CROI 2009
16th Conference on Retroviruses and Opportunistic Infections
Montreal, Canada
February 8-11, 2009

Abstract K-103
Poster # 571b

96-Week Results from BENCHMRK 1&2, Phase III Studies of Raltegravir (RAL) in Patients (pts) Failing Antiretroviral Therapy (ART) with Triple-Class Resistant HIV

R. T. Steigbigel[1], D. A. Cooper[2], J. E. Eron[3], J. M. Gatell[4], P. N. Kumar[5], J. K. Rockstroh[6], H. Wan[7], P. Sklar[7], H. Teppler[7], B-Y. Nguyen[7] for the BENCHMRK-1 and 2 Study Groups

[1]SUNY at Stony Brook, USA; [2]University of New South Wales, Sydney, Australia; [3]University of North Carolina, USA; [4]University of Barcelona, Spain; [5]Georgetown University Medical Center, Washington, DC, USA; [6]University of Bonn, Germany; [7]Merck Research Laboratories, West Point, PA, USA

Roy T. Steigbigel, MD
Division of Infectious Diseases
SUNY at Stony Brook
Stony Brook, NY 11794-8153

roy.steigbigel@stonybrook.edu
phone: 631-444-3490
fax: 631-444-7518

Abstract

Background: In 3 studies of HIV-infected pts with limited treatment options, RAL combined with optimized background therapy (OBT) was generally well tolerated and provided superior viral suppression for 48 weeks (wk) compared to OBT alone. Here we present the 96-wk results from BENCHMRK 1&2 (Protocols 018&019), ongoing, double-blind Phase III studies being conducted globally.

Methods: Pts failing ART with triple-class resistant HIV were randomized 2:1 to oral BID RAL 400 mg or placebo (PBO). All pts received OBT. Prespecified efficacy endpoints included % pts with HIV RNA levels <50 copies/mL and the mean change in CD4 cell counts from baseline.

Results (updated): Baseline characteristics in the combined studies were similar in the RAL and PBO groups. At baseline, median CD4 counts were 119 and 123 cells/mm³, and geometric mean viral loads were 4.7 and 4.6 $\log_{10}$ copies/mL in the RAL and PBO groups, respectively. Genotyping demonstrated that OBT contained <1 active drug (sensitivity score = 0) in 25% and 28% of pts in the RAL and PBO groups, respectively. Results from the 96-wk combined efficacy analyses are shown below along with 24-wk and 48-wk results:

	% pts (95% CI) with HIV RNA <50 copies/mL[‡]			Change from baseline CD4 cells/ cu mm[††]		
	24-wk	48-wk	96-wk	24-wk	48-wk	96-wk
RAL (N=462)	63 (58, 67)	62 (58, 67)	57 (52, 62)	84 (75, 92)	109 (97, 120)	123 (110, 137)
PBO (N=237)	34 (28, 40)	33 (27, 39)	26 (21, 32)	36 (27, 46)	45 (32, 57)	49 (35, 63)
RAL - PBO[†]	29* (21, 36)	29* (22, 36)	31* (23, 38)	47* (34, 60)	64* (47, 81)	74* (55, 94)

RAL and PBO were given with OBT; [†]Difference between RAL and PBO; a positive value favors RAL over PBO; *Nominal P<0.001; [‡]Non-Completer=Failure; [††]Baseline values carried forward for virologic failures

RAL was generally well tolerated with few discontinuations [4% (18 pts)] due to adverse events.

Conclusions: In these pivotal studies of pts failing ART with triple-class resistant HIV, RAL plus OBT demonstrated superior antiretroviral and immunological responses compared to OBT alone, that were sustained out to 96 wks.

Background

Raltegravir (MK-0518, ISENTRESS™) is an HIV-1 Integrase Strand-Transfer Inhibitor (InSTI)

- Clinical efficacy: percent of patients achieving HIV RNA <50 copies/mL when used in combination with other ART
 - in ART-naive patients (in combination with TDF/FTC or TDF/3TC):
 - 83% at Week 96 in a phase II dose-ranging study (Markowitz M, et al. 27th Int. AIDS Conference 2008, Abs TUAB02)
 - 86% at Week 48 in the phase III STARTMRK study (Lennox J, et al. ICAAC/IDSA 2008, Abs H-896a)
 - In patients failing therapy with triple class resistant virus (in combination with OBT):
 - 48% at Week 96 in a phase II dose-ranging study (Gatell J, et al. HIV DART 2008, Abs #35)
 - 63% at Week 48 in the phase III BENCHMRK-1 and 2 studies combined (Steigbigel R., et al. NEJM 359;4: 339-354, 2008)

Methods

BENCHMRK-1 & 2 Study Design

- Randomized, double-blind, placebo-controlled with Data and Safety Monitoring Board
- Primary analysis at Week 16; secondary analysis at Week 48; planned study duration of 256 weeks

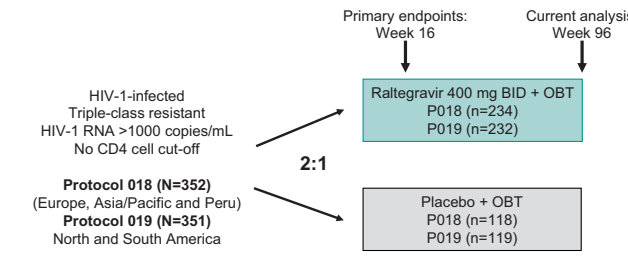
Primary endpoints: Week 16
Current analysis: Week 96
HIV-1-infected
Triple-class resistant
HIV-1 RNA >1000 copies/mL
No CD4 cell cut-off
Protocol 018 (N=352)
(Europe, Asia/Pacific and Peru)
Protocol 019 (N=351)
North and South America
2:1
Raltegravir 400 mg BID + OBT
P018 (n=234)
P019 (n=232)
Placebo + OBT
P018 (n=118)
P019 (n=119)

- OBT was selected by investigator based on baseline resistance testing and prior treatment history. Selected investigational ARTs, darunavir and tipranavir, were permitted.

Statistical Analyses - Efficacy

The following predefined endpoints were examined at Week 96:

- HIV RNA <50 copies/mL
- HIV RNA <400 copies/mL
 - for proportions over time analysis - Non-Completer = Failure (NC=F) approach was used
- Change from baseline in CD4 cell count (cells/mm³)
 - Observed Failure (OF) approach - assumes baseline value was carried forward for virologic failures
- For efficacy by subgroup - Observed Failure (OF) approach was used

Occurrence of AIDS-defining conditions (ADCs) prespecified as an exploratory endpoint

- All suspected ADCs were adjudicated under blinded conditions
- Analysis conducted as previously described (JE Eron et al, ICAAC/IDSA 2008)

Statistical Analyses - Safety

- Clinical adverse events and lab abnormalities – displayed in 2 ways
 - percent of patients (n/N) with event
 - exposure-adjusted event rates (number of patients with event /100 person-years exposure)
 - since ~ 50% greater duration of double-blind therapy for RAL vs Placebo (mean weeks 90 vs 59) due to more discontinuations from Placebo due to virologic failure
- Cancer events – exposure-adjusted analysis based on all treated patients as originally randomized

Results

Patient Disposition

	Raltegravir + OBT		Placebo + OBT	
	N	(%)	N	(%)
Randomized	466		237	
Treated	462	(99)	237	(100)
Continuing in Double-Blind phase	301	(65)	67	(28)
Entered Open-Label post VF* phase	91	(19)	129	(54)
Discontinued study	70	(15)	41	(17)
Discontinued due to adverse event	18	(4)	10	(4)

*Definition of virologic failure:
1) <1 $\log_{10}$ ↓ HIV RNA from baseline and HIV RNA >400 copies/mL at week 16, OR
2) virologic relapse: >1 $\log_{10}$ ↑ HIV RNA above nadir or >400 copies/mL from nadir after response <400 copies/mL (on 2 consecutive measurements at least 1 week apart).

Baseline Patient Characteristics

	Raltegravir + OBT N = 462	Placebo + OBT N = 237
Mean Age, yrs (SD)	46 (9)	45 (8)
% Male	88	89
% Caucasian	65	73
Median CD4 Count, cells/mm³	119	123
GM Viral Load, copies/mL ($\log_{10}$HIV RNA)	44897 (4.7)	39059 (4.6)
% with AIDS	92	91
Median yrs of prior ARTs (Mean # ART)	10 (12)	10 (12)
% Hepatitis B+/% Hepatitis C+/both	8/8/1	3/11/1
% GSS[†] 0/1	25/39	28/41
% PSS[†] 0/1	15/31	19/30
% new enfuvirtide in OBT	20	20
% new darunavir in OBT	36	38

[†]GSS/PSS = total ART in OBT to which patient's virus showed geno/phenotypic sensitivity by Phenosense GT assay. First use of enfuvirtide and darunavir were each counted at +1 active agent and added to GSS/PSS.

Primary Efficacy Endpoints at Week 96 (NC=F Approach)

	HIV RNA <400 copies/mL n/N (%)		HIV RNA <50 copies/mL n/N (%)	
	Raltegravir	Placebo	Raltegravir	Placebo
Total	283/460 (62)	67/237 (28)	262/460 (57)	62/237 (26)
BENCHMRK-1	146/231 (63)	39/118 (33)	134/231 (58)	35/118 (30)
BENCHMRK-2	137/229 (60)	28/119 (24)	128/229 (56)	27/119 (23)

Note: all patients also received OBT.

Acknowledgements

All patients who participated in BENCHMRK-1 and 2.

BENCHMRK-1 Investigators: *Australia:* Allworth A, Anderson J, Bloch M, Cooper DA, Hoy J, Moore RJ, Workman C; *Belgium:* Clumeck N, Colebunders R, Florence E, Moutschen M; *Denmark:* Gerstoft J, Larsen C, Mathiesen L, Pedersen C; *France:* Delfraissy JF, Dellamonica P, Katlama C, Molina JM, Raffi F, Reynes J, Vittecoq D, Yeni P; *Germany:* Arasteh K, Fatkenheuer G, Jaeger H, Rockstroh J, Stoehr A; *Italy:* Aiuti F, Carosi G, Cauda R, Chiodo F, Di Perri G, Filice G, Galli M, Lazzarin A, Mazzarom I, Vullo V; Peru: Castaneda M, Florez A, Mendo F, Paredes A, Salazar R, Ticona E; *Portugal:* Antunes R, Diniz A, Mansinho K, Saraiva da Cunha J, Sarmento R, Teofilo E, Vera J; *Spain:* Arrizabalaga J, Clotet Sala B, Domingo Pedrol P, Gatell Artigas J, Moreno Guillen S, Soriano Vazquez V; *Switzerland:* Hirschel B, Opravil M, Weber R; *Taiwan:* Lin H-H, Sheng W-H, Wang J-H; *Thailand:* Sungkanuparph S, Suwanagool S.

BENCHMRK-2 Investigators: *Brazil:* Grinsztejn, B., Madruga, JV., Schechter, M. *Canada:* Baril, J-G., Loutfy, MR., Montaner, JS., Tremblay, C., Tsoukas, CM., Vezina, S. *Colombia:* Cortes, JA., Mendoza, H., Velez, J. *Mexico:* Quintero Perez, N., Ramos, J., Rodriguez, E. *Puerto Rico:* Morales-Ramirez, JO., Sepulveda, GE. *US:* Aberg, J., Beatty, GW., Benson, P., Bolon, RK., Bredeek, UF., Bruno, C., Campbell, T., Campo, R., Coodley, GO., Corales, RB., DeJesus, E., Eron, JJ., Fessel, WJ., Fetchick, RJ., Gonzales, CJ., Hicks, C., Horberg, MA., Klein, DB., Kozal, MJ., Kumar, PN., LaMarca, A., Lennox, JL., Lichtenstein, KA., Liporace, R., Little, SJ., Luetkemeyer, A., Mariuz, P., Markowitz, M., McMahon, DK., Perez, G., Pierone, G., Reichman, RC., Rhame, F., Shalit, P., Short, W., Skolnik, PR., Steigbigel, RT., Tedaldi, EM., Ward, DJ., Wiznia, AA., Wright, DP.

Merck Research Laboratories: H. Wan, A. Rodgers, J. Zhao, C. Lu, D. Hazuda, M. Miller, R. Danovich, R. Rhodes, B. Jackson, K. Strohmaier, P. Sklar, R. Leavitt, H. Teppler, B-Y. Nguyen.

Overall Efficacy

Percent of Patients (95% CI) Achieving HIV RNA <400 Copies/mL (NC=F Approach)

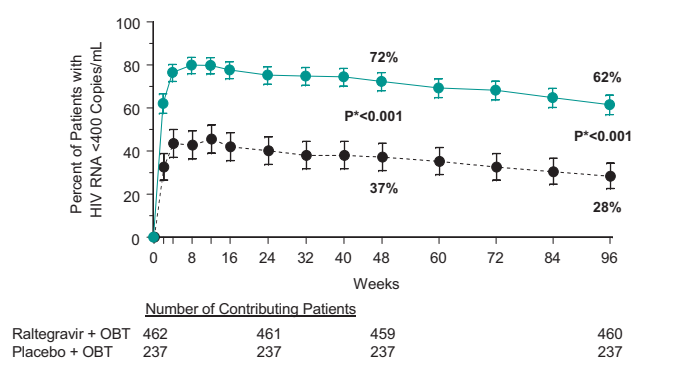
Percent of Patients with HIV RNA <400 Copies/mL
72%
62%
P*<0.001
P*<0.001
37%
28%
Weeks
Number of Contributing Patients
Raltegravir + OBT 462 461 459 460
Placebo + OBT 237 237 237 237

*p-value was derived from a logistic regression model adjusted for baseline HIV RNA level ($\log_{10}$), first enfuvirtide use in OBT, first darunavir use in OBT, active PI in OBT.

Percent of Patients (95% CI) Achieving HIV RNA <50 Copies/mL (NC=F Approach)

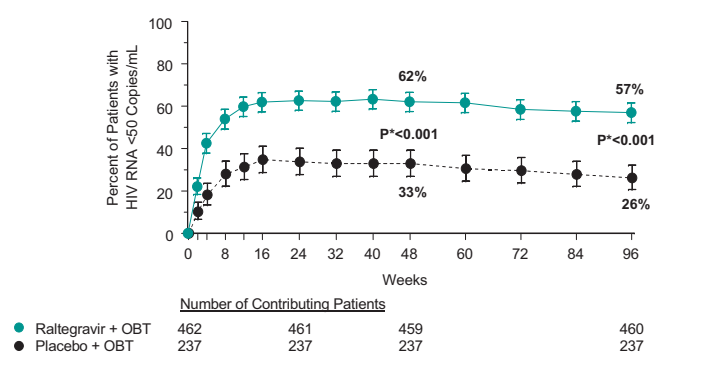
Percent of Patients with HIV RNA <50 Copies/mL
62%
57%
P*<0.001
P*<0.001
33%
26%
Weeks
Number of Contributing Patients
Raltegravir + OBT 462 461 459 460
Placebo + OBT 237 237 237 237

*p-value was derived from a logistic regression model adjusted for baseline HIV RNA level ($\log_{10}$), first enfuvirtide use in OBT, first darunavir use in OBT, active PI in OBT.

Change From Baseline in CD4 Cell Count (cells/mm³) (Observed Failure Approach)

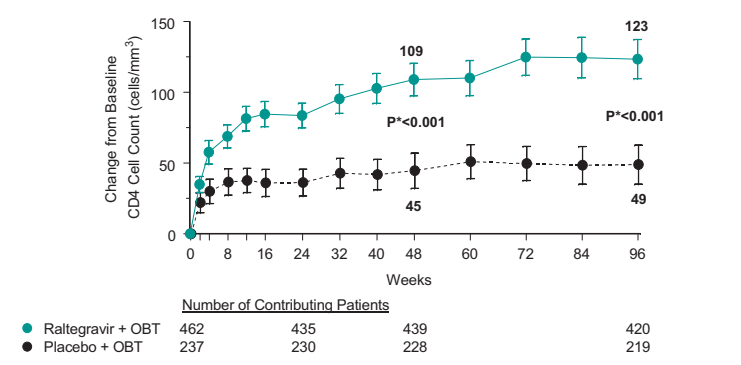
Change from Baseline CD4 Cell Count (cells/mm³)
109
123
P*<0.001
P*<0.001
45
49
Weeks
Number of Contributing Patients
Raltegravir + OBT 462 435 439 420
Placebo + OBT 237 230 228 219

*p-value was derived from a mixed-effects model adjusted for: baseline CD4 cell count, stratum, treatment, visit, interactions between visit and previous variables.

Percent of Patients with HIV RNA <50 copies/mL at Week 96 by Subgroup

Outcome by Baseline HIV RNA and CD4 Cell Count[†]

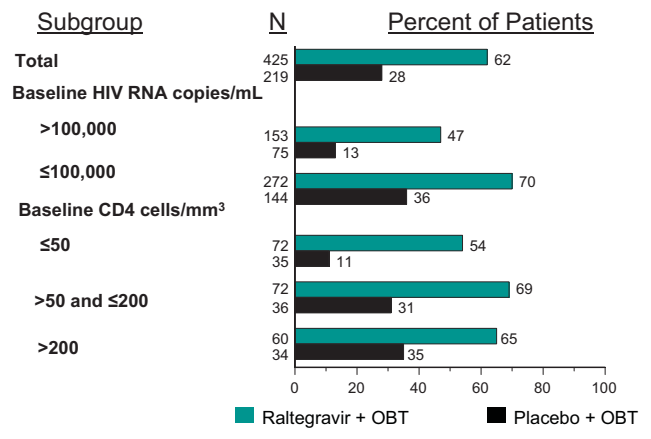
Subgroup
N
Percent of Patients
Total 425 62
219 28
Baseline HIV RNA copies/mL
>100,000 153 47
75 13
≤100,000 272 70
144 36
Baseline CD4 cells/mm³
≤50 72 54
35 11
>50 and ≤200 72 69
36 31
>200 60 65
34 35
Raltegravir + OBT
Placebo + OBT

[†]Virologic failures carried forward

Outcome by Genotypic Sensitivity Score (GSS)[†]

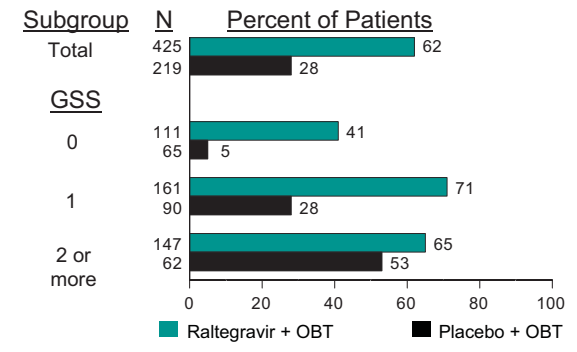
Subgroup
N
Percent of Patients
Total 425 62
219 28
GSS
0 111 41
65 5
1 161 71
90 28
2 or more 147 65
62 53
Raltegravir + OBT
Placebo + OBT

[†]Virologic failures carried forward.

In patients with baseline GSS=1, the active agents in OBT were: darunavir (52%, 52% in raltegravir and placebo groups, respectively), enfuvirtide (8%, 16%), tenofovir (12%, 6%), and tipranavir (11%, 11%).

Outcome by Selected ARTs in OBT[†]

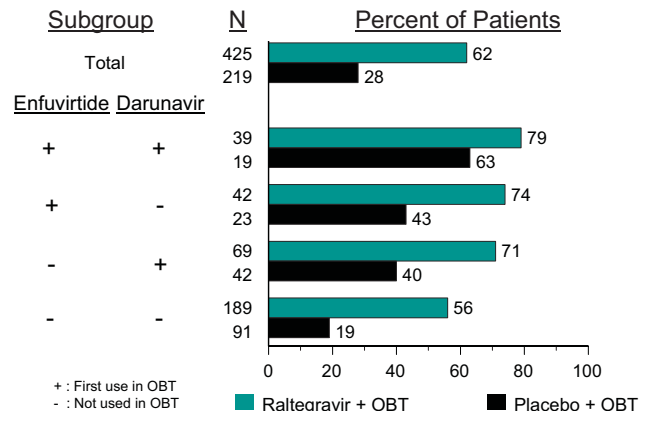
Subgroup
N
Percent of Patients
Total 425 62
219 28
Enfuvirtide Darunavir
+ + 39 79
19 63
+ - 42 74
23 43
- + 69 71
42 40
- - 189 56
91 19
+ : First use in OBT
- : Not used in OBT
Raltegravir + OBT
Placebo + OBT

[†]Virologic failures carried forward.

Exposure-Adjusted Rates and Relative Risk (95% CI) of Confirmed AIDS-Defining Conditions and Death, Week 96 (double-blind)

	Raltegravir + OBT (N=462)	Placebo + OBT (N=237)	Relative Risk
Endpoint	Cases/PYR[†] (Rate[‡])	Cases/PYR[†] (Rate[‡])	(95% CI)
New or Recurrent ADC[#]	18/807 (2.23)	11/267 (4.13)	0.54 (0.24, 1.27)
New ADC	8/817 (0.98)	6/267 (2.25)	0.44 (0.13, 1.52)
Death	13/824 (1.58)	7/269 (2.60)	0.61 (0.23, 1.80)
New or Recurrent ADC or Death	26/806 (3.22)	16/267 (6.00)	0.54 (0.28, 1.07)
New ADC or Death	18/817 (2.20)	12/267 (4.49)	0.49 (0.22, 1.12)

[†]Cases/PYR = No. of patients with endpoint/Person-years (PYR) at risk; [‡]Per 100 PYR; [#]Pre-specified analysis.

Time to first new or recurrent ADC [median (25th - 75th percentile)]

- Raltegravir: 9.3 weeks (7.4 – 19.1)
- Placebo: 15.0 weeks (2.7 - 16.9)

Raltegravir group had approx 50% longer duration on double-blind study therapy than placebo group. The rates of clinical endpoints occurring during the double-blind phase were lower for raltegravir than for placebo at week 96, regardless of endpoint. These differences did not reach statistical significance.

RAL resistance: Patients with virologic failure between Week 48 and Week 96 (VL >400 copies/mL)

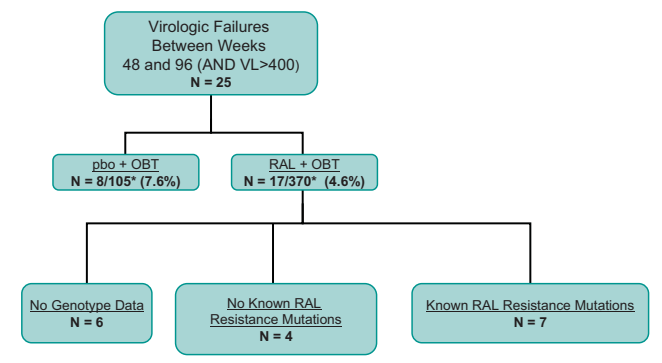
Virologic Failures Between Weeks 48 and 96 (AND VL>400)
N = 25
pbo + OBT
N = 8/105* (7.6%)
RAL + OBT
N = 17/370* (4.6%)
No Genotype Data
N = 6
No Known RAL Resistance Mutations
N = 4
Known RAL Resistance Mutations
N = 7

*Denominator = number of patients continuing on study at Week 48.

Raltegravir VF Patients with IN Mutations

Pt ID	Baseline vRNA (copies/mL)	Baseline OBT GSS/PSS	Week of VF	vRNA at VF	IN changes from baseline sequence[†]
1[‡]	46500	2/2	84	1190	**N155H, L74M,** F181L, G52E, R224R/W
2	1520	0/0	96	1110	**N155N/H, V151V/I, Q148H, G140S,** N120N/S
3[‡]	1410	0/0	84	801	**Q148R, E138K,** D253A, A282A/E, T115T/A
4	4330	2/2	72	3280	**Y143C, N155H, L74L/I/M,** A38A/T/D/N, G70R, Q216H, D288D/E
5	9350	2/2	96	1210	**N155H,** Q95K, E157D, G197G/R
6[‡]	256000	1/1	72	2840	**N155H,** Q95Q/R, A196A/P, G163R
7	726000	0/0	60	756	**N155H**

Known RAL resistance mutations are shown in bold.

[†]Data derived by population sequencing. All amino acid changes from baseline observed in any of multiple independent PCR reactions are listed without regard to linkage.

[‡]Patients 1, 3, and 6 all had vRNA <400 copies/mL subsequent to VF with no change in ART regimen.

Overall Safety

Clinical Adverse Events (AEs) Percent of Patients (Exposure Adjusted Rate†)

	Raltegravir + OBT N = 462	Placebo + OBT N = 237
Mean time on therapy (weeks)	93.0	59.3
Person-years (PYR) at risk	823.8	269.4
	% (rate[†])	% (rate[†])
Any AE	92.9 (52.1)	88.6 (78.0)
Drug-related[‡] AE	58.4 (32.8)	58.6 (51.6)
Serious AE	25.3 (14.2)	22.4 (19.7)
Serious drug-related AE	2.8 (1.6)	3.8 (3.3)
Deaths	2.8 (1.6)	3.0 (2.6)
Discontinued due to AE	3.7 (2.1)	5.1 (4.5)
Most common* drug-related AEs		
Abdominal distension	2.2 (1.2)	1.7 (1.5)
Diarrhea	3.2 (1.8)	5.1 (4.5)
Nausea	4.1 (2.3)	4.6 (4.1)
Vomiting	1.5 (0.8)	2.1 (1.9)
Fatigue	3.2 (1.8)	0.8 (0.7)
Pyrexia	0.9 (0.5)	2.5 (2.2)
Headache	4.8 (2.7)	5.1 (4.5)

[†]per 100 person-years (PYR); [‡]Determined by the investigator to be possibly, probably, or definitely related to raltegravir or placebo (alone or in combination with OBT); *incidence ≥ 2%, any intensity.

Grade 3 or 4 Laboratory Abnormalities Percent of Patients (Exposure Adjusted Rate[†])

Laboratory Test (Unit)	Toxicity Criteria*		Raltegravir + OBT N=462 (823.8 PYR) % (rate[†])	Placebo + OBT N=237 (269.4 PYR) % (rate[†])
ANC (10³/µl)	Grade 3	0.50 -0.749	3.0 (1.7)	3.4 (3.0)
	Grade 4	<0.50	1.3 (0.7)	1.3 (1.1)
Hemoglobin (gm/dL)	Grade 3	6.5 – 7.4	0.9 (0.5)	0.8 (0.7)
	Grade 4	<6.5	0.2 (0.1)	0.0 (0.0)
Platelet count (10³/µL)	Grade 3	25 – 49.999	0.6 (0.4)	0.4 (0.4)
	Grade 4	<25	0.9 (0.5)	0.4 (0.4)
Fasting LDL-C (mg/dL)	Grade 3	≥190	6.3 (3.4)	4.6 (3.7)
Fasting cholesterol (mg/dL)	Grade 3	>300	10.2 (5.7)	5.5 (4.8)
Fasting triglyceride (mg/dL)	Grade 3	751 - 1200	6.3 (3.5)	3.4 (3.0)
	Grade 4	>1200	3.7 (2.1)	2.1 (1.9)
Fasting glucose (mg/dL)	Grade 3	251 – 500	1.9 (1.1)	1.7 (1.5)
	Grade 4	>500	0.0 (0.0)	0.0 (0.0)
Serum creatinine (mg/dL)	Grade 3	1.9 – 3.4 x ULN	1.5 (0.8)	0.8 (0.7)
	Grade 4	≥3.5 x ULN	0.2 (0.1)	0.4 (0.4)
Total bilirubin (mg/dL)	Grade 3	2.6 – 5.0 x ULN	3.0 (1.7)	3.0 (2.6)
	Grade 4	>5.0 x ULN	0.9 (0.5)	0.0 (0.0)
AST (IU/L)	Grade 3	5.1 – 10.0 x ULN	4.3 (2.4)	3.0 (2.6)
	Grade 4	>10.0 x ULN	0.6 (0.4)	1.3 (1.1)
ALT (IU/L)	Grade 3	5.1 – 10.0 x ULN	4.1 (2.3)	2.5 (2.2)
	Grade 4	>10.0 x ULN	1.3 (0.7)	1.7 (1.5)
Alkaline phosphatase (IU/L)	Grade 3	5.1 – 10.0 x ULN	0.4 (0.2)	1.3 (1.1)
	Grade 4	>10.0 x ULN	0.6 (0.4)	0.4 (0.4)
Pancreatic amylase (IU/L)[§]	Grade 3	2.1 – 5.0 x ULN	5.0 (2.8)	3.0 (2.6)
	Grade 4	>5.0 x ULN	0.2 (0.1)	0.4 (0.4)
Lipase (IU/L)	Grade 3	3.1 – 5.0 x ULN	1.9 (1.1)	0.8 (0.7)
	Grade 4	>5.0 x ULN	0.0 (0.0)	0.0 (0.0)
Creatine kinase (IU/L)	Grade 3	10.0 – 19.9 x ULN	3.9 (2.2)	2.5 (2.2)
	Grade 4	≥20.0 x ULN	3.0 (1.7)	0.8 (0.7)

* Grades 3 and 4 per DAIDS toxicity criteria. [†]per 100 person-years (PYR).
[§] % defined as (number of patients meeting specific pancreatic amylase criteria) / (number of patients with amylase test result).

Cancer Events in Double-Blind Phase - Relative Risk (95% CI)

	Raltegravir + OBT		Placebo + OBT		Relative Risk
	N	Cases/PYR[†] (Rate[‡])	N	Cases/PYR[†] (Rate[‡])	(95% CI)
Total	462	24/801 (3.0)	237	7/264 (2.6)	1.1 (0.5, 3.1)
BENCHMRK-1	232	12/417 (2.9)	118	2/133 (1.5)	
BENCHMRK-2	230	12/384 (3.1)	119	5/131 (3.8)	

[†] Person-years at risk; [‡]Per 100 PYR.

For a comprehensive assessment of cancer risk, a similar analysis was done including all double blind data from Phase II and Phase III studies (Protocols 004, 005, BENCHMRK-1 and 2 and STARTMRK), which provides a malignancy rate of 1.7 /100 PYR for raltegravir and 2.2 /100 PYR for the comparator group, resulting in a relative risk (95% CI) of 0.8 (0.4, 1.5). (D. Cooper et al, CROI 2009 Abs R-106, Poster 859).

Conclusions

In HIV-infected, treatment-experienced patients failing antiretroviral therapy with triple-class resistant HIV:

- Raltegravir 400 mg b.i.d. plus OBT, compared to placebo plus OBT, had potent, superior, and durable antiretroviral and immunological efficacy sustained through Week 96.
 - 57% of patients receiving raltegravir maintained HIV RNA < 50 copies /mL
 - up to 79% in patients receiving new, active ART in OBT
- Virologic failure was generally associated with mutations at one of three primary residues, Q148, N155, or Y143, in combination with at least one other mutation.
- Rates of ADC and death during double-blind phase were lower for raltegravir than placebo at Week 96, regardless of endpoint, although these differences did not reach statistical significance.
- Raltegravir 400 mg b.i.d. plus OBT was generally well tolerated as compared to placebo in combination with OBT.
 - Few adverse experiences led to discontinuation
 - Risk of developing malignancy was comparable between raltegravir and comparator groups.

Copyright ©2009 Merck & Co., Inc., Whitehouse Station, New Jersey, USA, All Rights Reserved