2009 CROI
Poster #573

Subgroup Analyses from STARTMRK, a Phase III Study of Raltegravir (RAL)-Based vs Efavirenz (EFV)-Based Combination Therapy in Treatment-Naïve HIV-Infected Patients

Jeffrey Lennox[1], Edwin DeJesus[2], Adriano Lazzarin[3], Richard Pollard[4], Jose Valdez Ramalho Madruga[5], Jing Zhao[6], Anthony Rodgers[6], Bach-Yen Nguyen[6], Randi Leavitt[6], and Peter Sklar[6]

[1]Emory University Center for AIDS Research, Atlanta, GA; [2]Orlando Immunology Center, Orlando, FL; [3]Univ Vita-Salute San Raffaele, Milan, Italy; [4]University of California at Davis, Sacramento, CA; [5]Centro de Referencia Treinamento DST/AIDS, Sao Paulo, Brazil; [6]Merck & Co., Inc., West Point, PA

Dr. Peter Sklar
Merck Research Laboratories
PO Box 1000, UG3D-56
North Wales, PA 19454-1099, USA
Tel 267-305-1204
Fax 267-305-6477
peter_sklar@merck.com

ABSTRACT

Background: In treatment-naive patients given 48 weeks (wk) of therapy, RAL+tenofovir (TDF)/emtricitabine (FTC) had non-inferior antiretroviral activity compared to EFV+TDF/FTC and was associated with greater increases in CD4 cell counts.

Methods: Patients with HIV RNA levels >5000 c/mL and no resistance to EFV, TDF or FTC were eligible for a blinded randomized study of RAL (400 mg bid) vs EFV (600 mg qhs), each with TDF/FTC. Stratification was by screening RNA (≤/> 50,000 c/mL) and hepatitis status. Results for the primary and secondary end-points (% of patients achieving RNA <50 c/mL and <400 c/mL at Wk 48, and change from baseline in CD4 cell count at Wk 48) were summarized within several prespecified subgroups in order to further characterize the effect of RAL. Summary statistics [along with 95% confidence intervals (CI)] were provided by treatment group within each subgroup using the observed failure (OF) approach.

Results: Of 563 treated patients, 19% were female, 58% were non-white, and region of residence was Latin America (35%), North America (31%), EU/Australia (23%) and Asia (11%). Median age was 37 years. 53% had vRNA >100,000 c/mL, 48% had CD4 counts <200 cells/mm³ at entry, and 19% were infected with non-subtype B HIV. RAL and EFV had virologic efficacy that was generally consistent across baseline demographic factors. Table summarizes the proportion of patients with vRNA <50 c/mL at Wk 48 by selected baseline factors. RAL demonstrated greater CD4 cell increases than EFV at Wk 48 that was consistent across baseline demographic factors. RAL was in general well tolerated.

	Raltegravir Group		Efavirenz Group		Difference in Response Rates[1]
	n/N	% (95% CI[1])	n/N	% (95% CI[1])	% (95% CI[1])
Overall	241/263	91.6 (87.6, 94.7)	230/258	89.1 (84.7, 92.7)	2.5 (-2.6, 7.7)
Baseline Plasma HIV RNA					
≤100,000 copies/mL	111/120	92.5 (86.2, 96.5)	114/128	89.1 (82.3, 93.9)	3.4 (4.1, 11.0)
>100,000 copies/mL	130/143	90.9 (85.0, 95.1)	116/130	89.2 (82.6, 94.0)	1.7 (-5.6, 9.2)
Baseline CD4 Cell Counts					
≤50 cells/mm³	21/25	84.0 (63.9, 95.5)	24/28	85.7 (67.3, 96.0)	-1.7 (-23.0, 18.7)
>50 to ≤200 cells/mm³	85/95	89.5 (81.5, 94.8)	83/97	85.6 (77.0, 91.9)	3.9 (-5.7, 13.7)
>200 cells/mm³	135/143	94.4 (89.3, 97.6)	122/132	92.4 (86.5, 96.3)	2.0 (-4.1, 8.5)
missing	--	--	1/1	100.0 (2.5, 100.0)	--
Viral Subtype					
Clade B	186/206	90.3 (85.4, 94.0)	185/209	88.5 (83.4, 92.5)	1.8 (-4.3, 7.9)
Non-Clade B	52/54	96.3 (87.3, 99.5)	40/44	90.0 (78.3, 97.5)	5.4 (-4.9, 18.0)
missing	3/3	100.0 (29.2, 100.0)	5/5	100.0 (47.8, 100.0)	0.0 (-59.4, 46.8)

[1]Difference was calculated as the response rate in the raltegravir group minus the response rate in the efavirenz group.
N = Number of patients in each treatment group; n = number of patients in each subcategory.

Conclusion: RAL demonstrated consistent virologic and immunologic efficacy across demographic and all baseline prognostic factors, including baseline plasma vRNA level >100,000 c/mL, baseline CD4 count ≤50 cells/mm³, gender/racial groups, and viral subtypes.

STUDY DESIGN

- Double-blind, randomized (1:1), non-inferiority study
- RAL 400 mg b.i.d. vs EFV 600 mg q.h.s. both in combination with tenofovir/emtricitabine (TDF/FTC as TRUVADA™)
- Key inclusion criteria
 - no prior ART
 - HIV RNA level >5000 copies/mL
 - viral susceptibility to EFV, TDF, and FTC
- Endpoints
 - Efficacy: Proportion with HIV RNA levels < 50 copies/mL, change in CD4 cell counts
 - Safety/tolerability: adverse experiences; central nervous system (CNS) events; lipid changes from baseline

Hypotheses and Statistical Methods:
In treatment-naïve, HIV-infected patients:
- RAL + TDF/FTC will have non-inferior (12% bound) virologic efficacy compared to EFV + TDF/FTC
 - Proportion of patients with HIV RNA <50 copies/mL at Week 48 [non-completer = failure approach (NC=F)]
- RAL + TDF/FTC will have similar immunologic efficacy compared to EFV + TDF/FTC
 - Change from baseline in CD4 count at Week 48 [observed failure approach (OF)]
- RAL + TDF/FTC will be generally safe and well tolerated
 - RAL + TDF/FTC will be superior to EFV + TDF/FTC in terms of CNS adverse events up to Week 8

STUDY DESIGN (CONTINUED)

Objective of Subgroup Analyses:
- To assess the virologic and immunologic effects of RAL in subgroups based on demographic and prognostic factors at entry
 - viral load
 - CD4 cell count
 - hepatitis B or C co-infection
 - age, gender, race, region
 - viral subtype (clade B or non-B clades)

Statistical Approaches to Subgroup Analyses:
- Missing data handled using an observed-failure approach that purely assesses virologic effect
 - Discontinuations due to lack of efficacy considered as a failure thereafter
 - Baseline carried forward for failures as calculating change from baseline of CD4 counts

BASELINE CHARACTERISTICS

	Raltegravir Group (N = 281)	Efavirenz Group (N = 282)	All Treated Patients (N = 563)
Gender, n (%)			
Male	227 (81)	231 (82)	458 (81)
Female	54 (19)	51 (18)	105 (19)
Race/Ethnicity, n (%)			
White	116 (41)	123 (44)	239 (42)
Black	33 (12)	23 (8)	56 (10)
Asian	36 (13)	32 (11)	68 (12)
Hispanic	60 (21)	67 (24)	127 (23)
Native American	1 (0)	1 (0)	2 (0)
Multiracial	35 (12)	36 (13)	71 (13)
Region, n (%)			
Latin America	99 (35)	97 (34)	196 (35)
Southeast Asia	34 (12)	29 (10)	63 (11)
North America	82 (29)	90 (32)	172 (31)
Europe/Australia	66 (23)	66 (23)	132 (23)
Age, in years			
Mean (SD)	38 (9)	37 (10)	37 (9)
Median (range)	37 (19 - 67)	36 (19 - 71)	37 (19 - 71)
CD4 Cell Count*, cells/mm³			
Mean (SD)	219 (124)	217 (134)	218 (129)
Median (range)	212 (1 - 620)	204 (4 - 807)	208 (1 - 807)
AIDS History, n (%)			
Yes	40 (14)	42 (15)	82 (15)

	Raltegravir Group (N = 281)	Efavirenz Group (N = 282)	All Treated Patients (N = 563)
Plasma HIV RNA, log_{10} copies/mL			
Mean (SD)	5 (1)	5 (1)	5 (1)
Median (range)	5 (3 to 6)	5 (4 to 6)	5 (3 to 6)
Stratum, n (%)			
Screening vRNA level ≤50,000 copies/mL	74 (26)	80 (28)	154 (27)
Hepatitis B or C**	20 (7)	19 (7)	39 (7)
Viral Subtype, n (%)			
Clade B	219 (78)	230 (82)	449 (80)
Non-Clade B†	59 (21)	47 (17)	106 (19)
Missing	3 (1)	5 (2)	8 (1)
Baseline Plasma HIV RNA, n (%)			
≤50,000 copies/mL	79 (28)	84 (30)	163 (29)
>50,000 copies/mL	202 (72)	198 (70)	400 (71)
≤100,000 copies/mL	127 (45)	139 (49)	266 (47)
>100,000 copies/mL	154 (55)	143 (51)	297 (53)
Baseline CD4 Cell Counts, n (%)			
≤50 cells/mm³	27 (10)	31 (11)	58 (10)
>50 to ≤200 cells/mm³	104 (37)	105 (37)	209 (37)
>200 cells/mm³	150 (53)	145 (51)	295 (52)
Missing	0 (0)	1 (0)	1 (0)

*One patient in the efavirenz group with missing results was excluded.
**Hepatitis B surface antigen positivity or detectable hepatitis C RNA.
†Non-clade B subtypes (number of patients): clade A (4), A/C (1), A/G (2), A1(1), AE (29), AG (12), BF (6), C (37), D (2), F (2), F1 (5), G (2), and Complex (3).
N = Number of patients in each group; n (%) = number (percent) of patients in each category.

OVERALL STUDY RESULTS

(2008 ICAAC-IDSA Abstract H-896a):
- RAL provided potent and statistically non-inferior viral suppression compared to EFV
- RAL exerted a greater immunological effect than EFV, measured by the increase CD4 cell counts

Percent of Patients with HIV RNA <50 Copies/mL (NC = F[1])

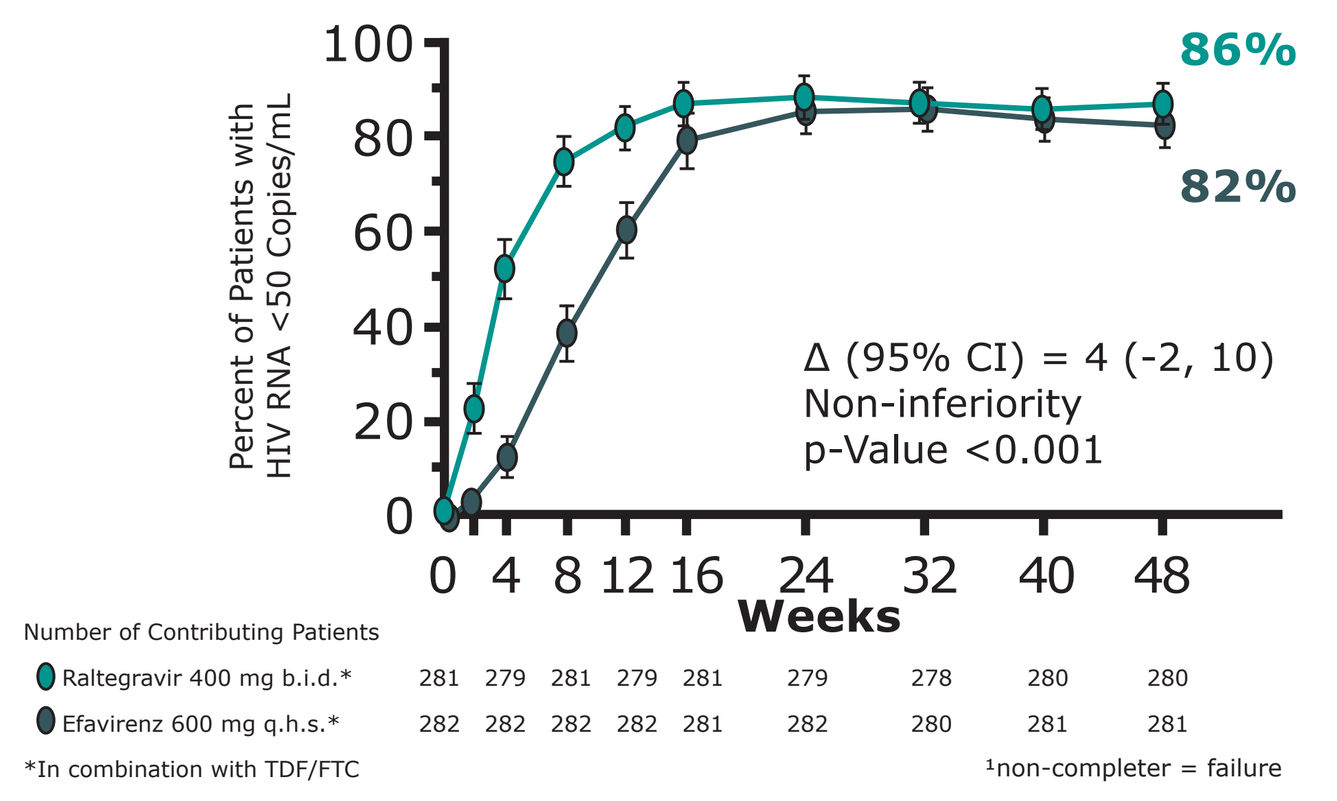

100
80
60
40
20
0
Percent of Patients with HIV RNA <50 Copies/mL
86%
82%
Δ (95% CI) = 4 (-2, 10)
Non-inferiority
p-Value <0.001
0 4 8 12 16 24 32 40 48
Weeks
Number of Contributing Patients
Raltegravir 400 mg b.i.d.* 281 279 281 279 281 279 278 280 280
Efavirenz 600 mg q.h.s.* 282 282 282 282 281 282 280 281 281
*In combination with TDF/FTC
[1]non-completer = failure

Mean Change from Baseline CD4 Cell Count (cells/mm³) (OF[2])

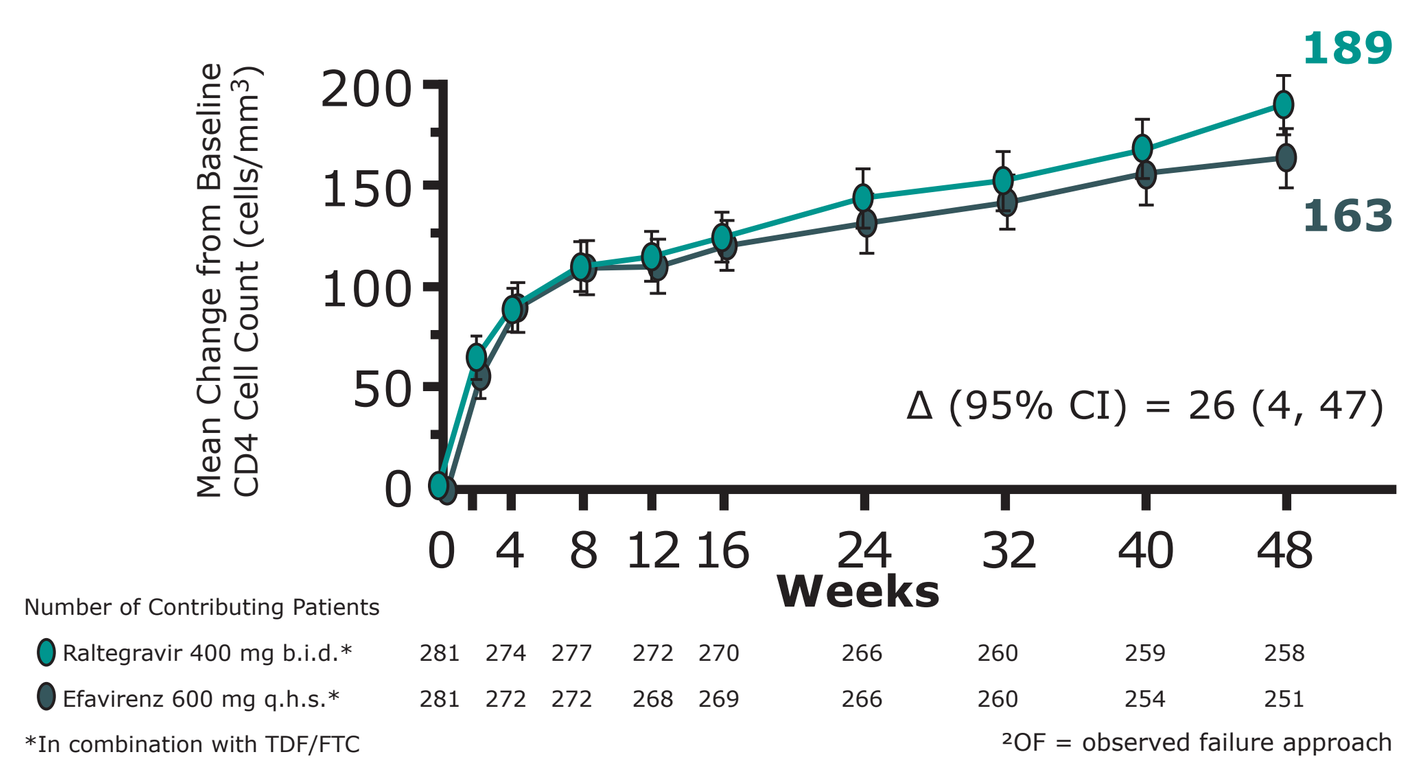

200
150
100
50
0
Mean Change from Baseline CD4 Cell Count (cells/mm³)
189
163
Δ (95% CI) = 26 (4, 47)
0 4 8 12 16 24 32 40 48
Weeks
Number of Contributing Patients
Raltegravir 400 mg b.i.d.* 281 274 277 272 270 266 260 259 258
Efavirenz 600 mg q.h.s.* 281 272 272 268 269 266 260 254 251
*In combination with TDF/FTC
[2]OF = observed failure approach

- RAL was generally better tolerated than EFV
 - Significantly fewer overall and drug-related clinical adverse events
 - Significantly lower percentages of patients with CNS side-effects
 - Safety profile was similar in subjects with and without hepatitis B and/or hepatitis C virus co-infection

FOREST PLOTS SHOWING THE VIROLOGIC AND IMMUNOLOGIC EFFICACY OF RALTEGRAVIR RELATIVE TO EFAVIRENZ AT WEEK 48 BY BASELINE FACTORS (OBSERVED-FAILURE APPROACH)

Subgroup		Raltegravir n/N (%)	Efavirenz n/N (%)
		% of Patients with HIV RNA Levels <50 Copies/mL	
Overall		241/263 (92)	230/258 (89)
Baseline Plasma HIV RNA	≤50,000 copies/mL	71/75 (95)	68/78 (87)
	>50,000 copies/mL	170/188 (90)	162/180 (90)
	≤100,000 copies/mL	111/120 (93)	114/128 (89)
	>100,000 copies/mL	130/143 (91)	116/130 (89)
Screening Plasma HIV RNA	≤50,000 copies/mL	66/70 (94)	61/70 (87)
	>50,000 copies/mL	175/193 (91)	169/188 (90)
Baseline CD4 Cells	≤50 cells/mm³	21/25 (84)	24/28 (86)
	>50 to ≤200 cells/mm³	85/95 (89)	83/97 (86)
	>200 cells/mm³	135/143 (94)	122/132 (92)
Hepatitis B/C Status	B or C positive*	16/17 (94)	14/16 (88)
	Both B and C negative	225/246 (91)	216/242 (89)
Age (years)**	≤ Median	126/137 (92)	129/145 (89)
	> Median	115/126 (91)	101/113 (89)
Gender	Male	194/214 (91)	188/213 (88)
	Female	47/49 (96)	42/45 (93)
Race/Ethnicity	White	100/109 (92)	100/107 (93)
	Black	24/27 (89)	20/22 (91)
	Asian	31/34 (91)	26/30 (87)
	Hispanic	54/58 (93)	53/62 (85)
	Multiracial	31/34 (91)	30/36 (83)
Region	Latin America	88/97 (91)	81/96 (84)
	Southeast Asia	30/32 (94)	26/28 (93)
	North America	63/70 (90)	70/76 (92)
	Europe/Australia	60/64 (94)	53/58 (91)
Viral Subtype	Clade B	186/206 (90)	185/209 (89)
	Non-Clade B	52/54 (96)	40/44 (91)

Difference in Response Rates

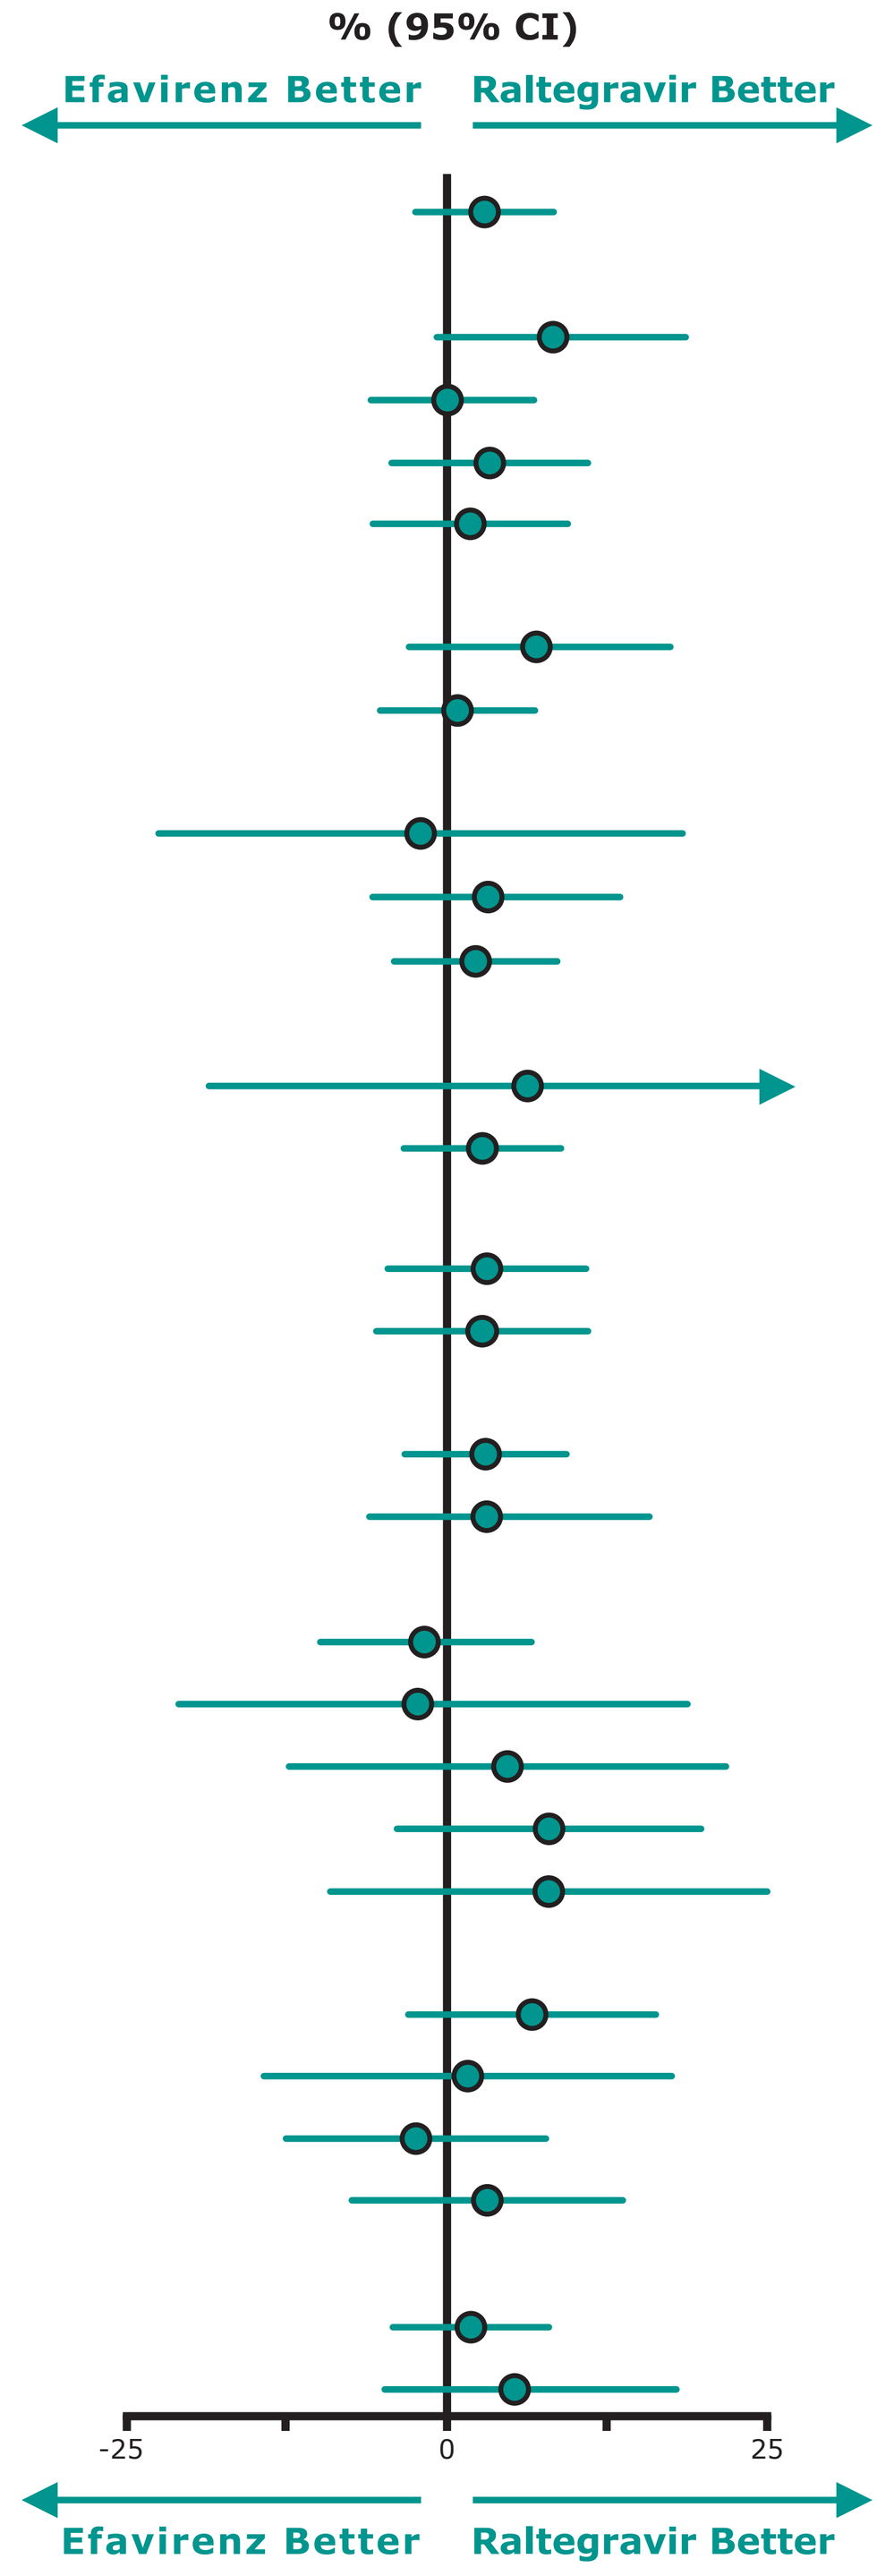

% (95% CI)
Efavirenz Better
Raltegravir Better
-25
0
25
Efavirenz Better
Raltegravir Better

*An arrow end implies the upper limit of 95% CI exceeding the maximum of x-axis
**Median age: 37 for Raltegravir group vs. 36 for Efavirenz

Subgroup		Raltegravir cells/mm³ (95% CI)	Efavirenz cells/mm³ (95% CI)
		Mean Change in CD4 Cells from Baseline	
Overall		189 (174, 204)	163 (148, 178)
Baseline Plasma HIV RNA	≤50,000 copies/mL	169 (145, 194)	142 (119, 166)
	>50,000 copies/mL	197 (178, 216)	172 (153, 191)
	≤100,000 copies/mL	180 (160, 200)	134 (115, 153)
	>100,000 copies/mL	196 (174, 219)	192 (169, 214)
Screening Plasma HIV RNA	≤50,000 copies/mL	182 (155, 209)	134 (106, 162)
	>50,000 copies/mL	192 (173, 210)	174 (156, 192)
Baseline CD4 Cells	≤50 cells/mm³	170 (122, 218)	152 (123, 180)
	>50 to ≤200 cells/mm³	193 (169, 217)	175 (151, 198)
	>200 cells/mm³	190 (168, 212)	157 (134, 181)
Hepatitis B/C Status	B or C positive*	188 (98, 279)	183 (130, 235)
	Both B and C negative	189 (174, 204)	162 (146, 178)
Age (years)**	≤ Median	192 (171, 212)	171 (149, 193)
	> Median	186 (163, 209)	154 (133, 174)
Gender	Male	193 (176, 211)	162 (146, 179)
	Female	170 (139, 201)	168 (133, 203)
Race/Ethnicity	White	195 (172, 218)	178 (154, 203)
	Black	163 (123, 203)	125 (87, 164)
	Asian	185 (148, 223)	152 (101, 204)
	Hispanic	196 (165, 228)	150 (122, 179)
	Multiracial	182 (121, 242)	168 (125, 211)
Region	Latin America	193 (165, 220)	169 (146, 192)
	Southeast Asia	185 (145, 225)	157 (105, 209)
	North America	181 (153, 208)	154 (127, 182)
	Europe/Australia	195 (163, 226)	168 (131, 205)
Viral Subtype	Clade B	187 (170, 204)	164 (147, 181)
	Non-Clade B	189 (153, 225)	156 (121, 190)

Difference in Mean Change

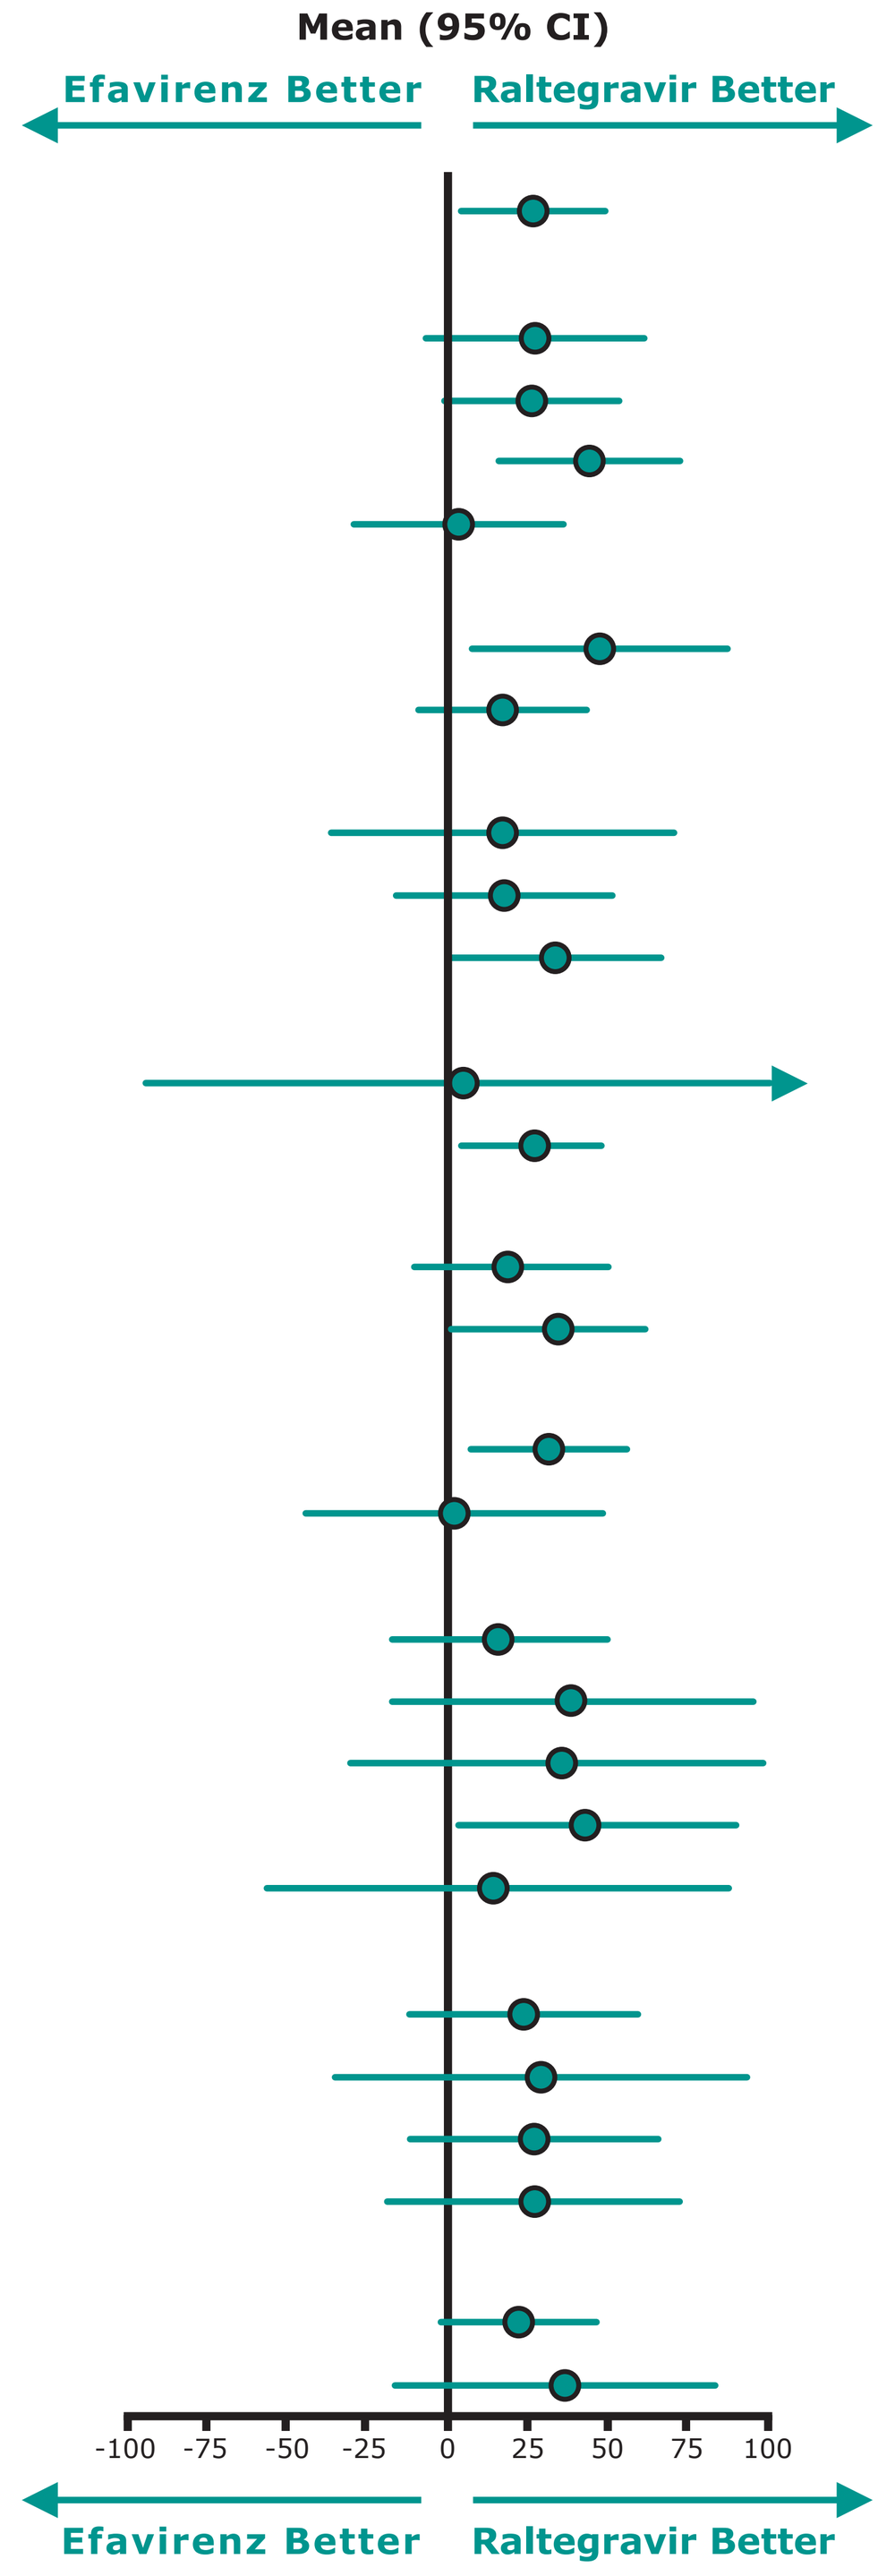

Mean (95% CI)
Efavirenz Better
Raltegravir Better
-100 -75 -50 -25 0 25 50 75 100
Efavirenz Better
Raltegravir Better

*An arrow end implies the upper limit of 95% CI exceeding the maximum of x-axis
**Median age: 37 for Raltegravir group vs. 36 for Efavirenz

CONCLUSIONS

- In the STARTMRK trial of combination antiretroviral therapy in previously untreated patients, RAL with TDF/FTC demonstrated consistent virologic and immunologic efficacy relative to EFV with TDF/FTC across demographic and baseline prognostic factors, including:
 - Baseline plasma vRNA level >100,000 copies/mL
 - ◇ Both RAL and EFV demonstrated comparable efficacy in patients with high baseline viral loads (>100,000 copies/mL) compared to patients with lower viral loads
 - Baseline CD4 count ≤50 cells/mm³
 - ◇ Response rates for both RAL and EFV recipients were numerically slightly lower in patients with low baseline CD4 cells (≤200 cells/mm³) compared to patients with higher CD4 cells
 - Demographic groups (including age, gender, region, and race)
 - Viral subtypes (comparing non-clade B as a group to clade B)

ACKNOWLEDGEMENTS: Special thanks to all patients who participated in Protocol 021. **STARTMRK Protocol 021 Study Team Investigators:** Dr. D. Berger, Dr. E. DeJesus, Dr. T. Friel, Dr. C. Hicks, Dr. M. Kozal, Dr. P. Kumar, Dr. J. Lennox, Dr. R. Liporace, Dr. S. Little, Dr. J. Morales-Ramirez, Dr. R. Novak, Dr. R. Pollard, Dr. M. Saag, Dr. S. Santiago, Dr. S. Schneider, Dr. R. Steigbigel, Dr. W. Towner, Dr. D. Wright, Dr. G. Pialoux, Dr. Y. Yazdanpanah, Dr. L. Cotte, Dr. D. Salmon Ceron, Dr. P. M. Girard, Dr. D. Cooper, Dr. C. Beltran, Dr. A. Afani, Dr. J. Perez Godoy, Dr. J. Santamaria Juaregui, Dr. J. Portilla Sogorb, Dr. A. Rivero-Roman, Dr. F. Smaill, Dr. S. Sungkanuparph, Dr. A. Vibhagool, Dr. W. Manosuthi, Dr. K. Supparatpinyo, Dr. S. Esser, Dr. G. Fatkenheuer, Dr. J. Rockstroh, Dr. H. J. Stellbrink, Dr. R. Schmidt, Dr. J. Sierra, Dr. J. Andrade, Dr. N. Quintero Perez, Dr. I. Torres, Dr. G. Carosi, Dr. G. Reyes, Dr. C. Viscoli, Dr. A. Lazzarin, Dr. A. Chiranni, Dr. R. Esposito, Dr. C. Kovacs, Dr. J. G. Baril, Dr. R. Zajdenverg, Dr. J. Madruga, Dr. E. Martins Netto, Dr. J. Velez, Dr. J. Tamara, Dr. A. Arango, Dr. M. Angela, Dr. R. Salazar, Dr. E. Gotuzzo, Dr. R. Cabello, Dr. J. Lama, Dr. O. Srivastava, Dr. J. Rajendran, Dr. A. Pazare, Dr. M. Dinaker. **Merck Research Laboratories:** P. Sklar, R. Leavitt, B.-Y. Nguyen, J. Zhao, X. Xu, A. Rodgers, A. Williams-Diaz, R. Isaacs, K. Gottesdiener, C. Gilbert, S. Rawlins, M. Cahill, S. Foley, T. Finn, L. Wenning, M. Miller, R. Barnard, D. Hazuda, M. DiNubile, K. Davis.

Copyright © 2009 Merck & Co., Inc., Whitehouse Station, New Jersey, USA, All Rights Reserved