Inhibitors of the RNase H Activity of Reverse Transcriptase as an Approach to New HIV-1 Antiretroviral Agents

P. D. Williams,[a] D. D. Staas,[a] S. Venkatraman,[a] T. M. Booth,[a] H. M. Loughran,[a] R. D. Ruzek,[a] J. S. Wai,[a] J. P. Vacca,[a] S. Prasad,[b] S. K. Munshi,[b] C. Bahnck,[c] G. Dornadula,[c] R. C. Hrin,[c] A. L. Himmelberger,[c] M. D. Miller,[c] D. J. Hazuda,[d] J. A. Grobler,[c]

Departments of [a]Medicinal Chemistry, [b]Structural Biology, [c]Antiviral Research, [d]Infectious Diseases Administration, Merck & Co., Inc., West Point, PA 19486 USA

Peter D. Williams
Merck & Co., Inc.
West Point PA 19438
peter_williams@merck.com
215-652-3003

Abstract

A novel series of HIV-1 RNase H inhibitors has been developed. A prototype inhibitor was shown to bind to two manganese ions in the RNase H active site of full length RT by X-ray crystallography. Systematic structure-activity studies around the core structure identified substituents which improved potency and selectivity in biochemical assays and antiviral activity in cell culture. An optimal compound from this work, inhibitor **5**, showed good potency and selectivity in biochemical assays (RT RNase H IC_{50} = 0.045 uM, RT Polymerase IC_{50} = 13 uM; Integrase Strand Transfer IC_{50} = 24 uM) and antiviral effects in cell culture (IC_{50} = 0.19 uM) with a 17-fold window with respect to cytotoxicity (CC_{50} = 3.3 uM).

Introduction

Nucleoside and non-nucleoside HIV-1 reverse transcriptase (RT) inhibitors are widely prescribed agents for the treatment of HIV infection. Both drug classes work by interfering with the polymerase function of RT. In addition to the polymerase active site, RT contains a ribonuclease H (RNase H) active site which hydrolyzes RNA phosphodiester bonds in RNA-DNA duplexes produced during reverse transcription. Viruses with inactivating mutations in the RNase H active site fail to replicate, thus demonstrating an essential role of the RNase H activity and raising considerable interest in the identification of an RNase H inhibitor as a mechanistically novel antiretroviral agent.

We recently identified a novel series of HIV-1 RNase H inhibitors based on a 1-hydroxy-1,8-naphthyridin-2-one scaffold. Lead compound **1** exhibited good activity as an RNase H inhibitor in a biochemical assay (IC_{50} = 0.1 uM).

1

IC_{50} = 0.1 uM

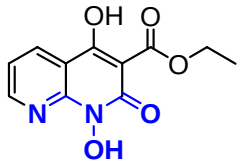

Herein we report optimization efforts and mechanism-of-action studies with inhibitors in this novel structural class.

Methods and Results

Figure 1

X-ray crystal structure showing binding of compound **1** to two Mn^{2+} ions (purple) in the RNase H active site of HIV-1 reverse transcriptase.

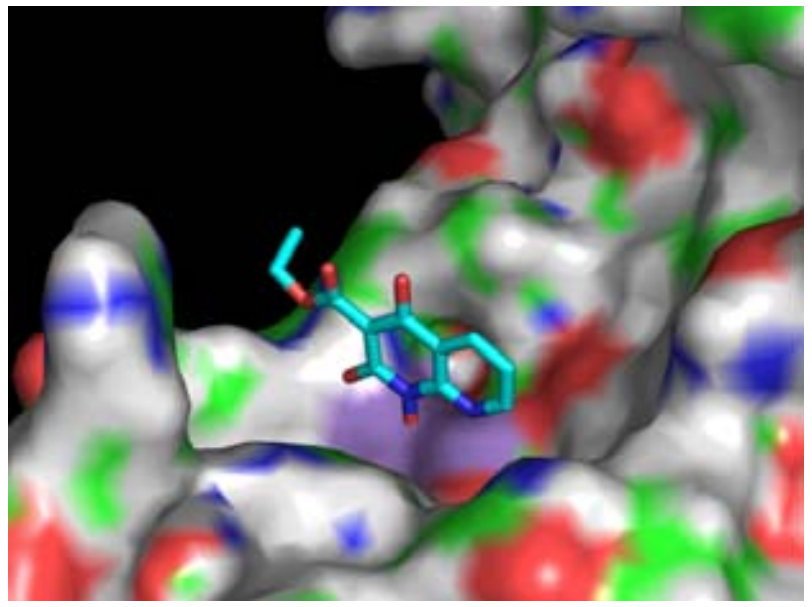

Figure 2

Double reciprocal plot of velocity vs. substrate concentration showed mixed-type inhibition of RNase H activity by compound **1**, indicating that **1** binds to an enzyme-substrate or enzyme-product complex.

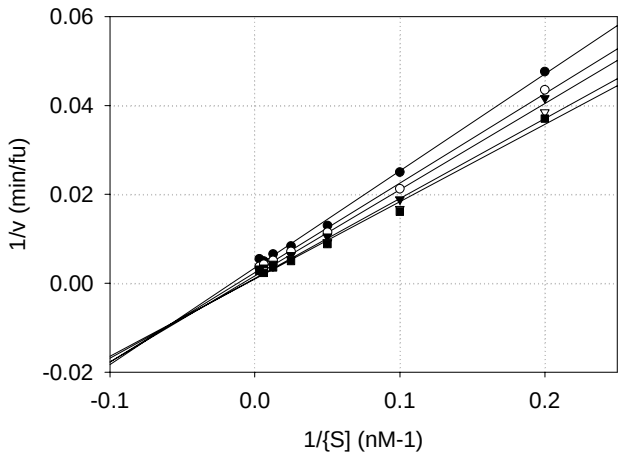
0.06
0.04
0.02
0.00
-0.02
1/v (min/fu)
-0.1
0.0
0.1
0.2
1/[S] (nM-1)

Figure 3

Structure-activity relationships of 1-hydroxy-1,8-naphthyrin-2-ones.

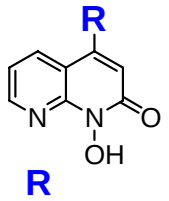

cmpd	R	IC_{50} (uM) Biochemical Assays: RT RNase H	RT Polymerase	Integrase Strand Transfer	Cellular Assays: HIV-1 Replication	Cytotoxicity CC_{50}
2	H	1.4	>50	15	4.5	20
3	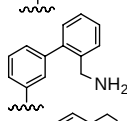	0.67	36	18	2.8	9.1
4	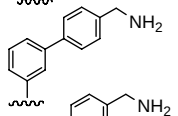	0.13	16	7.8	0.62	2.3
5	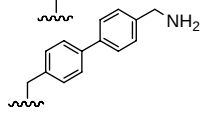	0.045	13	24	0.19	3.3

- Optimization of core structure **2** was possible with certain mixed polar/lipophilic substituents at position 4.
- Potency of antiviral activity in cell culture tracked with potency of inhibition of RNase H activity in biochemical assays.
- The window between antiviral activity in cell culture and cytotoxicity ranged from 3- to 17-fold.

Figure 4

Dose-response curves for compound **5** showing window between antiviral effects (red) and cytotoxicity measured with Alamar Blue assay (blue).

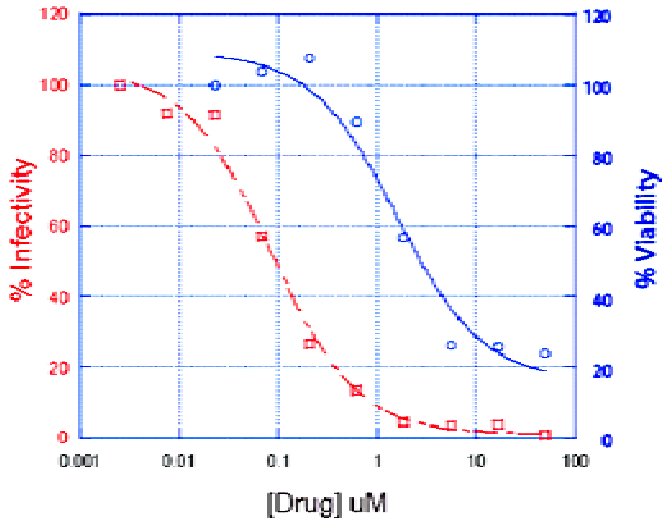
% Infectivity
% Viability
120
100
80
60
40
20
0
0.001
0.01
0.1
1
10
100
[Drug] uM

Figure 5

Equivalent dose-response curves using wild-type HIV-1 (blue) and VSV-g pseudotyped virus (red) indicated that the antiviral effects of compound **5** are not due to inhibition of virus entry into the cell.

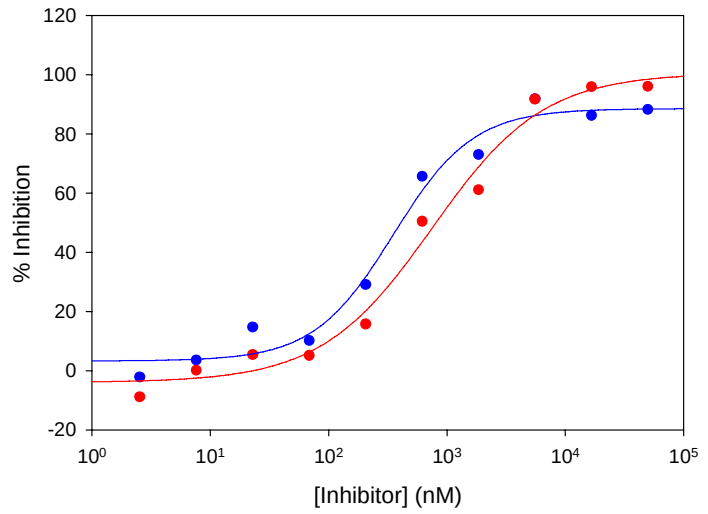
120
100
80
60
40
20
0
-20
% Inhibition
10^0
10^1
10^2
10^3
10^4
10^5
[Inhibitor] (nM)

Conclusions

- A prototype compound in a novel series of HIV-1 RNase H inhibitors based on a 1-hydroxy-1,8-naphthyridin-2-one scaffold was shown to bind to active site metals by X-ray crystallography and to bind to an enzyme-substrate or enzyme-product complex using kinetics experiments.
- From structure-activity studies, potency for inhibiting RNase H activity was optimized by introducing mixed polar/lipophilic groups at a position on the scaffold which may be oriented toward nucleic acid (substrate or product).
- Compound **5** showed good potency for inhibiting RNase H activity in biochemical assays (IC_{50} = 0.045 uM) with >100-fold selectivity vs. two other Mg-dependent viral enzymes.
- Infectivity studies with wild type and VSV-g pseudotyped virus suggested that the antiviral effects of compound **5** in cell culture were not due to inhibition of virus entry into the cell.
- Compound **5** inhibited viral replication in cell culture at sub-micromolar concentrations (IC_{50} = 0.19 uM) and with a 17-fold window with respect to cytotoxic effects (CC_{50} = 3.3 uM).

Copyright © 2006 Merck & Co., Inc., Whitehouse Station, New Jersey, USA, All Rights Reserved