Comparison of Phenotypic (Trofile®) and Genotypic (SensiTrop II) Assays to Determine HIV Tropism in Treatment-Naïve Subjects

Michael C. McCarthy, Regis Vilchez, Jorge Quiroz, Julie Strizki, Jennifer Liberi, and Lisa M. Dunkle

Schering-Plough Research Institute, Kenilworth, NJ, USA

Abstract

Background: CXCR4 viruses are believed to be largely restricted to the later stages of HIV infection. Recent data indicate that CXCR4 viruses can be detected at various stages in the disease and that CCR5 and CXCR4 populations can coexist in a dynamic process. Since the presence of X4-tropic virus may impact the magnitude of viral load reduction achieved with a regimen containing a CCR5 antagonist, the performance and accuracy of HIV tropism testing is important for patient management.

Methods: Paired plasma samples from 252 HIV-infected treatment-naïve individuals from Africa, EU, Latin America and USA were independently evaluated in a blinded fashion by both the standard Trofile® (Monogram Biosciences) and the SensiTrop II (Pathway Diagnostics) assays. The Trofile® assay determines tropism phenotypically by infection of coreceptor-expressing cell lines with reporter viruses expressing patient-derived gp160. The SensiTrop II assay determines tropism genotypically using a combination of gp120 sequencing and heteroduplex complex formation. The marketed turn-around times for these assays are 14-18 days for Trofile® and 4 days for SensiTrop II.

Results: Inconclusive results were obtained in 33 (13%) specimens by Trofile® and 11 (4%) by SensiTrop II. Inconclusive results from Trofile® were related to Non-B Clade virus and higher viral load (10,000-100,000 and >100,000). Exclusive CCR5 and mixed CCR5/CXCR4 viruses were detected in 189 (75%) and 30 (12%), and 186 (74%) and 55 (22%) when using Trofile® and SensiTrop II, respectively. Of 211 samples with reportable results for both assays, 166 (79%) were concordant for tropism. Among the 45 (21%) samples discordant for tropism, 33 (16%) subjects had detectable CCR5/CXCR4 by SensiTrop II, but exclusively CCR5 by Trofile® (95% CI, 10.7%-20.5%). Conversely, 12 (6%) had CCR5/CXCR4 detected by Trofile® but exclusively CCR5 by SensiTrop (95% CI, 3%-9%). Results were generally received within 4 weeks for Trofile® and within 5 days for SensiTrop II.

Conclusion: A significant proportion of treatment-naïve subjects harbor CXCR4 viruses. SensiTrop II assay appears to detect CXCR4 virus frequently and to provide results more quickly. Inconclusive results appear to be more frequent with Trofile® than with SensiTrop II.

Background/Introduction

- Vicriviroc is a novel CCR5 antagonist in clinical development for patients with CCR5-tropic HIV.
- Current guidelines dictate that tropism testing should be performed to assure that CCR5 antagonists are prescribed only for individuals with R5-only infection.
- The first commercially available tropism assay was the Trofile® assay (Monogram Biosciences). This assay has a reported sensitivity to detect minor populations of X4-tropic viruses present in plasma at 5% with about 85% certainty.[1]
- New assays, including the enhanced Trofile® assay (Trofile® ES) and the SensiTrop II assay (Pathway Diagnostics), have been or are being developed to assess tropism more satisfactorily.
- These new assays utilize different technologies to determine viral tropism; thus, the differences between the assays and their performance in clinical samples must be evaluated.
- We conducted an independent evaluation comparing the performance of the phenotypic Trofile® and the genotypic SensiTrop II assays in a population of HIV-infected patients.

Study Objective

- To compare the results of tropism determinations by the standard Trofile® assay and the SensiTrop II assay using plasma samples from treatment-naïve individuals in a Phase II trial.

Study Subjects and Methods

- Tropism was determined at screening via standard Trofile® and SensiTrop II assays using aliquots of a single plasma specimen from each of 252 individuals enrolled in Study PO4875.
- Study P04875 is designed to evaluate the efficacy and safety of vicriviroc versus emtricitabine/tenofovir, both combined with atazanavir/ritonavir, in R5-only treatment-naïve patients.
- Conducted in South Africa, Europe, Latin America, and the United States.
- Key inclusion criteria:
 - HIV-1 RNA ≥5000 copies/mL at screening
 - CD4+ cell count ≥200 cells/mm³ at screening

Assay Methodologies

Trofile® is a phenotypic assay based on infection of coreceptor-expressing cell lines with reporter viruses expressing patient-derived gp160 (**see Figure 1**).

Figure 1: Trofile® HIV Coreceptor Tropism Assay (Monogram Biosciences)

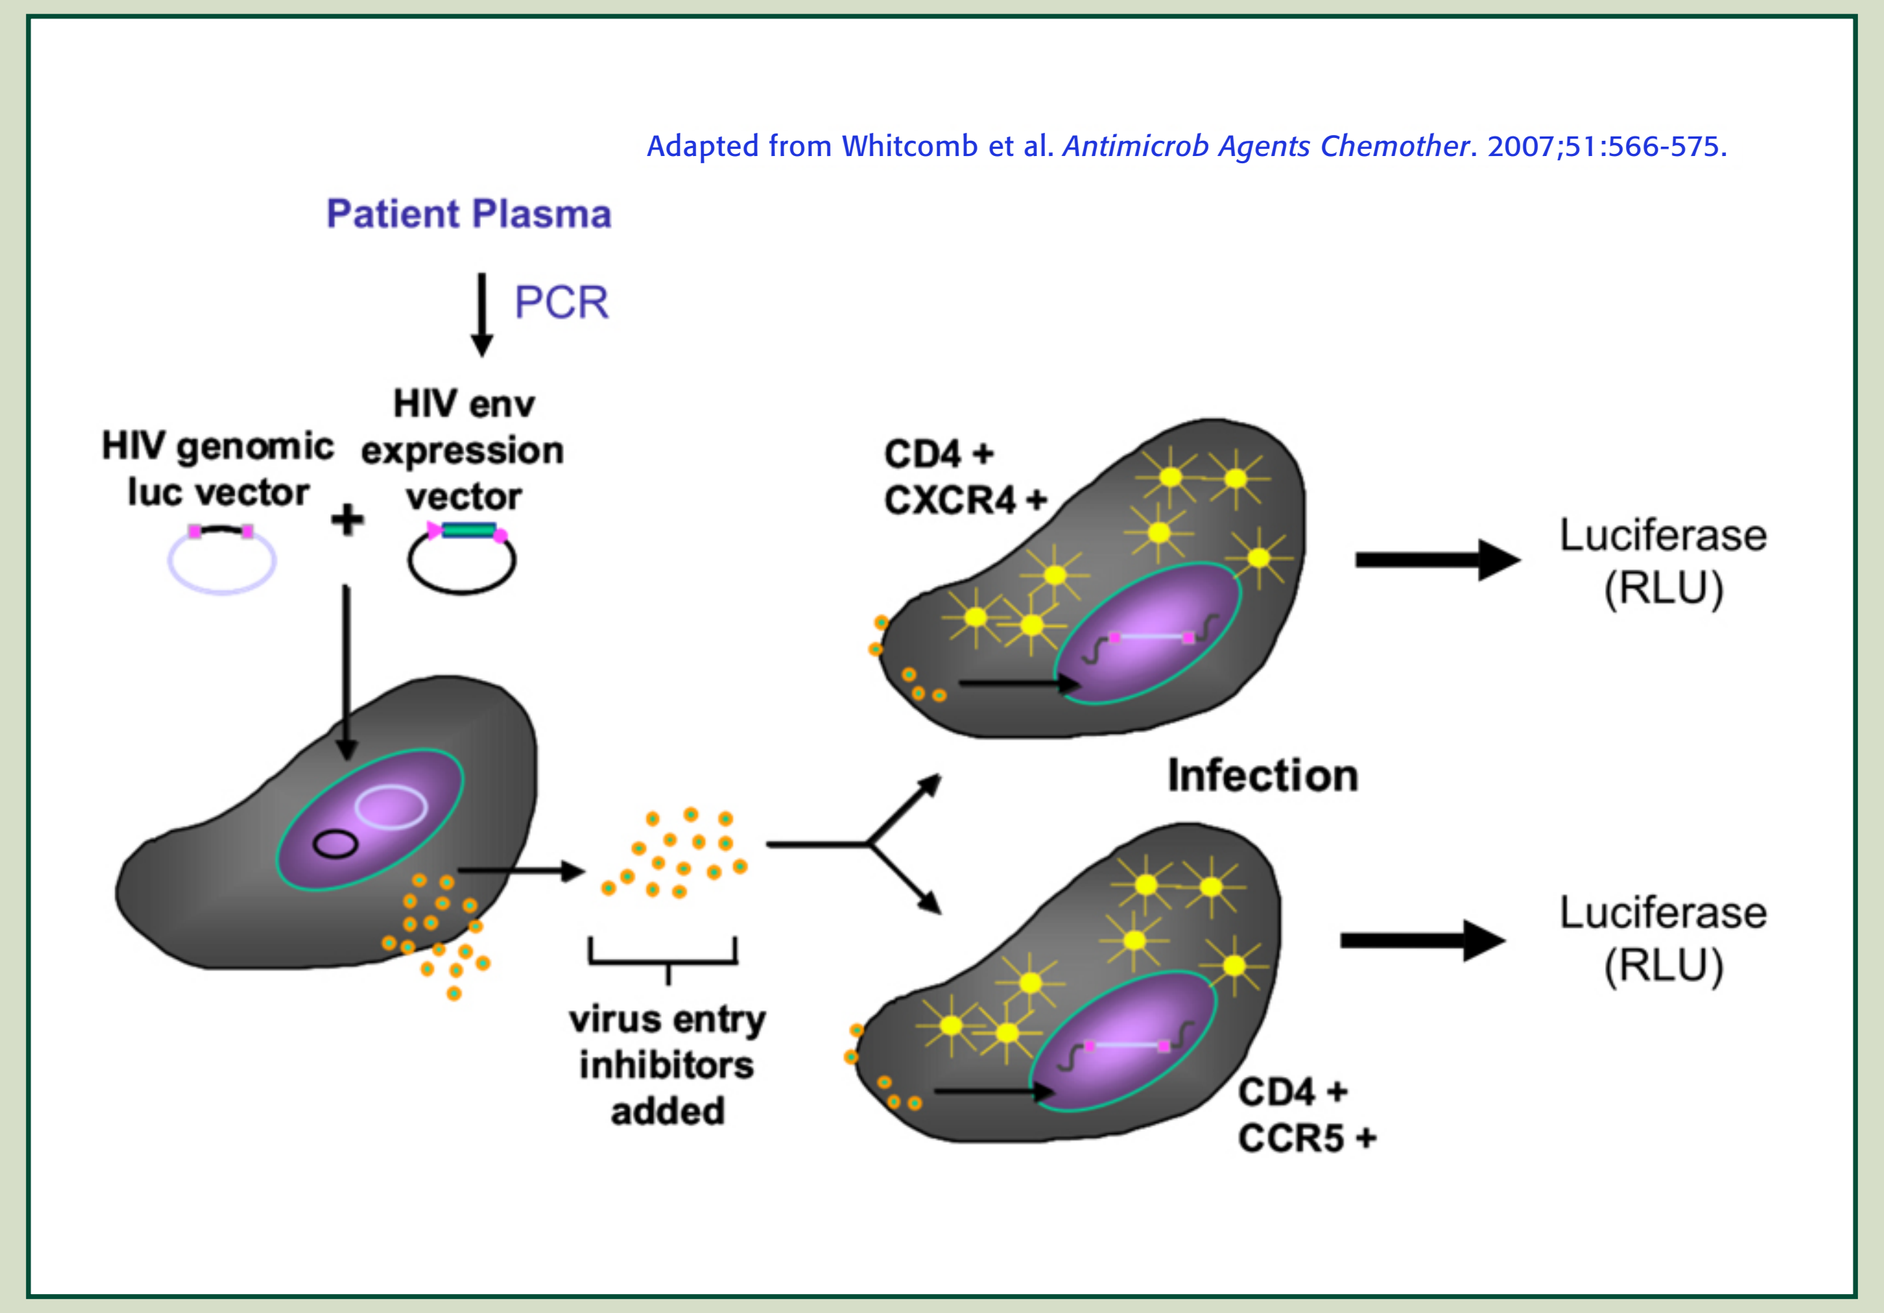

Adapted from Whitcomb et al. Antimicrob Agents Chemother. 2007;51:566-575.
Patient Plasma
PCR
HIV genomic luc vector
HIV env expression vector
CD4 + CXCR4 +
Luciferase (RLU)
Infection
virus entry inhibitors added
CD4 + CCR5 +
Luciferase (RLU)

- Method:
 - Plasma samples collected in EDTA are aliquoted, frozen, and shipped to Monogram Biosciences.
 - HIV gp160 sequences are amplified by PCR from plasma-derived RNA, gel purified, and cloned into an expression vector.
 - Pooled envelope clones are cotransfected with a full-length HIV expression vector lacking a functional gp160 gene and containing a luciferase reporter gene to replication-defective pseudovirus particles containing patient-derived gp160.
 - The pseudoviruses are separately incubated with U87-CCR5 and U87-CXCR4 CD4+ target cells for 72 hours and viral infection is determined by measurement of luciferase activity in cell lysates.
- Note: all assays were performed using the standard Trofile® assay, as the enhanced Trofile® (Trofile® ES) was not available at the time of study initiation.

- SensiTrop II is a genotypic assay based on a combination of gp120 V3-loop sequencing and heteroduplex complex formation (**see Figure 2**).

Figure 2: SensiTrop II Molecular Coreceptor Tropism Assay (Pathway Diagnostics)

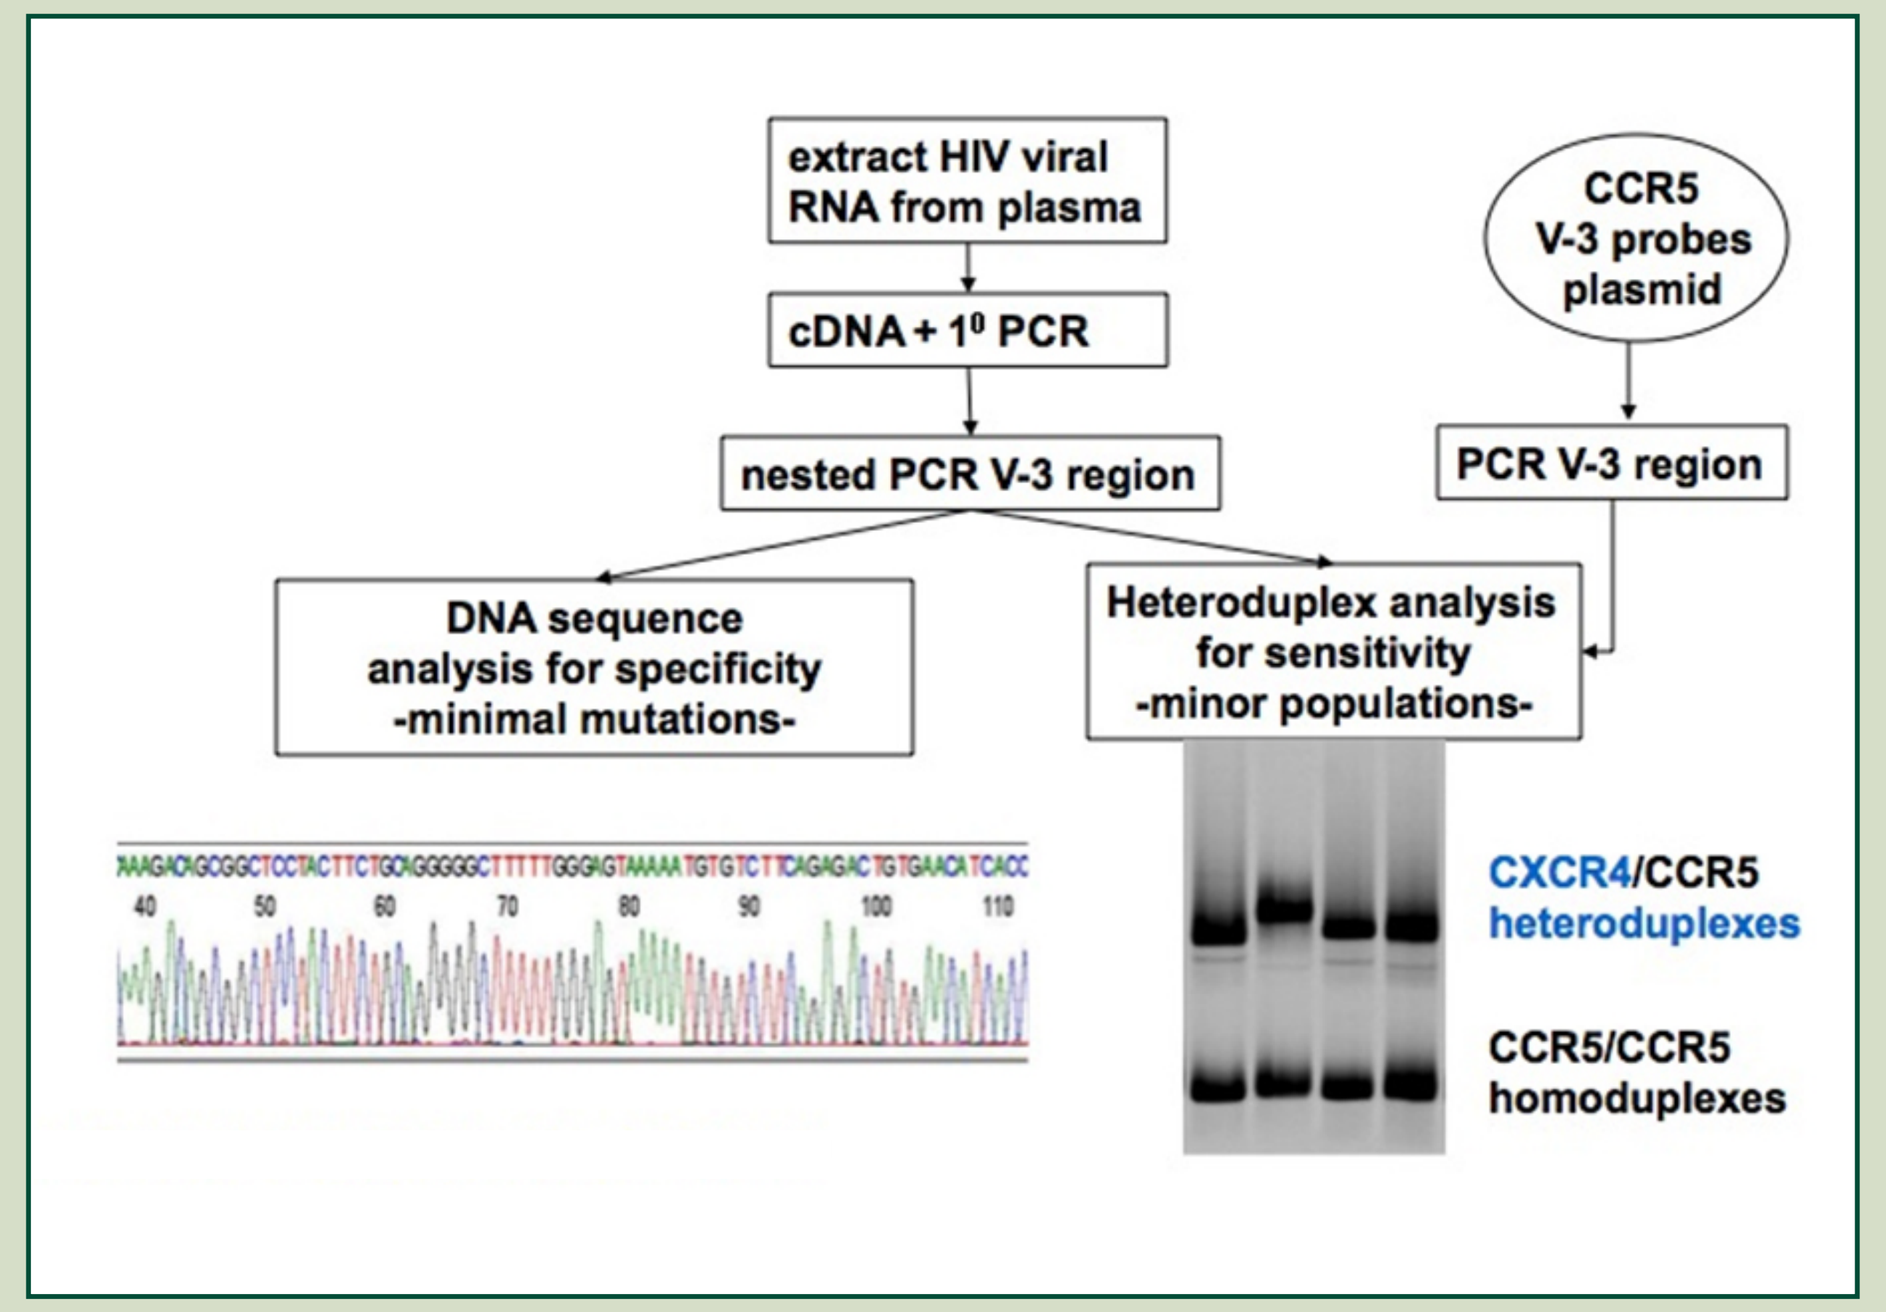

extract HIV viral RNA from plasma
cDNA + 1⁰ PCR
nested PCR V-3 region
CCR5 V-3 probes plasmid
PCR V-3 region
DNA sequence analysis for specificity -minimal mutations-
Heteroduplex analysis for sensitivity -minor populations-
CXCR4/CCR5 heteroduplexes
CCR5/CCR5 homoduplexes

- Method:[2]
 - Viral RNA is isolated from patient plasma, converted to cDNA and the V3-loop is amplified using nested PCR.
 - The PCR amplicon is sequenced and the sequence is analyzed for tropism using two algorithms: (1) the web-based PSSM tropism algorithm and (2) the "11/24/25 rule".[3]
 - The amplicon is further analyzed using a heteroduplex tracking assay in which the V3-loop DNA sequences are denatured and mixed with radiolabeled V3-loop DNA probes from 4 different clade B R5-tropic strains and allowed to anneal. The mixtures are resolved by non-denaturing polyacrylamide gel electrophoresis (PAGE) and analyzed for the presence of homo- and/or heteroduplex complex formation. The formation of heteroduplexes indicates the presence of X4-tropic virus.
 - Population sequencing and HTA are utilized to determine tropism. Samples with predicted X4-tropic genotype and/or heteroduplex formation are reported as R5/X4-tropic.

Statistical Methods

- The marginal distribution of a single assay is the relative frequency of tropism determination by that assay. We evaluated the homogeneity of the marginal distributions of the two assays to compare the results of tropism determinations by the Trofile® and SensiTrop II assays.

Results

Table 1: Summary of Screening Characteristics (N=252)

Characteristic	
Median age, yr (range)	37 (18-71)
Geographic area	
United States	69
Europe	75
Latin America	45
Africa	63
Mean HIV-1 RNA, $\log_{10}$ copies/mL (range)	4.68 (2.14-5.87)
Mean CD4+ cell count, cells/mm³ (range)	297 (10-994)
HIV clade	
B	167
Non-B	80
Missing	5

Comparative characteristics of the two tropism assays are shown in **Table 2**.

Table 2: Comparison of the Trofile® and SensiTrop II Assays

Feature	Trofile®	SensiTrop II
Method of tropism determination	Infection-based phenotype	Genotype HTA combined with population sequence
Sample required	Frozen plasma	Frozen plasma
Sample handling	Plasma collected, aliquoted and frozen on site, shipped to Monogram for analysis	Blood samples shipped to local lab for plasma prep and shipped to Pathway Diagnostics for analysis
Reported assay sensitivity for detection of minor X4 populations	5-10% X4 virus	1% X4 virus
Stated turnaround time	14-18 days	4 days

Results of tropism determinations by the two assays are shown in **Table 3**.

Table 3: Comparison of Tropism Results with the Trofile® and SensiTrop II Assays on Paired Samples

		SensiTrop II			
		Dual/Mixed	CCR5	Inconclusive	Total (MD)
Trofile®	Dual/Mixed	18	12	0	30 (12%)
	CCR5	33	148	8	189 (75%)
	Inconclusive	4	26	3	33 (13%)
	Total (MD)	55 (22%)	186 (74%)	11 (4%)	252

(MD = Marginal distribution)

- The test of marginal distribution homogeneity indicated that the results of the Trofile® and SensiTrop II assays were statistically different ($p<0.0001$).

Tropism results were reported by both assays for 211 samples (**Figure 3**).

Figure 3: Comparison of Reportable Results for the Trofile® and SensiTrop II Assays

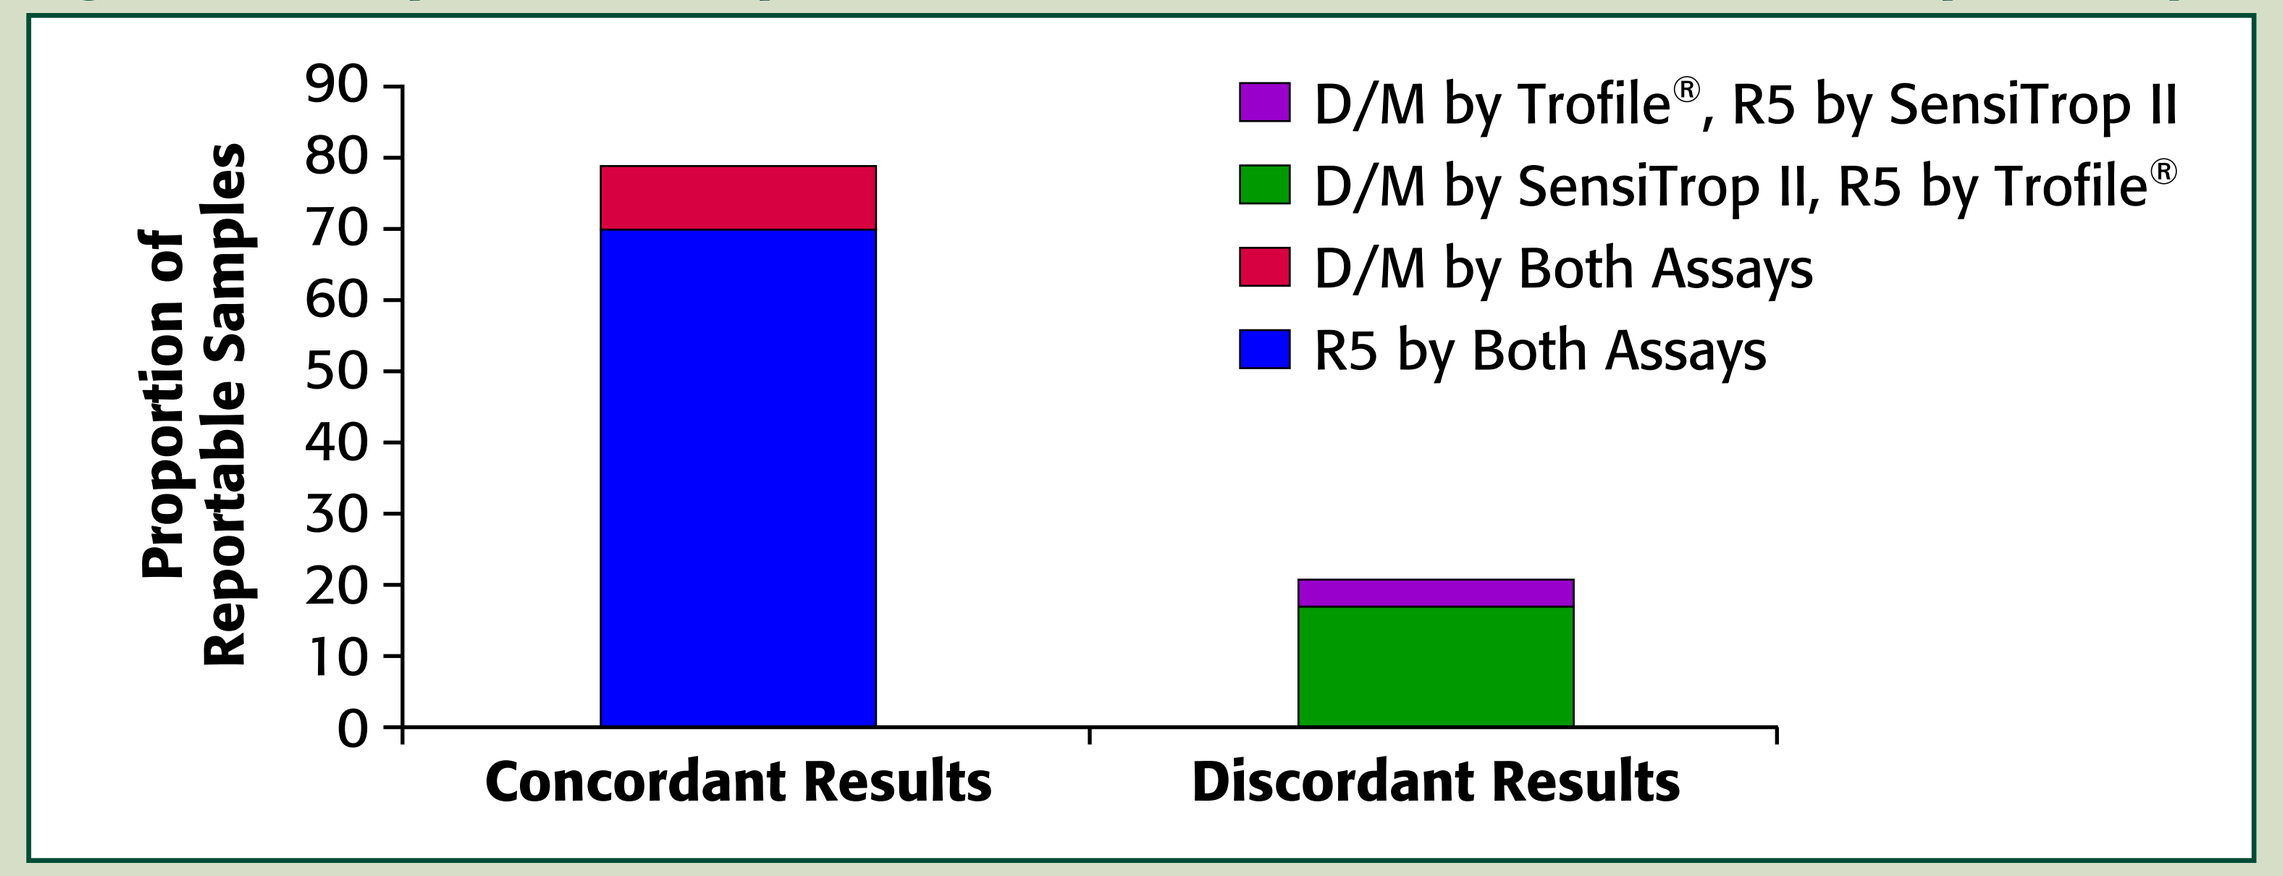

Proportion of Reportable Samples
90
80
70
60
50
40
30
20
10
0
Concordant Results
Discordant Results
D/M by Trofile®, R5 by SensiTrop II
D/M by SensiTrop II, R5 by Trofile®
D/M by Both Assays
R5 by Both Assays

- Among these samples, 166 (79%) had concordant tropism results and 45 (21%) were discordant.
 - Among discordant samples, the SensiTrop II assay reported a greater proportion of samples as D/M (33/211) [16%; 95% CI: 11%-21%], compared with 12/211 (6%; 95% CI: 3%-9%) as determined by the Trofile® assay.
- The number of inconclusive results was 13% for the Trofile® assay and 4% for the SensiTrop II assay.
 - Reasons for inconclusive results included sample integrity and assay performance issues.
- Subset analyses for viral load and HIV clade were performed to assess possible association between these parameters and assay performance.

Figure 4: Tropism Determinations by Viral Load

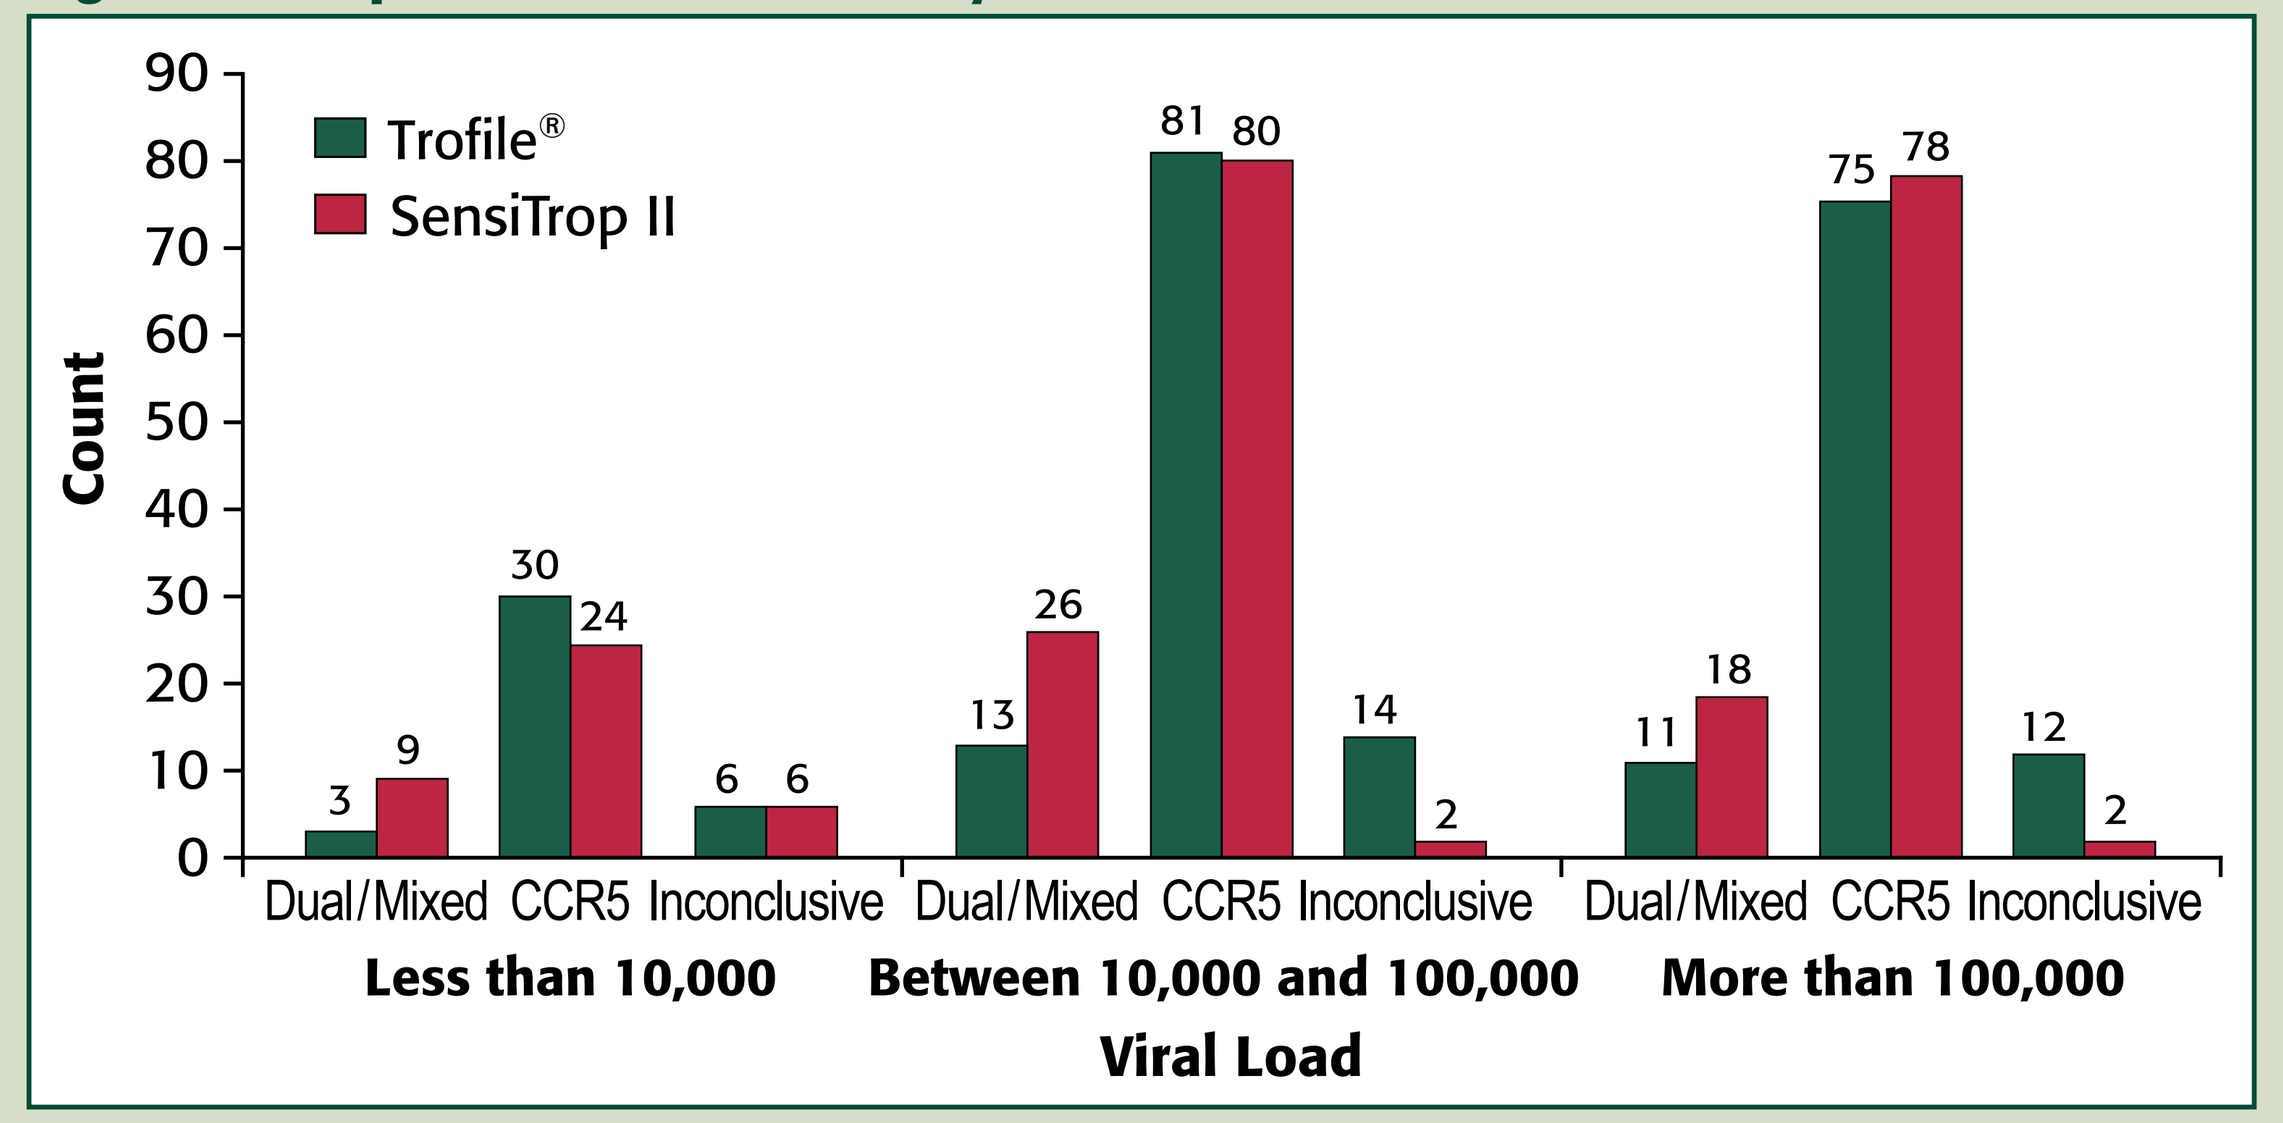

Count
90
80
70
60
50
40
30
20
10
0
Trofile®
SensiTrop II
3
9
30
24
6
6
13
26
81
80
14
2
11
18
75
78
12
2
Dual/Mixed CCR5 Inconclusive
Dual/Mixed CCR5 Inconclusive
Dual/Mixed CCR5 Inconclusive
Less than 10,000
Between 10,000 and 100,000
More than 100,000
Viral Load

- Overall, SensiTrop II reported D/M tropism more frequently than Trofile®, regardless of baseline viral load.

Table 4: Proportions of Samples Yielding Inconclusive Results

Viral load level (copies/mL)	Trofile®	SensiTrop II
<10,000	15%	15%
10,000 – 100,000	13%	2%
>100,000	12%	2%

- Inconclusive results were reported by Trofile® in similar proportions over all ranges of baseline viral load.
- SensiTrop II reported significantly fewer inconclusive results than Trofile® in subjects with HIV RNA >10,000 copies/mL.

Figure 5: Tropism Determinations by Clade Type

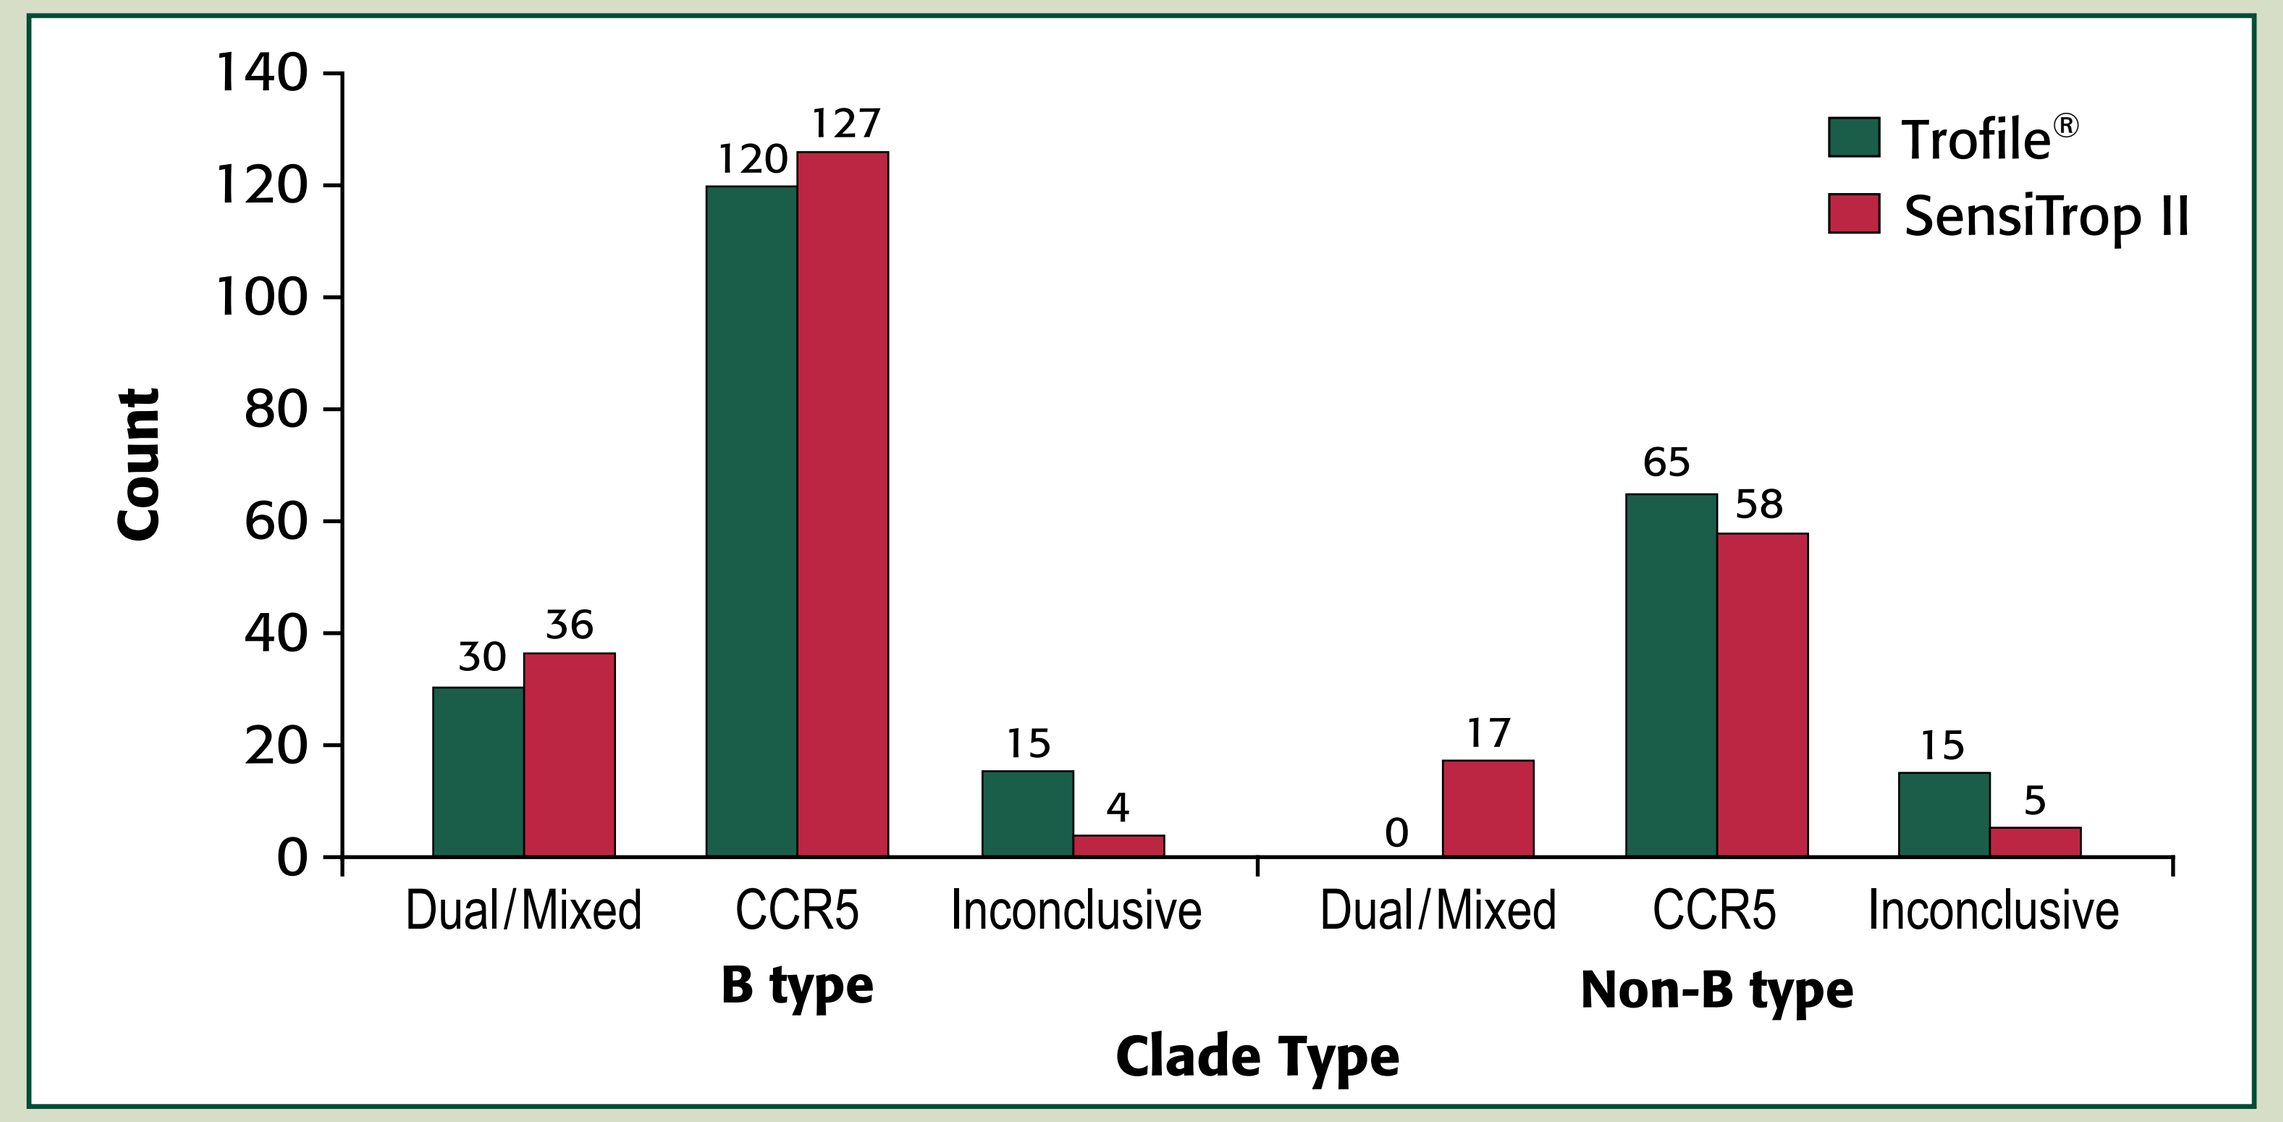

Count
140
120
100
80
60
40
20
0
Trofile®
SensiTrop II
30
36
120
127
15
4
0
17
65
58
15
5
Dual/Mixed CCR5 Inconclusive
Dual/Mixed CCR5 Inconclusive
B type
Non-B type
Clade Type

- For clade B, there was concordance between assays ($p=0.08$).
- Trofile® reported no D/M results from non-clade B samples (**Table 5**).

Table 5: Proportion of D/M Results by Clade Type

	Trofile®		SensiTrop II	
	D/M	Inconclusive	D/M	Inconclusive
Clade B	18%	9%	22%	2%
Non-clade B	0%	19%	21%	6%

- Similarly, Trofile® yielded inconclusive results more frequently than SensiTrop II in non-clade B samples.

Conclusions

- **Comparison of the standard Trofile® assay and the SensiTrop II assay showed concordance between the two assays in 79% (166/211) of samples with reportable results.**
- **SensiTrop II detected D/M viruses in 22% (55/252) of samples, compared with 12% (30/252) for Trofile®.**
- **The number of inconclusive test results from Trofile® was 13%, compared with 4% from SensiTrop II.**
- **In this analysis, viral load and HIV clade affected the measurement of tropism in the two assays.**
- **Clinical outcome studies are needed to determine the predictive value of different tropism assays with respect to long-term benefit of CCR5 antagonists.**

References

1. Whitcomb JM, Huang W, Fransen S, et al. Development and characterization of a novel single-cycle recombinant-virus assay to determine human immunodeficiency virus type 1 coreceptor tropism. *Antimicrob Agents Chemother*. 2007;51:566-575.
2. Weiser B, Philpott S, Klimkait T, et al. HIV-1 coreceptor usage and CXCR4-specific viral load predict clinical disease progression during combination antiretroviral therapy. *AIDS*. 2008;22:469-479.
3. Cardozo T, Kimura T, Philpott S, et al. Structural basis for coreceptor selectivity by the HIV type 1 V3 loop. *AIDS Res Hum Retroviruses*. 2007;23:415-426.

Presented at the 16th Conference on Retroviruses and Opportunistic Infections, Montréal, Canada, February 8-11, 2009