 2002 [1] using a Poisson model to estimate the total number of HIV+ women of childbearing age (aged 13-44) in the United States.
- The number of births to HIV+ women in 2006 was estimated as the product of two parameters: the total number of HIV+ women of childbearing age, and the pregnancy rate for HIV + women.
- The total number of HIV+ women was estimated from 3 numbers using data reported to the U.S. national HIV/AIDS reporting system (HARS):

(1) The number of women (aged 13-44) living with diagnosed HIV (not AIDS) was estimated from 33 HIV reporting states and then extrapolated to all other states based on a Poisson regression model.

Figure1: 33 States with Name-Based HIV Reporting since 1996, United States

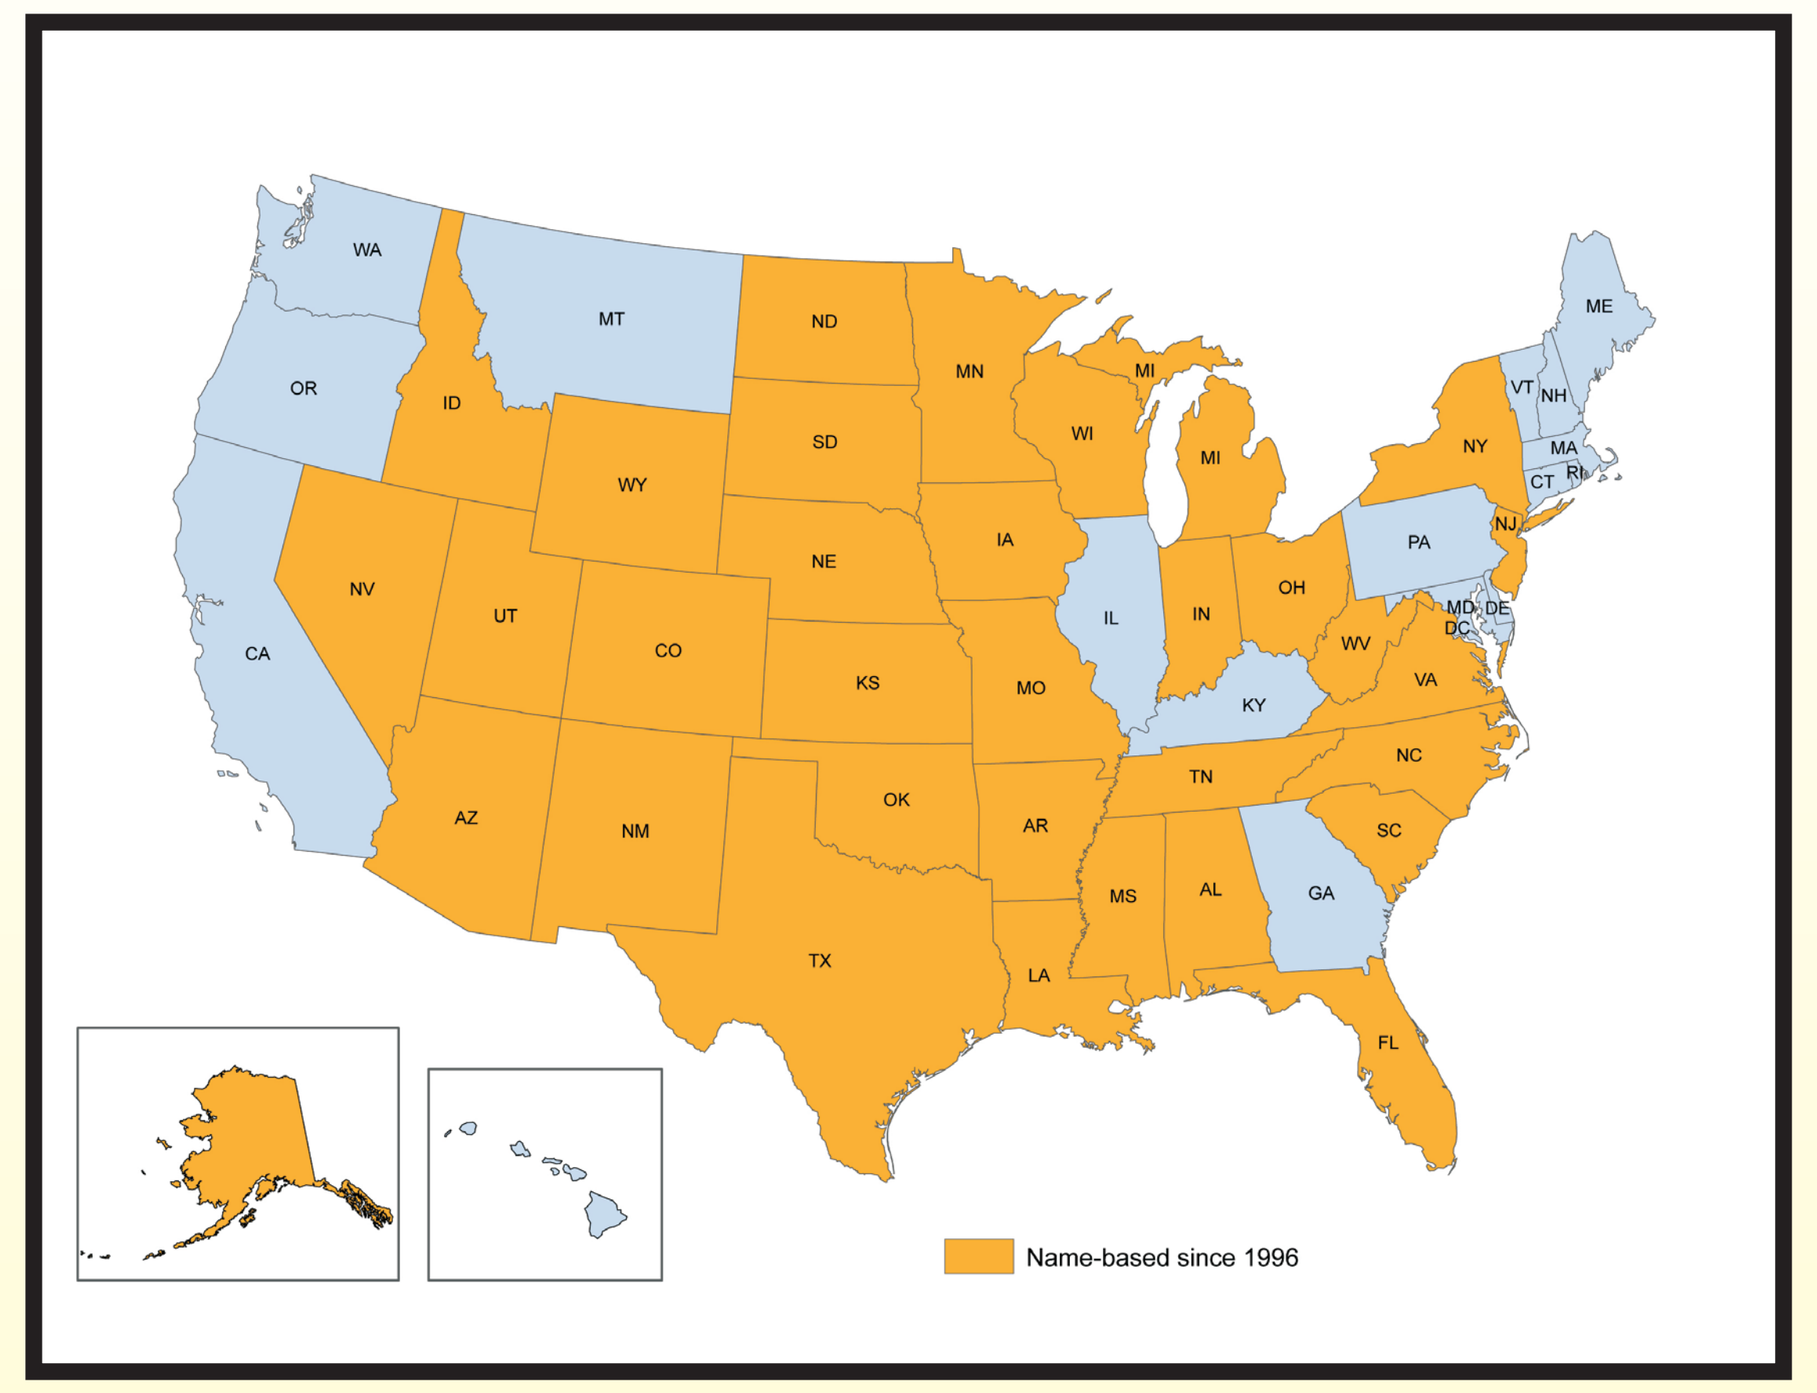
Name-based since 1996

(2) The number of HIV + women of childbearing age, whose infection has not yet been diagnosed, was estimated using a back-calculation method. Back-calculation models have been described in determining both the incidence of HIV infection in the United States and HIV prevalence estimates for 2006 [2, 3].

(3) The total number of women of childbearing age, living with AIDS (immunologic AIDS, and clinical AIDS) was computed directly from the reported data of AIDS diagnosis from 50 states and the District of Columbia, adjusting for reporting delay.

- The pregnancy rate of HIV+ women was estimated from the Adult/adolescent Spectrum of Disease (ASD) project conducted in over 100 clinics in 11 U.S. cities.
- Finally, by using the observed pregnancy rates from ASD, births were estimated for the total number of women living with HIV diagnosis (not AIDS), immunologic AIDS, and clinical AIDS. All analyses were conducted using SAS Version 9.1.

Results

- As of 2006, a total of 111,332 women aged 13-44 years were diagnosed and living with HIV (not AIDS) in the United States.
- By using back-calculation, we obtained the undiagnosed proportions for HIV infection among women of childbearing age by race/ethnicity. After stratification of the proportion of women with undiagnosed HIV infection for each race group, the data were then summarized to get the total number of undiagnosed cases for 2006.

Results (cont)

- After adjustment to estimate the number of women with undiagnosed HIV infection, we estimated that 125,050-128,653 women were living with HIV (not AIDS) in 2006. Fleming et al. reported that 80,293-86,080 women were living with HIV (not AIDS) in 2000 [1], which represents an increase of approximately 33-36% of women of childbearing age living with HIV (not AIDS) from 2000 to 2006.
- We calculated the pregnancy rate for women aged 15-44 per 100 person years of observation during 1998-2001, stratified by sex, race/ethnicity, age group, risk category, and clinical status for HIV (not AIDS) and AIDS. There were an estimated 7,021-7,272 pregnancies for women of childbearing age, living with HIV (not AIDS) in 2006.
- For the total of 56,822 women, aged 13-44 living with AIDS in 2006, 55% had immunologic AIDS (CD4 counts<200, 899 pregnancies), and 45% presented with clinical AIDS (730 pregnancies), showing signs of opportunistic infections.

Tables and Figures

Table 1: Total number of women, aged 13-44 living with AIDS and HIV (not AIDS) in 2000 and 2006, U.S.

	Low Estimate	Estimate	High Estimate
Living with AIDS 2000	N/A	49,238	N/A
Living with HIV (not AIDS) 2000	80,293	N/A	86,080
Living with AIDS 2006	N/A	56,822	N/A
Living with HIV (not AIDS) 2006	125,050	N/A	128,653

- The estimated number of births in 2006 to all women living with HIV infection was 8,650-8,900, approximately 30% higher than the estimated number of total births (6,075-6,422 in 2000) reported by Fleming et al. in 2002.

Table 2: The estimated number of births to women aged 13-44 living with HIV disease in 2000 and 2006, U.S.

	Low Estimate	High Estimate
Estimated number of births in 2000	6,075	6,422
Estimated number of births in 2006	8,650	8,900

Figure2: Flowchart of methods for calculating the estimated number of births to HIV-positive women, United States, 2006

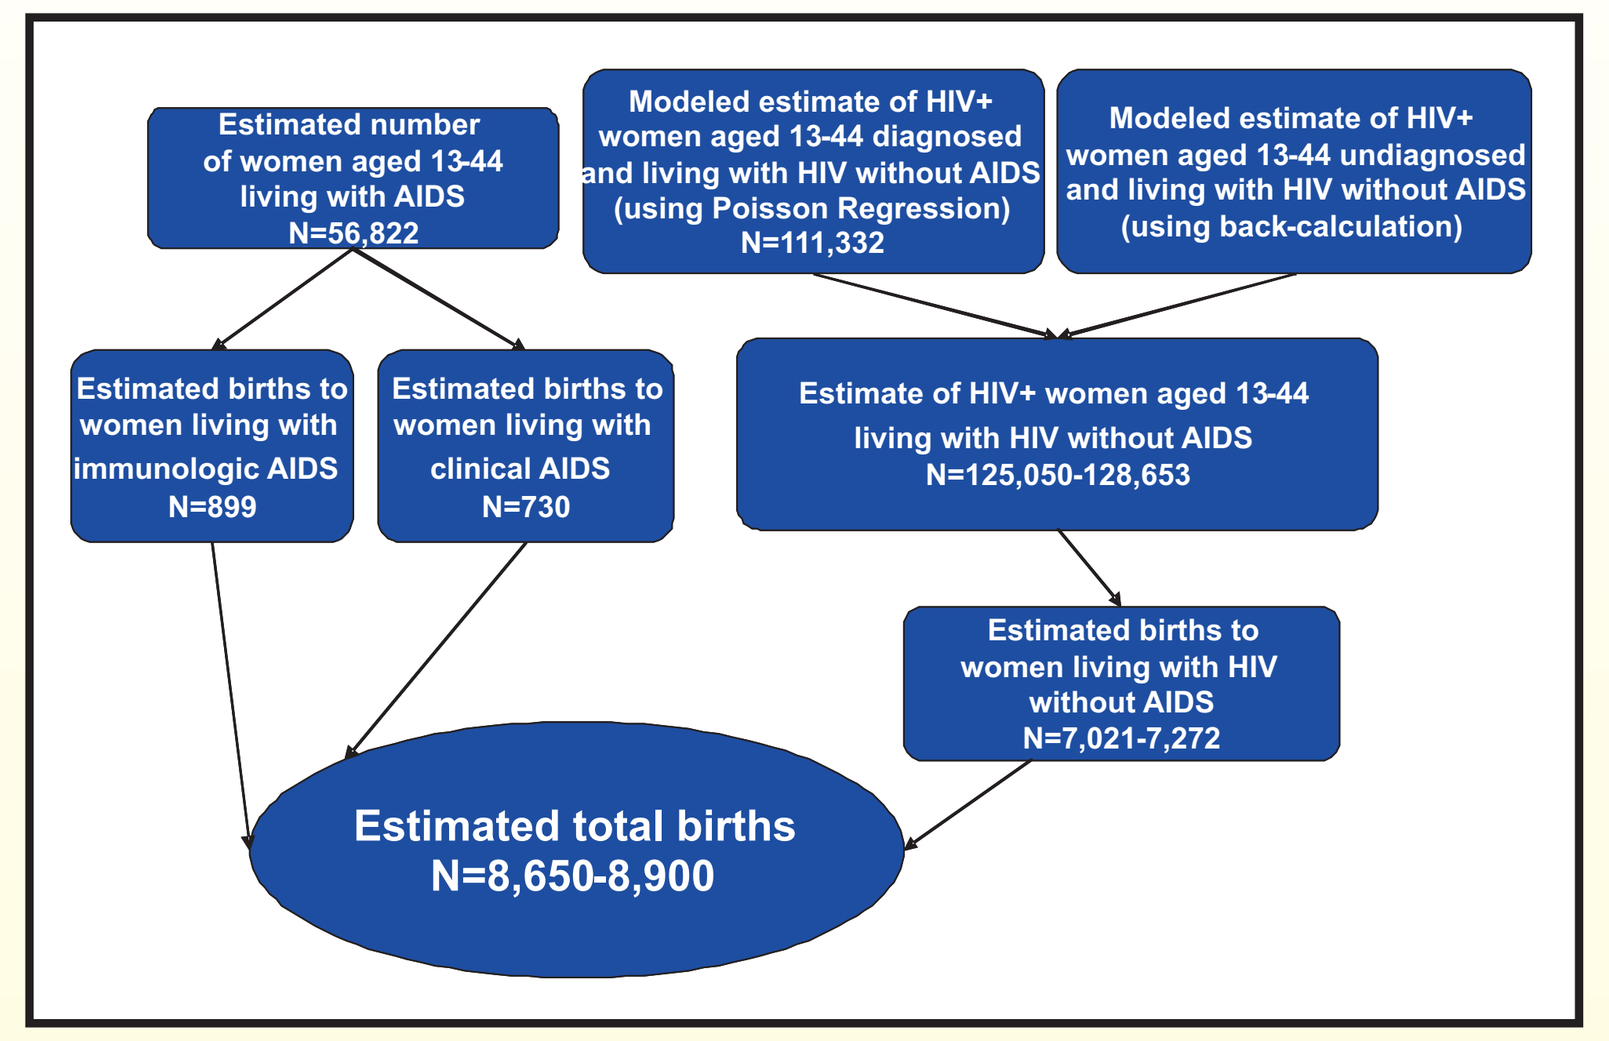
Estimated number of women aged 13-44 living with AIDS N=56,822
Modeled estimate of HIV+ women aged 13-44 diagnosed and living with HIV without AIDS (using Poisson Regression) N=111,332
Modeled estimate of HIV+ women aged 13-44 undiagnosed and living with HIV without AIDS (using back-calculation)
Estimated births to women living with immunologic AIDS N=899
Estimated births to women living with clinical AIDS N=730
Estimate of HIV+ women aged 13-44 living with HIV without AIDS N=125,050-128,653
Estimated births to women living with HIV without AIDS N=7,021-7,272
Estimated total births N=8,650-8,900

Comment / Discussion

- Although the incidence of HIV infection through perinatal transmission has declined over 95% in the past decade with adherence to USPHS recommendations, the estimated numbers of HIV-positive women of childbearing age giving birth in the United States increased from 2000 to 2006.
- HIV-infected women of childbearing age are living longer, have a wider range of reproductive choices (including assisted reproductive technologies for HIV discordant couples), and leading healthier lives.
- A number of assumptions were made in this study:

(1) The prevalent number of HIV-positive women aged 13-44 follows a Poisson distribution; and the number of HIV infections in the 33 states with HIV data is a proportion of the HIV infection in the United States (51 areas with AIDS data) and may not accurately represent the entire nation.

(2) The Adult Spectrum of Disease study does not systematically record pregnancy outcome; we assumed that all pregnancies resulted in single live births, which may overestimate or underestimate the number of live births to women with AIDS.

(3) In addition, there are a number of assumptions made in using back-calculation for estimation of the number of women with undiagnosed HIV (not AIDS) for women of childbearing age. Information on the limitations for back-calculation models has been documented in determining both the incidence of HIV infection in the United States and HIV prevalence estimates for 2006 [2, 3].

- Universal HIV perinatal exposure reporting has been recommended by CDC, the Council of State and Territorial Epidemiologists, and the American Academy of Pediatrics for all jurisdictions [4, 5].
- Without universal HIV perinatal exposure reporting, indirect extrapolation such as performed in this study is the only available method to estimate the total number of HIV-exposed infants in the U.S.

References

(1) Estimated number of perinatal HIV infections, U.S., 2000. Fleming PL, Lindegren ML, Byers R, Hammett T, Harris N, Schulte J, Janssen R; *Int Conf AIDS*. 2002 Jul 7-12; 14: abstract no. TuPeC4773. CDC, Atlanta, United States

(2) ML Campsmith, DDS, P Rhodes, PhD, HI Hall, PhD, T Green, HIV Prevalence Estimates -- United States, 2006 , MMWR Weekly Report, October 3, 2008 / 57(39);1073-1076

(3) Hall HI, Song R, Rhodes P, et al. Estimation of HIV incidence in the United States. JAMA 2008; 300:520 -- 9.

(4) Committee on Pediatric AIDS, Surveillance of pediatric HIV infection, *Pediatrics* 101 (1998), pp. 315319.

(5) Council of State and Territorial Epidemiologists, Surveillance for perinatal HIV exposure (2002) Atlanta, Ga; Position Statement 02-ID-04.

Acknowledgements

Phil Rhodes, PhD and Janet Blair, PhD,
Division of HIV/AIDS Prevention
National Center for HIV/AIDS, Viral Hepatitis, STD, and TB Prevention
Centers for Disease Control and Prevention,
Atlanta, GA

Patricia L. Fleming, PhD, MS
Dept. of Preventive Medicine & Community Health
New Jersey Medical School
University of Medicine & Dentistry of New Jersey

Dan Hazelwood
Graphic Services Branch, Division of Creative Services,
National Center for Health Marketing
Centers for Disease Control and Prevention,
Atlanta, GA

The findings and conclusions in this report are those of the authors and do not necessarily represent the official position of the Centers for Disease Control and Prevention.

CS124748