16th Conference on Retroviruses and Opportunistic Infections ● February 8–11, 2009 ● Montréal, Canada

Poster M-199

CCR5-tropic Resistance to Maraviroc is Uncommon Even Among Patients on Functional MVC Monotherapy or with Ongoing Low-level Replication

R Jubb, M Lewis, P Simpson, C Craig, M Haddrick, M Perros, and M Westby
Pfizer Global Research and Development, Sandwich, UK
For further information, please contact Becky Jubb at becky.jubb@pfizer.com

Background

- Maraviroc (MVC) is a first-in-class CCR5 antagonist that is approved for use in the management of treatment-experienced patients with CCR5-tropic (R5) HIV-1 infection.
- In the Phase 3 MOTIVATE 1 and 2 studies, MVC (QD or BID) plus optimized background therapy (OBT) demonstrated significantly greater virologic and immunologic efficacy and a similar safety profile compared to placebo + OBT at 48 weeks in treatment-experienced patients with R5 virus.[1] These findings were durable through Week 96.[2]
- As with other studies of new agents in treatment-experienced patients[3,4] preplanned subgroup analyses of the MOTIVATE studies[5] showed that more patients who received a higher number of potentially active drugs in their OBT according to baseline susceptibility scores achieved virologic suppression.
 - Subsequent *post-hoc* analyses of the MOTIVATE virologic outcomes population have shown that an alternative methodology, weighted OBT susceptibility score (wOBTSS), is a better measure of OBT activity than simply counting active drugs (X4 or D/M).[6]
- Data from the MOTIVATE studies demonstrated that virologic failure in treatment-experienced patients receiving MVC-containing regimens can occur with either MVC-resistant R5 virus or as a result of 'un-masking' of pre-existing CXCR4-using virus.[7]
 - MVC resistance is different to resistance to ARVs that act on viral proteins; because MVC acts on a host protein rather than a viral protein the virus cannot mutate to prevent the drug from binding to its target. Rather, MVC resistance arises when the virus mutates to make use of drug-bound CCR5.
 - Plateaus in maximum drug inhibition, rather than shifts in IC_{50}, are phenotypic markers of MVC resistance.[8,9]
 - A signature mutation for MVC has not been identified. V3 loop mutations were identified in viruses that showed a plateau in maximal percentage inhibition below 95%, but the pattern of amino acid changes was different between patients.[9,10]
- Functional monotherapy, where a new agent is combined with an inactive background regimen, is typically associated with rapid emergence of resistance to new agents in treatment-experienced patients.

Objective

- To investigate the influence of OBT regimen activity, as assessed by the wOBTSS algorithm, on the development of phenotypic resistance to MVC in antiretroviral-experienced patients who had virologic failure with R5 virus.

Methods

Study population

- The virologic outcomes (VO) population previously defined for wOBTSS-based analyses of MOTIVATE[6] is a 636 patient subgroup of the full analysis set (Figures 1 and 2).
- The VO population excluded patients who failed study treatment through 48 weeks for non-virologic reasons (n=145) and patients for whom a baseline OBT activity score may not have accurately represented the drugs received throughout their period of randomized therapy (n=268).
- For the purpose of this substudy of MVC patients experiencing virologic failure with R5 virus, the VO population was further refined to exclude patients randomized to receive placebo (n=118), patients with non-R5 virologic failure (n=163)—including those with non-reportable tropism results and with viral load levels too low (<500 copies/mL) for assay at time of failure—and patients without paired baseline and at-failure phenotypic MVC susceptibility data (n=24).

Figure 1. Patients included in the analyses

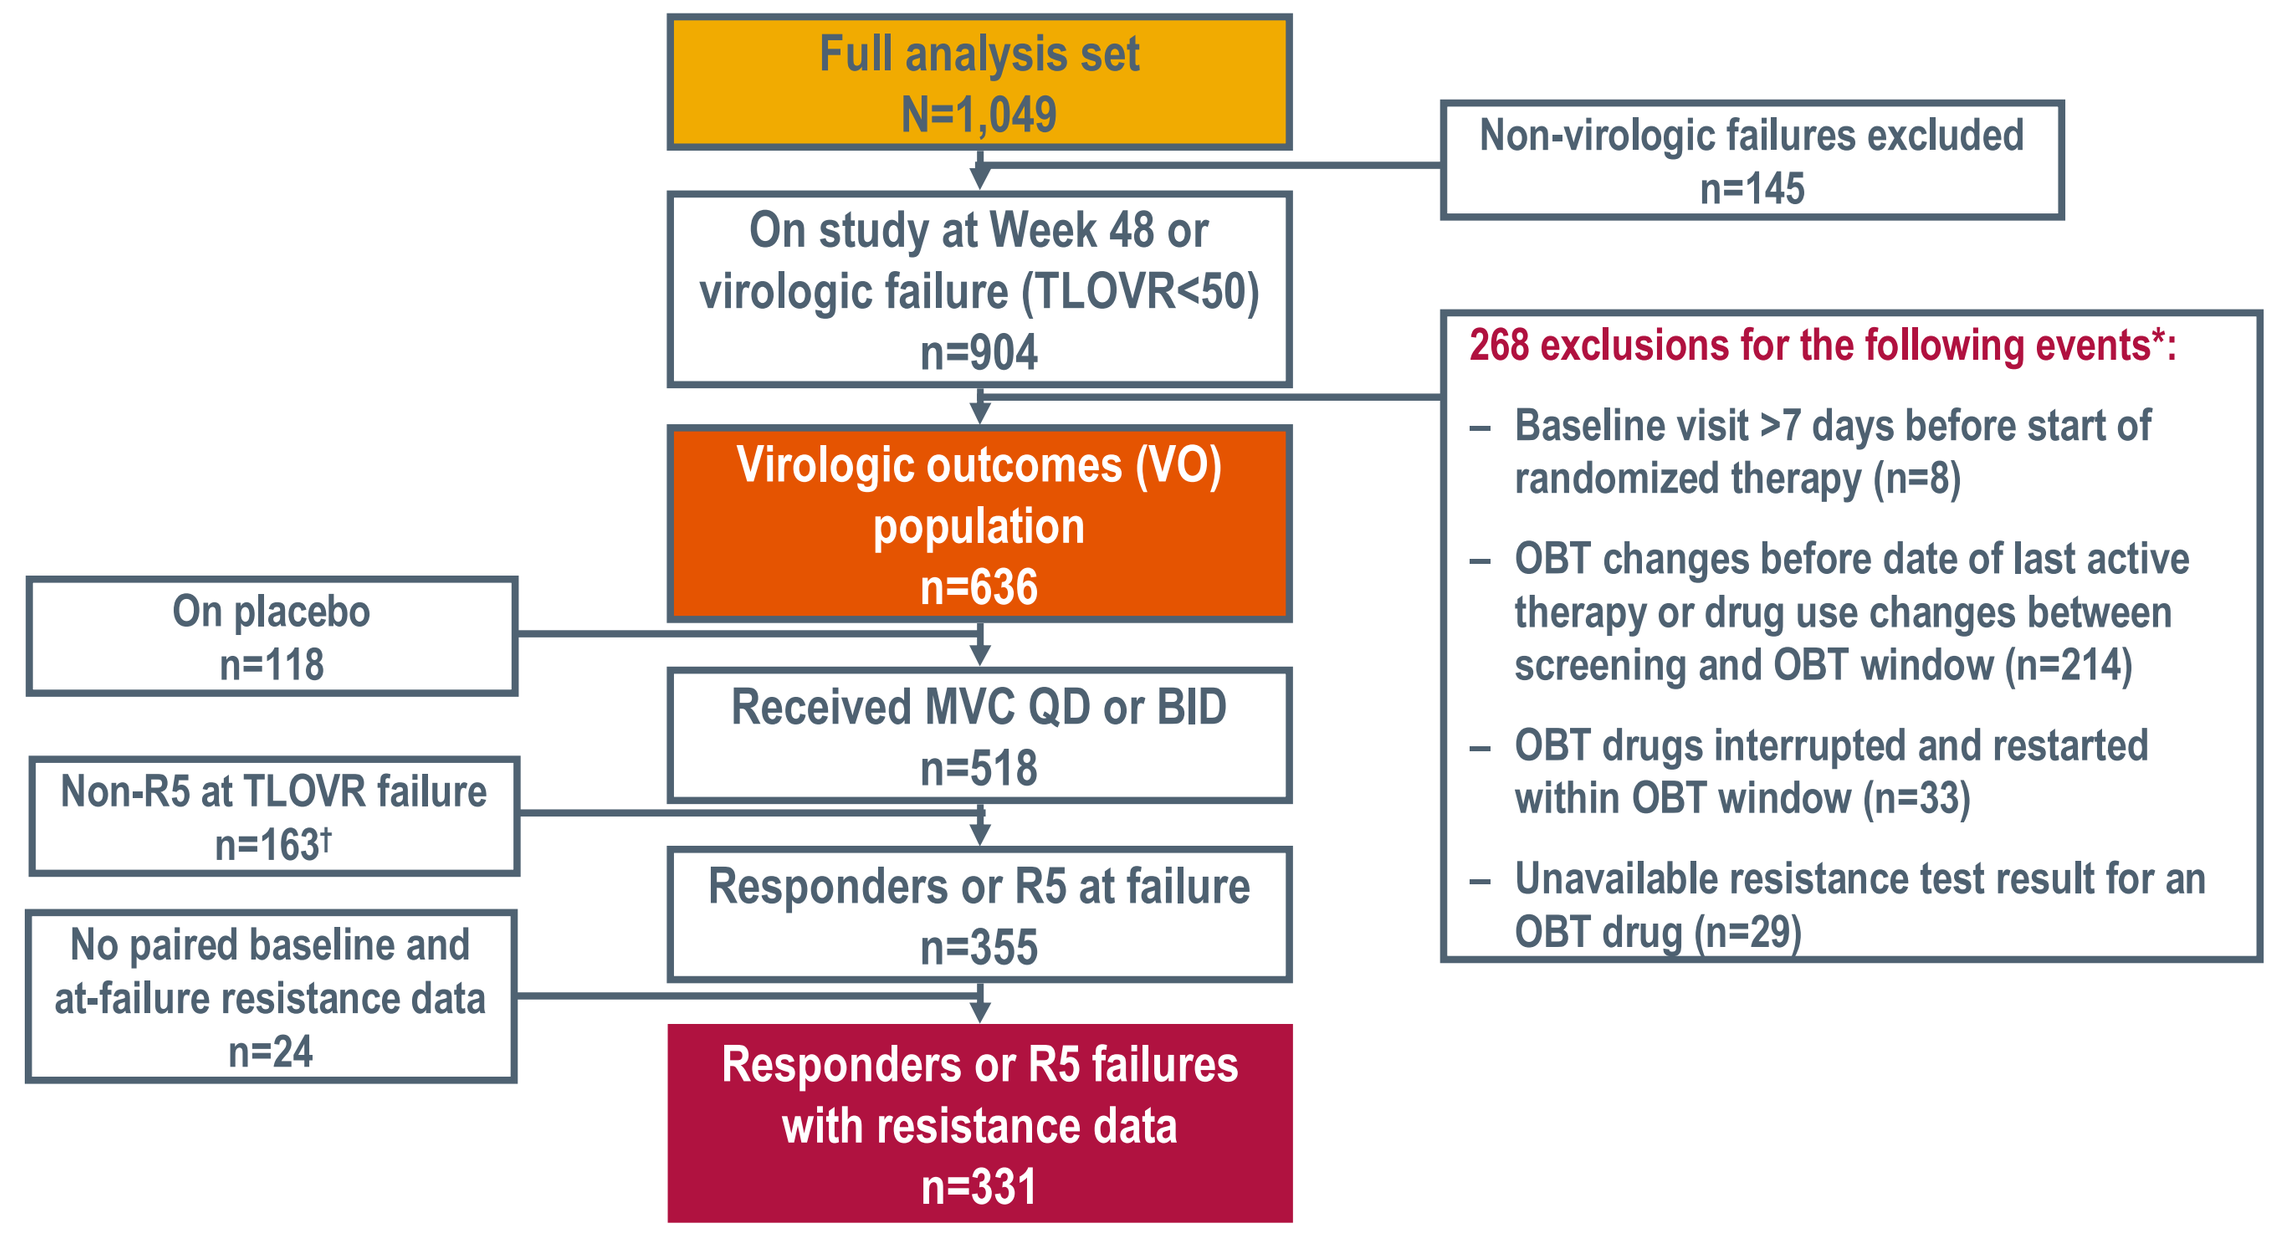
Full analysis set
N=1,049
Non-virologic failures excluded
n=145
On study at Week 48 or virologic failure (TLOVR<50)
n=904
268 exclusions for the following events*:
– Baseline visit >7 days before start of randomized therapy (n=8)
– OBT changes before date of last active therapy or drug use changes between screening and OBT window (n=214)
– OBT drugs interrupted and restarted within OBT window (n=33)
– Unavailable resistance test result for an OBT drug (n=29)
Virologic outcomes (VO) population
n=636
On placebo
n=118
Received MVC QD or BID
n=518
Non-R5 at TLOVR failure
n=163†
Responders or R5 at failure
n=355
No paired baseline and at-failure resistance data
n=24
Responders or R5 failures with resistance data
n=331

†Dual/mixed virus n=65; X4 virus n=30; non-reportable n=12; below limit of quantitation n=56
*More than one event could have occurred in a given patient

Figure 2. Allowed patterns of OBT drug use for inclusion in the VO population

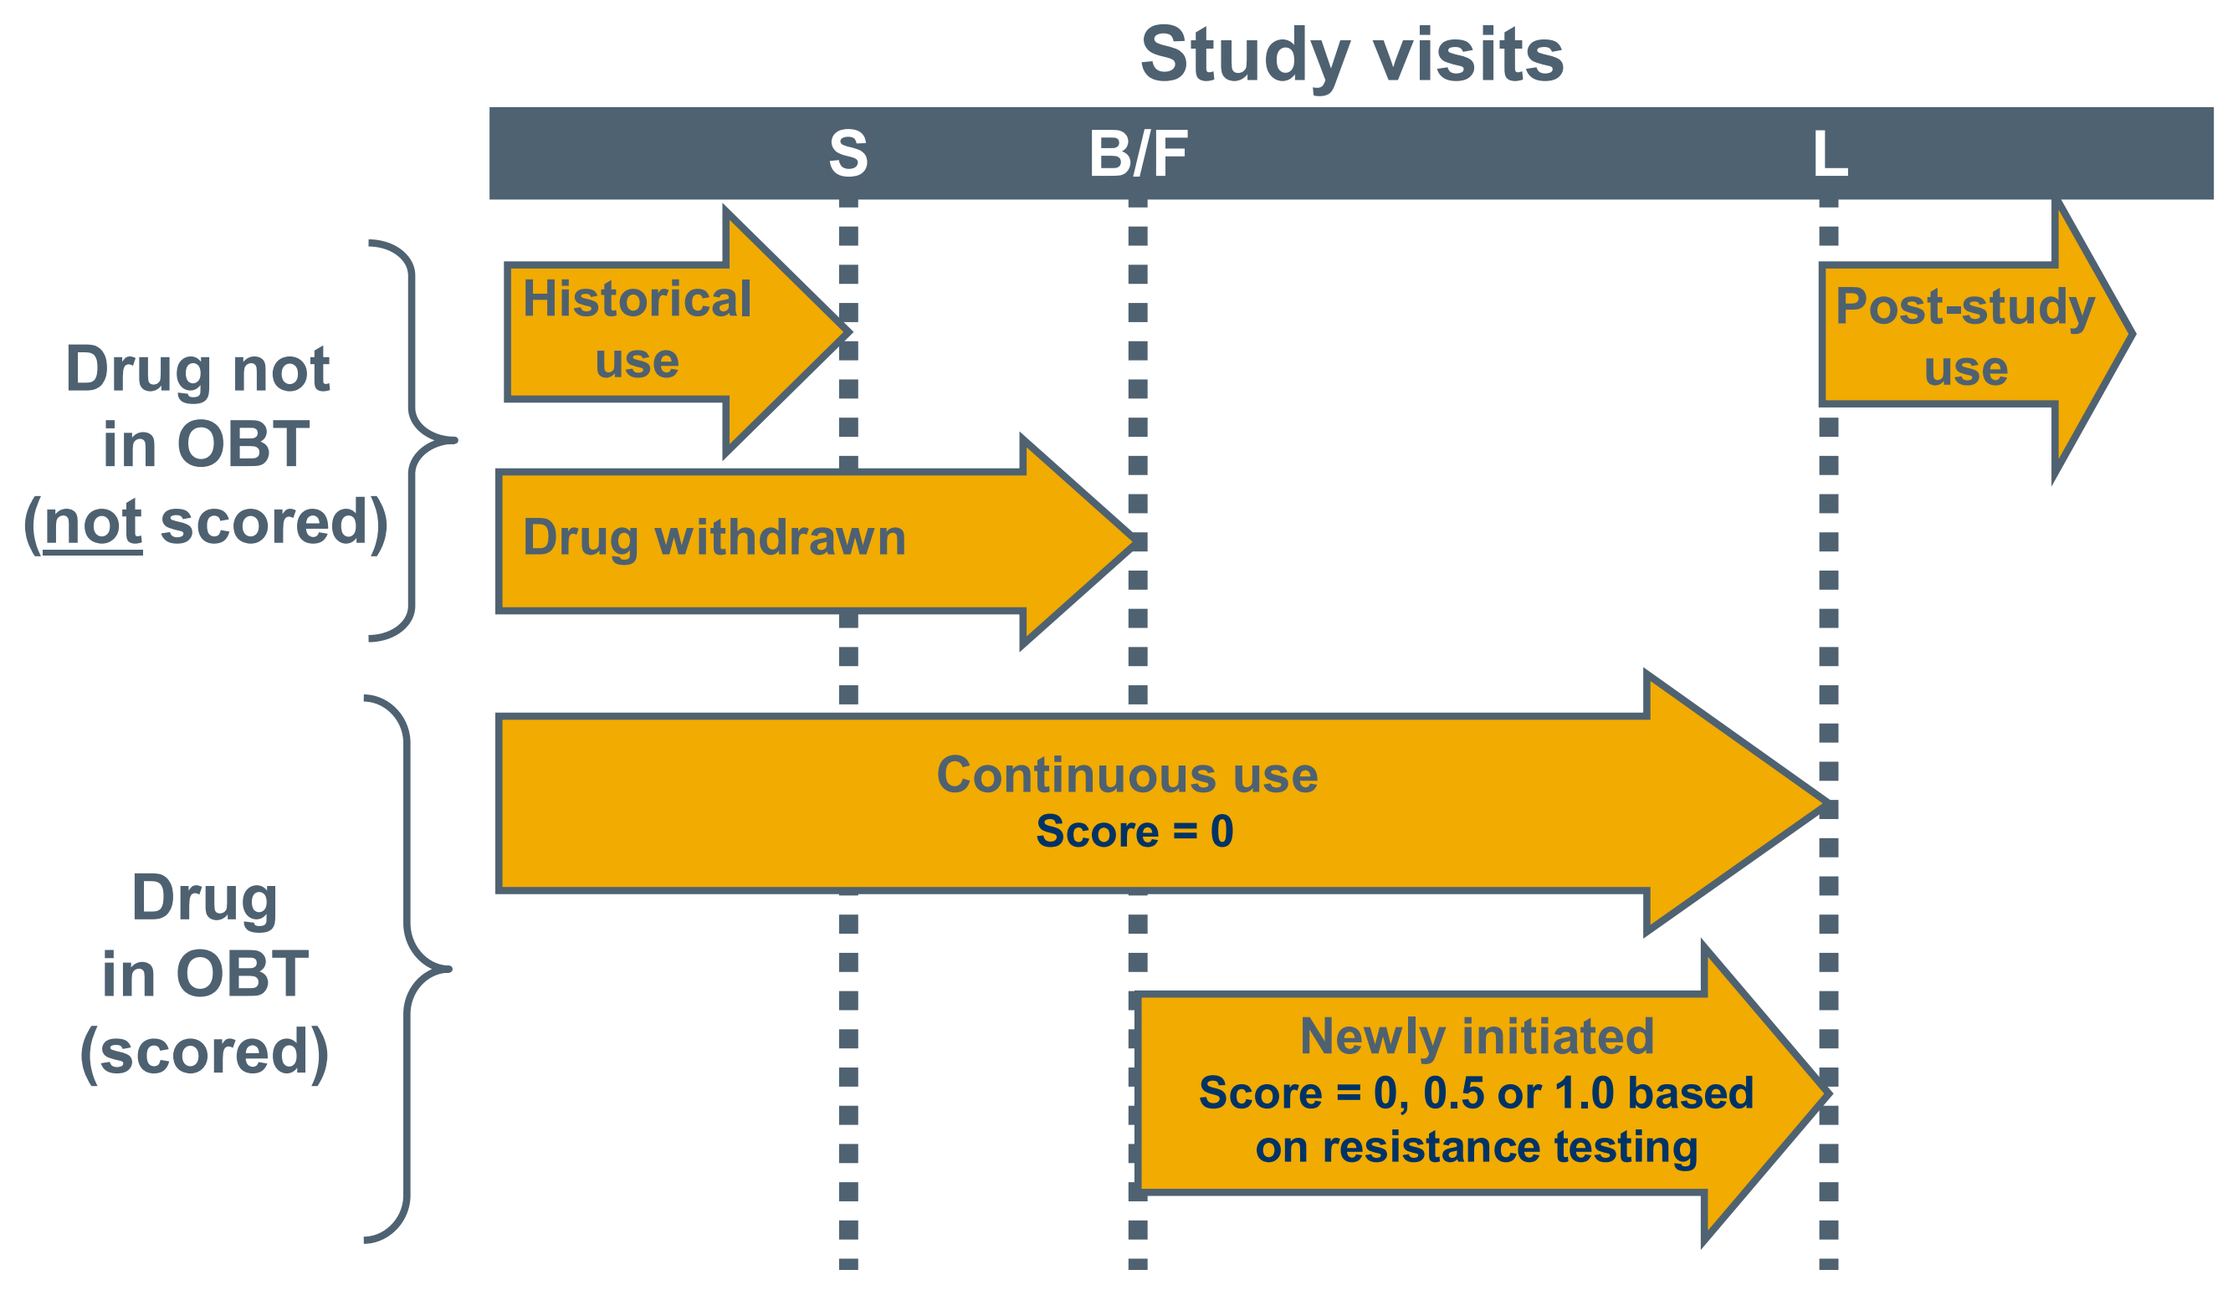
Study visits
S
B/F
L
Drug not in OBT (not scored)
Historical use
Drug withdrawn
Post-study use
Drug in OBT (scored)
Continuous use
Score = 0
Newly initiated
Score = 0, 0.5 or 1.0 based on resistance testing

S, screening visit; **B**, baseline visit; **F**, first randomized therapy date (maximum 7 days post baseline); **L**, last randomized therapy date (Week 48 visit or virologic failure)

Definitions

- Virologic response was determined by the TLOVR (time to loss of virologic response) <50 algorithm, which defined responders as those patients who achieved and maintained <50 copies/mL through Week 48.
- Three patterns of virologic failure are defined by the algorithm:
 - **Non-responder (NR)**: Never achieved <50 copies/mL and discontinued before Week 48 for lack of efficacy.
 - **Never suppressed (NS)**: Never achieved <50 copies/mL but still on study at Week 48.
 - **Rebound (R)**: Two consecutive viral load measurements ≥50 copies/mL after a confirmed <50 copies/mL result.
- The last active therapy date on study was the Week 48 visit for responders and those who never suppressed, and the date of study discontinuation for rebounders and non-responders.
- The wOBTSS algorithm used was:
 - Drugs in continuous use pre-screening to last active therapy date were scored as 0, since drugs already present in a failing baseline (pre-MOTIVATE) regimen do not contribute to post-baseline responses.
 - Viral susceptibility or resistance was defined by phenotype only (PhenoSense™ HIV assay, Monogram Biosciences) or by genotype for enfuvirtide (BC Centre for Excellence in HIV/AIDS, Vancouver, Canada).
 - Active NRTIs score 0.5 each, other active agents score 1.0. This is based on data showing active nucleosides contributing only ~50% of the virologic activity of drugs from other classes in treatment-experienced patients.[11]
- Phenotypic MVC resistance was assessed in baseline and on-treatment samples at Week 48 or time of virologic failure, as appropriate, by the PhenoSense Entry™ assay (Monogram Biosciences).
- The presence of reduced susceptibility to maraviroc among R5 failure samples was assessed by wOBTSS and by pattern of virologic failure. The analysis was descriptive and no formal statistical testing was performed.

Note: Numbers quoted in these analyses may differ slightly from those in the printed abstract due to a subsequent update to the wOBTSS censoring algorithm.

Results

- Virologic outcomes are shown in Figure 3a for the full MVC-treated set of 518 patients, classified by wOBTSS.
- Patients who were either TLOVR<50 responders or who experienced virologic failure with R5 HIV-1 and had paired baseline and on-therapy MVC susceptibility data were then studied further (n=331).
- Of these, 62/331 (18.7%) patients experienced virologic failure with R5 HIV-1, of whom 22 had virus with evidence of phenotypic resistance to MVC (Figure 3b).
- In the analysis of the 331 patients, the percentage of responders increased with higher wOBTSS values; 88% of MVC patients with a wOBTSS ≥2 were responders, compared to 63% of MVC patients with a wOBTSS <1 (i.e. patients receiving functional monotherapy or one active NRTI) (Figure 4).
- The percentage of patients failing with MVC-resistant R5 virus was highest in the subgroup of patients with a wOBTSS <1, while no patient with a wOBTSS ≥2 failed with MVC-resistant R5 virus (Figure 4).
 - 16 patients failed with MVC-resistant vs 14 patients with MVC-susceptible R5 virus in the functional monotherapy or one active NRTI subgroup (wOBTSS<1).

Figure 3. Treatment outcomes at Week 48 a) according to OBT regimen activity for all MVC-treated patients (n=518), and b) for TLOVR<50 responders or R5 virologic failures

a)

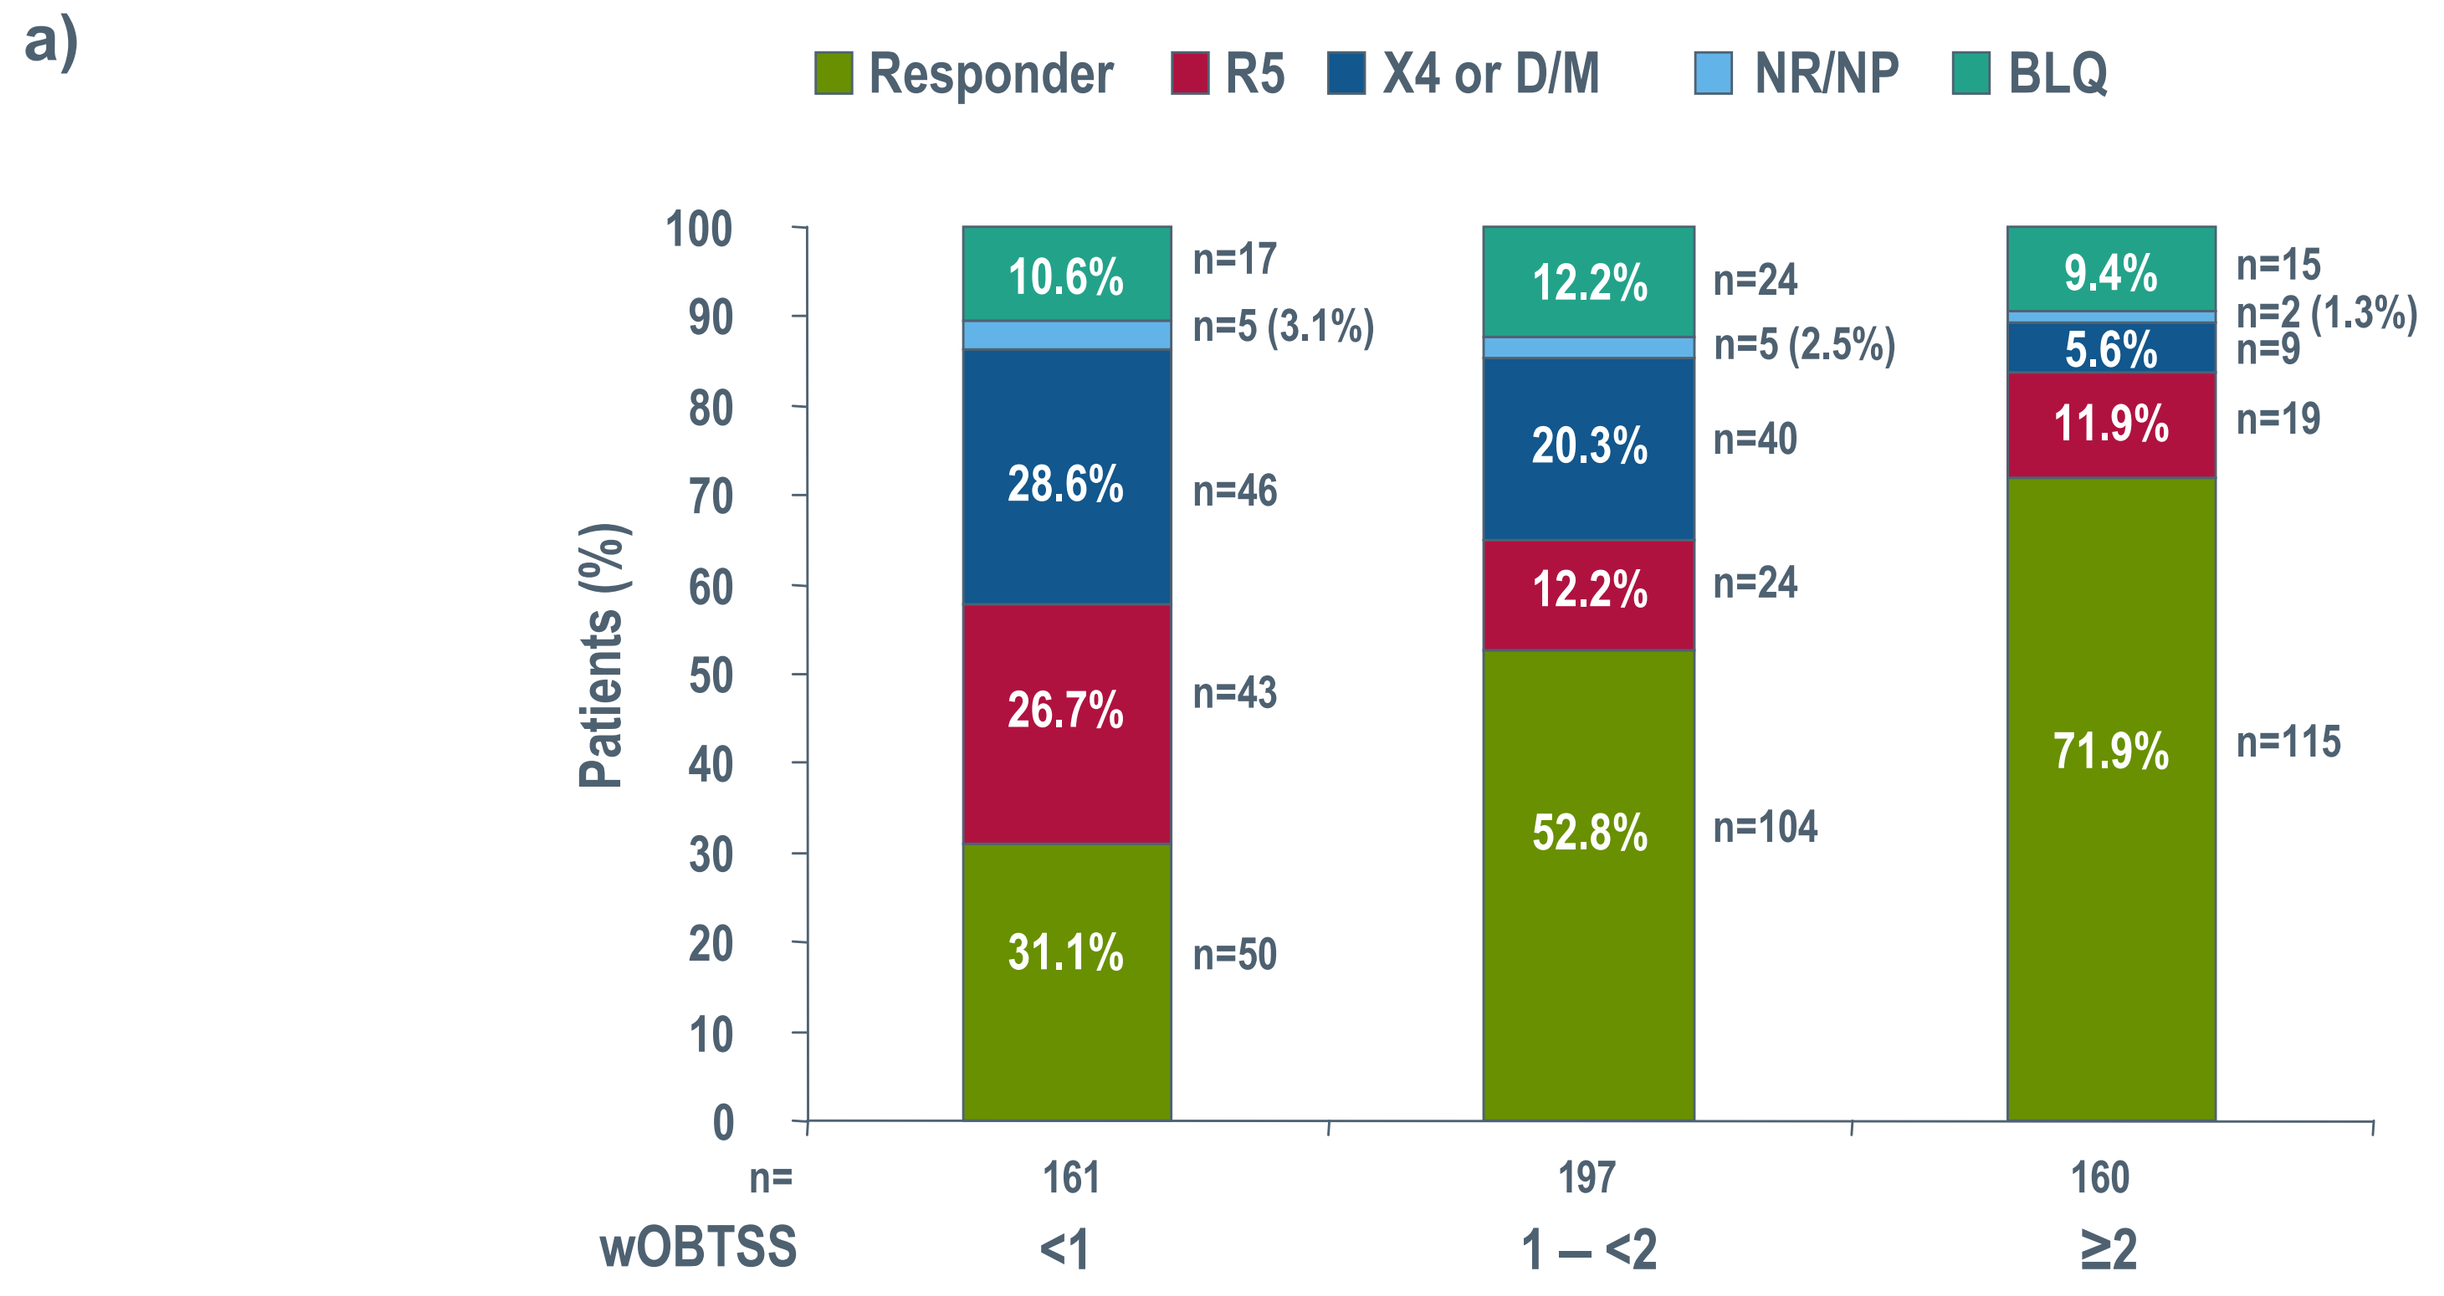
Responder
R5
X4 or D/M
NR/NP
BLQ
Patients (%)
10.6% n=17
n=5 (3.1%)
28.6% n=46
26.7% n=43
31.1% n=50
12.2% n=24
n=5 (2.5%)
20.3% n=40
12.2% n=24
52.8% n=104
9.4% n=15
n=2 (1.3%)
5.6% n=9
11.9% n=19
71.9% n=115
n= 161 197 160
wOBTSS <1 1 – <2 ≥2

NR/NP = Non-reportable/non-phenotypeable
BLQ = below limit of quantitation (<500 HIV-1 RNA copies/mL)

b)

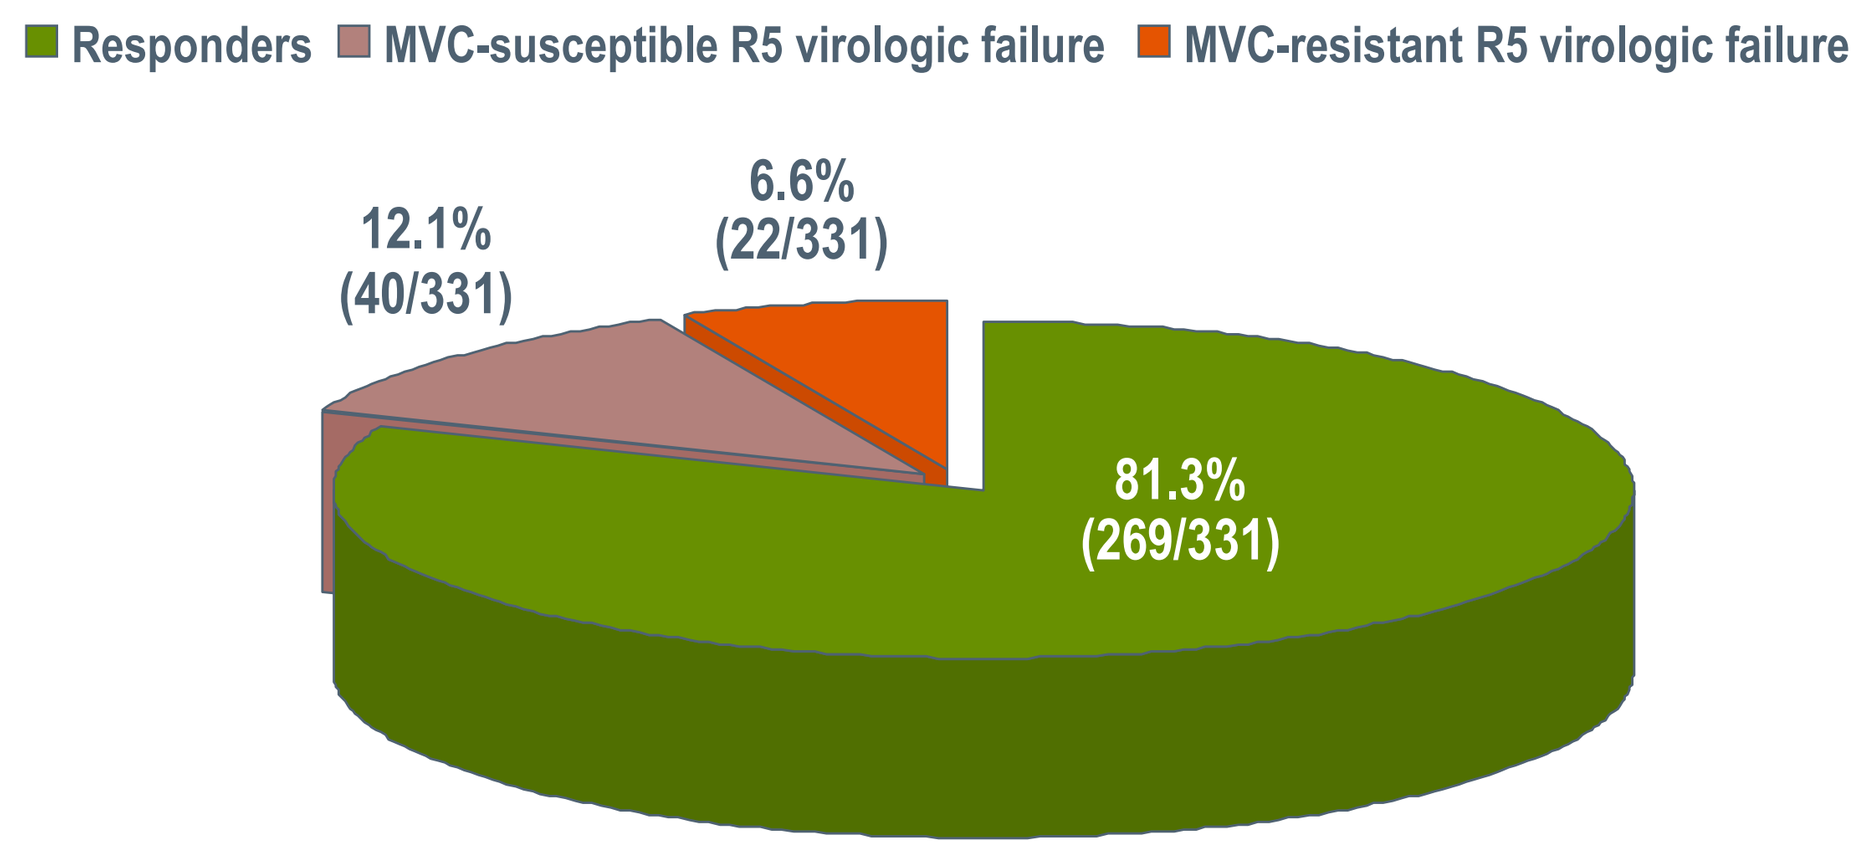
Responders
MVC-susceptible R5 virologic failure
MVC-resistant R5 virologic failure
12.1% (40/331)
6.6% (22/331)
81.3% (269/331)

Figure 4. Treatment outcomes (TLOVR<50) at Week 48 according to OBT regimen activity

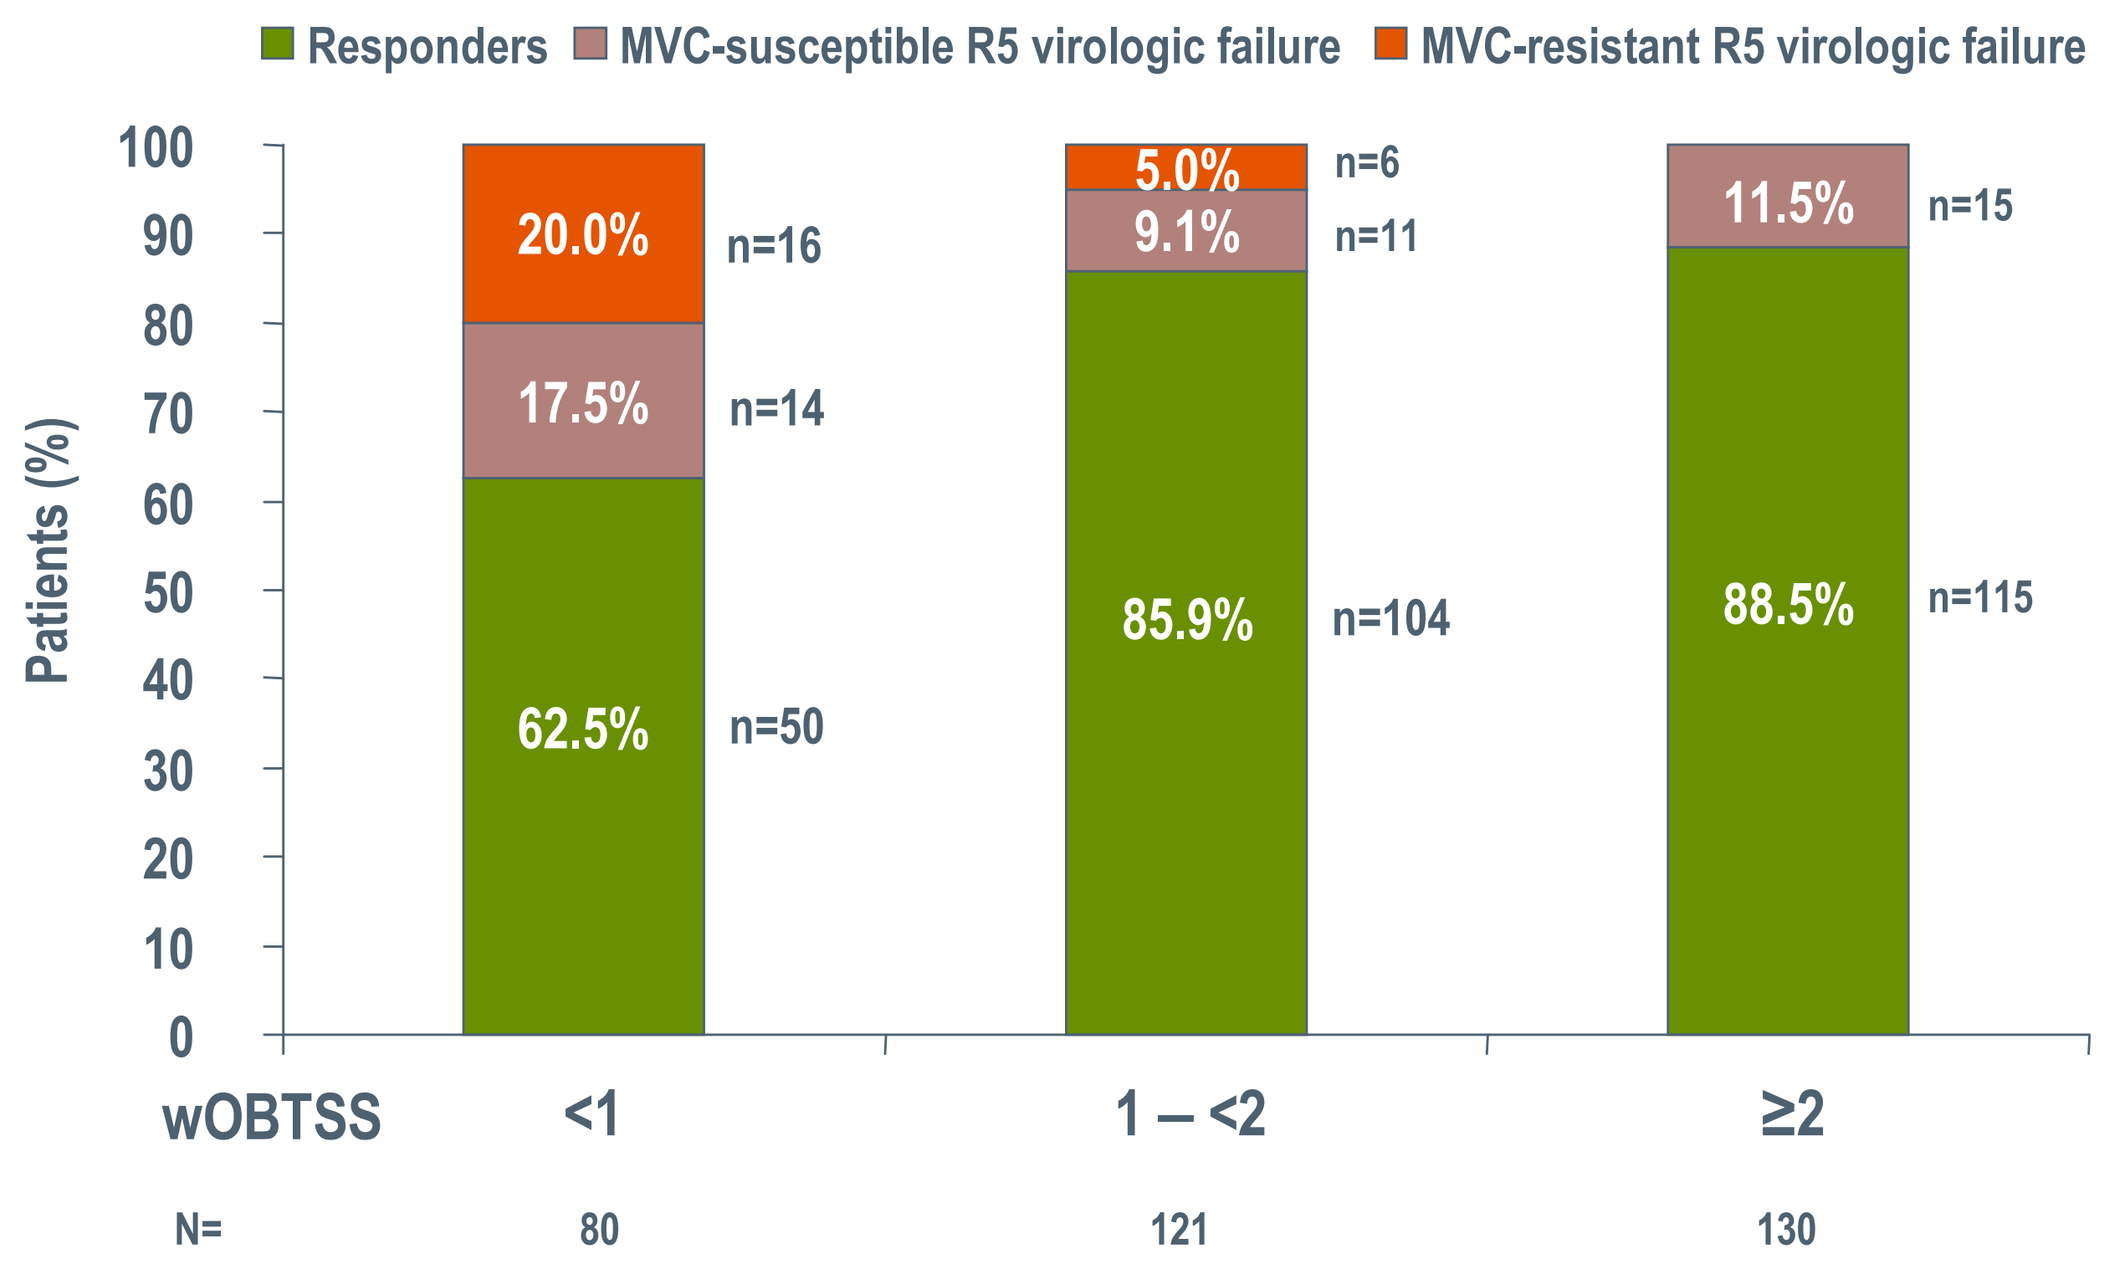
Responders
MVC-susceptible R5 virologic failure
MVC-resistant R5 virologic failure
Patients (%)
20.0% n=16
17.5% n=14
62.5% n=50
5.0% n=6
9.1% n=11
85.9% n=104
11.5% n=15
88.5% n=115
wOBTSS <1 1 – <2 ≥2
N= 80 121 130

- Of 22 MVC-resistant R5 virologic failures by the TLOVR<50 algorithm, 16/22 (73%) were on functional maraviroc monotherapy or receiving a single active nucleoside in the OBT (wOBTSS <1) with the remaining 6/22 (27%) occurring in patients with an intermediate wOBTSS of 1 or 1.5 (Figure 5a).
- By individual TLOVR<50 failure pattern, 15/34 (44%) non-responders (never <50 copies/mL and discontinued) and 7/20 (35%) patients who were never suppressed on treatment but were still on therapy at Week 48 had evidence of MVC resistance (Figure 5b). All eight patients who experienced virologic rebound following suppression to <50 RNA copies/mL had virus that was susceptible to MVC.
- Only in the subgroup of patients who had a wOBTSS <1 and were non-responders was R5 virologic failure predominantly associated with MVC resistance (11 patients with MVC-resistant R5 virus vs 6 patients with MVC-susceptible R5 virus) (Figure 6).

Figure 5. a) Percentage of MVC-resistant R5 virologic failures through Week 48 according to OBT regimen activity, and b) Proportions of MVC-susceptible and MVC-resistant R5 virologic failures by TLOVR<50 failure class

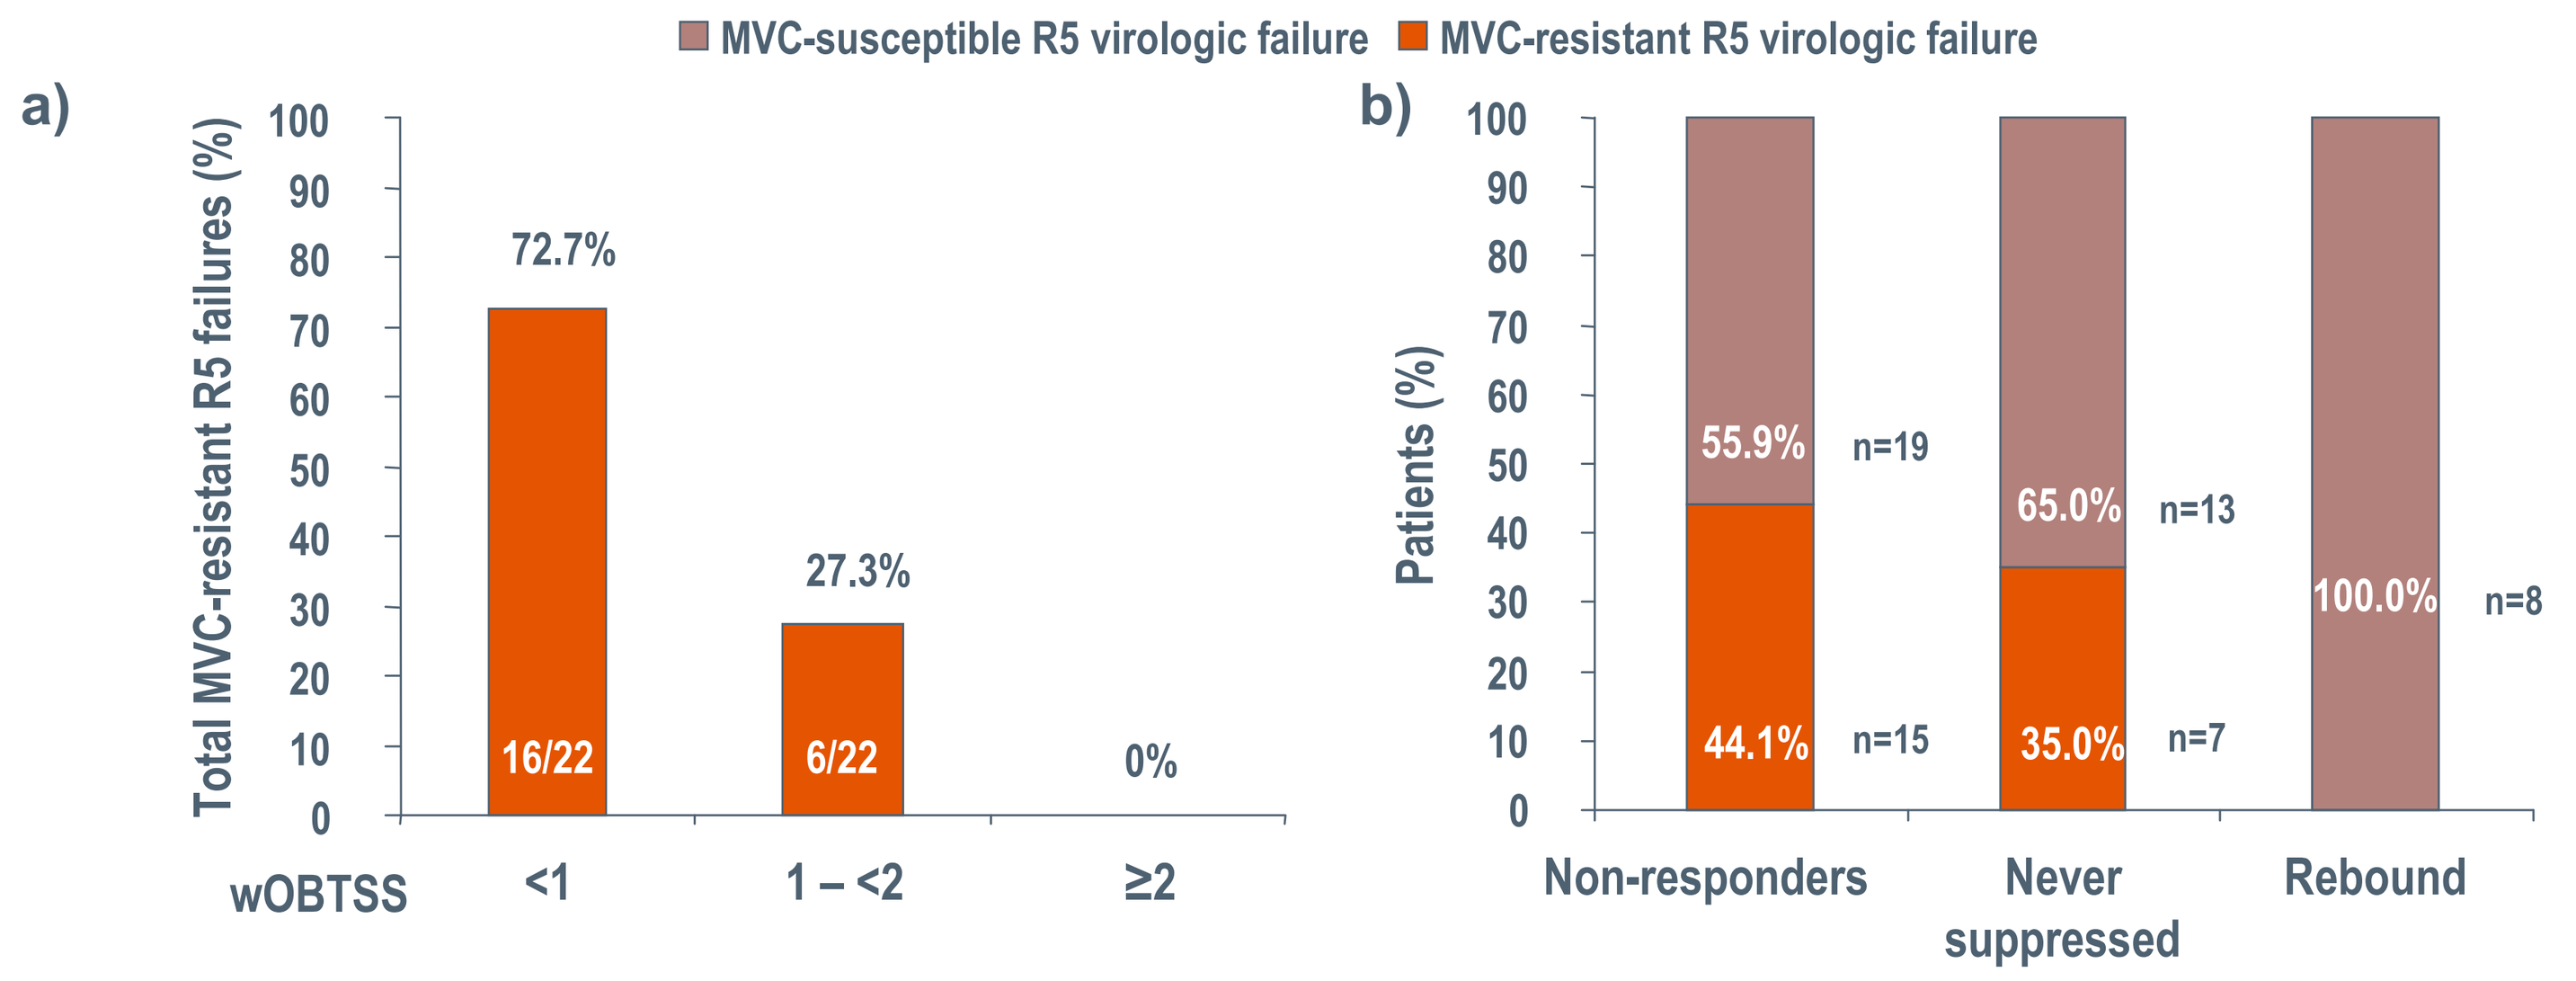
MVC-susceptible R5 virologic failure
MVC-resistant R5 virologic failure
a)
Total MVC-resistant R5 failures (%)
72.7%
16/22
27.3%
6/22
0%
wOBTSS <1 1 – <2 ≥2
b)
Patients (%)
55.9% n=19
44.1% n=15
65.0% n=13
35.0% n=7
100.0% n=8
Non-responders
Never suppressed
Rebound

Figure 6. Influence of OBT regimen activity on MVC resistance emergence in patients with R5 virologic failure

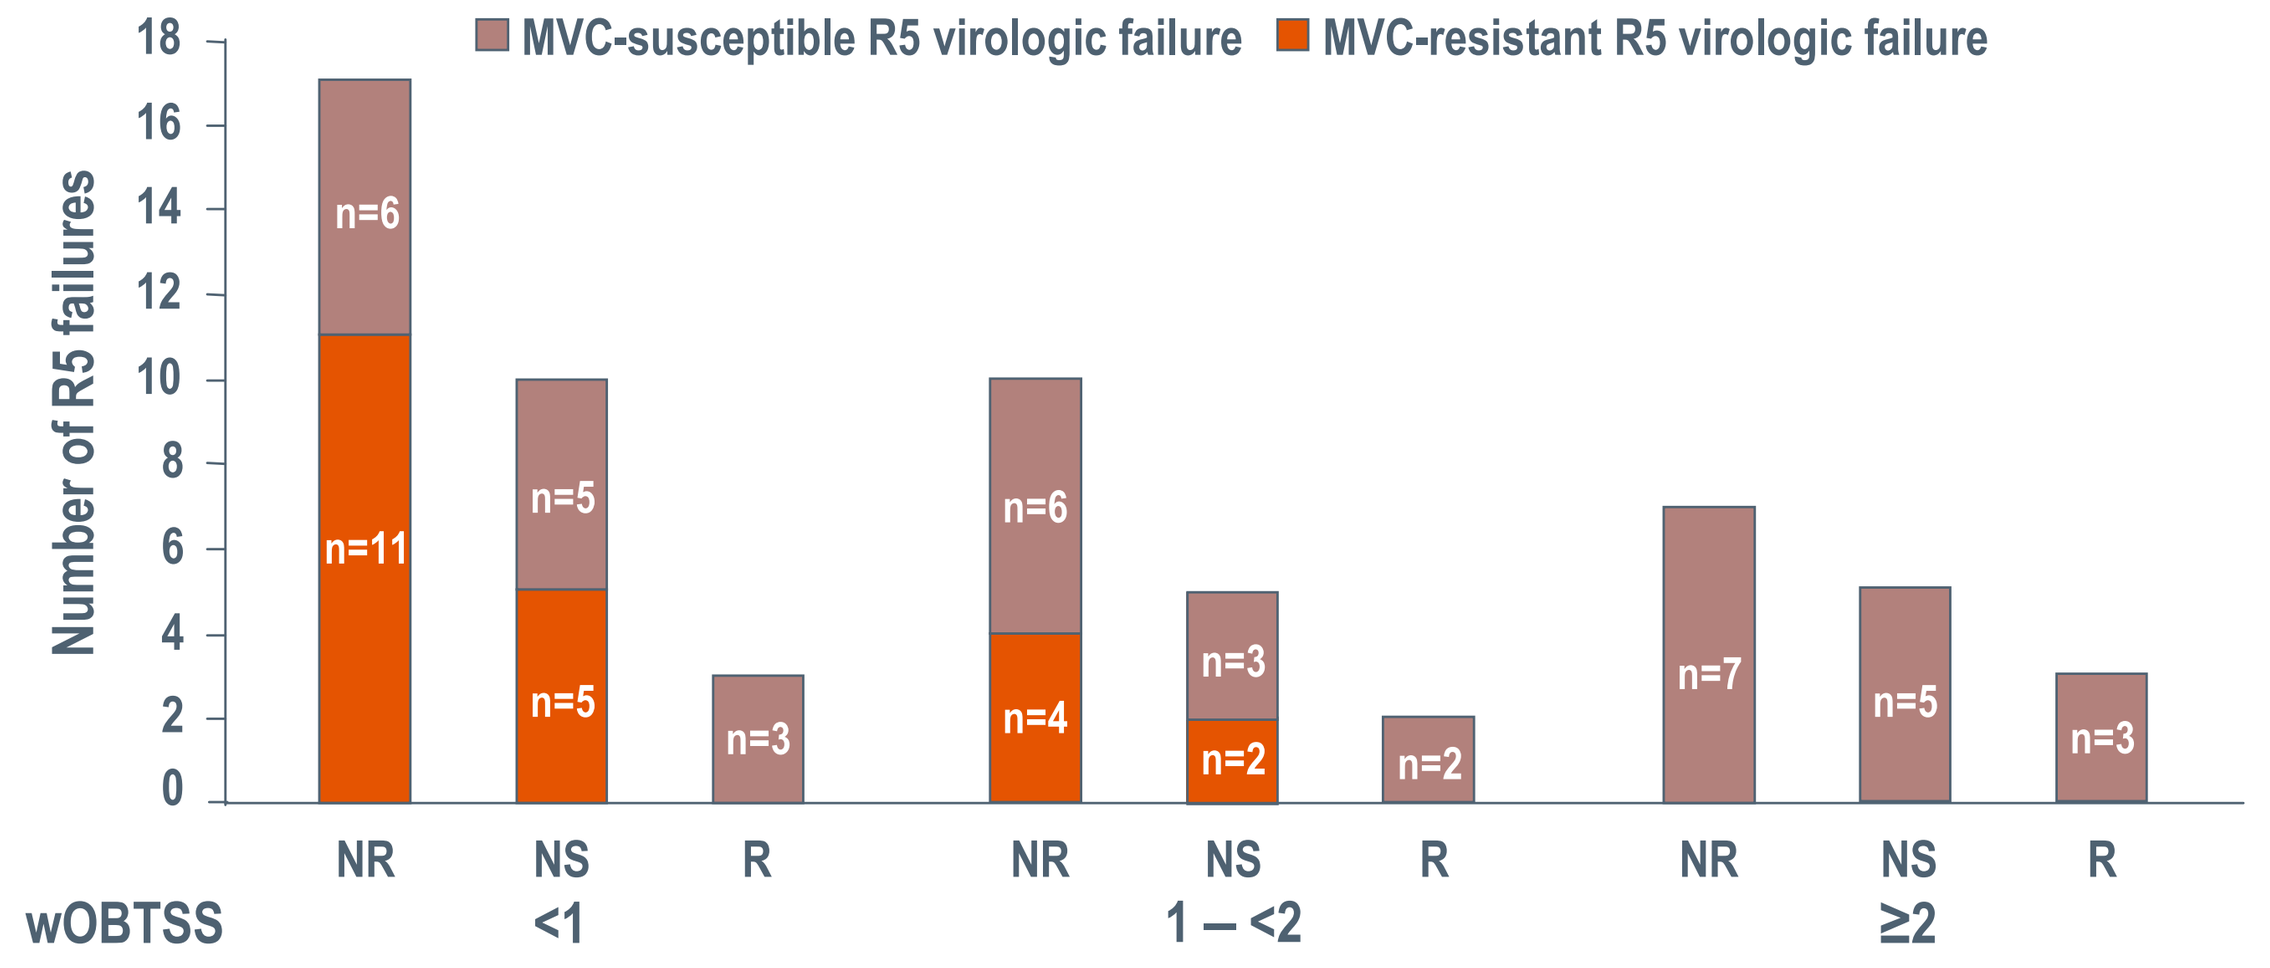
MVC-susceptible R5 virologic failure
MVC-resistant R5 virologic failure
Number of R5 failures
n=6
n=11
n=5
n=5
n=3
n=6
n=4
n=3
n=2
n=2
n=7
n=5
n=3
NR NS R NR NS R NR NS R
wOBTSS <1 1 – <2 ≥2

NR = Non-responders; NS = Never suppressed to <50 copies/mL; R = Rebound after suppression to <50 copies/mL

Conclusions

- 115/160 (72%) of patients receiving MVC with a wOBTSS ≥2 were TLOVR<50 responders with a viral load <50 HIV-1 RNA copies/mL at Week 48.
- Excluding non-R5 failures and those without matched MVC susceptibility data, R5 virologic failure occurred in 62/331 (19%) patients with MVC-resistant HIV-1 seen in 22/62 (35%) of these.
- Functional monotherapy or a single active NRTI (wOBTSS<1) accounted for 16/22 (73%) MVC resistance-associated failures, and no MVC-resistant virus was found in patients with a wOBTSS of ≥2 who failed MVC treatment with an R5 tropism result.
 - 16/80 (20%) patients who received functional monotherapy or a single active NRTI experienced MVC resistance-associated R5 virologic failure, and 50/80 (63%) of these patients responded through Week 48.
- The low incidence of MVC resistance in patients failing with virologic rebound or who never achieved viral suppression is consistent with a high barrier to resistance in these patients. Virologic failure in these patients is likely to be associated with other factors.

References

1. Gulick RM, *et al. N Engl J Med* 2008; **359**:1429–1441.
2. Hardy WD, *et al. HIV9* 2008. Presentation O425.
3. Clotet B, *et al. Lancet* 2007; **369**:1169–1178.
4. Cooper DA, *et al. N Engl J Med* 2008; **359**:355–365.
5. Fätkenheur G, *et al. N Engl J Med* 2008; **359**:1442–1455.
6. Valdez H, *et al. 48th ICAAC/46th IDSA* 2008. Poster H-1221.
7. Lewis M, *et al. XVI Int HIV Drug Res Wkshp* 2007. Abstract 56.
8. Westby M, *et al. J Virol* 2007; **81**:2359–2371.
9. Mori J, *et al. XVI Int HIV Drug Res Wkshp* 2007; Abstract 10.
10. Lewis M, *et al. 15th CROI* 2008; Poster 871.
11. Swanstrom R, *et al. J Infect Dis* 2004; **190**:886-893.

Acknowledgements

- Investigators and study site staff
- Patients who participated in the trial
- Colleagues from Pfizer: Hernan Valdez, Christophe Delogne, Douglass Chapman, Elna van der Ryst, Julie Mori and Lynn McFadyen

Pfizer HIV/AIDS